ABSTRACT
232

An antibody-drug conjugate carrying a microtubule inhibitor and a DNA alkylator exerts both mechanisms of action on tumor cells

Jeremy R. Duvall, Marc Damelin, Mariya V. Kozytska, Barrett J. Nehilla, Marina Protopopova, Patrick R. Conlon, LiuLiang Qin, Mark Nazzaro, Josh D. Thomas, Qingxiu Zhang, Dorin Toader, Timothy B. Lowinger

Mersana Therapeutics, Inc., Cambridge, MA

Abstract

Antibody-drug conjugates (ADCs) consist of a tumor-targeted antibody, a drug (payload) with specified mechanism of action, and the chemical framework for attaching them to each other. By selective delivery of the payload to the tumor and not to normal tissues, ADCs can provide greater efficacy and tolerability than systemic chemotherapies, which can translate to longer duration of treatment and response. Conceptually, dual-payload ADCs (DP-ADCs)[1] could extend these advantages into combination therapy, which is standard-of-care in clinical oncology.

We have engineered a dual-payload ADC that delivers two mechanistically distinct payloads to a single target cell. To build a precision DP-ADC, we leveraged our Synthemer platform, which enables us to chemically attach the payloads to a synthetic scaffold in a defined manner, and then to chemically attach the loaded scaffold to the antibody. Our prototype DP-ADC combines auristatin F hydroxypropyl amide (AF-HPA) with a DNA monoalkylator (I-BiP). The DP-ADC maintained antigen binding and exhibited potent cytotoxicity, and we observed both of the expected mechanisms of action in the target cells.

Although AF-HPA and I-BiP did not exhibit synergy in cytotoxicity studies in cancer cell lines, their combination in a DP-ADC is expected to confer potential clinical benefit over either single-payload ADC across patient populations due to the concept of Independent Action.[2]

Therapeutic Hypothesis & Payload Pairing

Strategies for payload pairing:

- Synergy / Additivity
- Independent Action

While synergy / additivity is often cited as desired, independent action can explain the results of most combination regimens in clinical and preclinical oncology. [2]

- Payloads with non-cooperating mechanisms of action could confer a therapeutic benefit through:
 - Less inherent & acquired resistance
 - Greater response rate across a population

Payloads with Synergy / Additivity

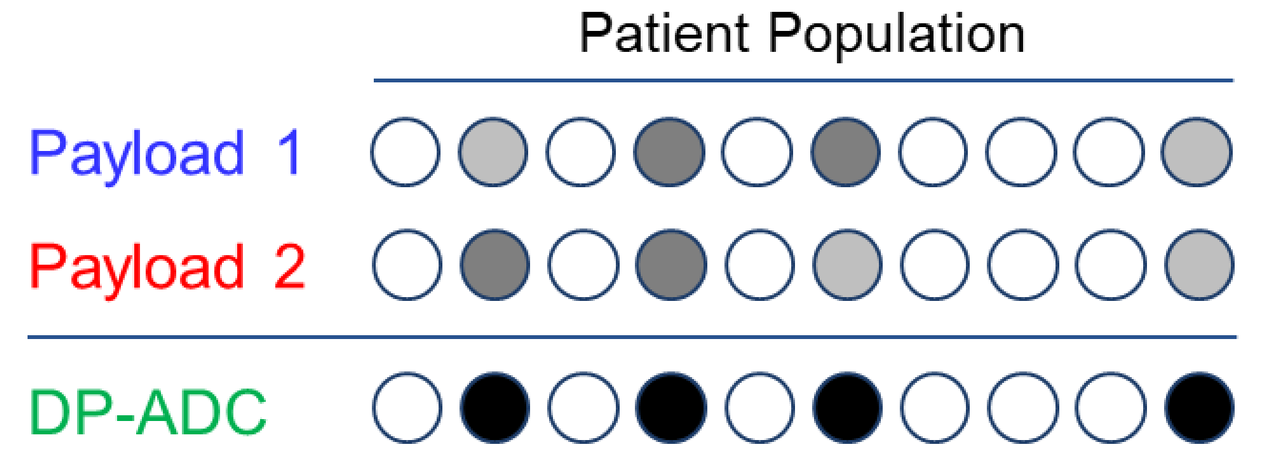
Patient Population
Payload 1
Payload 2
DP-ADC

Payloads with Independent Action

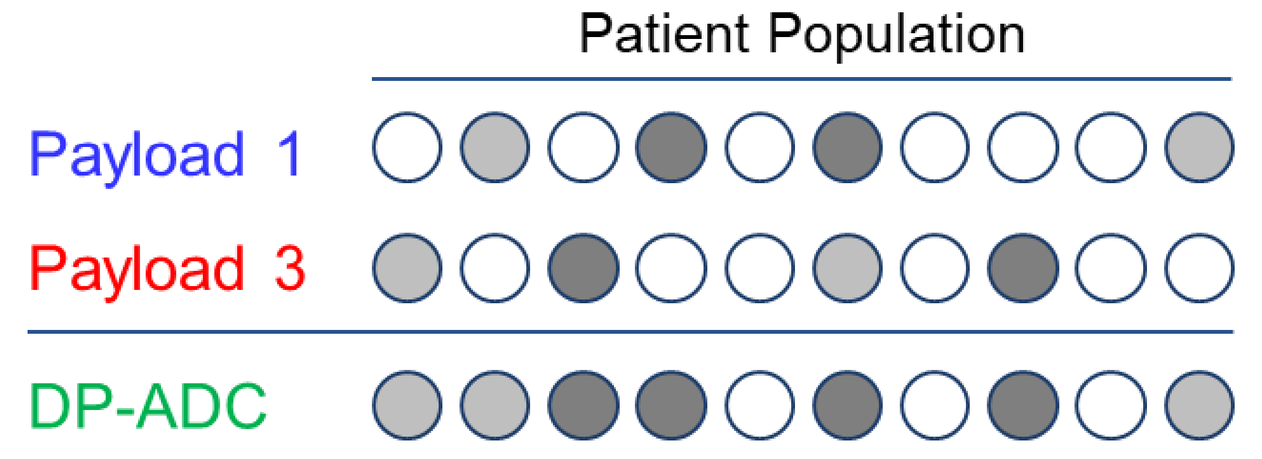
Patient Population
Payload 1
Payload 3
DP-ADC

Considerations

- Synergy / Additivity may have more liability for target-dependent toxicities
- Independent Action may have more liability for target-independent toxicities
- Optimal payload pairing may depend on target

Dual-Payload ADC Design

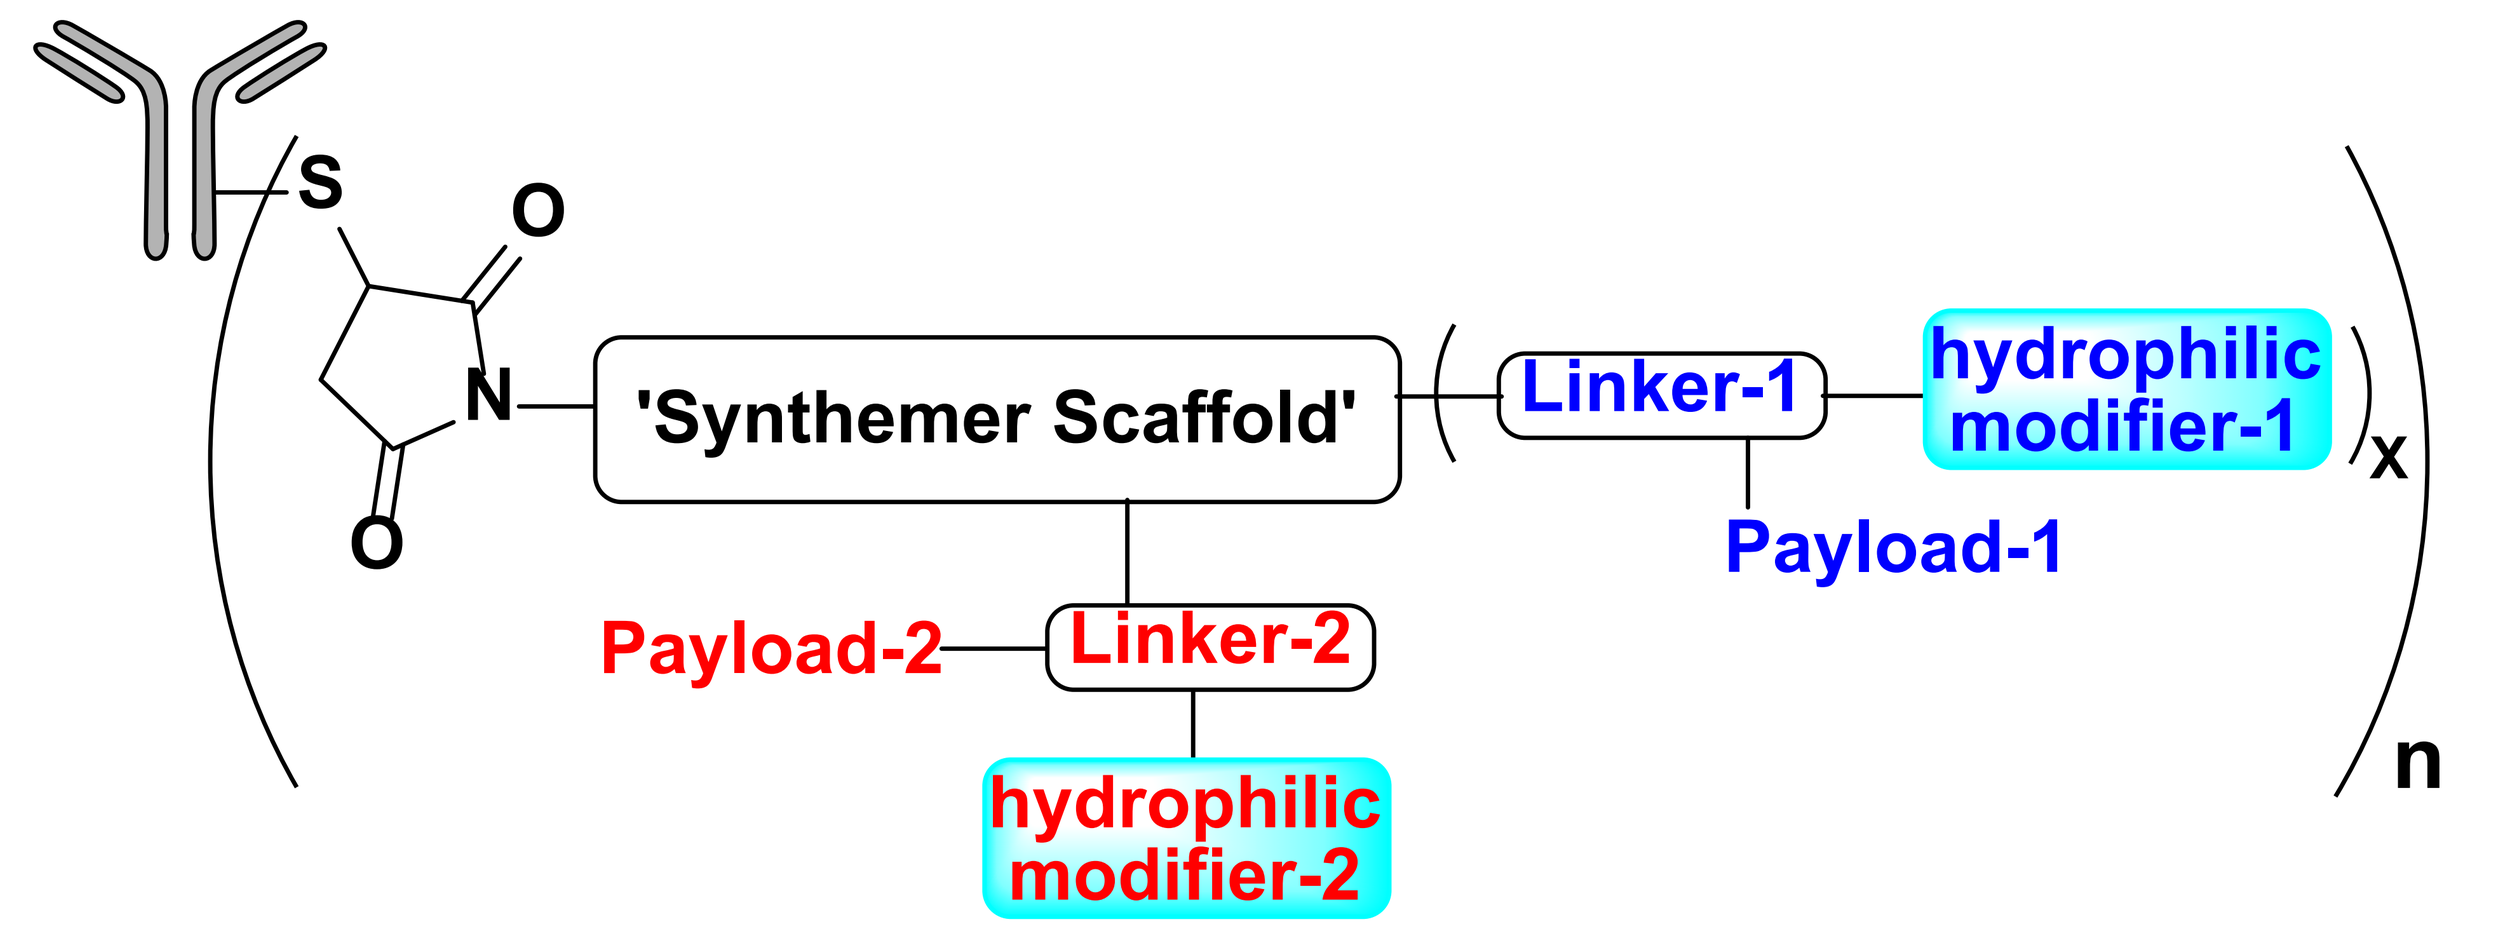
S
O
N
O
'Synthemer Scaffold'
Linker-1
hydrophilic modifier-1
x
Payload-1
Payload-2
Linker-2
hydrophilic modifier-2
n

Synthemer allows for flexibility and precise control of dual-payload ADCs.

Synthemer[3] is a novel proprietary, synthetic platform that enables:

- Tuning of the drug:antibody ratio (DAR): 2-8 Synthemer moieties per mAb
 - Creation of high DAR ADCs
- Incorporation of multiple payloads in a fixed ratio, e.g. 1:1, 2:1, 3:1
 - Solubilizing group and linker group matched to each payload
- Site-specific or stochastic conjugation

Selection & Profiling of Payloads

DNA alkylator

I-BiP

Indole-Biaryl Pyrrolobenzodiazepines[4]

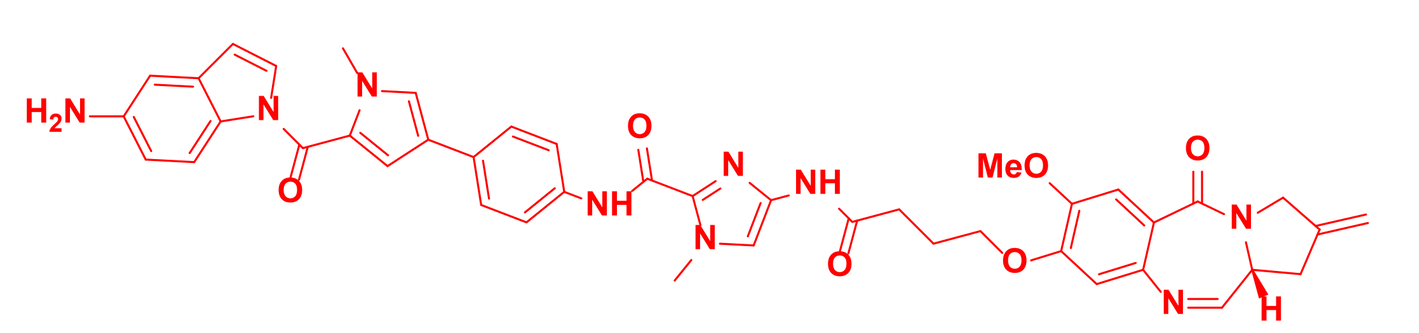

Microtubule inhibitor

AF-HPA

Auristatin F-hydroxypropyl amide[5]

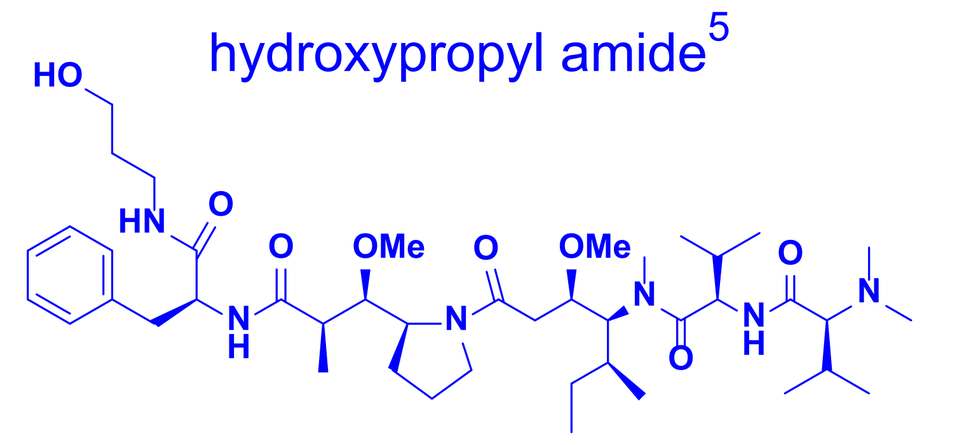

AF-HPA and I-BiP do not exhibit synergy / additivity *in vitro*.

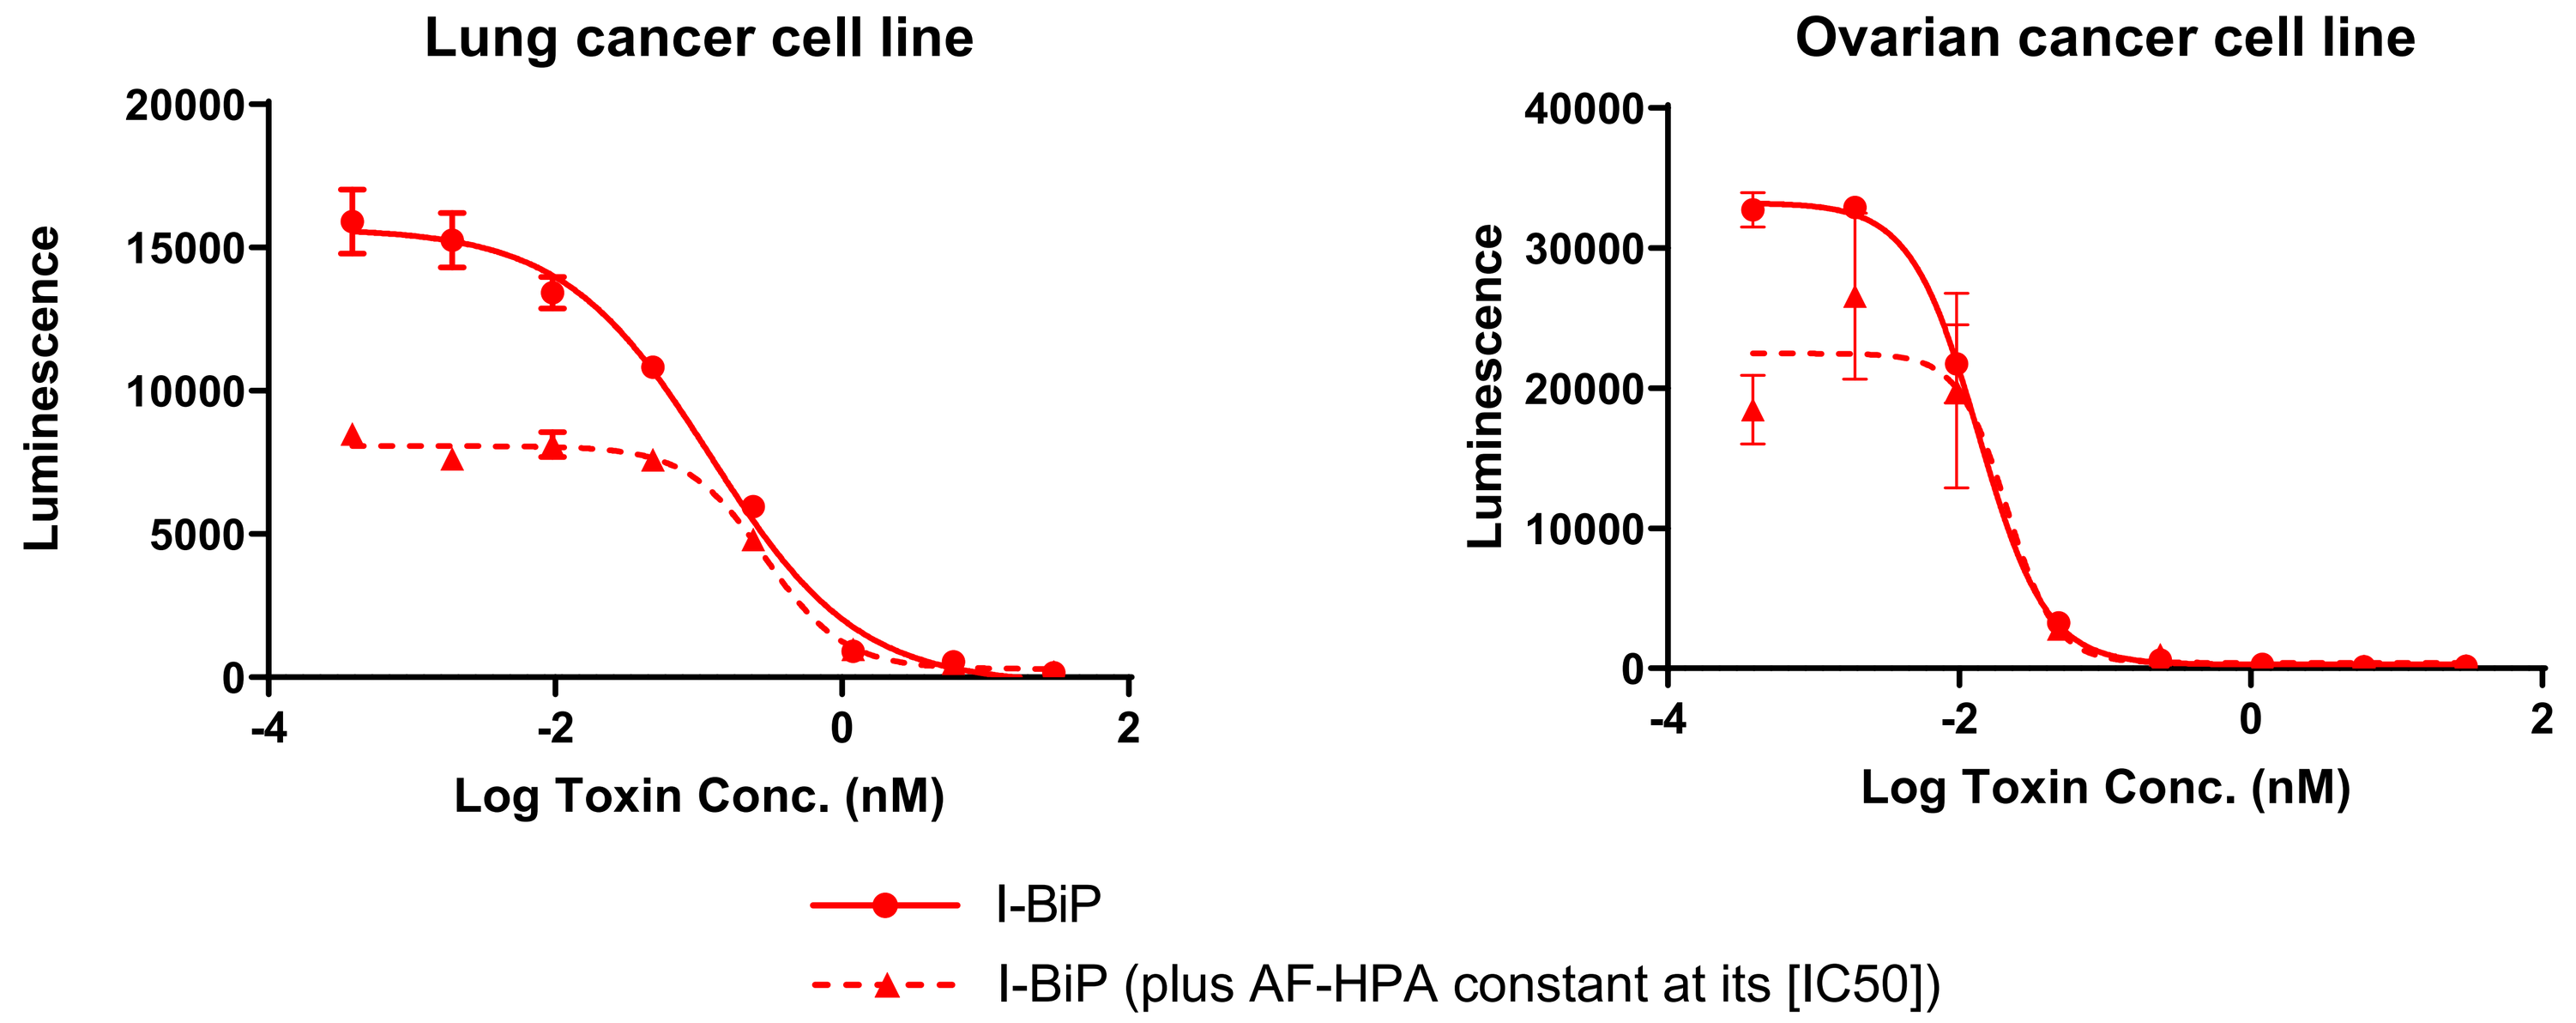
Lung cancer cell line
Luminescence
20000
15000
10000
5000
0
-4
-2
0
2
Log Toxin Conc. (nM)
Ovarian cancer cell line
Luminescence
40000
30000
20000
10000
0
-4
-2
0
2
Log Toxin Conc. (nM)
I-BiP
I-BiP (plus AF-HPA constant at its [IC50])

Generation of the Dual-Payload ADC

Dual-Payload ADC (DP-ADC) generated with Synthemer platform:

- 3 equivalents of AF-HPA & 1 equivalent of I-BiP within each Synthemer moiety
- Total DAR average = 18 – 20
- Minimal aggregation observed on SEC

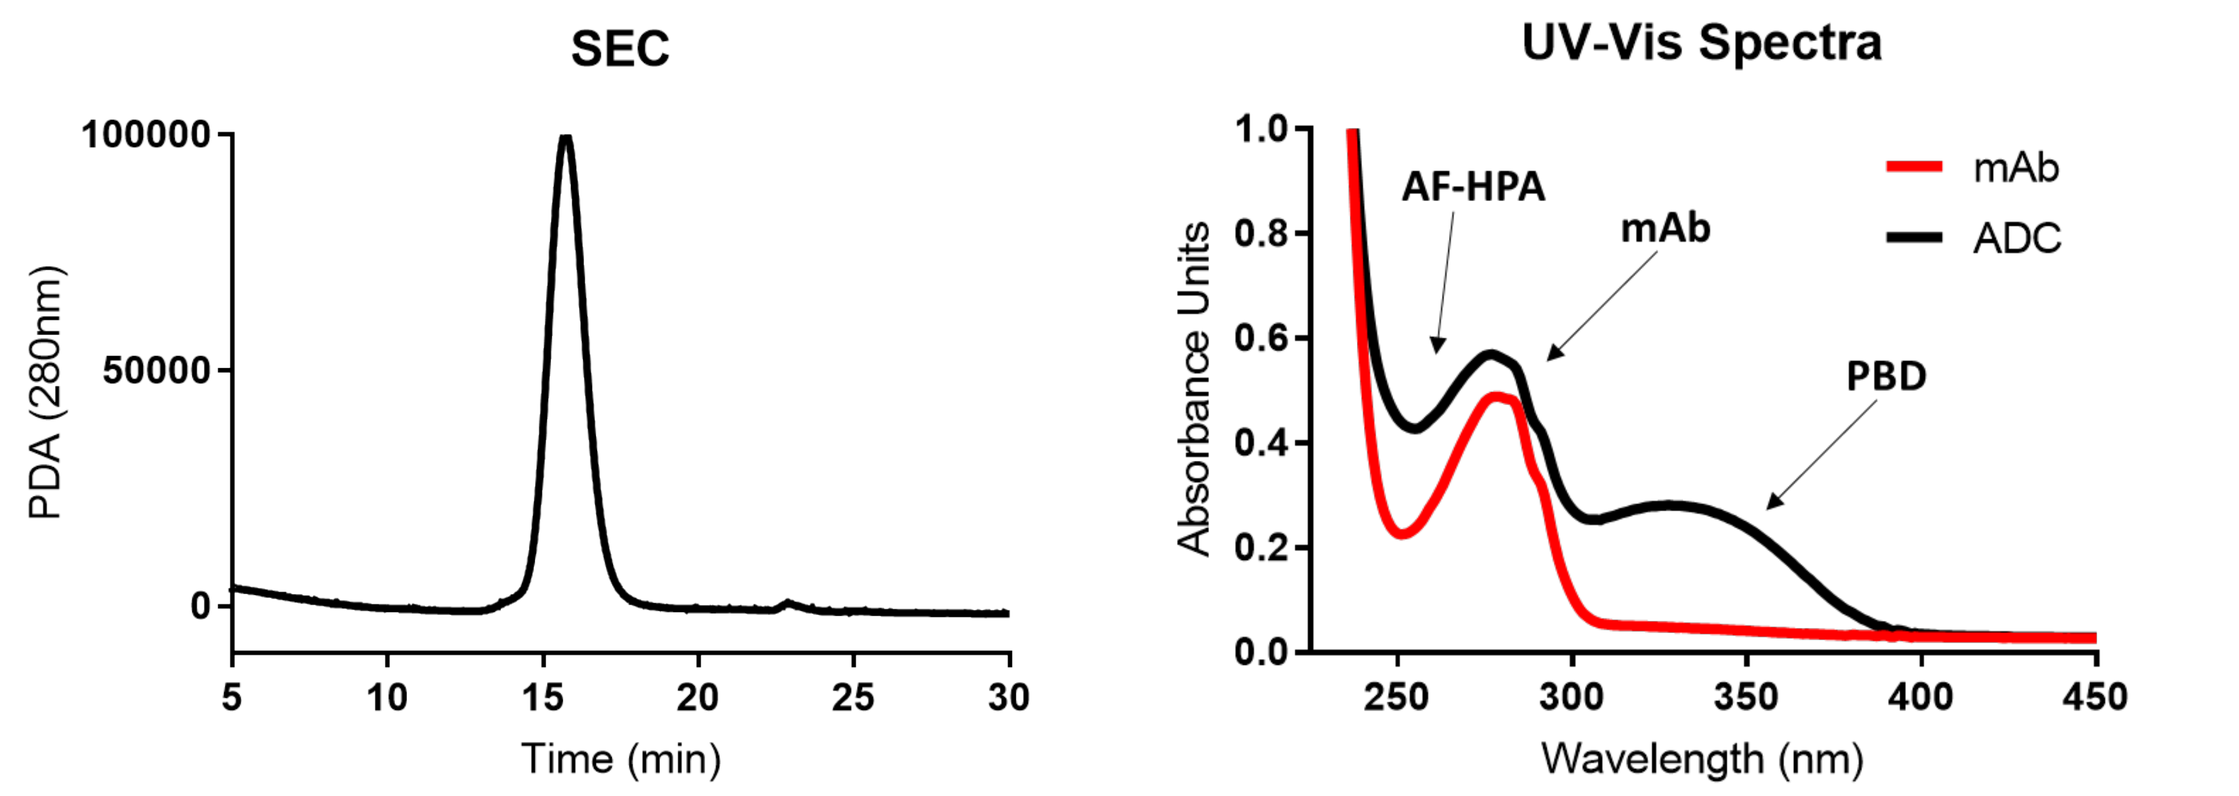
SEC
PDA (280nm)
100000
50000
0
5
10
15
20
25
30
Time (min)
UV-Vis Spectra
Absorbance Units
1.0
0.8
0.6
0.4
0.2
0.0
AF-HPA
mAb
PBD
mAb
ADC
250
300
350
400
450
Wavelength (nm)

Attribute	Value
mAb (mg/mL)	2.0
Unconjugated mAb (%)	0.7
DAR (hydrolysis method)	20
DAR (UV-Vis method)	18
Free AF-HPA (%)	1.2
HMW (%)	1.8

ADC concentration was measured by UV-Vis; unconjugated mAb was measured by HIC; free AF-HPA was measured by LC-MS/MS.

Single-payload ADCs using the same antibody were generated for comparison:

- AF-HPA ADC: average DAR = 15
- I-BiP ADC: average DAR = 5

DP-ADC Maintains Cell Binding

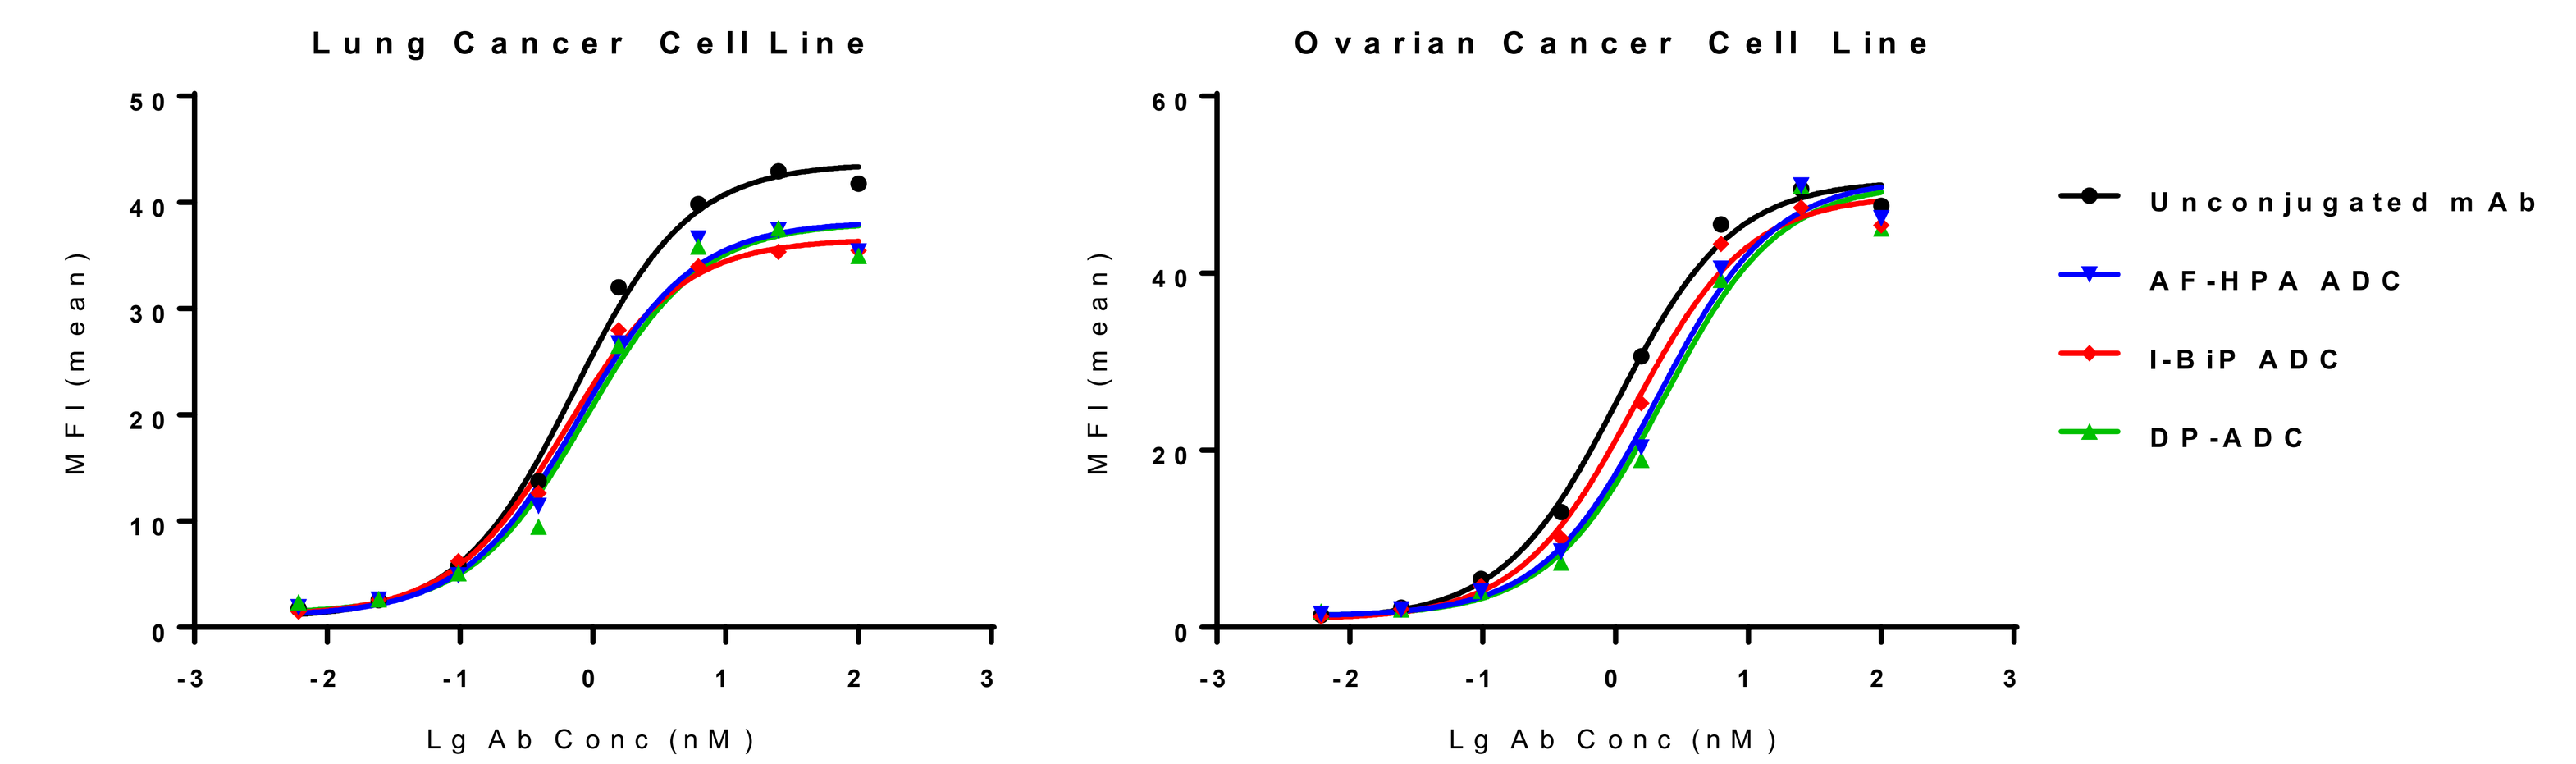
Lung Cancer Cell Line
MFI (mean)
50
40
30
20
10
0
-3
-2
-1
0
1
2
3
Lg Ab Conc (nM)
Ovarian Cancer Cell Line
MFI (mean)
60
40
20
0
Lg Ab Conc (nM)
Unconjugated mAb
AF-HPA ADC
I-BiP ADC
DP-ADC

Cells were incubated with ADC on ice for 2 hours, washed and incubated on ice for 1 hour with Alex647-Donkey anti-hIgG (H+L) (Jackson ImmunoResearch), and then signal was measured on a flow cytometer.

DP-ADC Exhibits Potent Cytotoxicity

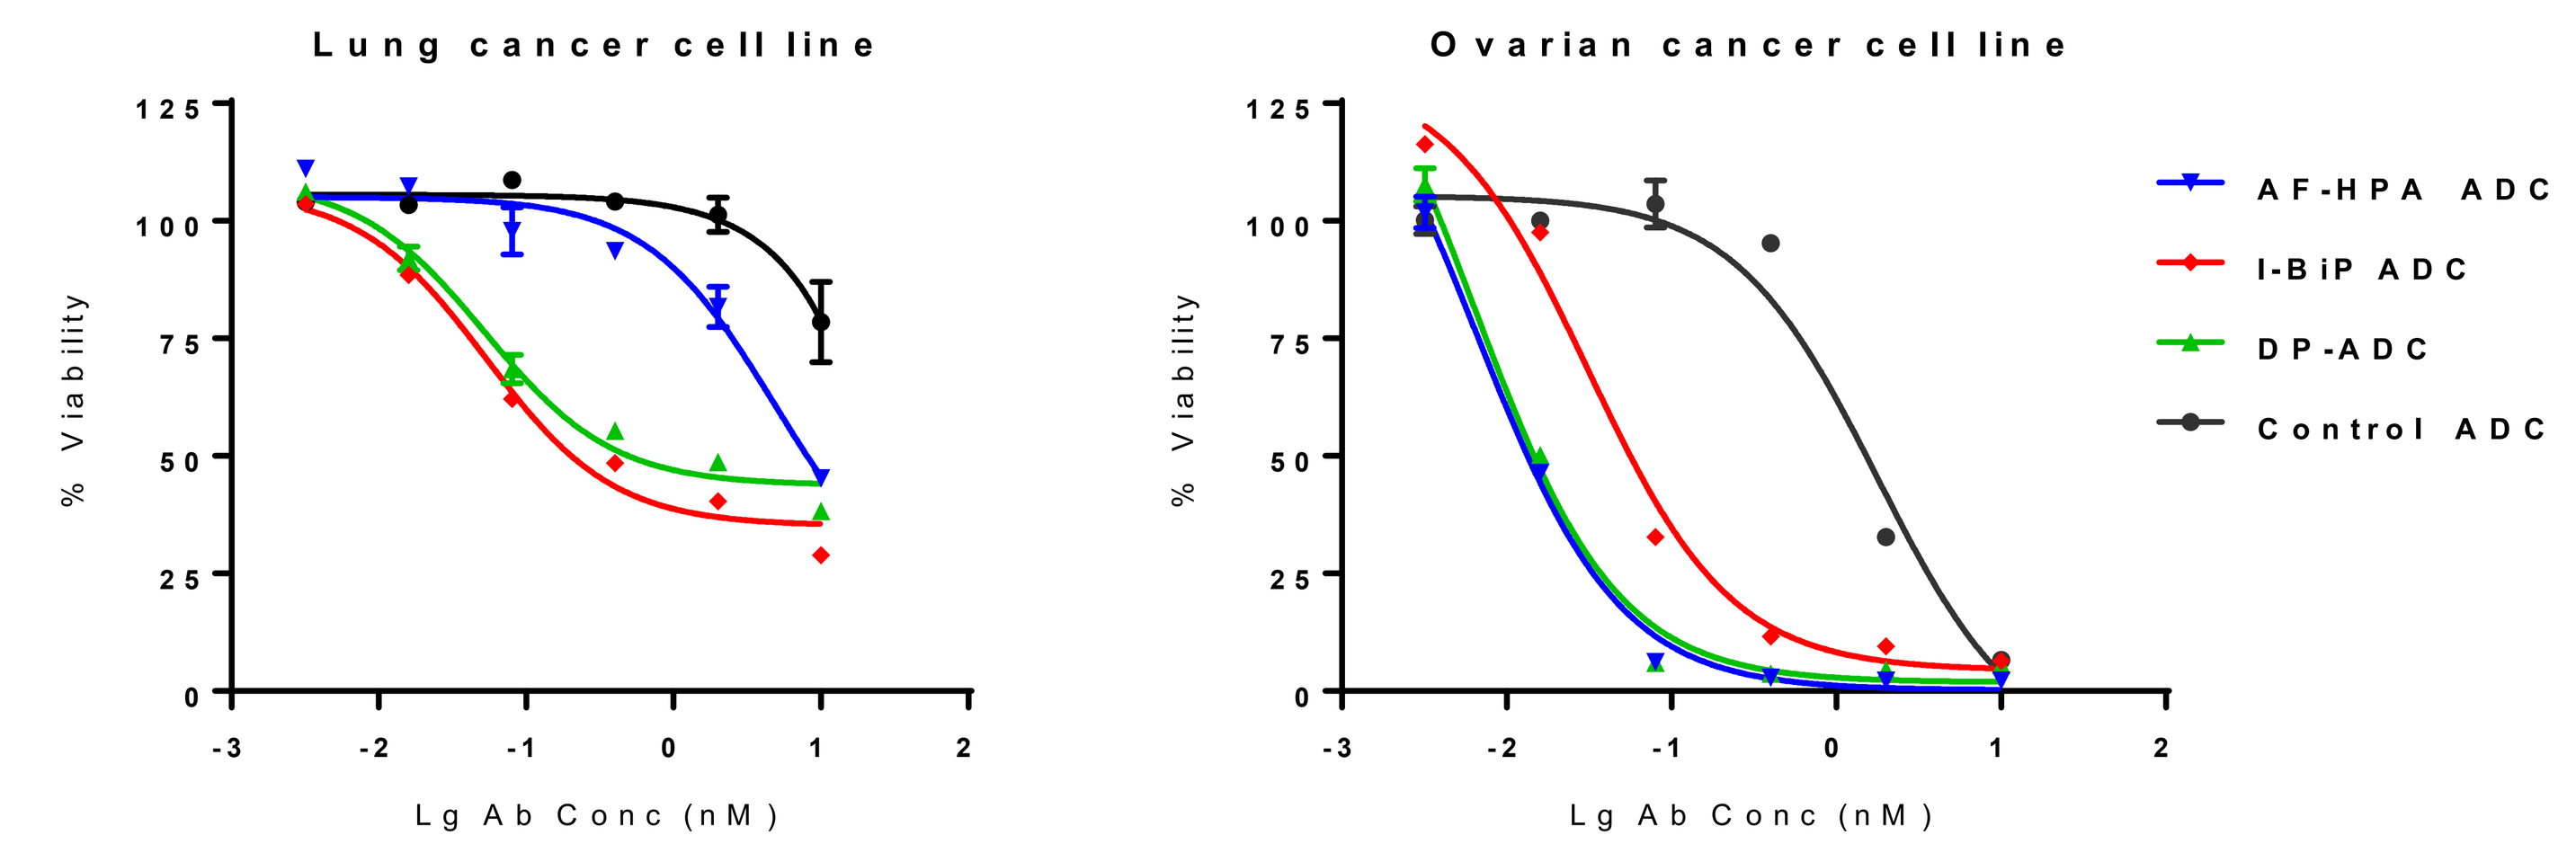
Lung cancer cell line
% Viability
125
100
75
50
25
0
-3
-2
-1
0
1
2
Lg Ab Conc (nM)
Ovarian cancer cell line
% Viability
Lg Ab Conc (nM)
AF-HPA ADC
I-BiP ADC
DP-ADC
Control ADC

Cells were treated with ADC for 5 days, and then viability was measured with CellTiter-Glo (Promega).

DP-ADC Exerts Both Mechanisms of Action

Analysis of mitotic spindle assembly

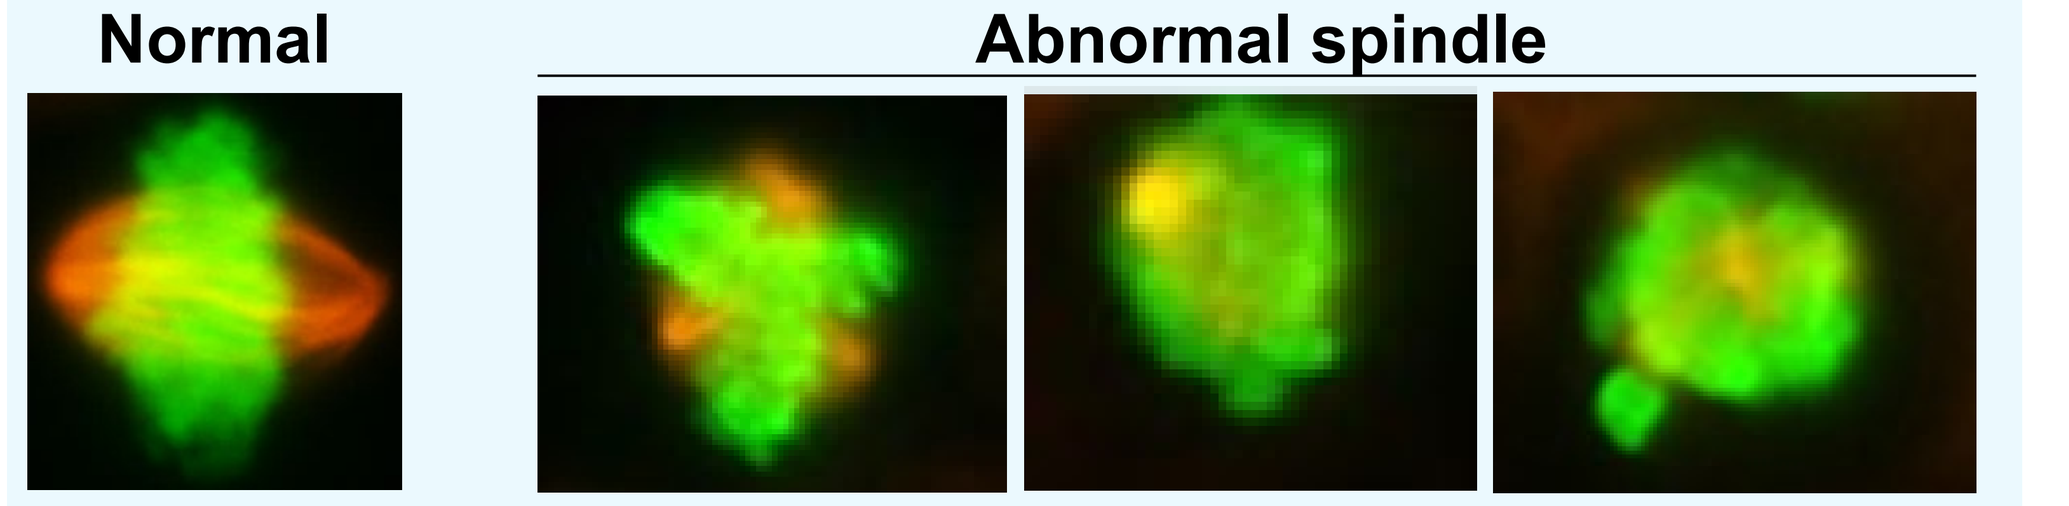
Normal
Abnormal spindle

Cells were fixed and stained with antibodies against phospho-HistoneH3 (green) #3465 & microtubules (orange) #2116, Cell Signaling. Spindles were categorized as normal or abnormal.

Analysis of DNA Damage

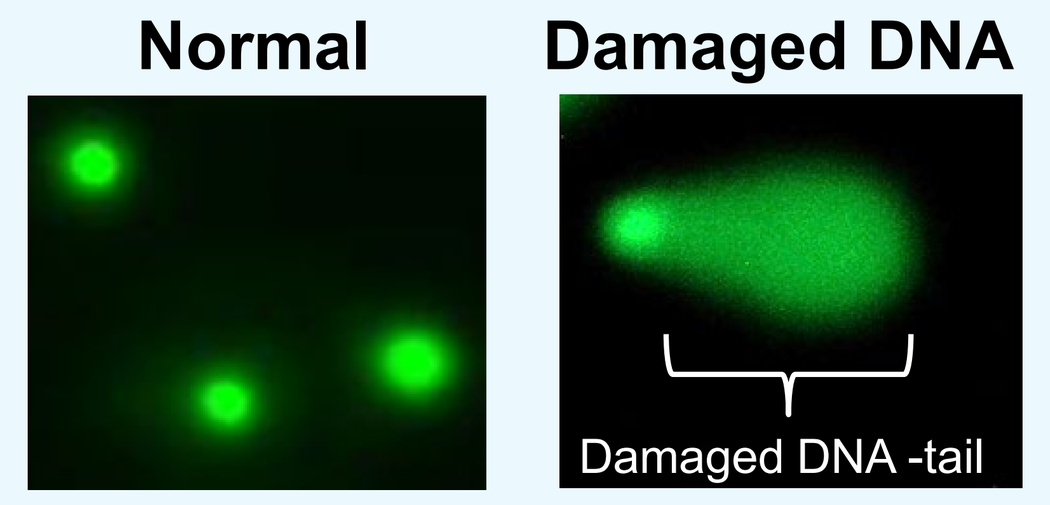
Normal
Damaged DNA
Damaged DNA -tail

Comet assay detects single-stranded DNA damage (# STA-350, Cell Biolabs, Inc)

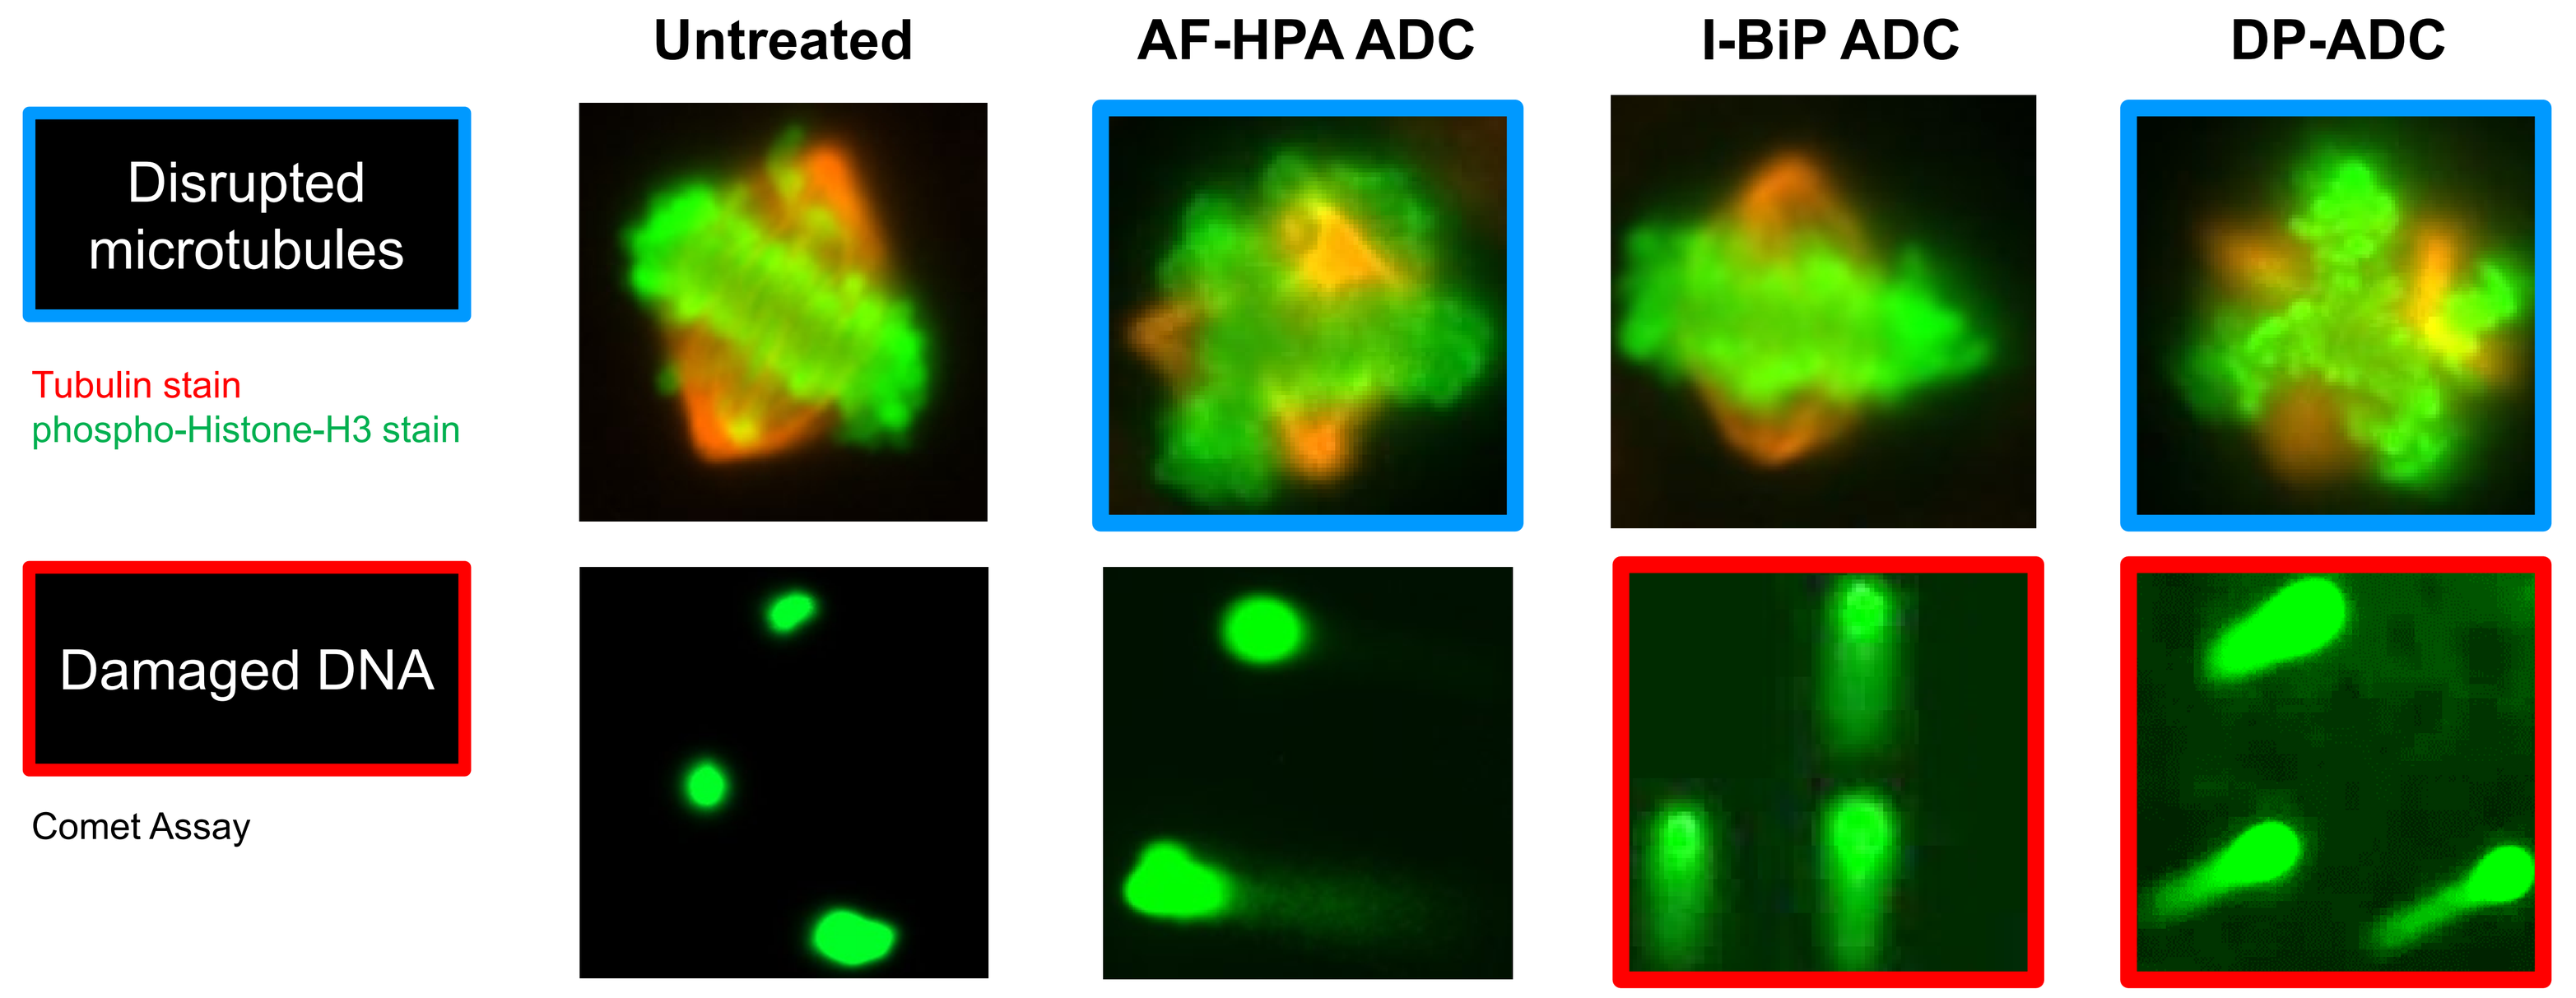
Untreated
AF-HPA ADC
I-BiP ADC
DP-ADC
Disrupted microtubules
Tubulin stain
phospho-Histone-H3 stain
Damaged DNA
Comet Assay

Cancer cells were treated with 10nM ADC for 24 hours and then processed for both assays. For the Comet assay, cells were electrophoresed under alkaline conditions at 33V/300mA, 15 min and then evaluated under a Zeiss fluorescence microscope.

Summary & Discussion

- The Synthemer platform provides both **flexibility** and **precise control** to the engineering of dual-payload ADCs.
- Our prototype dual-payload ADC based on a microtubule disruptor and a DNA monoalkylator maintained cell binding and exhibited potent cytotoxicity – and both mechanisms of action were observed in target cells.
- DP-ADCs based on synergy / additivity *may* result in greater efficacy than single-agent ADCs in select patient populations, with potential liabilities for target-dependent toxicities.
- DP-ADCs based on independent action *may* result in broader activity across patient populations, with potential liabilities due to additive target-independent toxicities.

References

1. *(a)* Levengood *et al.* Angew. Chem. Int. Ed. 2017 56:733-737.
 (b) Kumar *et al.* Bioorg. Med. Chem. Lett. 2018 28:3617-3621.
2. Palmer and Sorger. Cell 2017 171:1678-1691.
3. Kozytska *et al.* EORTC-NCI-AACR Symposium, Nov 2018, abstract 272. *Available on www.mersana.com/our-technology*
4. Thomas *et al.* EORTC-NCI-AACR Symposium, Nov 2018, abstract 173. *Available on www.mersana.com/our-technology*
5. Bodyak *et al.* AACR Annual Meeting, Apr 2015, abstract 641. *Available on www.mersana.com/our-technology*