ABSTRACT
7067

XMT-1592, a Site-Specific Dolasynthen-Based NaPi2b-Targeted Antibody-Drug Conjugate for the Treatment of Ovarian Cancer and Lung Adenocarcinoma

Shawn P. Fessler, Anouk Dirksen, Scott D. Collins, Ling Xu, Winnie Lee, Jason Wang, Ron Eydelloth, Elena Ter-Ovanesyen, Jeffrey Zurita, Elizabeth Ditty, Barrett Nehilla, Susan Clardy, Tyler Carter, Kenneth Avocetien, Mark Nazzaro, Nam Le, Kalli C. Catcott, Alex Uttard, Bingfan Du, Chen-Ni Chin, Rebecca Mosher, Kelly Slocum, LiuLiang Qin, David Lee, Dorin Toader, Marc Damelin, Timothy B. Lowinger
Mersana Therapeutics, Inc., Cambridge, MA

Abstract

The Dolasynthen platform incorporates the DolaLock payload, which is a potent auristatin F-HPA (AF-HPA) that exerts a controlled bystander effect. The platform enables the synthesis of antibody-drug conjugates (ADCs) with precise control of the drug-to-antibody ratio (DAR) and site-specific bioconjugation. XMT-1592 is a novel ADC comprised of an anti-NaPi2b antibody and Dolasynthen, which are conjugated in a site-specific manner to yield DAR 6.

NaPi2b, also known as SLC34A2, is a transmembrane sodium-phosphate transporter that is broadly expressed on tumor cells in ovarian carcinoma, lung adenocarcinoma and other tumor types. Recent studies have shown that NaPi2b expression is enriched in the EGFR and KRAS mutant subtypes of lung adenocarcinoma.

Binding studies showed a specific, high-affinity interaction of XMT-1592 with NaPi2b that was not affected by the bioconjugation process. XMT-1592 elicited potent target-dependent *in vitro* cytotoxicity against NaPi2b-expressing ovarian carcinoma cells. XMT-1592 exhibited potent target-dependent in vivo activity against an ovarian tumor xenograft and a lung adenocarcinoma patient-derived xenograft (PDX) that express NaPi2b. Consistent with the targeted delivery benefits of the ADC approach, XMT-1592 yielded high and sustained concentrations of AF-HPA to tumors vs. normal tissues.

To evaluate the benefits of site-specific bioconjugation of Dolasynthen, we conducted *in vitro* and *in vivo* comparisons of XMT-1592 to a stochastically conjugated version of the ADC. XMT-1592 had improved *in vivo* activity, pharmacokinetics, and clinical pathology relative to its stochastic counterpart. Taken together, these results support XMT-1592 as a development candidate for the treatment of NaPi2b-expressing tumors.

Site-Specific vs. Stochastic Bioconjugation of Dolasynthen

Dolasynthen is a novel, fully synthetic ADC platform with these key features:
- DolaLock payload with controlled bystander effect
- Ability to precisely modulate drug-to-antibody (DAR)
- Compatibility with site-specific bioconjugation

In these studies we compared two NaPi2b-targeted Dolasynthen ADCs with the same targeting antibody and linker-payload, and with comparable DAR:
- XMT-1592 = site-specific bioconjugation at glycan-remodeled Asn297; DAR = 6.
- Stochastic ADC = stochastic bioconjugation at cysteines; DAR ~ 6.5.

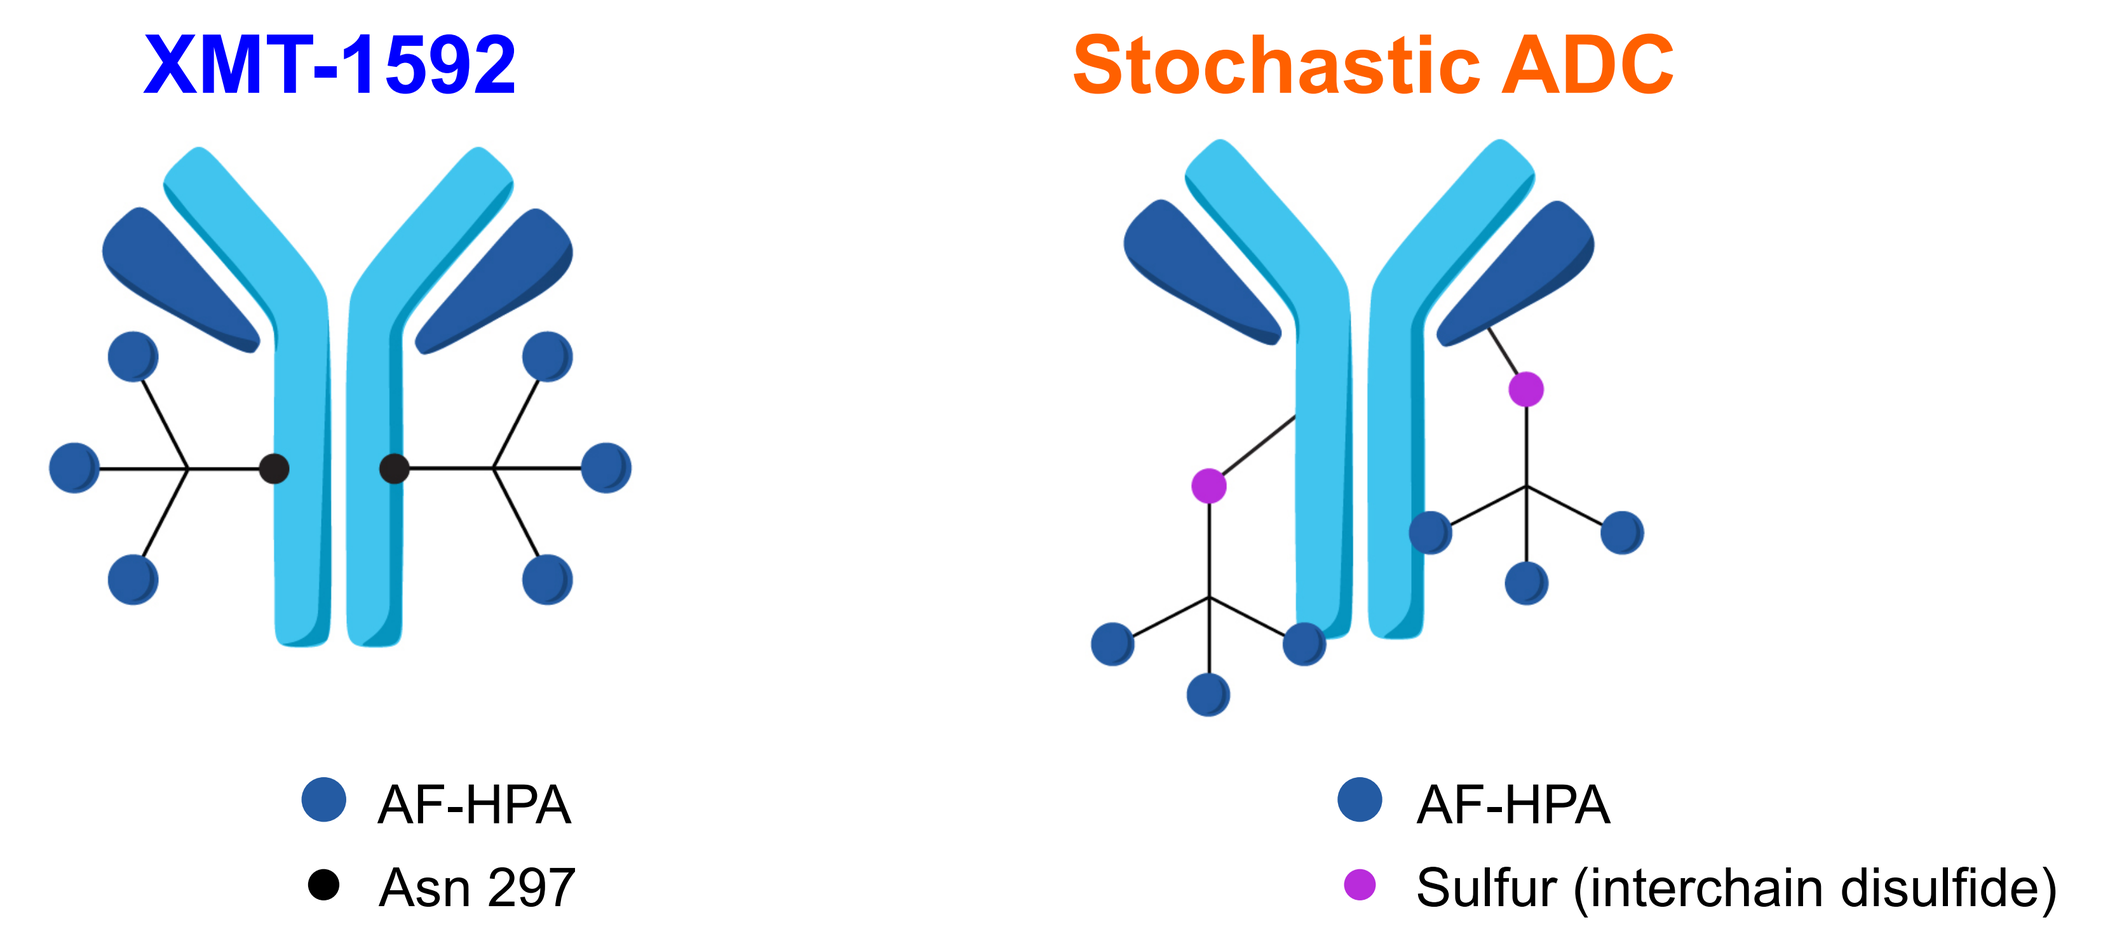
XMT-1592
Stochastic ADC
AF-HPA
Asn 297
AF-HPA
Sulfur (interchain disulfide)

Structure of XMT-1592

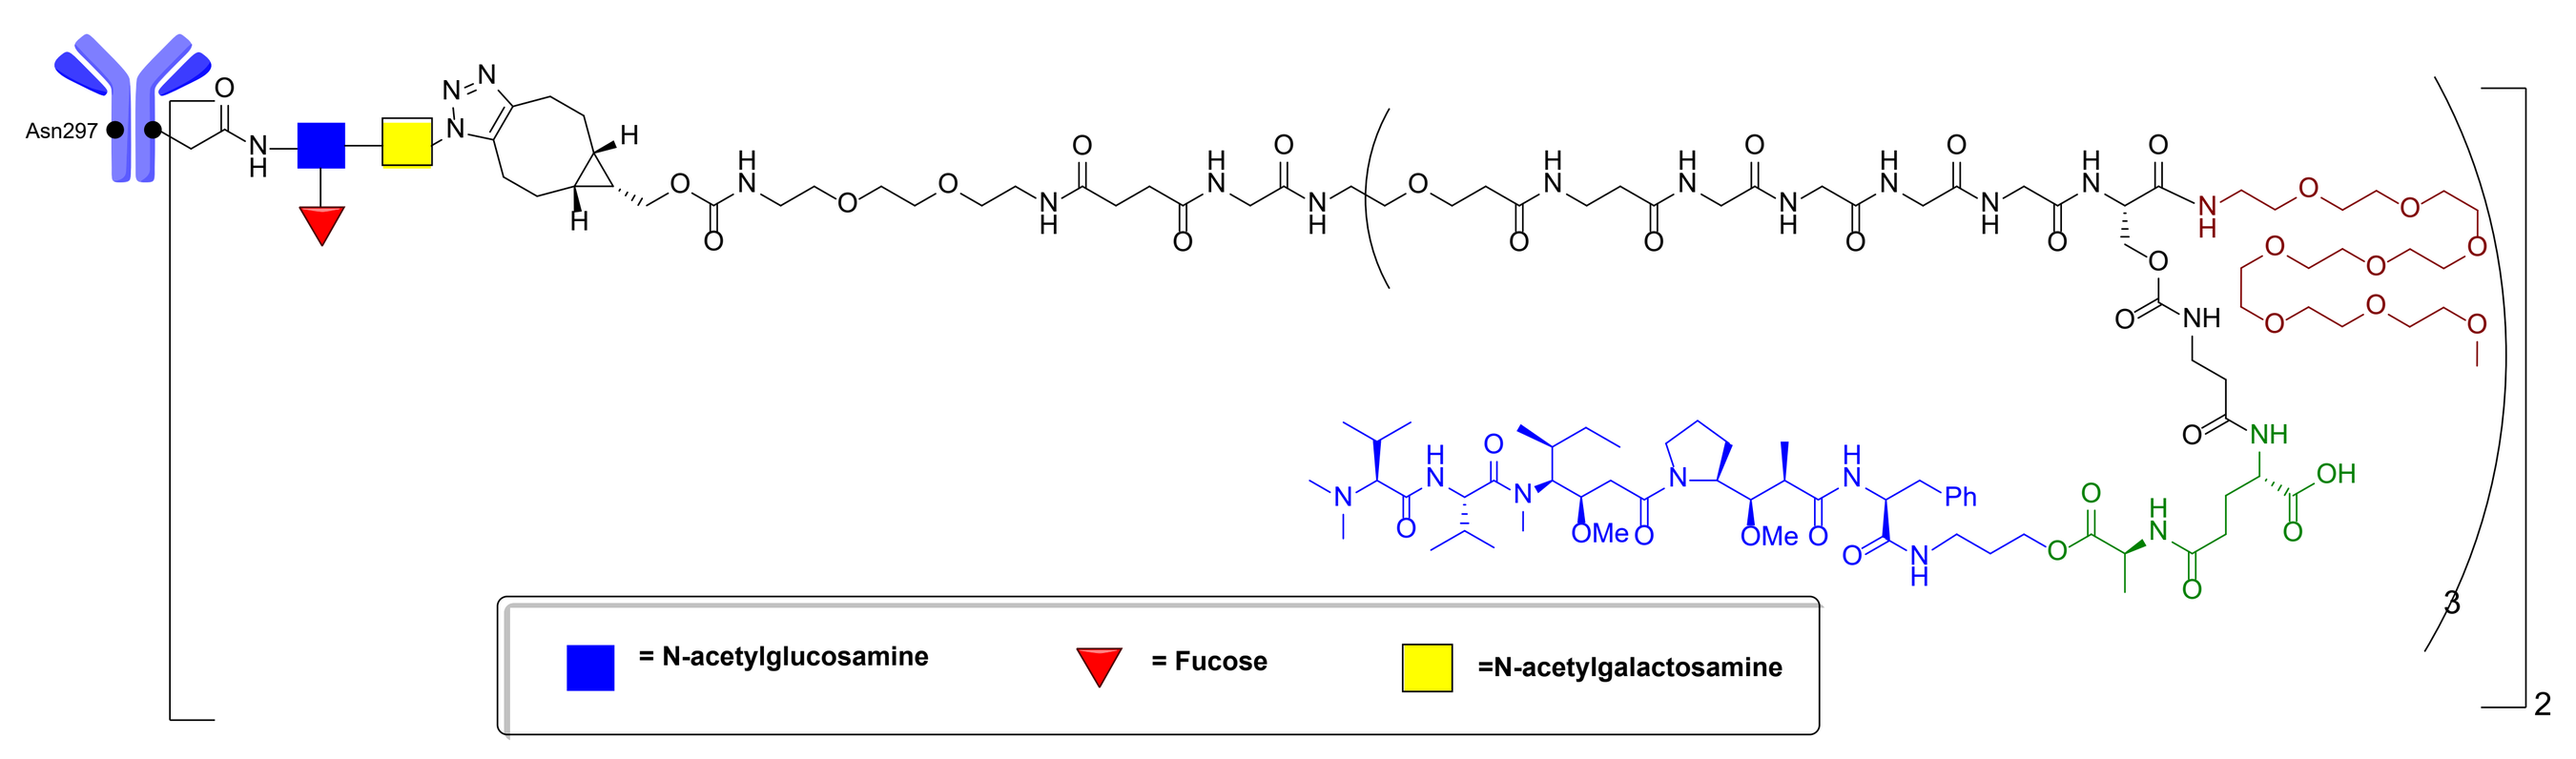
Asn297
= N-acetylglucosamine
= Fucose
=N-acetylgalactosamine

Figure 1: Structure of XMT-1592. Two molecules of Dolasynthen, each bearing three DolaLock payloads (AF-HPA; shown in blue), are conjugated to anti-NaPi2b monoclonal hIgG$_1$ antibody via click chemistry at Asn297 after glycan remodeling with Synaffix GlycoConnectTM technology. The result is a site-specific ADC with drug-to-antibody ratio (DAR) of 6.

Potent, Target-Dependent Cytotoxicity

Figure 2: Binding to NaPi2b antigen by ELISA. XMT-1592 and the stochastic ADC retain high potency binding to antigen.

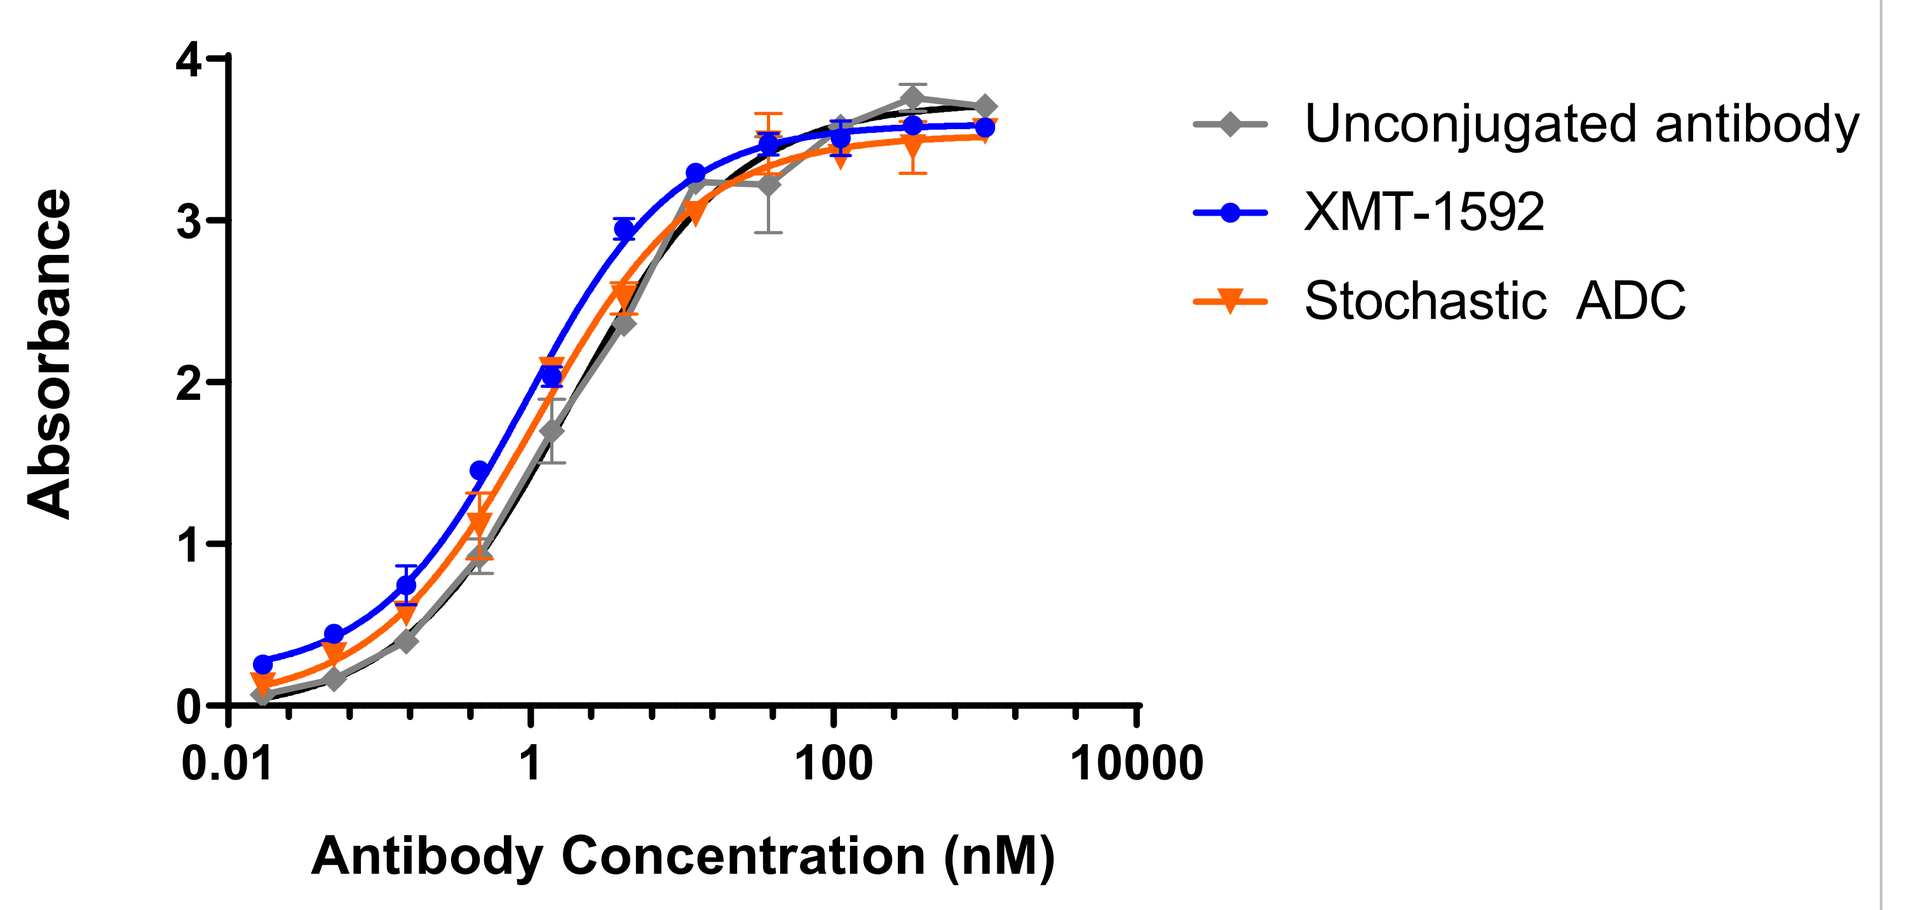
Absorbance
Antibody Concentration (nM)
Unconjugated antibody
XMT-1592
Stochastic ADC

Figure 3: Binding to ovarian cancer cells that express NaPi2b. XMT-1592 and the stochastic ADC retain high potency binding to antigen-expressing OVCAR-3 cancer cells.

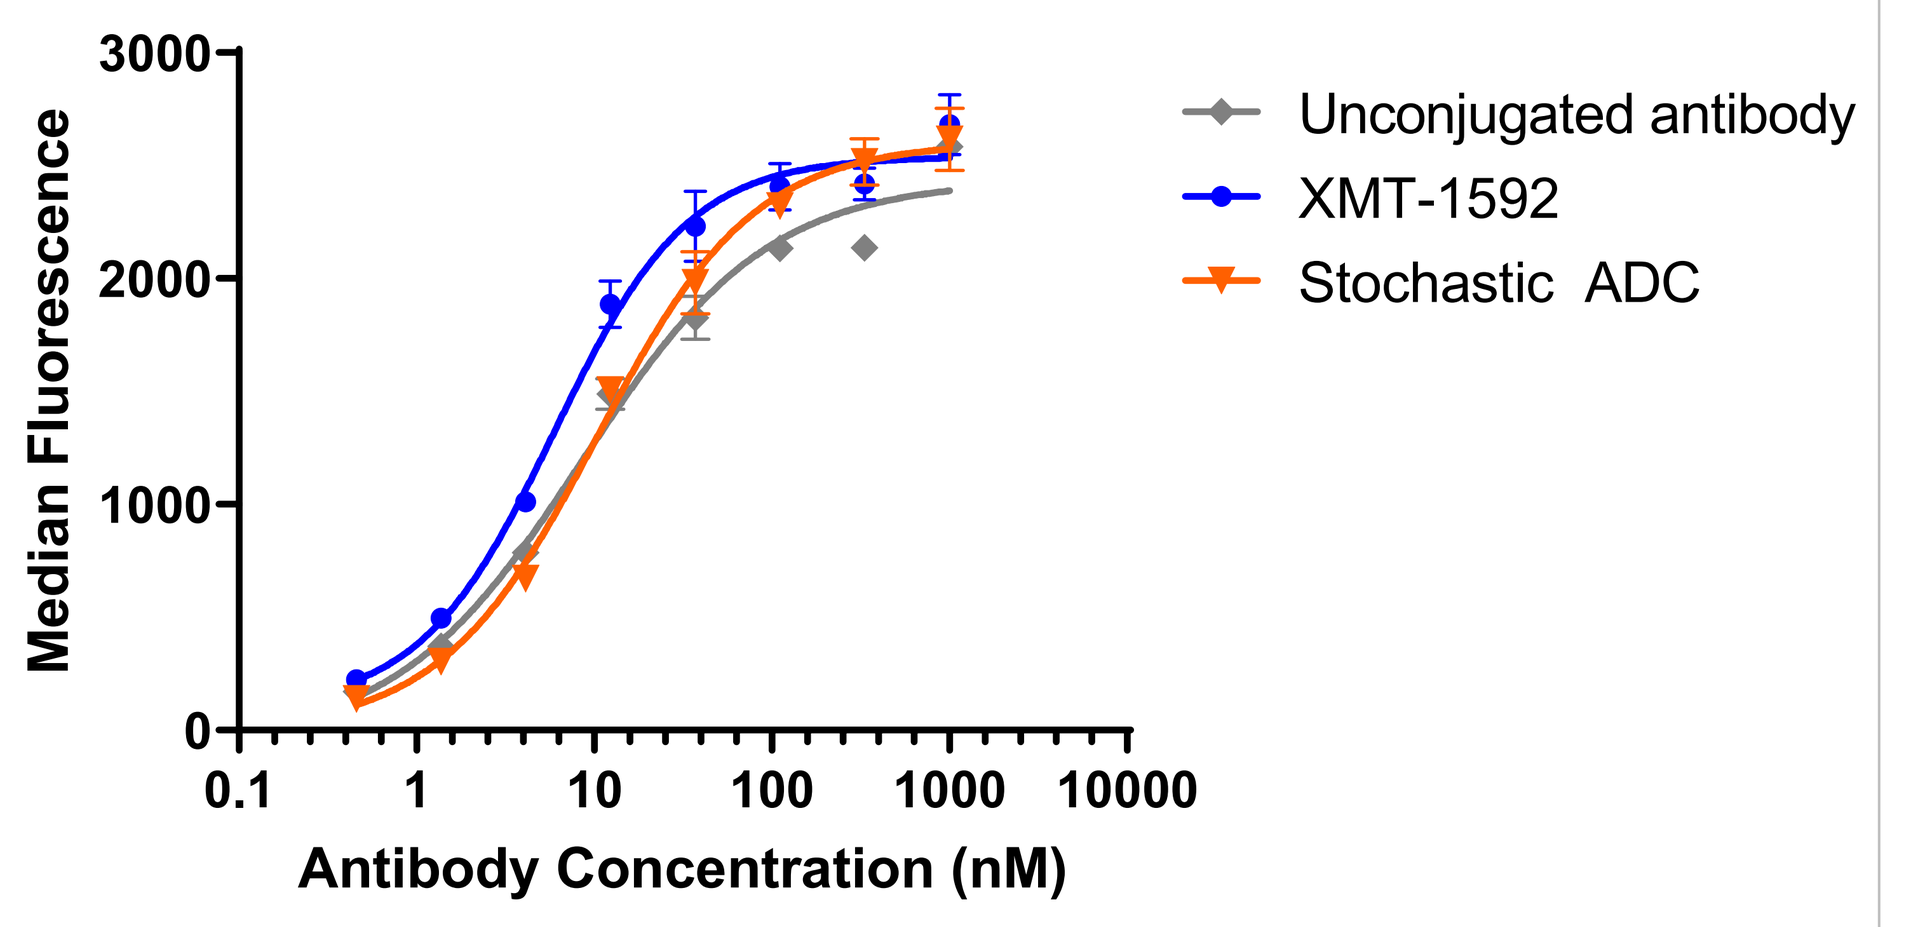
Median Fluorescence
Antibody Concentration (nM)
Unconjugated antibody
XMT-1592
Stochastic ADC

Figure 4: Potent cytotoxic activity. XMT-1592 and the stochastic ADC elicit potent and target-dependent cytotoxicity against OVCAR-3 cancer cells. The control ADC in Figures 4-6 is a non-binding antibody conjugated to Dolasynthen (site-specific).

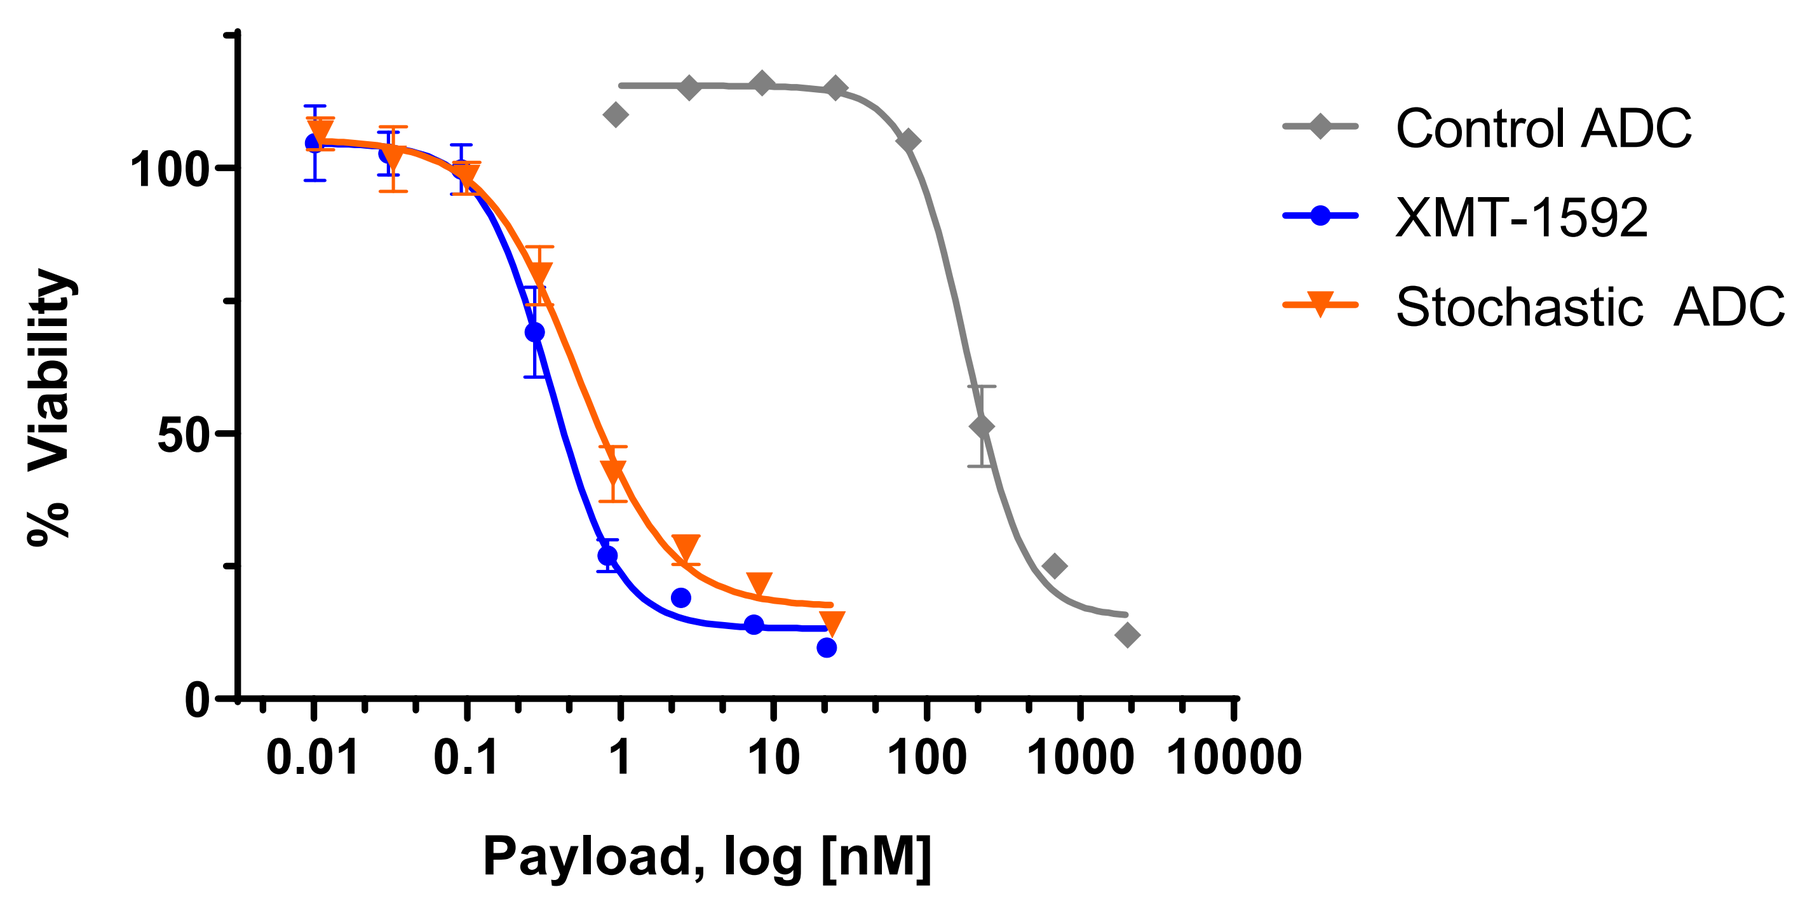
% Viability
Payload, log [nM]
Control ADC
XMT-1592
Stochastic ADC

Sustained Tumor Regression in an Ovarian Cancer Xenograft

Figure 5: Anti-tumor activity in OVCAR-3 xenografts. Athymic (*nu/nu*) mice bearing OVCAR-3 tumors were randomized into groups and administered one dose (black arrowhead) of XMT-1592, the stochastic ADC, or the control ADC. XMT-1592 outperformed the stochastic ADC at matched payload doses.

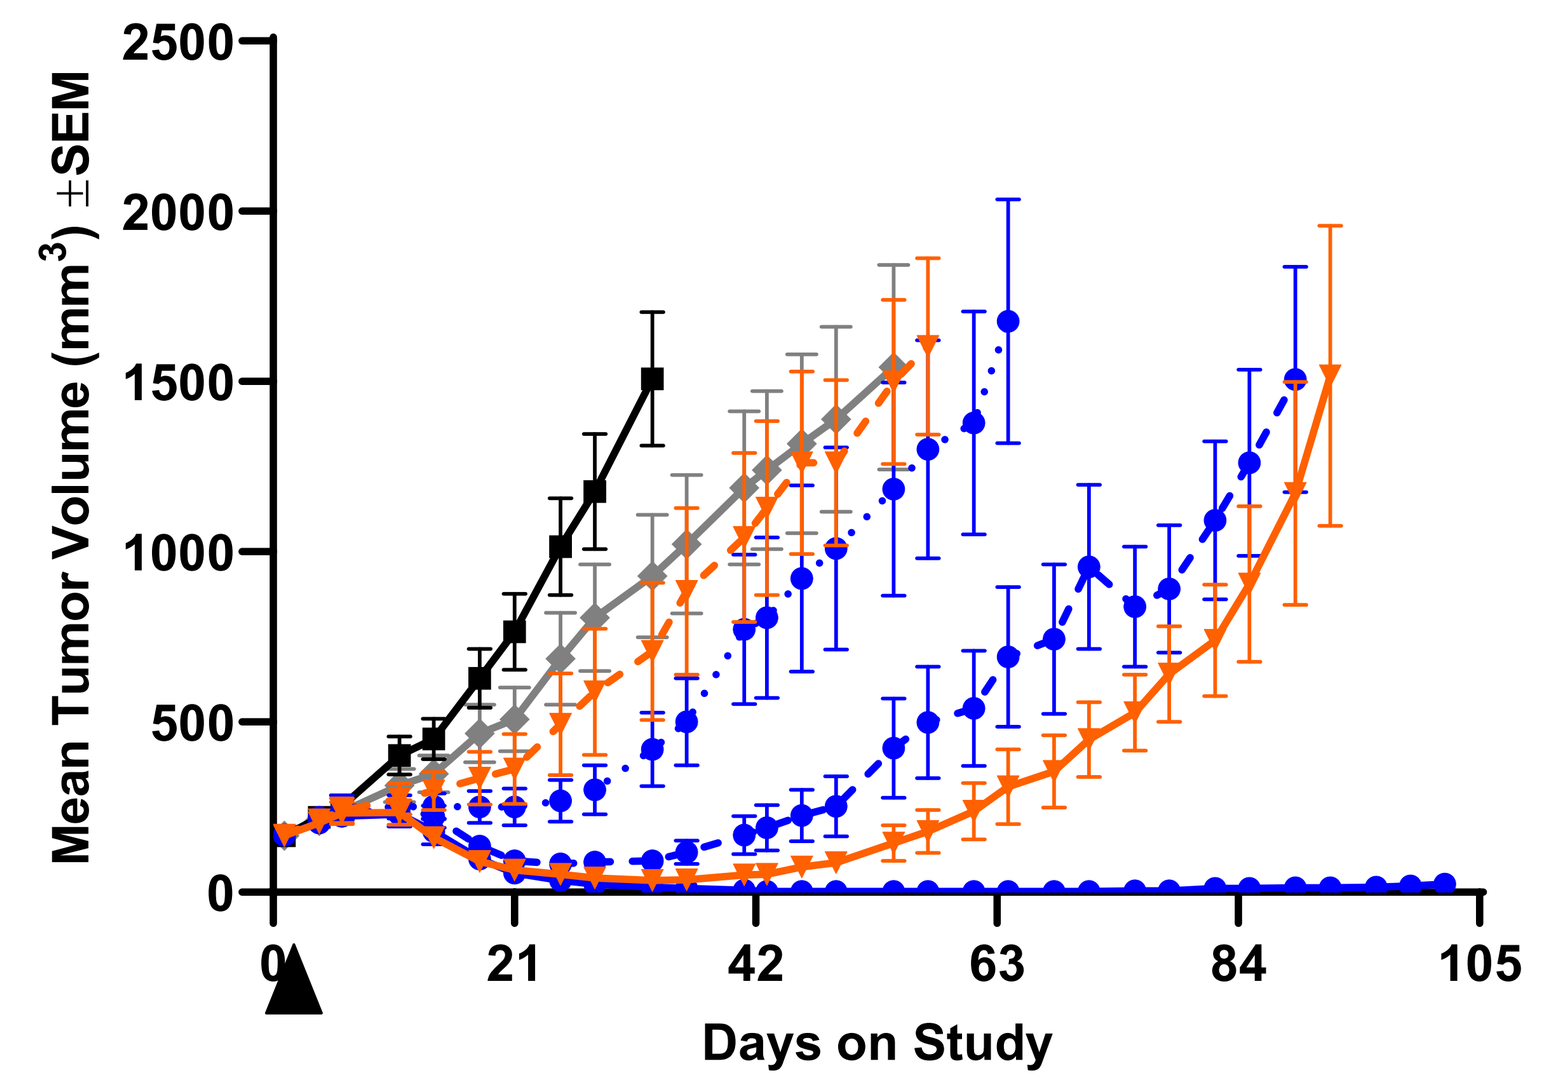
Mean Tumor Volume (mm³) ±SEM
Days on Study

	Test Article	AF-HPA dose (mg/kg)	Antibody dose (mg/kg)
	Vehicle	0	0
	Control ADC	0.1	3.0
	XMT-1592	0.1	3.0
		0.05	1.5
		0.025	0.75
	Stochastic ADC	0.1	2.6
		0.05	1.3

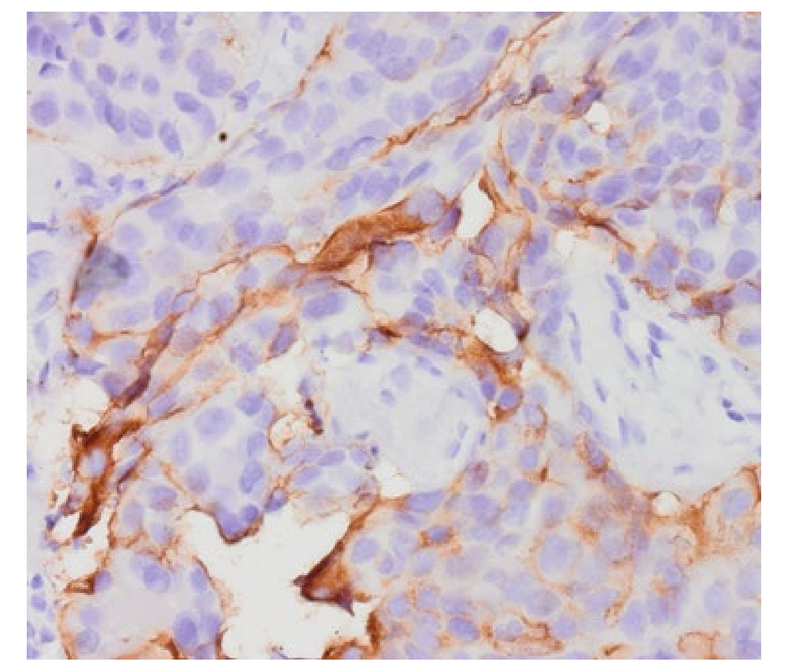

NaPi2b IHC in OVCAR-3 tumor.

Sustained Tumor Regression in a Lung Adenocarcinoma PDX

Figure 6: Anti-tumor activity in CTG-0852 lung adenocarcinoma xenografts. Athymic (*nu/nu*) mice bearing CTG-0852 tumors were randomized into groups and administered one dose (black arrowhead) of XMT-1592 or the control ADC. [The stochastic ADC was not included in this study.]

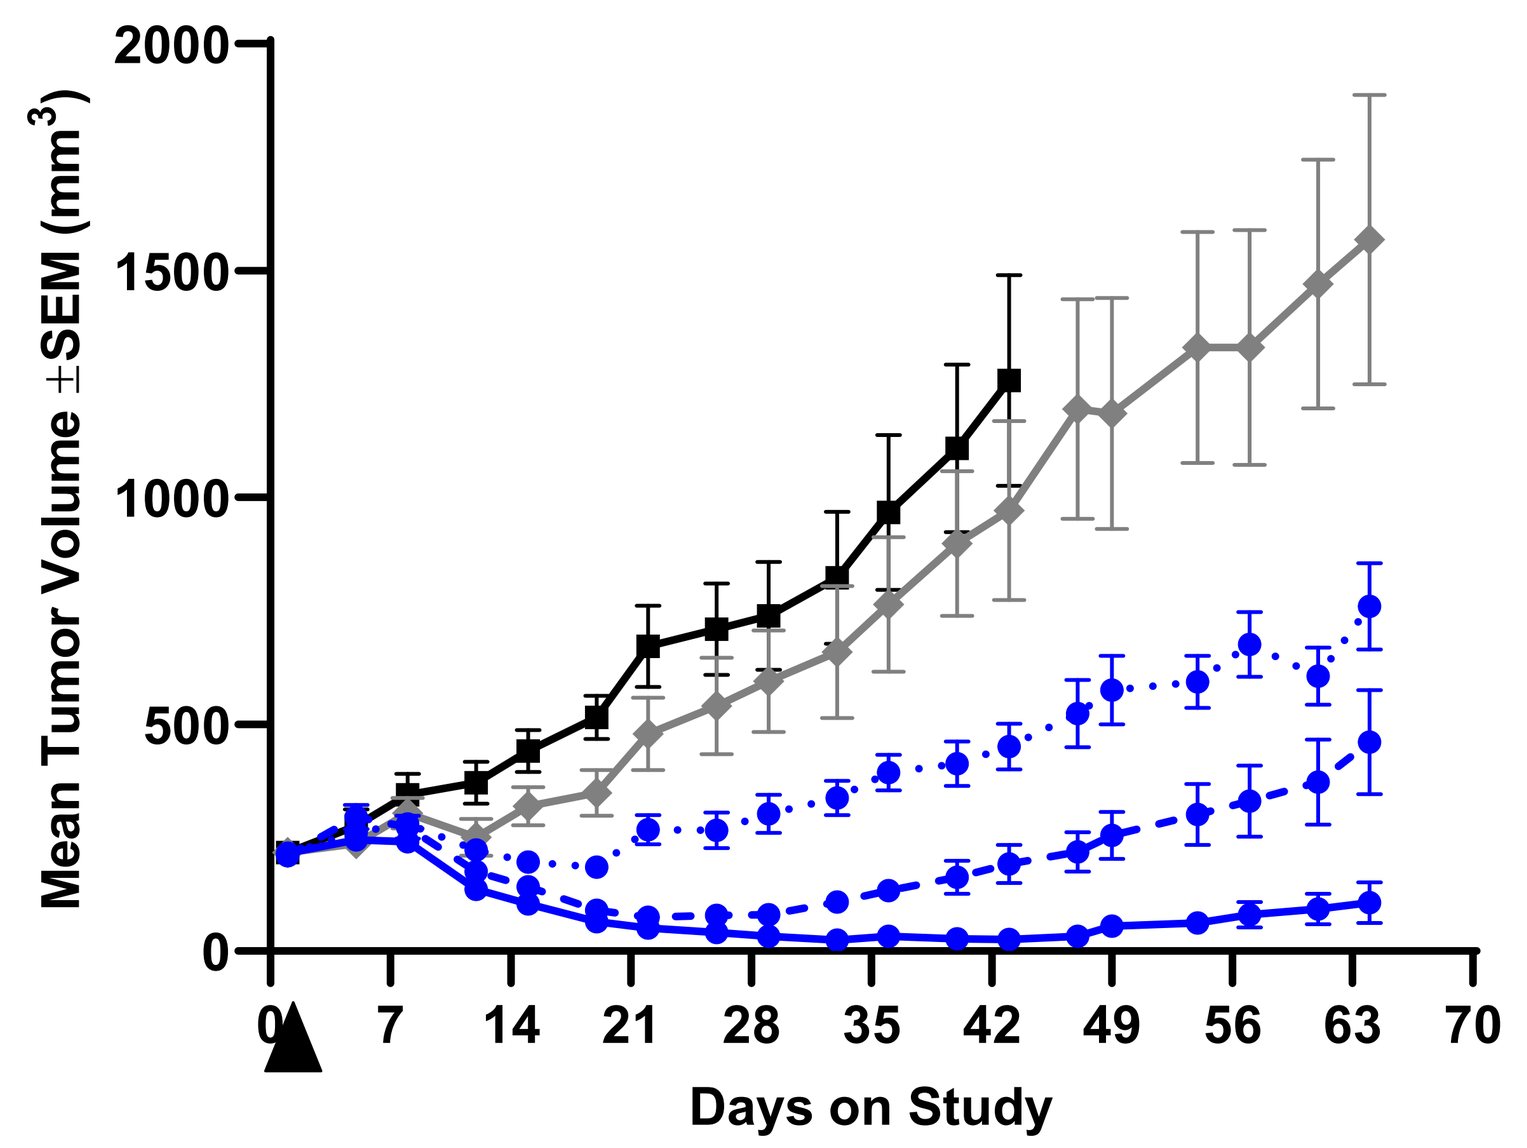
Mean Tumor Volume ±SEM (mm³)
Days on Study

	Test Article	AF-HPA dose (mg/kg)	Antibody dose (mg/kg)
	Vehicle	0	0
	Control ADC	0.1	3.0
	XMT-1592	0.1	3.0
		0.05	1.5
		0.025	0.75

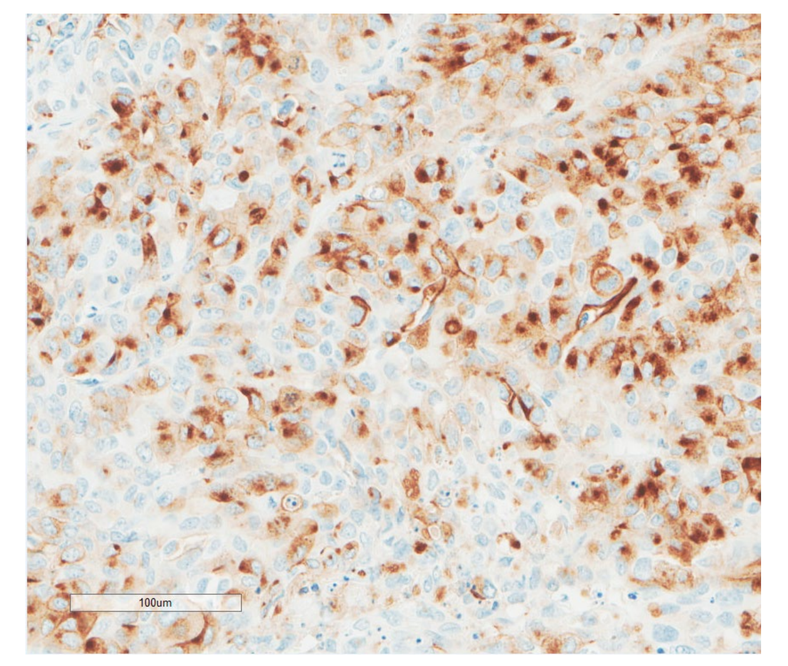

NaPi2b IHC in CTG-0852 PDX

Payload Accumulation in Tumor and Not in Normal Tissues

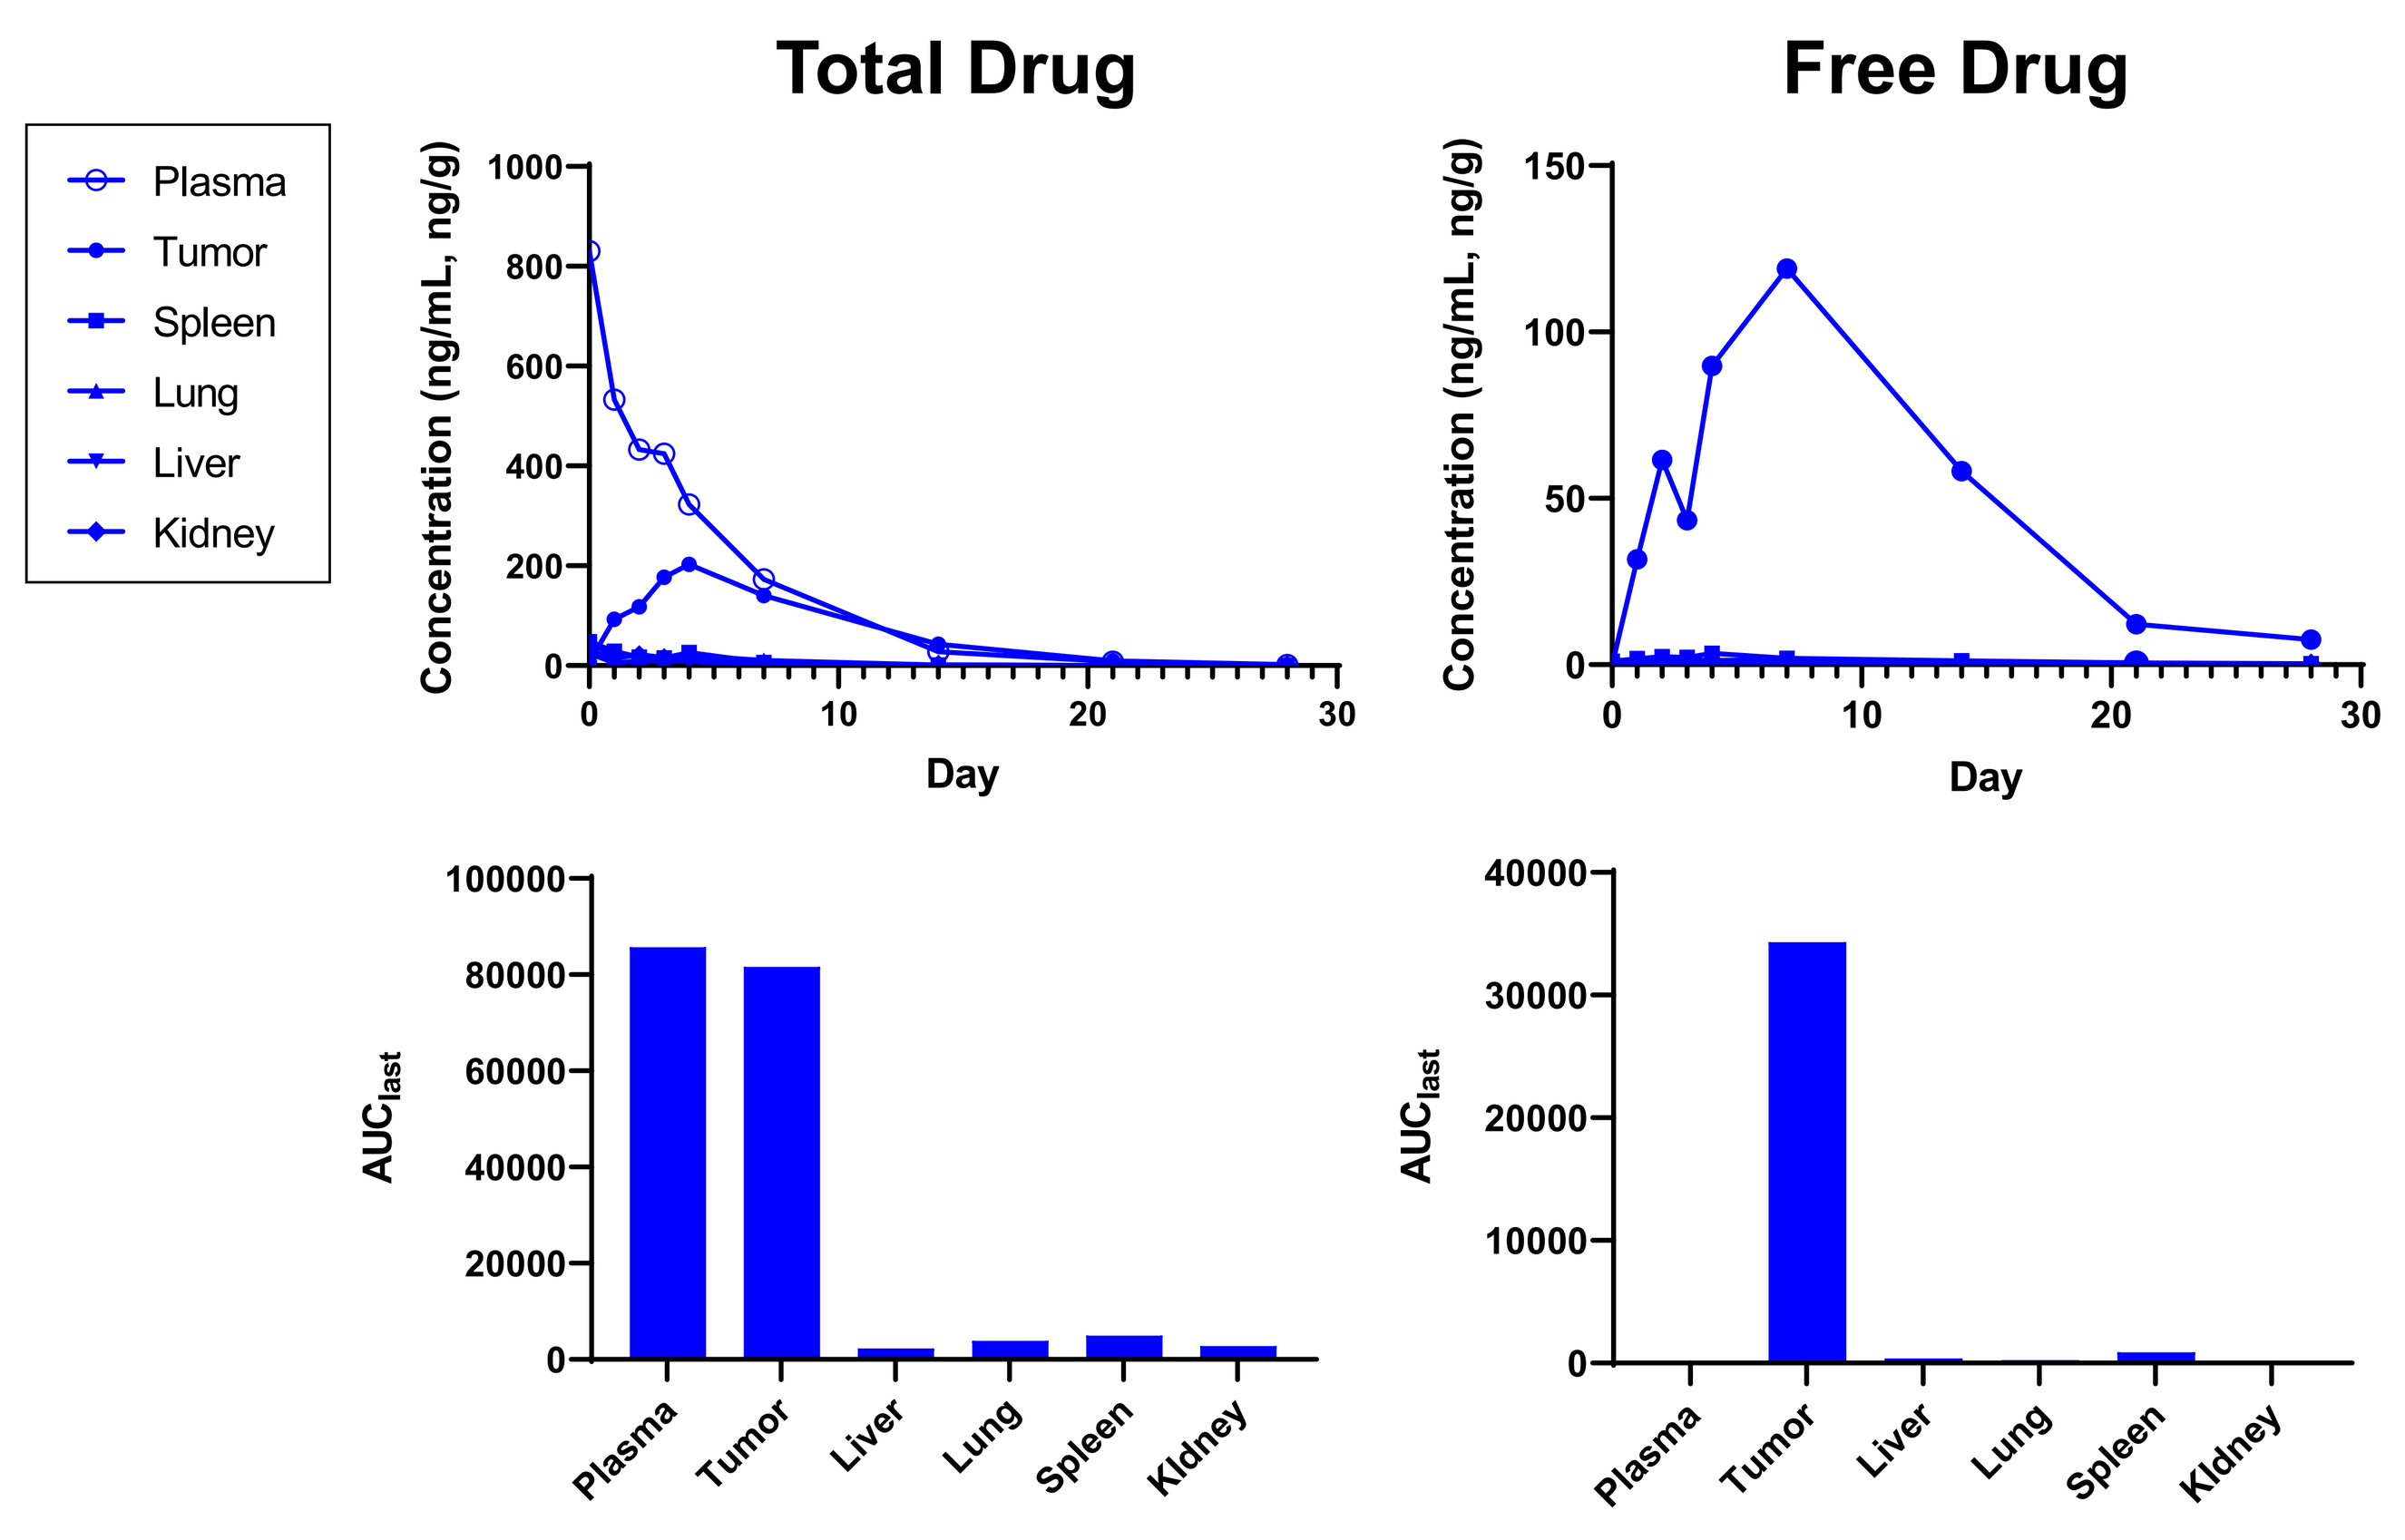
Total Drug
Free Drug
Plasma
Tumor
Spleen
Lung
Liver
Kidney
Concentration (ng/mL, ng/g)
Day
AUClast
Plasma
Tumor
Liver
Lung
Spleen
Kidney

Figure 7: Tissue biodistribution. Athymic (*nu/nu*) mice bearing OVCAR-3 tumors were administered one dose of XMT-1592 at 1.5/0.05 mg/kg antibody/payload; tissues were harvested from 4 mice at each time point and homogenized, and then total drug (after base hydrolysis) and free drug (without base hydrolysis) were measured by LC/MS-MS. The upper panels show the measured concentrations at each time point, and the bottom panels show the area under the curve (AUC).

Improved Pharmacokinetics Profile of XMT-1592 vs. Stochastic ADC

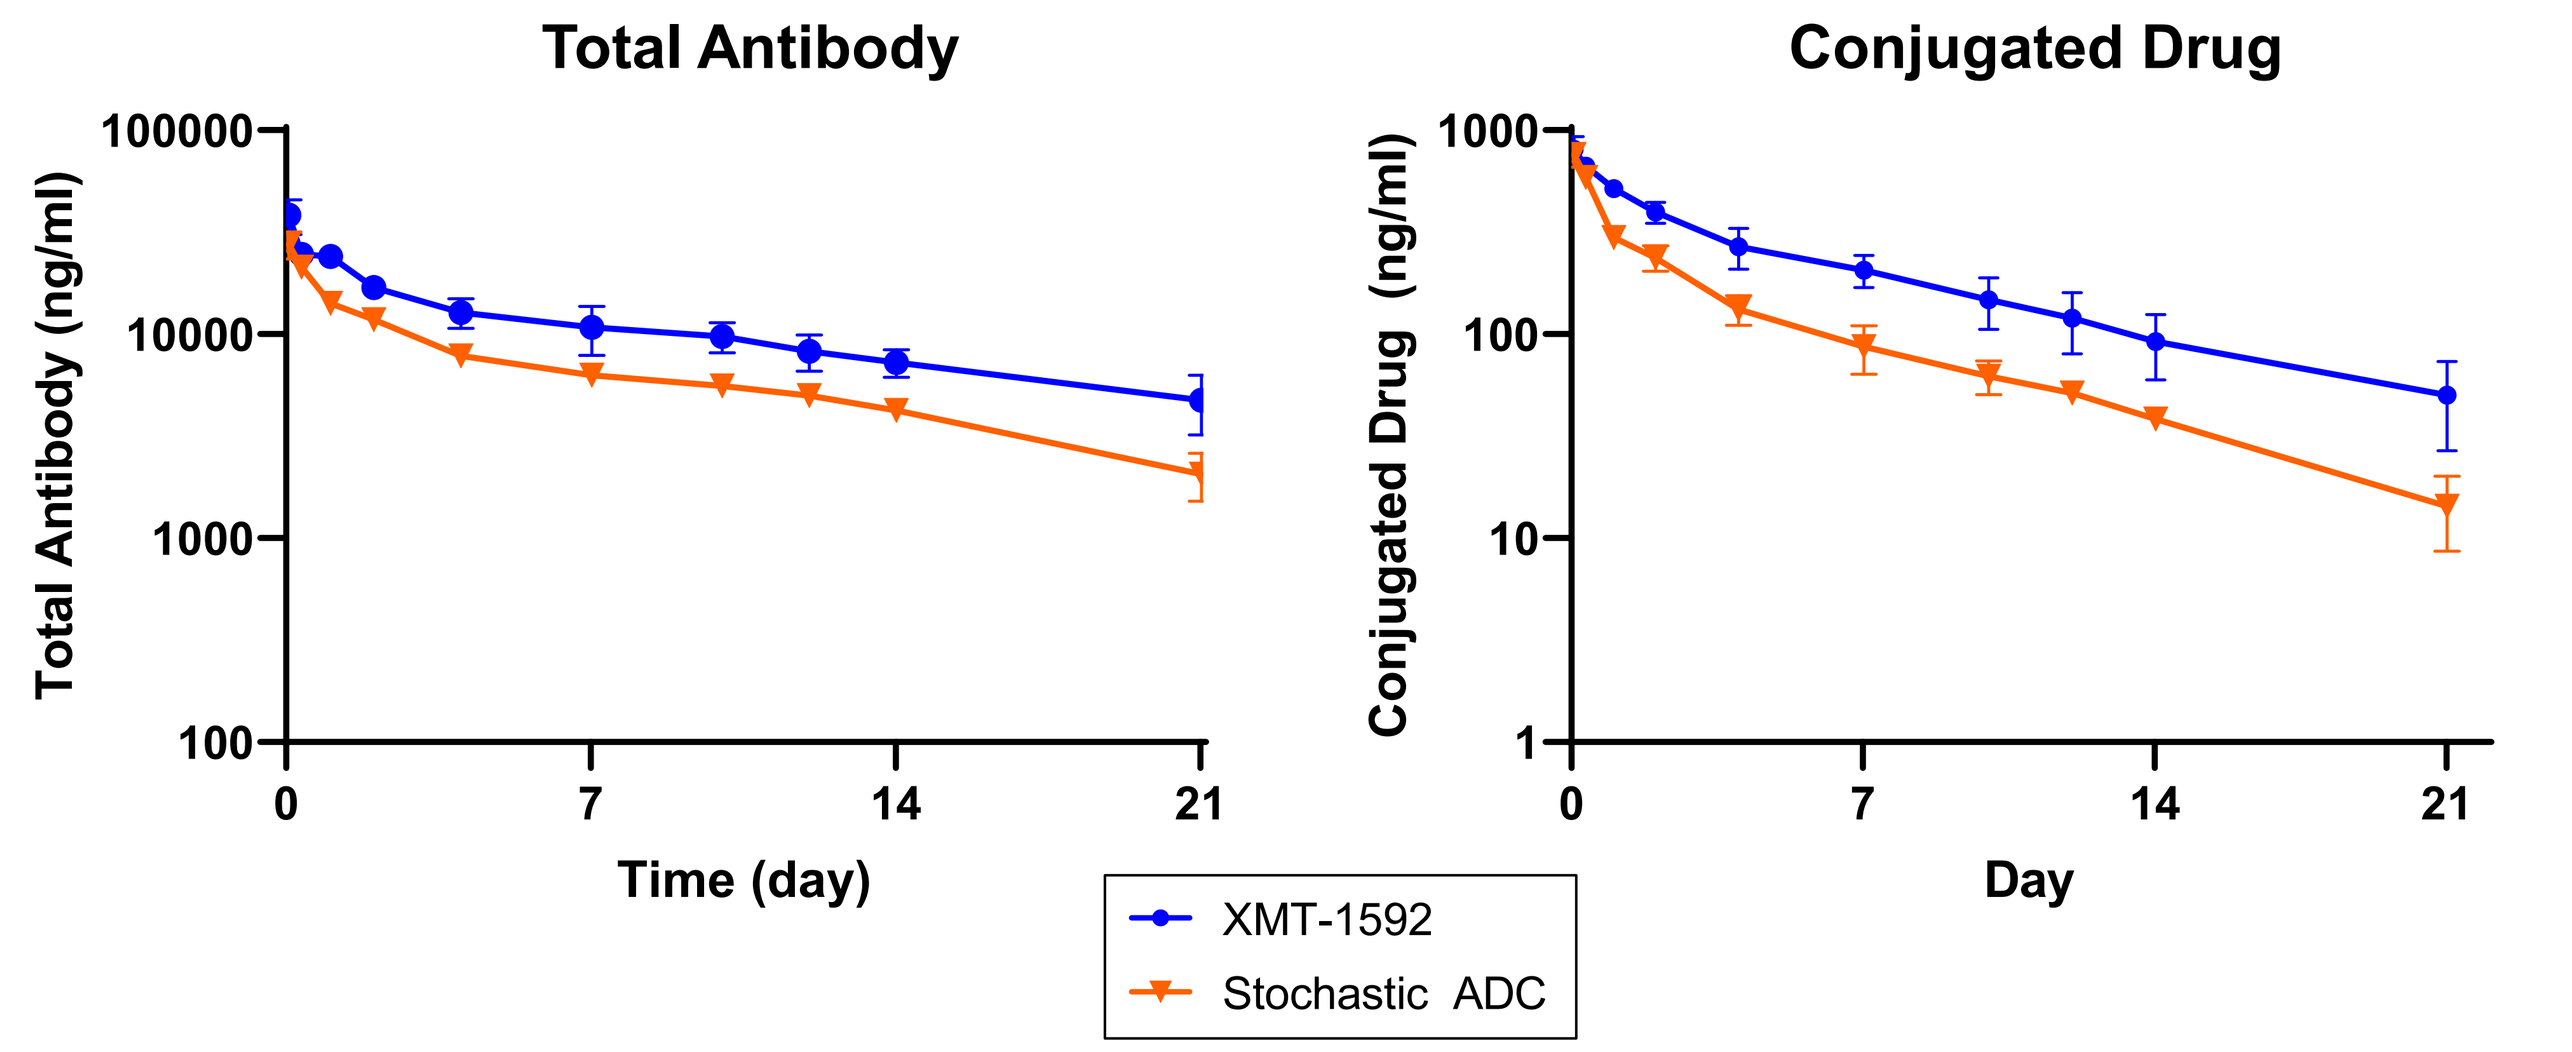
Total Antibody
Conjugated Drug
Total Antibody (ng/ml)
Conjugated Drug (ng/ml)
Time (day)
Day
XMT-1592
Stochastic ADC

Figure 8: Pharmacokinetics in non-human primates (NHP). Cynomologus monkeys were administered one dose of XMT-1592 or Stochastic ADC at 1 mg/kg antibody. Samples were collected at various time points and processed for the measurement of pharmacokinetics analytes including total antibody (left panel), conjugated drug (right panel), and free drug. XMT-1592 exhibited slower clearance of total antibody and conjugated drug. Free drug was minimal in all samples, which indicated the high stability of the ADCs.

Improved Clinical Pathology Profile of XMT-1592 vs. Stochastic ADC

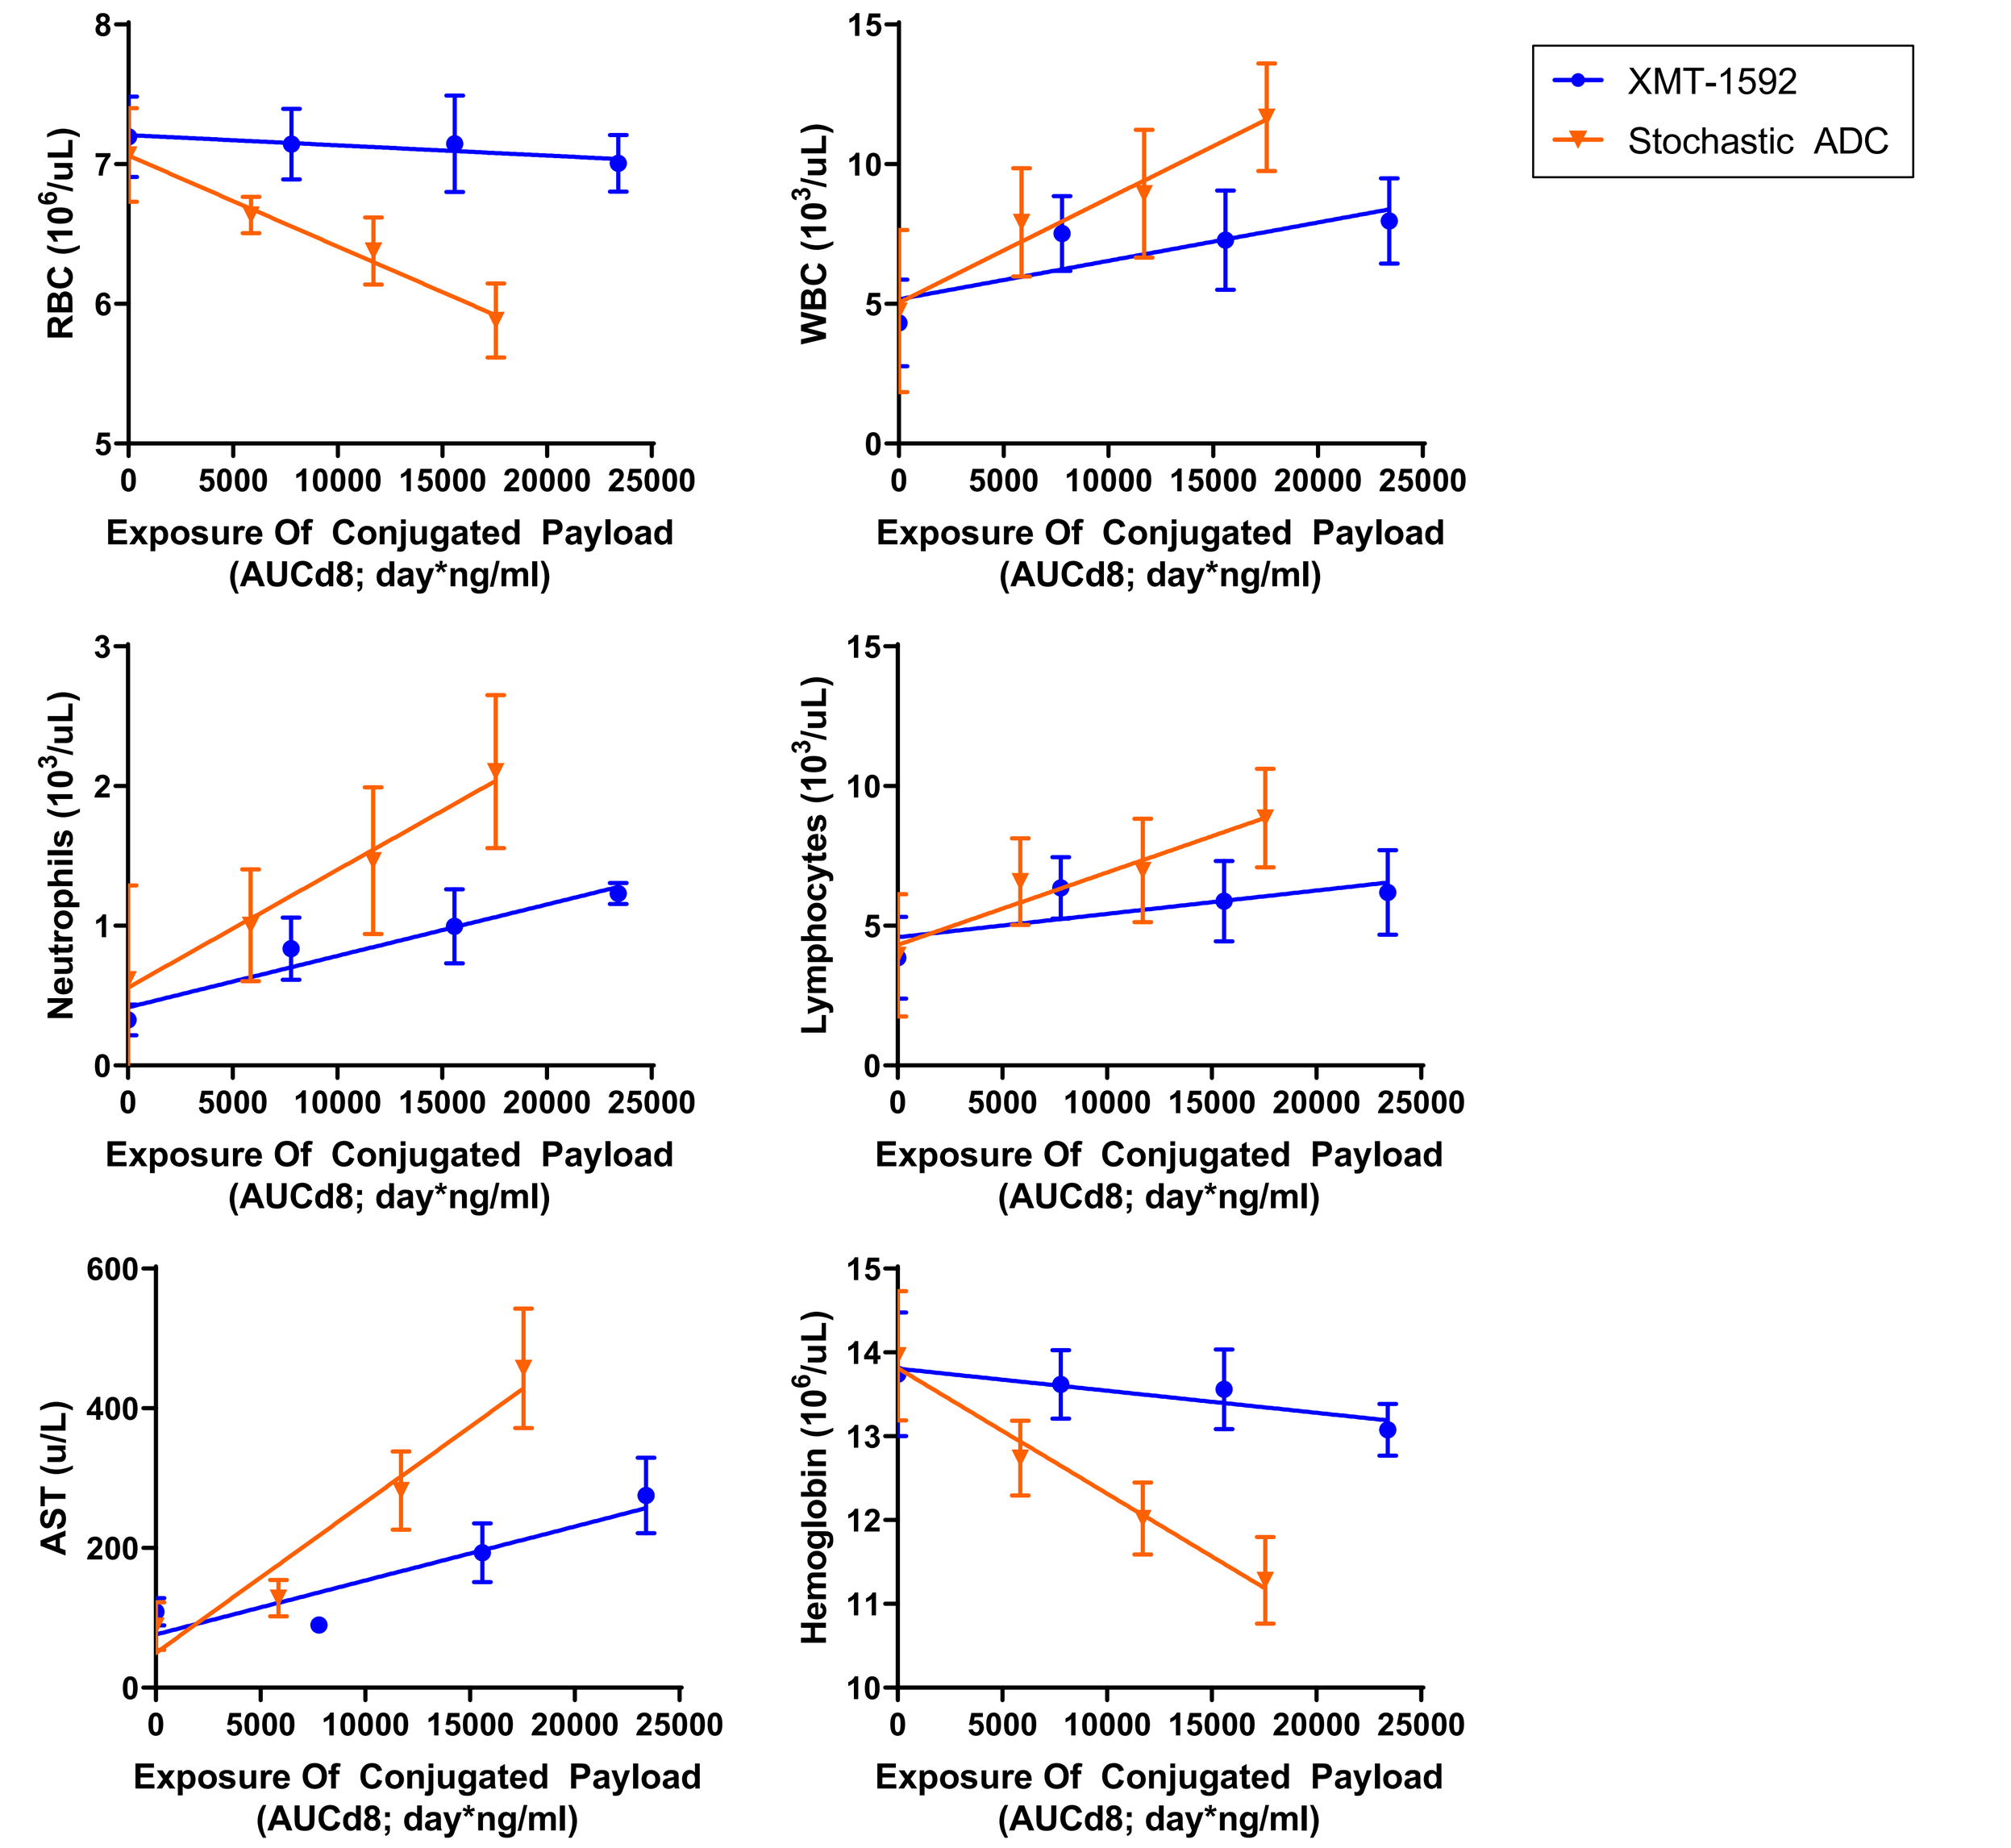
XMT-1592
Stochastic ADC
RBC (10⁶/uL)
WBC (10³/uL)
Neutrophils (10³/uL)
Lymphocytes (10³/uL)
AST (u/L)
Hemoglobin (10⁶/uL)
Exposure Of Conjugated Payload (AUCd8; day*ng/ml)

Figure 9: Clinical pathology in Sprague-Dawley rats. Animals were administered equal payload doses of XMT-1592 or the Stochastic ADC, and samples were taken for toxicokinetics analysis at multiple time points and for clinical pathology at Day 8. XMT-1592 induced significantly less change in several clinical pathology parameters, plotted here as a function of plasma exposure of conjugated drug. Selected parameters are shown.

Conclusions

1. XMT-1592 is a NaPi2b-targeted site-specific ADC, DAR 6, that uses the novel AF-HPA-based Dolasynthen platform and the Synaffix GlycoConnectTM platform.
2. XMT-1592 induced sustained tumor regressions in an ovarian tumor xenograft and a lung adenocarcinoma patient-derived xenograft.
3. XMT-1592 outperformed a stochastically conjugated ADC with the same antibody, payload, and DAR, exhibiting improved anti-tumor activity, pharmacokinetics, and clinical pathology.
4. A Phase 1 dose escalation clinical study of XMT-1592 was initiated in May 2020 (NCT04396340)

References

D. Toader, *et al*. Dolasynthen: A Novel, Homogeneous Auristatin F Hydroxypropyl Amide Antibody-Drug Conjugate Platform. 2019 AACR Annual Meeting, Atlanta GA.

K. Lin, *et al.* Preclinical Development of an Anti-NaPi2b (SLC34A2) Antibody-Drug Conjugate as a Therapeutic for Non-Small Cell Lung and Ovarian Cancers. Clin Cancer Res. 2015;**21**(22):5139-50.

R. van Geel, *et al*. Chemoenzymatic Conjugation of Toxic Payloads to the Globally Conserved N-Glycan of Native mAbs Provides Homogeneous and Highly Efficacious Antibody-Drug Conjugates. Bioconjugate Chem 2015: **26**, 2233-2242.