TPS2606

A phase Ib, first-in-human, dose escalation and expansion study of XMT-1522, a novel antibody-drug conjugate (ADC) directed against HER2, in patients with advanced breast cancer and other advanced tumors expressing HER2

Howard A. Burris[1,2], Minal A. Barve[3], Erika Paige Hamilton[1,2], Aditya Bardia[4], Hatem Hussein Soliman[5], Donna Jarlenski[6], Rebecca Mosher[6], Donald Alan Bergstrom[6]

[1]Sarah Cannon Research Institute and [2]Tennessee Oncology, PLLC, Nashville, TN; [3]Texas Oncology, Dallas, TX; [4]Massachusetts General Hospital Cancer Center, Boston, MA; [5]Moffitt Cancer Center, Tampa, FL; [6]Mersana Therapeutics, Cambridge, MA

Background

XMT-1522 is an ADC consisting of a novel human IgG1 anti-HER2 monoclonal antibody conjugated to a proprietary auristatin-based cytotoxic payload (AF-HPA). An average of 12 AF-HPA molecules is conjugated to each antibody via a biodegradable polymer. In pre-clinical xenograft experiments XMT-1522 achieved complete, durable tumor regressions in models of HER2-positive and HER2 1+/2+ breast cancer, HER2 2+/3+ NSCLC, and HER2-positive and HER2 1+ gastric cancer.

Deep Tumor Regression

Complete or Near-Complete Regression:
11/15 models

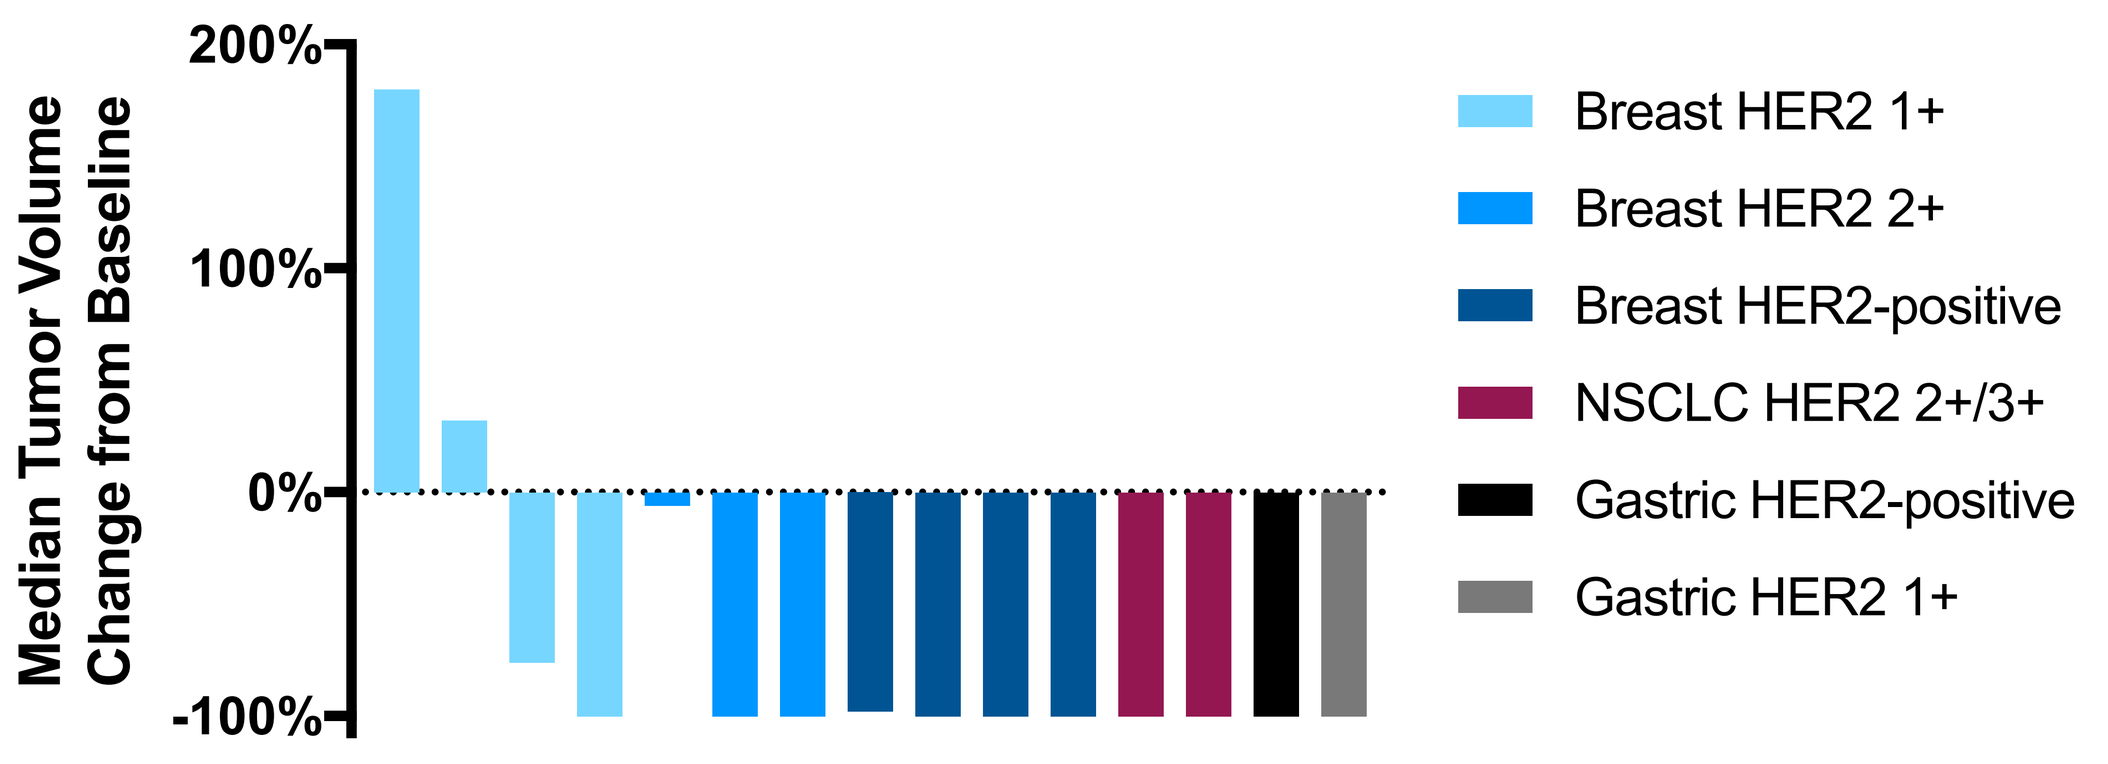

Median Tumor Volume Change from Baseline
200%
100%
0%
-100%
Breast HER2 1+
Breast HER2 2+
Breast HER2-positive
NSCLC HER2 2+/3+
Gastric HER2-positive
Gastric HER2 1+

Durable Tumor Regression

Models with CR Maintained to Day 60:
10/11 models
45-60 Days Elapsed Since Last XMT-1522 dose

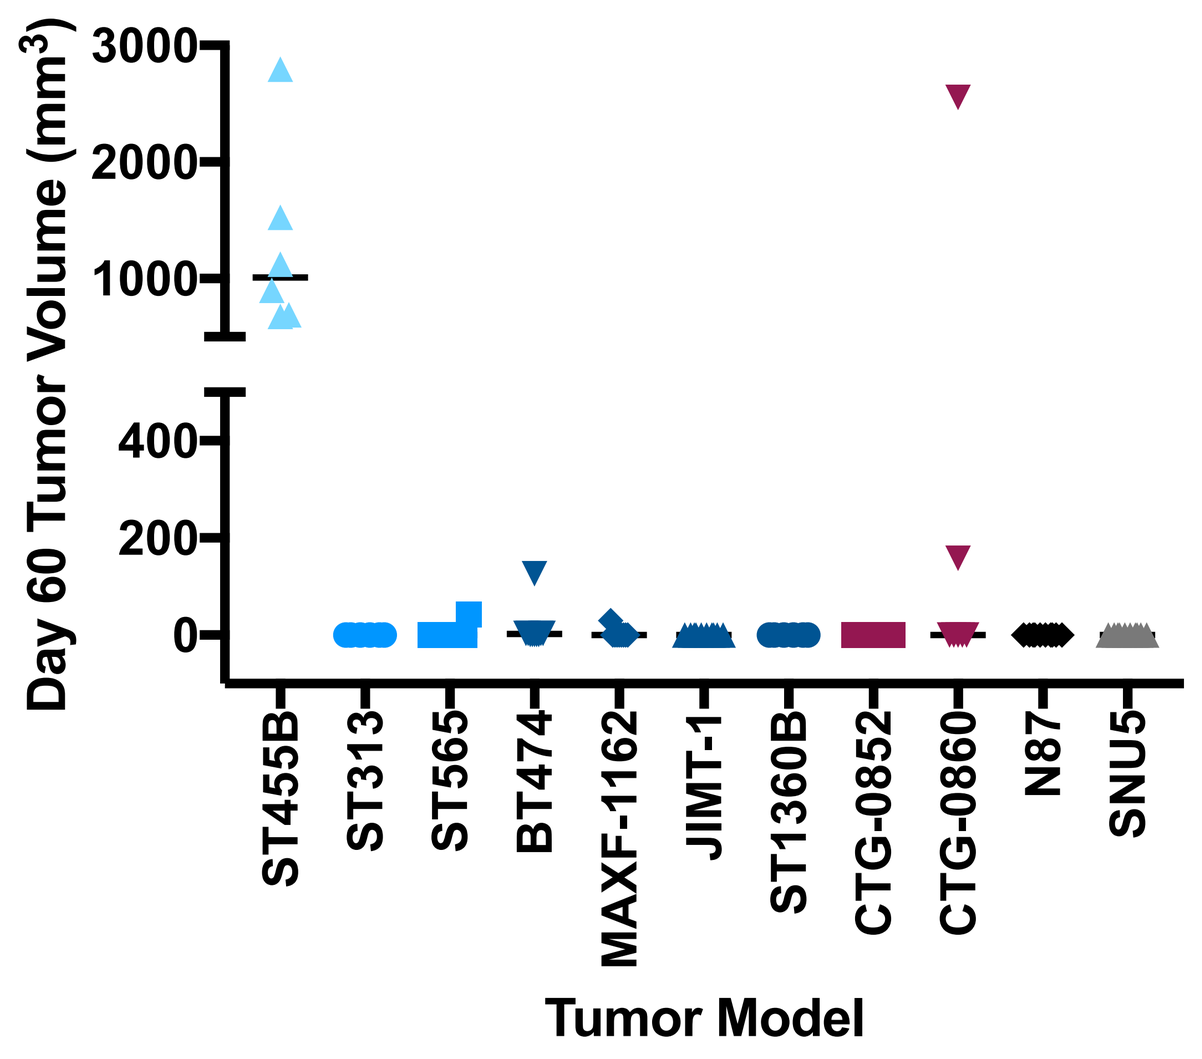

Day 60 Tumor Volume (mm³)
3000
2000
1000
400
200
0
ST455B
ST313
ST565
BT474
MAXF-1162
JIMT-1
ST1360B
CTG-0852
CTG-0860
N87
SNU5
Tumor Model

Study Design

Dose Escalation (DES) N=24-30

HER2 (IHC) ≥ 1+ Breast Cancer; HER2 2+/3+ NSCLC; HER2+ Gastric Cancer

Phase 1 Cohort Expansion (EXP) N=20

- Breast Cancer, HER2 IHC 1+/2+
- NSCLC, HER2 IHC 2+/3+
- Gastric Cancer, HER2+, post-trastuzumab
- Breast Cancer, HER2+, post T-DM1

Objectives

PRIMARY OBJECTIVES

Dose Escalation (DES):
- Determine the maximum tolerated dose (MTD) and recommended Phase 2 dose (RP2D) of XMT-1522 administered intravenously once every three weeks
- Assess the safety and tolerability of XMT-1522

Expansion (EXP):
- Assess further the safety and tolerability of XMT-1522 administered at the MTD/RP2D identified in the DES
- Assess the preliminary anti-neoplastic activity of XMT-1522

SECONDARY OBJECTIVES

DES and EXP:
- Assess the pharmacokinetics (PK) of XMT-1522, its release product, and selected metabolites
- Assess the development of anti-drug antibodies to XMT-1522

DES Only:
- Assess the preliminary anti-neoplastic activity of XMT-1522

EXPLORATORY OBJECTIVE
- Retrospectively evaluate the association of objective response and alternative assays for measurement of HER2 expression, expression of other genes, or patient subsets identified by tumor genetic mutations

Inclusion/Exclusion Criteria

KEY INCLUSION CRITERIA

1. Females and males, age ≥ 18 years old.
2. Histologically or cytologically confirmed adenocarcinoma of the breast with unresectable locally advanced disease or metastatic disease; OR histologically or cytologically confirmed unresectable locally advanced or metastatic gastric cancer; OR histologically or cytologically confirmed Stage IIIb or IV non-small cell lung cancer (NSCLC).
3. Breast cancer with HER2 IHC 1+, 2+ or 3+ by local laboratory assessment, or positive for HER2 gene amplification by local laboratory assessments; OR HER2-positive gastric cancer by local immunohistochemistry and/or gene amplification assessment; OR NSCLC with HER2 gene amplification by local laboratory assessment, or HER2 IHC 2+ or 3+ by local laboratory assessment.
4. Patient has progressed following all standard of care therapies for advanced breast cancer, gastric cancer, or NSCLC.
5. ECOG performance status 0 or 1.
6. Measurable disease as per RECIST, version 1.1.
7. Resolution of all acute toxic effects of prior therapy or surgical procedures to Grade ≤1 (except alopecia).
8. Cardiac left ventricular ejection fraction (LVEF) ≥ 50% or the institution's lower limit of normal by either ECHO or MUGA scan.
9. Adequate organ function
10. Confirmed availability (prior to Cycle 1, Day 1) of tumor tissue blocks (strongly recommended) or freshly cut tissue slides for confirmatory central laboratory HER2 status testing and other exploratory assessments. Tissue specimens must be submitted within 60 days after the first dose of study drug.

DISEASE SPECIFIC INCLUSION CRITERIA FOR EXP

Cohort 1: HER2 1+/2+ advanced breast cancer with 2-3 prior chemotherapy regimens
Cohort 2: HER2-positive advanced breast cancer with prior pertuzumab and ado-trastuzumab emtansine (T-DM1)
Cohort 3: HER2-positive advanced gastric cancer with prior trastuzumab
Cohort 4: HER2 2+/3+ NSCLC with at least 1 prior platinum regimen

KEY EXCLUSION CRITERIA

1. Major surgery or radiation therapy within 28 days of starting study treatment; OR systemic anti-cancer therapy within the lesser of 28 days or 5 half-lives of the prior therapy of before starting study treatment.
2. Progressive or untreated brain metastases, or leptomeningeal disease
3. Peripheral neuropathy of Grade 2 or greater within 3 weeks prior to the first study therapy.
4. History of exposure to cumulative doxorubicin dose ≥ 360 mg/m^2.
5. History of clinically significant cardiac dysfunction.
6. Current severe, uncontrolled systemic disease (e.g., clinically significant cardiovascular, pulmonary, or metabolic disease).
7. Severe dyspnea at rest, due to complications of advanced malignancy, or requiring supplementary oxygen therapy.
8. Pregnant or nursing women.
9. History of other malignancy within the last 5 years, except for appropriately treated carcinoma in situ of the cervix, non-melanoma skin carcinoma, or other malignancy with a similar expected curative outcome.

Dosing

Dose Escalation	Start	100%	100%	50%	33%	33%	33%
Dose Level (DL)	1	2	3	4	5	6	7
Planned dose, mg/m^2	2.0	4.0	8.0	12.0	16.0	21.3	28.3

- Intravenous administration, every three weeks until progression
- 3+3 dose escalation with option for fourth patient at each dose level
- Dosing after dose level 7 will proceed at 33% increments
- Intrapatient dose escalation allowed

Key Post-Dose Assessments

- 3 week DLT evaluation period
- CT scans read per RECIST v1.1 at baseline then every 2 cycles
- ECHO/MUGA at end-of-cycle 1 (EOC1), EOC3, then every 3 cycles
- Ophthalmologic exam (slit lamp) at baseline, EOC1, EOC2, then every 2 cycles
- Complete PK profile in Cycle 1 for monoclonal antibody, total and free AF-HPA payload, and auristatin F, the primary active metabolite of AF-HPA; limited PK sampling during subsequent cycles
- Anti-drug antibodies at baseline, EOC1, EOC2 then every 2 cycles

Enrollment Update

- Cohorts 1 and 2 have been completed without DLT
- Cohort 3 opened for enrollment May 2017
- Enrolling Centers: Sarah Cannon Research Institute (Nashville, TN); Mary Crowley Cancer Research (Dallas, TX); Massachusetts General Hospital (Boston, MA), Moffitt Cancer Center (Tampa, FL); South Texas Advanced Research Therapeutics (San Antonio, TX)
- Clinical trial information: NCT02952729