Abstract ID: 1773

Tumor cell-intrinsic STING pathway activation leads to robust induction of Type III Interferons and contributes to the anti-tumor activity elicited by STING agonism

Naniye Malli Cetinbas, Travis Monnell, Kalli C. Catcott, Winnie Lee, Pamela Shaw, Kelly Slocum, Keith Bentley, Susan Clardy, Eoin Kelleher, Rebecca Mosher, Marina Protopopova, Joshua D. Thomas, Dorin Toader, Jeremy Duvall, Raghida A. Bukhalid, Marc Damelin, Timothy B. Lowinger

Mersana Therapeutics, Inc., Cambridge, MA

Abstract

The STING pathway plays a critical role in inducing anti-tumor immunity by upregulating Type 1 Interferon (IFN) and IFN-stimulated genes within the tumor microenvironment in response to cytosolic nucleic acid ligands. Therefore, STING pathway agonism has emerged as a potential therapeutic mechanism to stimulate an anti-tumor innate immune response. Intratumorally injected free STING-agonists that are currently being evaluated in the clinic by others have shown limited effects in non-injected lesions. Antibody-drug conjugates (ADCs) constitute a proven therapeutic modality that enables tumor-targeted drug delivery with systemic administration. We have previously demonstrated that the tumor cell-intrinsic STING pathway can be activated in the presence of cues from immune cells and contributes to the anti-tumor activity of tumor cell-targeted Immunosynthen STING-agonist ADCs, in which a STING-agonist payload is conjugated to a tumor cell-targeting antibody. Here we investigated the nature of the STING pathway activation in tumor cells and its contribution to the anti-tumor activity elicited by STING agonism. Leveraging ADCs with a wild type (wt) or mutant Fc (deficient in Fcγ receptor -FcγR- binding), we delivered a STING-agonist simultaneously to both tumor-resident immune and cancer cells or only to cancer cells through FcγR-mediated and/or tumor antigen-mediated ADC internalization. We utilized these ADCs in *in vivo* human tumor xenograft models and STING wild type (wt) or knock out (ko) cancer cell:immune cell co-cultures and evaluated gene expression, cytokine production, and anti-tumor activities induced by STING-agonist ADCs. Surprisingly, Nanostring analysis of the human tumor xenografts from mice treated with tumor cell-targeted STING-agonist ADCs revealed human tumor cell-specific activation of Type III IFNs. In human cancer cell:immune cell co-cultures, treatment with tumor cell-targeted STING-agonist ADCs also led to marked upregulation of Type III IFNs, which was significantly reduced in STING ko cancer cell:immune cell co-cultures, suggesting that the cancer cells may contribute the majority of the Type III IFNs downstream of STING pathway activation. Blocking Type III IFNs with neutralizing antibodies in cancer cell:immune cell co-cultures inhibited the production of key cytokines, including Type I IFN, and nearly abolished tumor cell-killing in response to STING-agonist ADC treatment, indicating that the Type III IFNs play an important role in the anti-tumor activity induced by STING activation. These studies reveal a previously underappreciated mechanism of STING agonist anti-tumor activity. The ability of tumor cell-targeted STING-agonist ADCs to activate STING in both tumor cells and in tumor-resident immune cells may represent a significant therapeutic advantage of an Immunosynthen ADC approach to STING agonism.

Background

Tumor cell-targeted Immunosynthen STING agonist ADCs

- ❖ Systemically administered
- ❖ Tumor targeted delivery of STING agonist
- ❖ Efficacious at a single dose across multiple tumor models
- ❖ Well-tolerated at multiple doses in multiple non-clinical species
- ❖ Minimal systemic induction of inflammatory cytokines
- ❖ Dramatically greater efficacy compared to a systemically administered free STING agonist

Please refer to our additional poster (AACR 2021 #1738) and www.mersana.com for more information about our Immunosynthen platform

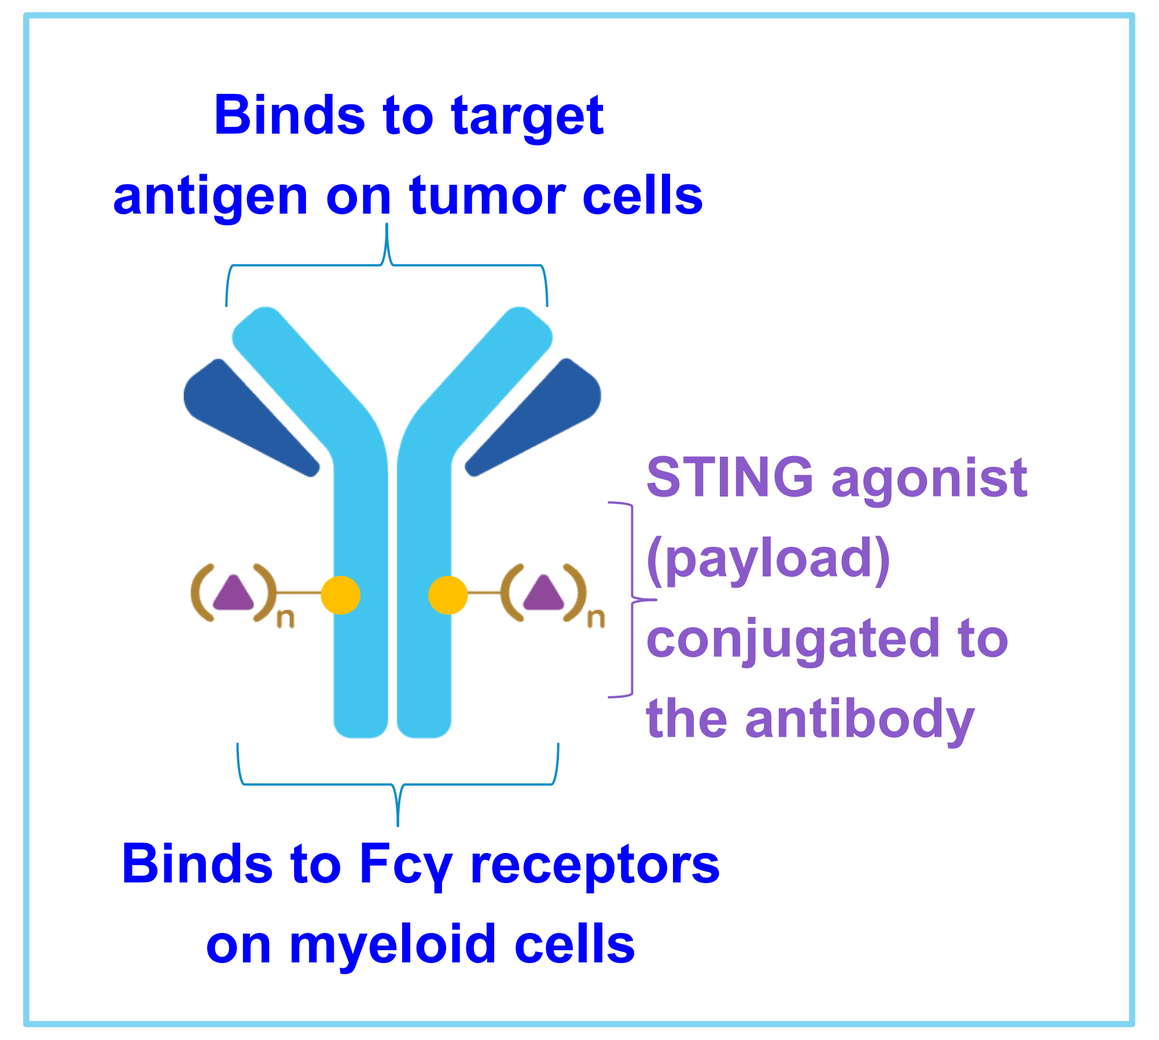
Binds to target
antigen on tumor cells
STING agonist
(payload)
conjugated to
the antibody
Binds to Fcγ receptors
on myeloid cells

Proposed mechanism of action of tumor cell-targeted STING agonist ADCs

A.

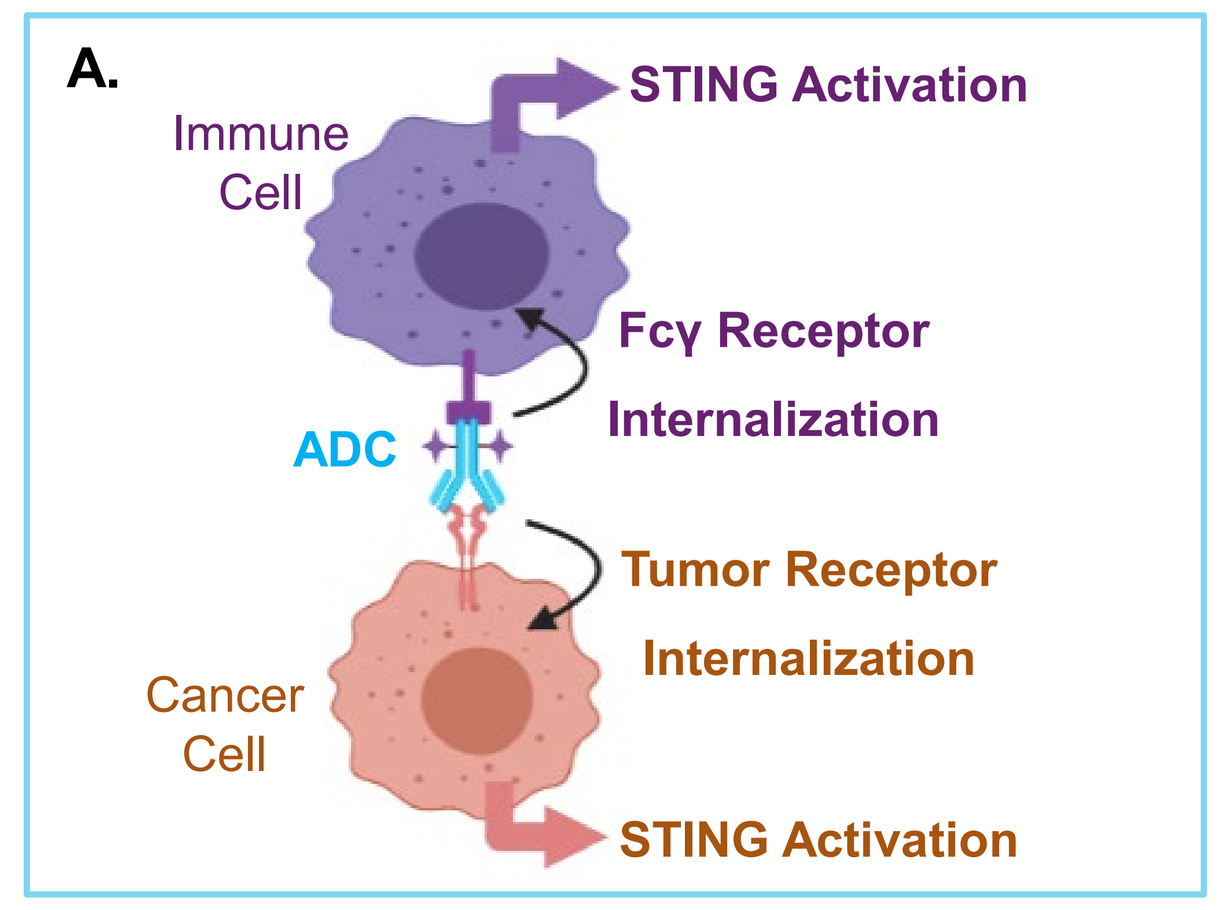
STING Activation
Immune
Cell
Fcγ Receptor
Internalization
ADC
Tumor Receptor
Internalization
Cancer
Cell
STING Activation

B. **Antibody variants used to determine the MOA**

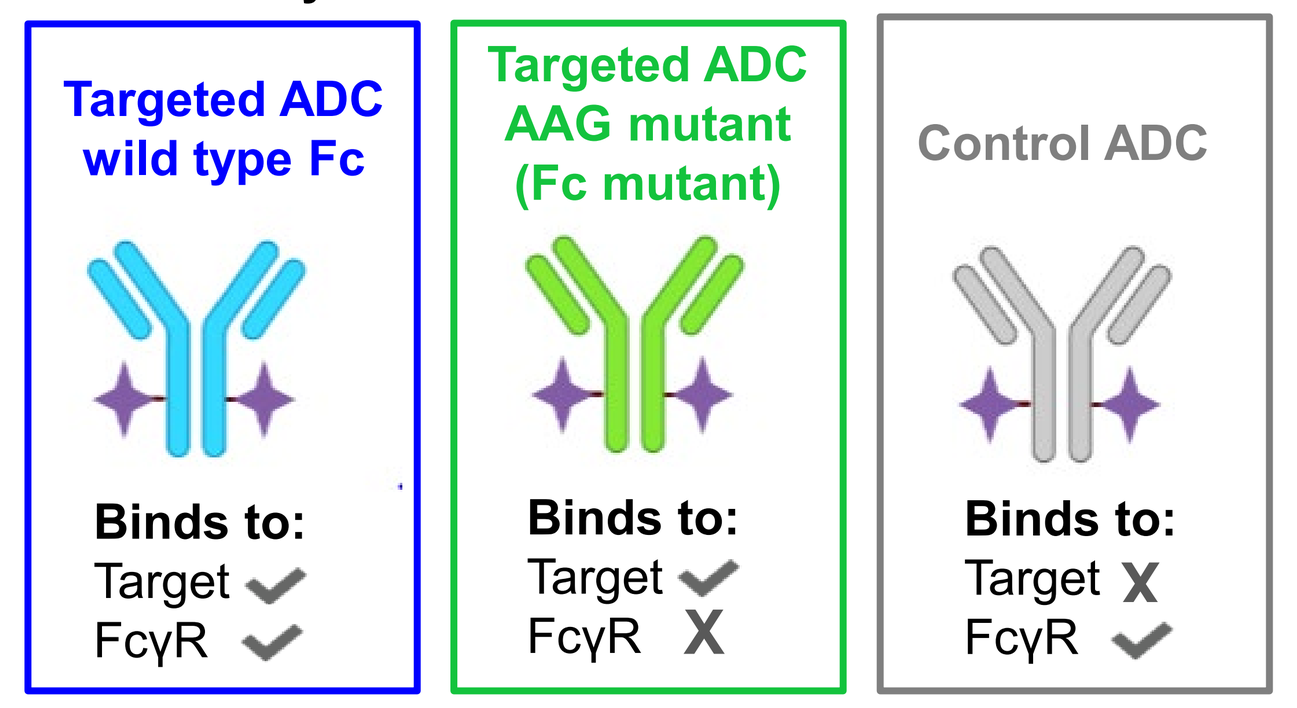
Targeted ADC
wild type Fc
Binds to:
Target ✓
FcγR ✓
Targeted ADC
AAG mutant
(Fc mutant)
Binds to:
Target ✓
FcγR X
Control ADC
Binds to:
Target X
FcγR ✓

Figure 1. Delivery of a STING agonist into tumor cells and myeloid cells via a tumor cell-targeted ADC.

- When bound to their target antigens on tumor cells ADCs strongly interact and crosslink Fcγ receptors and internalize into immune cells resulting in robust activation of the STING pathway.
- Tumor cell-targeted Immunosynthen ADCs are also internalized into tumor cells and can activate tumor intrinsic STING pathway in the presence of cues from immune cells[1].
- In this study we investigated the specific contribution of the tumor cell-intrinsic STING activation to the anti-tumor activity of tumor cell-targeted STING agonist ADCs by utilizing ADCs engineered to deliver STING agonist specifically either into both immune cells and cancer cells, or only into cancer cells in *in vitro* co-cultures of immune cells with STING wt or ko cancer cells, and in *in vivo* human tumor xenograft models.

RESULTS

Tumor cell-intrinsic STING pathway is activated *in vitro* in the presence of cues from primary human immune cells

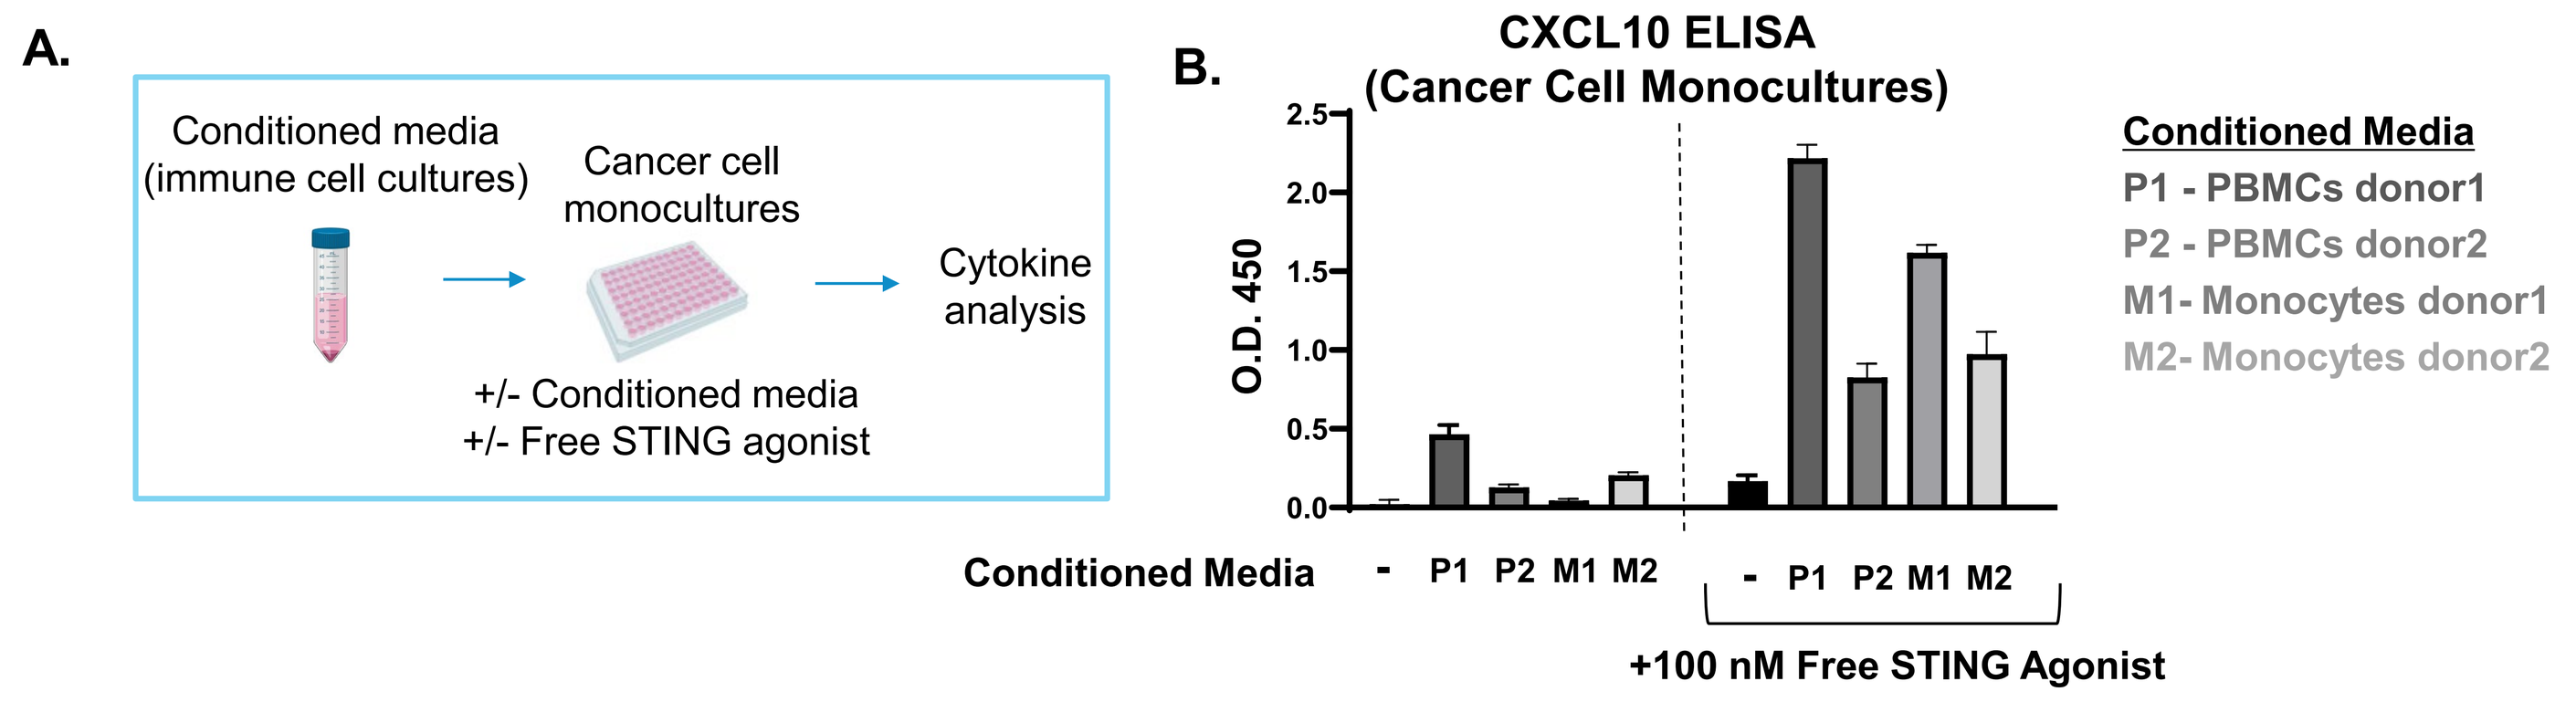
A.
Conditioned media
(immune cell cultures)
Cancer cell
monocultures
Cytokine
analysis
+/- Conditioned media
+/- Free STING agonist
B.
CXCL10 ELISA
(Cancer Cell Monocultures)
O.D. 450
Conditioned Media
+100 nM Free STING Agonist
Conditioned Media
P1 - PBMCs donor1
P2 - PBMCs donor2
M1- Monocytes donor1
M2- Monocytes donor2

Figure 2. STING pathway is activated in cancer cell monocultures in the presence of cues from immune cells. (A) Description of the immune cell-conditioned media experiment. **(B)** Supernatants (conditioned media) from untreated cultures of human PBMCs and isolated primary monocytes were harvested (spun down to remove immune cells) and added to cancer cell monocultures followed by free STING agonist treatment (100 nM). After 24 hours STING pathway activation was determined by human CXCL10 ELISA assay. STING agonist treatment led to potent activation of STING pathway in cancer cell monocultures only in the presence of conditioned media from PBMCs and monocytes supporting the hypothesis that STING pathway can be activated in cancer cells in the presence of cues from primary immune cells

Tumor cell-targeted Fc mutant STING agonist ADCs exhibit significant activity in cancer cell : primary human immune cell co-cultures and in *in vivo* tumor models

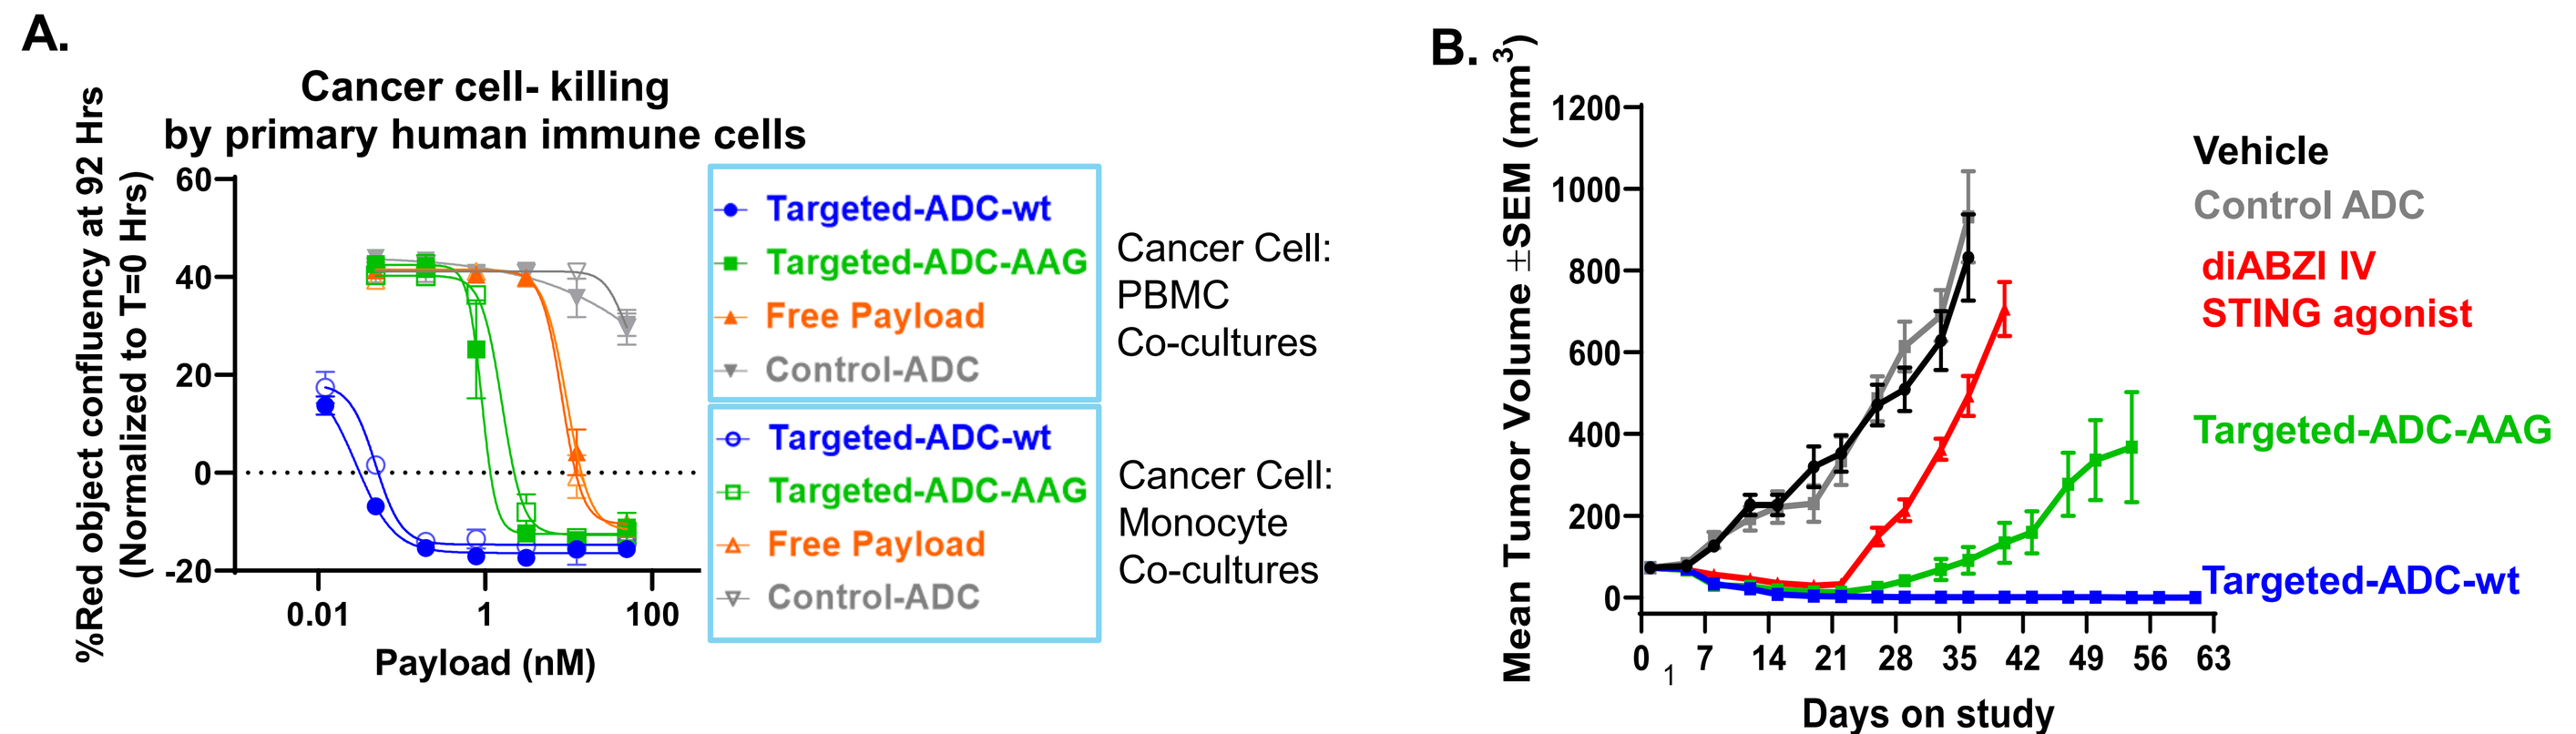
A.
Cancer cell- killing
by primary human immune cells
%Red object confluency at 92 Hrs
(Normalized to T=0 Hrs)
Payload (nM)
Targeted-ADC-wt
Targeted-ADC-AAG
Free Payload
Control-ADC
Cancer Cell:
PBMC
Co-cultures
Cancer Cell:
Monocyte
Co-cultures
B.
Mean Tumor Volume ±SEM (mm3)
Days on study
Vehicle
Control ADC
diABZI IV
STING agonist
Targeted-ADC-AAG
Targeted-ADC-wt

Similar results were obtained with other tumor antigens

Figure 3. Tumor cell-targeted Fc mutant STING agonist ADC exhibits significant activity in cancer : primary immune cell co-cultures and in *in vivo* tumor models. (A) Cancer cells expressing the ADC-target antigen were co-cultured with human PBMCs or monocytes in the presence of the indicated treatments and cancer cell growth was traced over time in an IncuCyte instrument. **(B)** Single doses of intravenously administered ADCs targeting an antigen expressed by the tumors led to complete and durable regressions, unlike the diABZI IV agonist[2], which was administered at a ~50x higher payload dose of the ADC. Non-targeted control ADC showed no efficacy. Consistent with cancer cell : primary immune cell co-culture results (Fig. 3A), Fc mutant targeted ADC exhibited lower but significant efficacy relative to the targeted ADC with wt Fc, suggesting that the tumor-intrinsic STING activation contributes to the tumor-targeted STING agonist ADC anti-tumor activity *in vivo*.

-Cancer cell lines used in these assays do not respond to STING agonism in monocultures (data not shown).

-Naked targeted antibodies have very minimal impact in cancer cell killing by primary immune cells (data not shown).

CRISPR knock out of STING in tumor cells confirms the contribution of tumor cell-intrinsic STING to the ADC activity

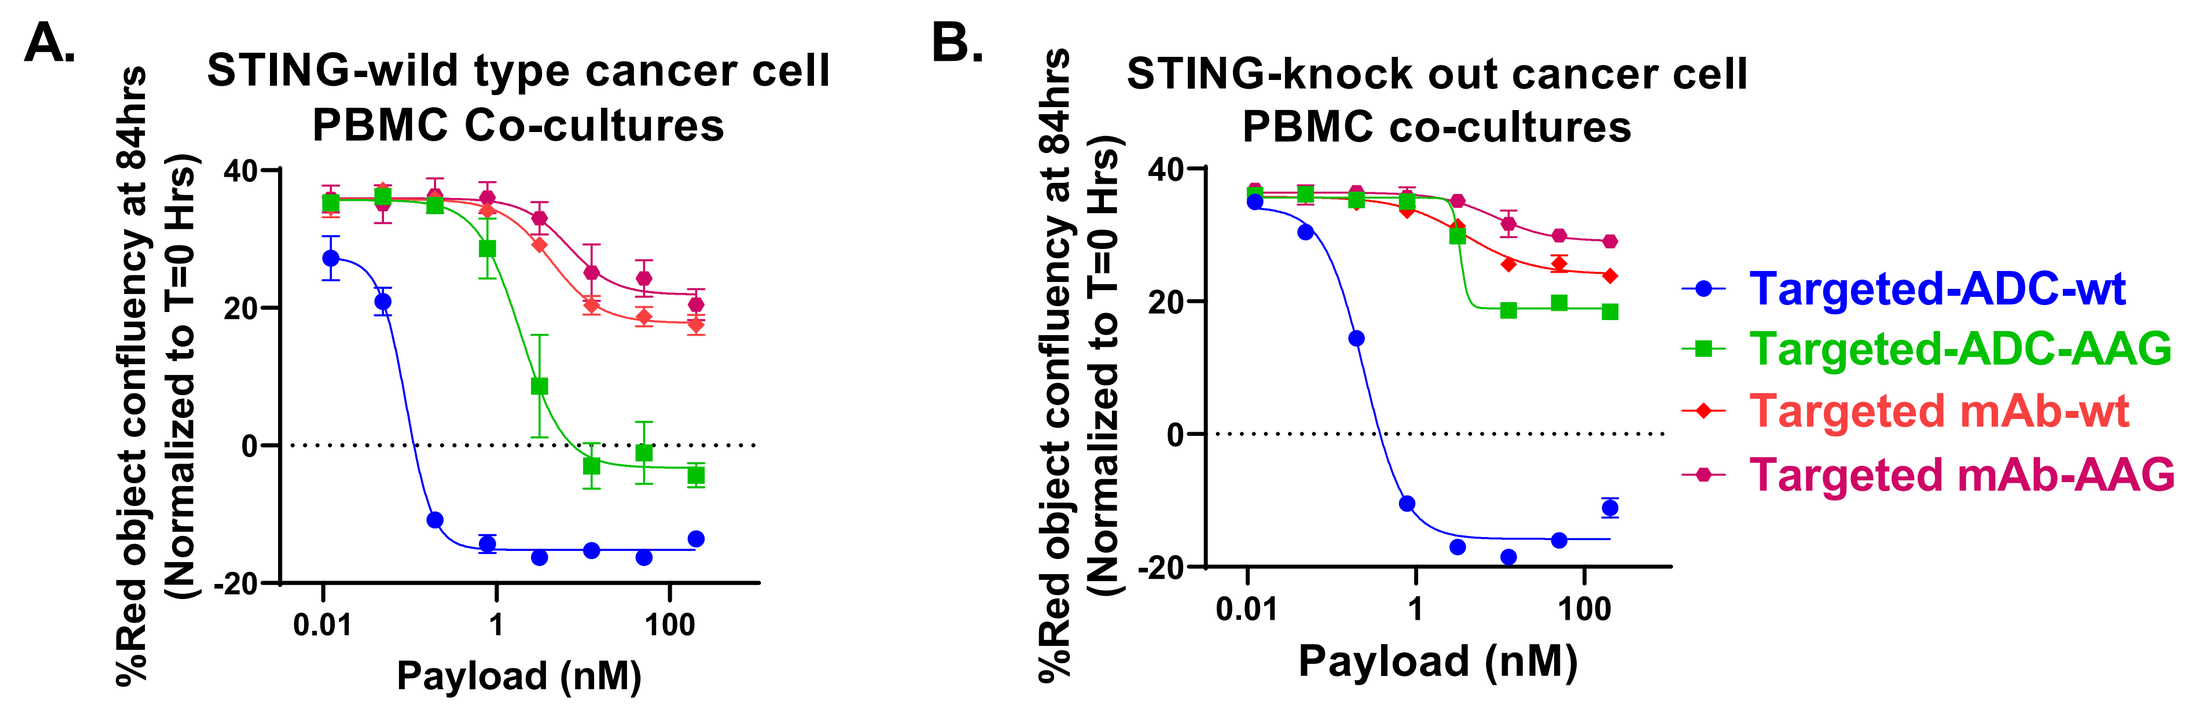
A.
STING-wild type cancer cell
PBMC Co-cultures
B.
STING-knock out cancer cell
PBMC co-cultures
%Red object confluency at 84hrs
(Normalized to T=0 Hrs)
Payload (nM)
Targeted-ADC-wt
Targeted-ADC-AAG
Targeted mAb-wt
Targeted mAb-AAG

Figure 4. Tumor cell-targeted Fc mutant ADC activity is diminished in STING knock out cancer : PBMC co-cultures. STING wt **(A)** and ko **(B)** cancer cells were co-cultured with PBMCs and cancer cell killing activity of the targeted Fc wt or Fc mutant ADCs, and naked targeted antibodies with wt or AAG-mutated Fc were assessed by IncuCyte analysis as described in Fig. 3A. Consistent with the results shown in Fig. 3A, targeted ADC with wt Fc induced significant killing of both STING wt and ko cancer cells in PBMC co-cultures, whereas Fc mutant targeted ADC induced significant killing of only STING wt cells but had diminished activity in STING ko cancer cell : PBMC co-cultures. Similar results were obtained with STING ko cancer cells generated with two other sgRNAs (2 clones each, data not shown). These data demonstrate the contribution of tumor-intrinsic STING pathway activation to the anti-tumor activity of the tumor cell-targeted STING agonist ADCs in *in vitro* co-cultures of human cancer cell : primary immune cell co-cultures.

- Unconjugated antibodies dosed at the corresponding antibody concentrations of the ADC.

Nanostring analysis of human tumor xenografts reveals tumor cell specific induction of Type III Interferons by tumor cell-targeted STING agonist ADCs

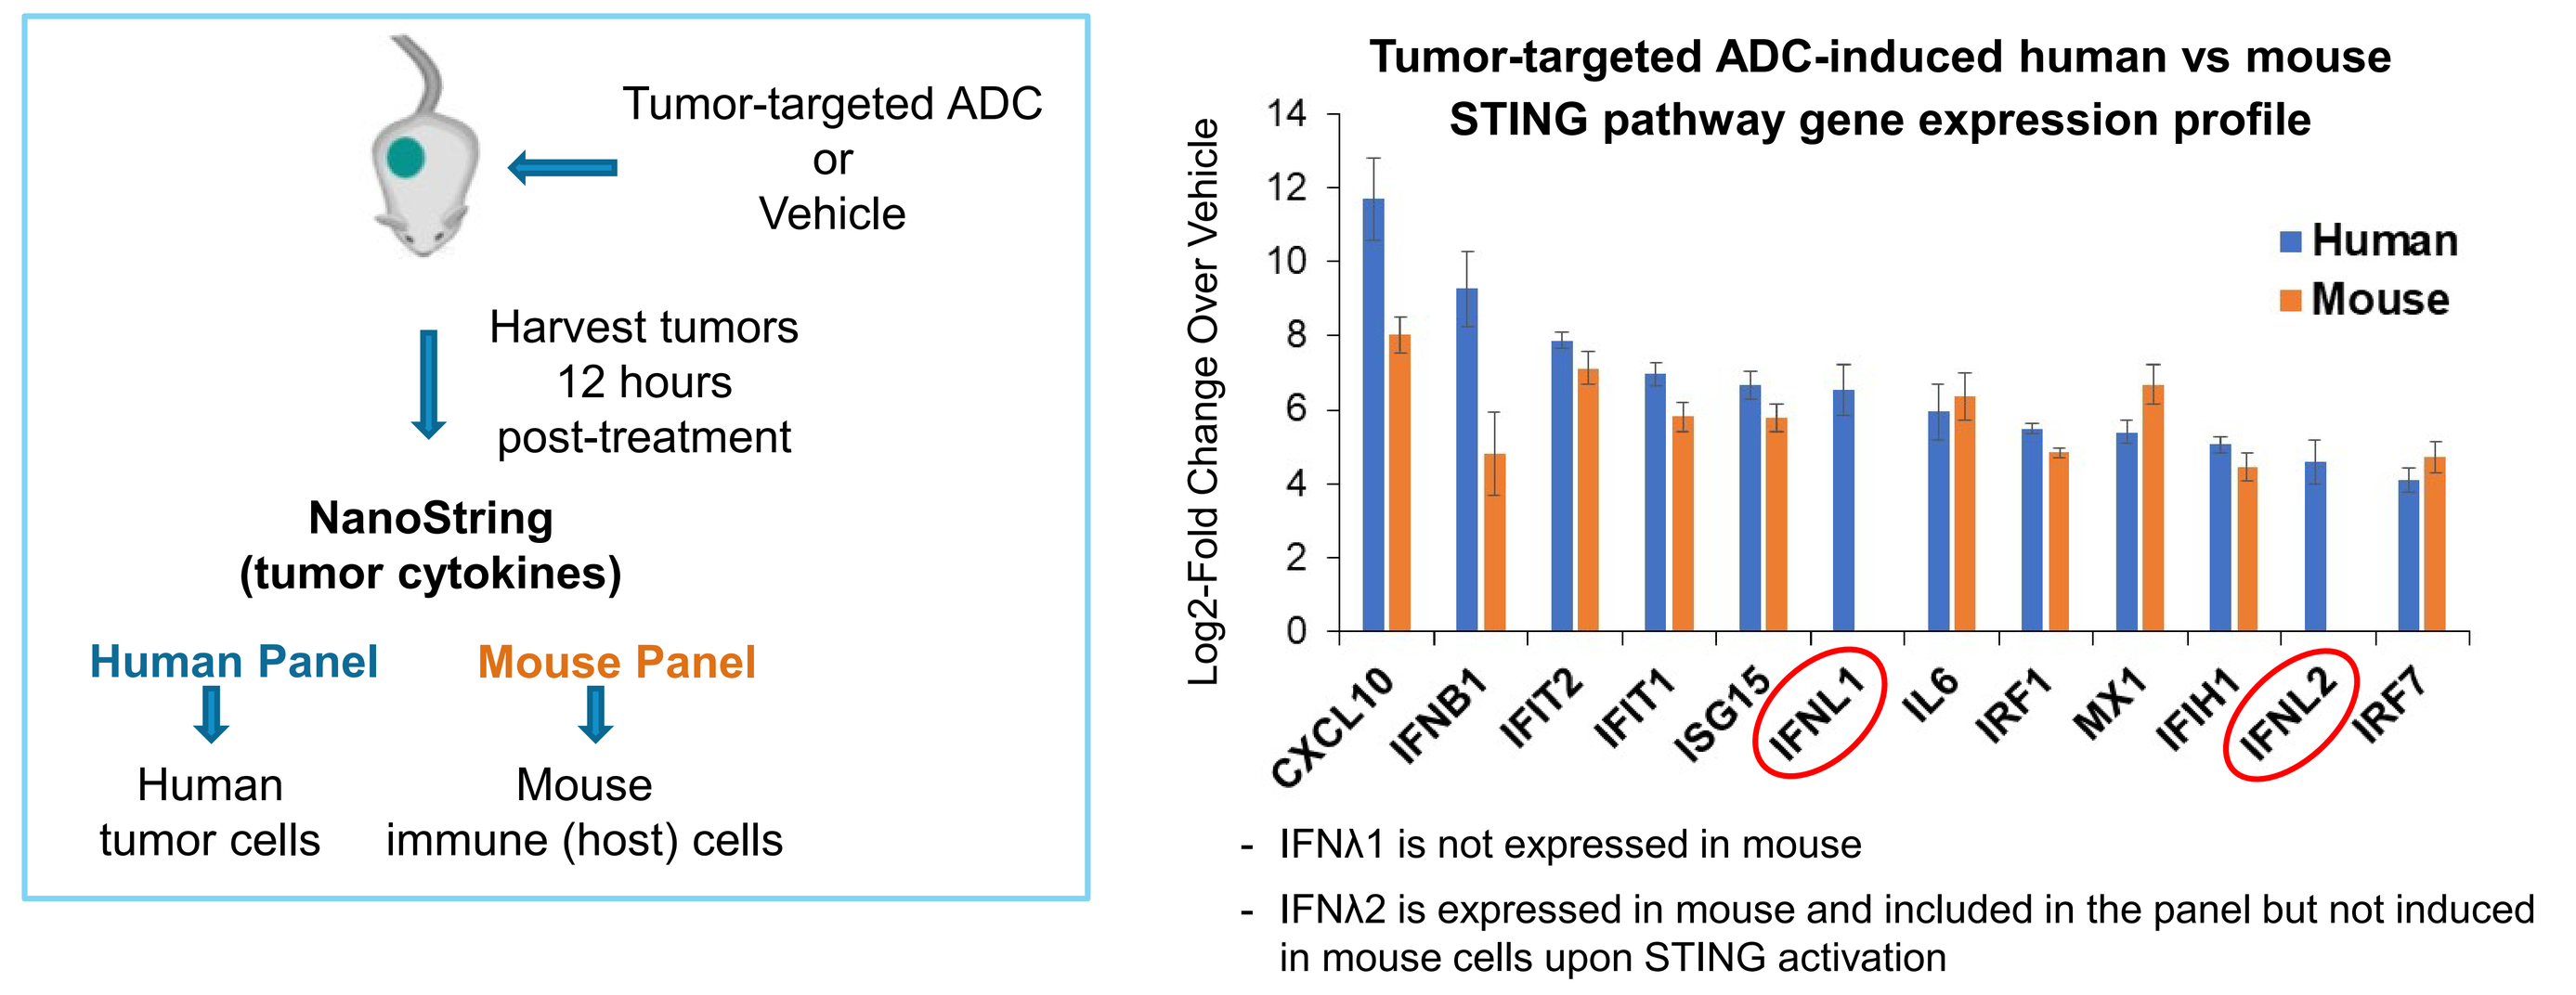
Tumor-targeted ADC
or
Vehicle
Harvest tumors
12 hours
post-treatment
NanoString
(tumor cytokines)
Human Panel
Mouse Panel
Human
tumor cells
Mouse
immune (host) cells
Tumor-targeted ADC-induced human vs mouse
STING pathway gene expression profile
Log2-Fold Change Over Vehicle
Human
Mouse
CXCL10
IFNB1
IFIT2
IFIT1
ISG15
IFNL1
IL6
IRF1
MX1
IFIH1
IFNL2
IRF7

- IFNλ1 is not expressed in mouse
- IFNλ2 is expressed in mouse and included in the panel but not induced in mouse cells upon STING activation

Figure 5. NanoString analysis of human tumor xenografts from SCID mice treated with tumor cell-targeted ADC using either human or mouse gene panels. Tumor-bearing CB.17 SCID mice were injected (IV) with a single dose of tumor cell-targeted STING agonist ADC or vehicle (as in Fig. 3B). Tumors were harvested 12 hours later and processed into FFPE. RNA was extracted and analyzed by NanoString using the human or mouse pan-cancer immune gene expression panels. Targeted ADC treatment led to marked induction of both mouse and human STING pathway genes, suggesting that the tumor-targeted STING agonist ADCs may induce tumor-intrinsic STING pathway activation in tumors *in vivo*. Interestingly, significant upregulation of type III Interferons (IFNλ1 and IFNλ2) were detected only by the human gene panel, suggesting that the type III Interferons are induced by the tumor cell-targeted ADCs specifically in the tumor cells in this xenograft model.

Tumor cell-targeted STING agonist ADC treatment induces Type III Interferons in cancer cell : PBMC co-cultures *in vitro*

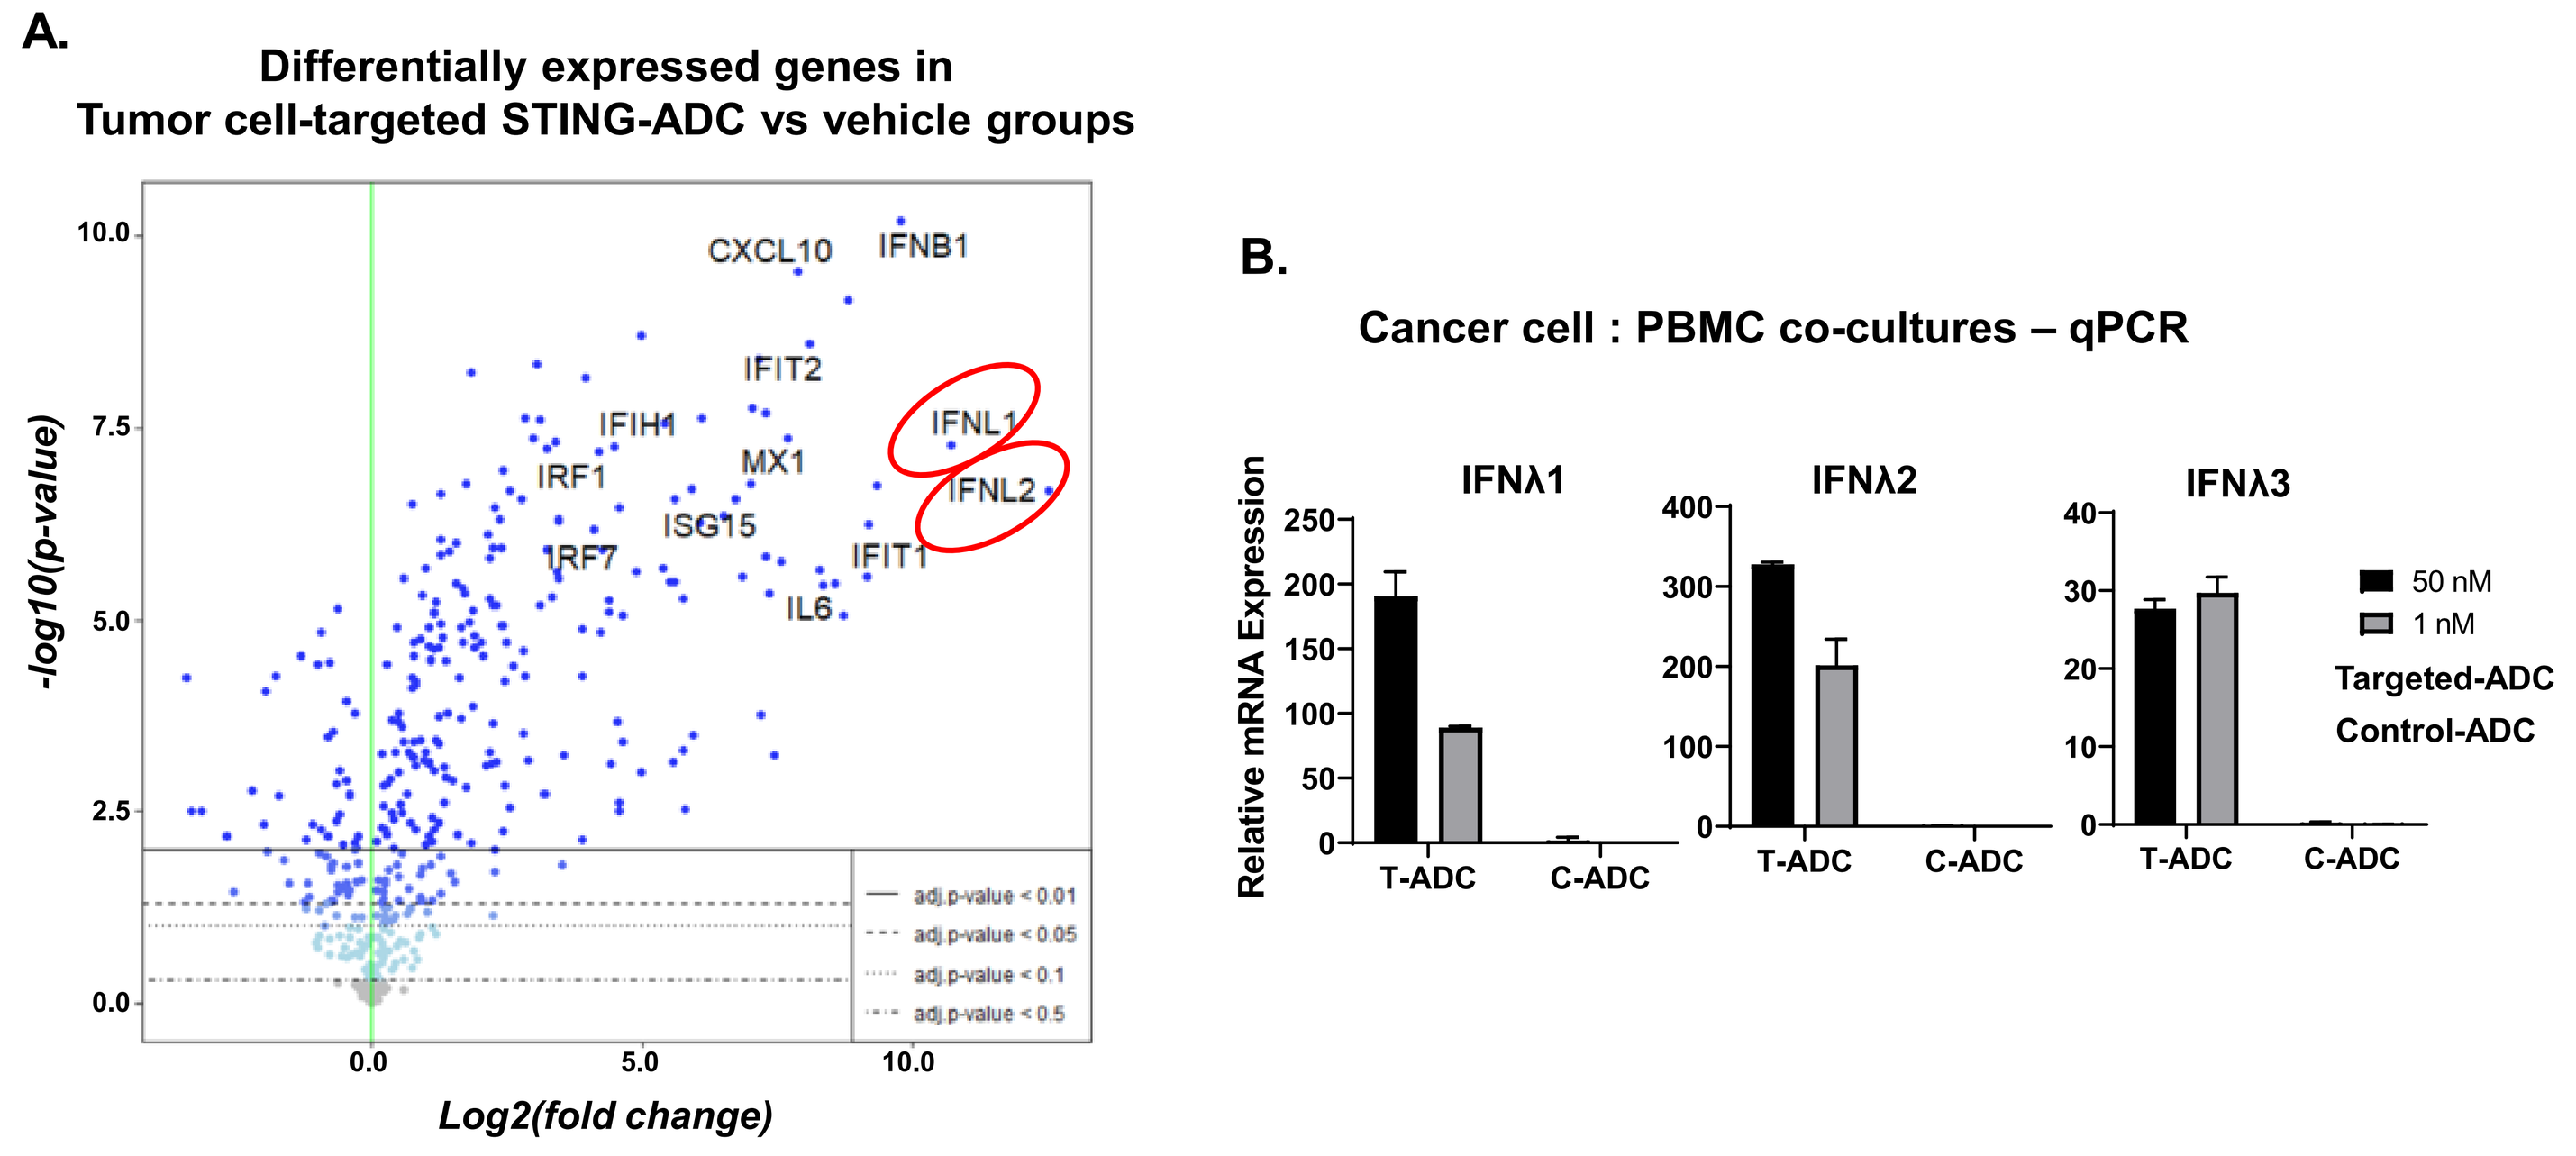
A.
Differentially expressed genes in
Tumor cell-targeted STING-ADC vs vehicle groups
-log10(p-value)
Log2(fold change)
CXCL10
IFNB1
IFIT2
IFIH1
IRF1
MX1
IFNL1
IFNL2
ISG15
IRF7
IFIT1
IL6
adj.p-value < 0.01
adj.p-value < 0.05
adj.p-value < 0.1
adj.p-value < 0.5
B.
Cancer cell : PBMC co-cultures – qPCR
IFNλ1
IFNλ2
IFNλ3
Relative mRNA Expression
T-ADC
C-ADC
50 nM
1 nM
Targeted-ADC
Control-ADC

C. ***Fc mutant targeted ADC induces similar levels of Type III IFNs compared to the wt ADC***

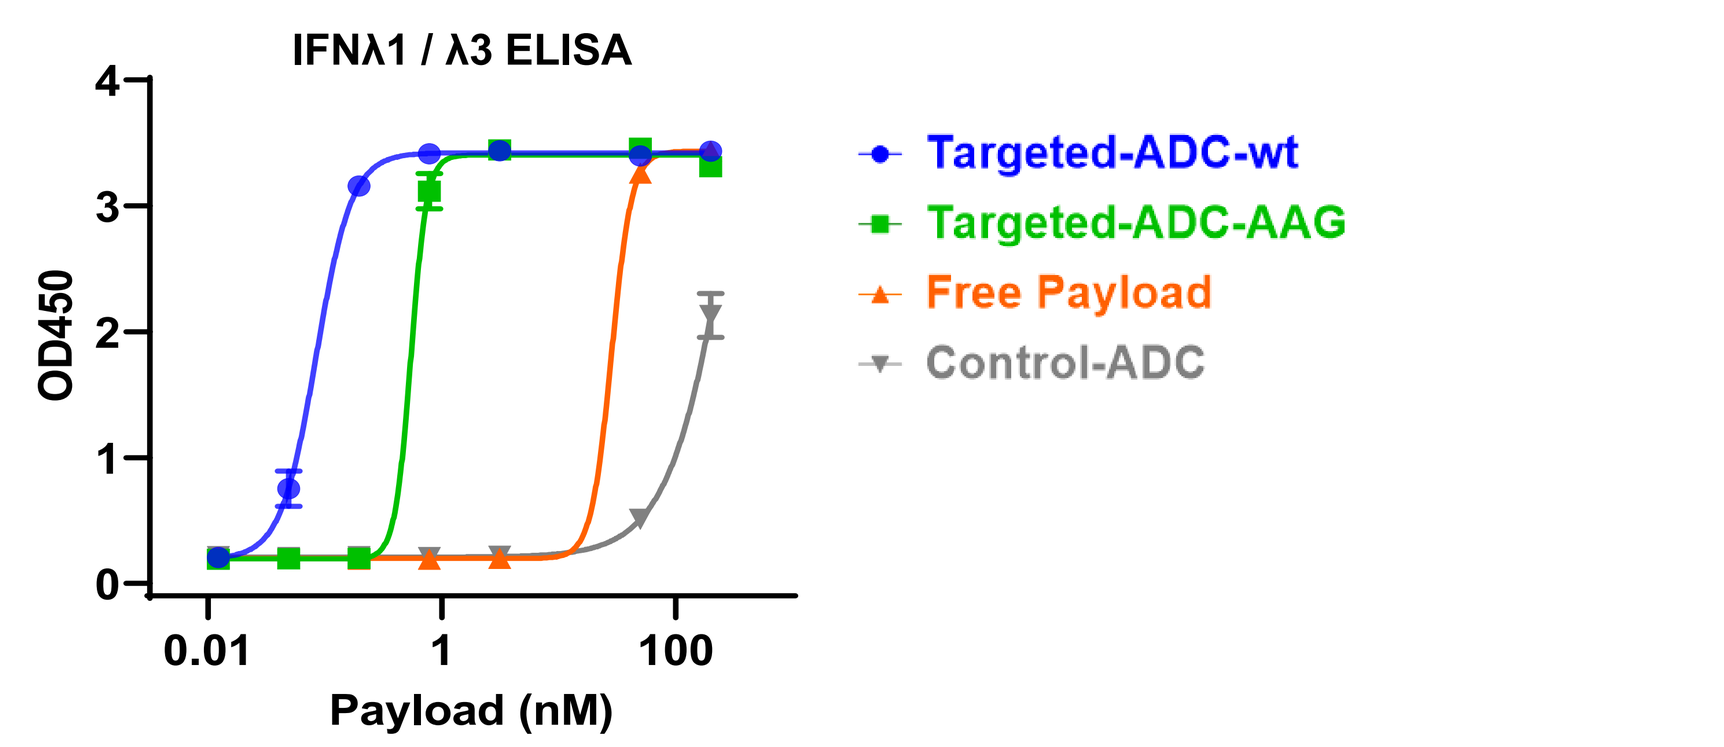
IFNλ1 / λ3 ELISA
OD450
Payload (nM)
Targeted-ADC-wt
Targeted-ADC-AAG
Free Payload
Control-ADC

Similar results were obtained with other tumor antigens/cancer cell lines

Figure 6. Tumor cell-targeted STING ADCs induce Type III Interferons in cancer cell : PBMC co-cultures in vitro. (A) cancer cell : PBMC co-cultures were treated with 50 nM (based on payload) tumor cell-targeting STING ADC for 5 hours. RNA was extracted and analyzed by Nanostring Human PanCancer Immune Profiling Panel. Data analysis was performed using the nSolver Advanced Software. Volcano plot shows the upregulation of STING pathway genes. The Type III Interferons (IFNλ1 and IFNλ2) are highlighted by the red circles. **(B)** Cancer cell PBMC co-cultures were treated with 50 nM or 1 nM (based on payload) Targeted or control ADCs for 5 hours and RNA was harvested followed by qPCR analysis of the Type III Interferons (IFNλ1, IFNλ2, IFNλ3). **(C)** Cancer cell : PBMC co-cultures were treated with the indicated test articles for 24 hours and supernatants were analyzed by IFNλ1 / λ3 ELISA assay. Fc mutant ADC induced similar levels of IFNλ1 / λ3 compared to the wt ADC suggesting that tumor-intrinsic STING pathway activation maybe contributing the majority of the Type III Interferons in the cancer cell : PBMC co-cultures.

Tumor cell-intrinsic STING is required for a robust Type III Interferon Induction in cancer cell : PBMC co-cultures

IFNλ1 / λ3 ELISA

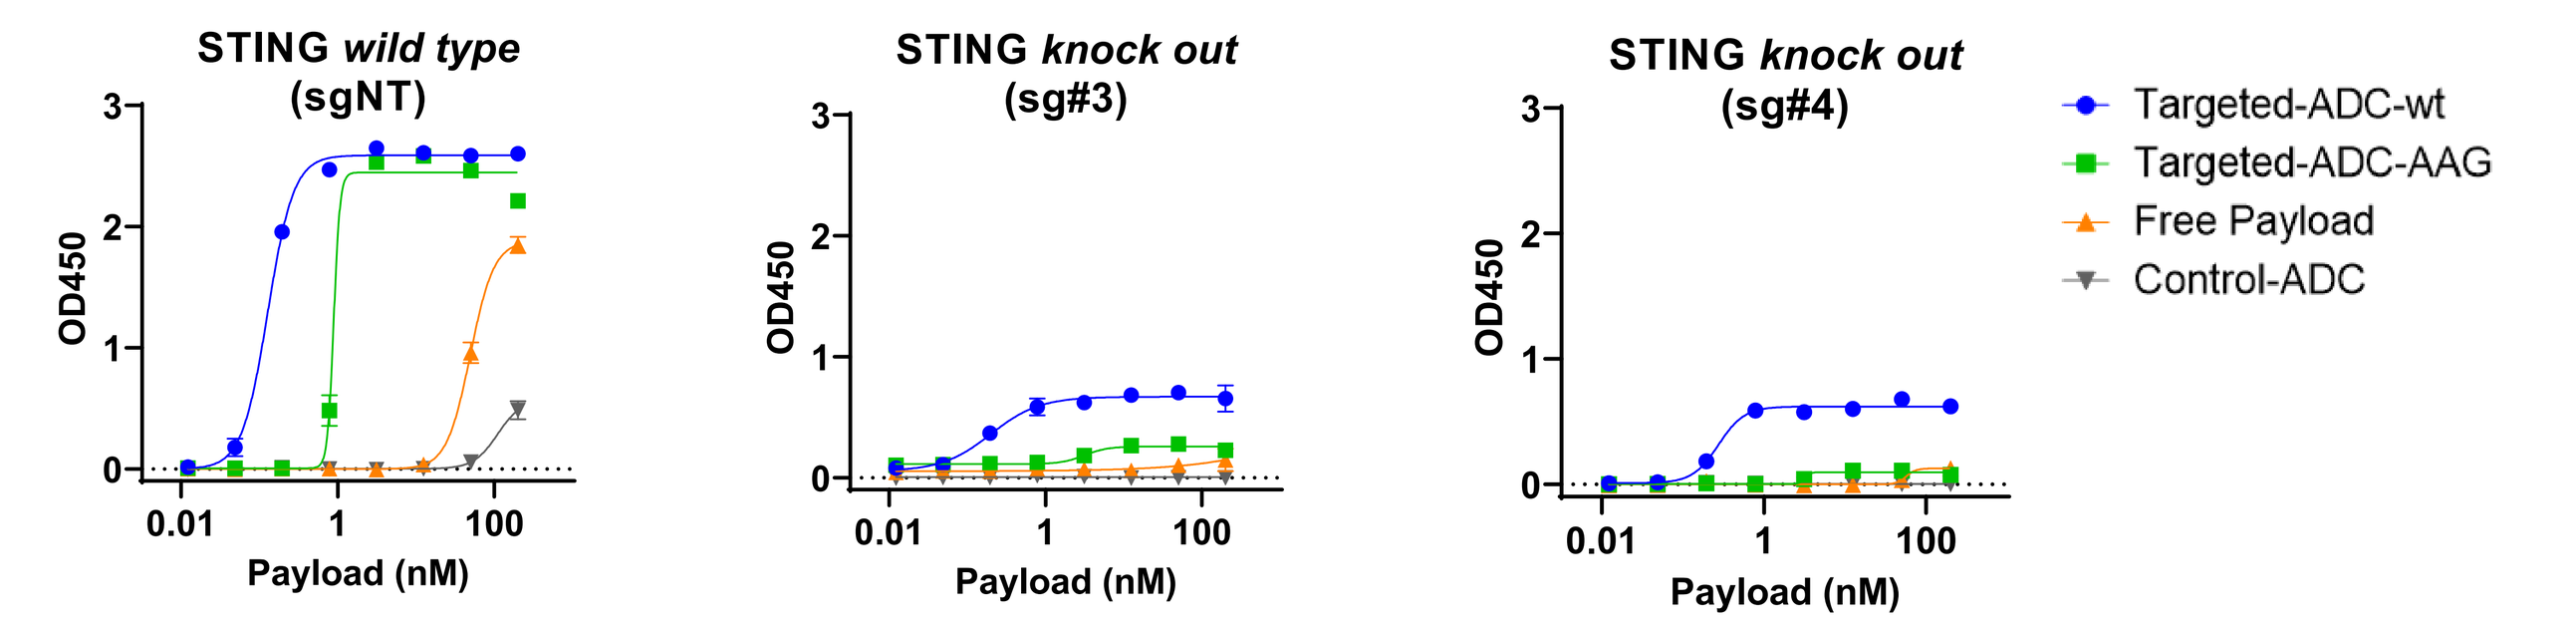
STING wild type
(sgNT)
STING knock out
(sg#3)
STING knock out
(sg#4)
OD450
Payload (nM)
Targeted-ADC-wt
Targeted-ADC-AAG
Free Payload
Control-ADC

Figure 7. Type III Interferon production is significantly reduced in STING knock out cancer cell : PBMC co-cultures. STING wt and ko cancer cells were co-cultured with PBMCs and treated with the indicated test articles for 24 hours. IFNλ1/ IFNλ3 cytokine levels in the supernatants were determined using a human IFNλ1/ IFNλ3 ELISA kit. IFNλ1/ IFNλ3 cytokine production was significantly reduced in STING knock out cancer cell : PBMC co-cultures, suggesting that the tumor cell-intrinsic STING activation is required for a robust Type III Interferon production.

IFNλ1 is important for cancer cell killing and production of key cytokines in PBMC co-cultures in response to STING-agonist ADCs

IFN λ1-neutralizing antibody treatment inhibits STING-ADC-induced cancer cell killing by PBMCs

Efficient neutralization of 0.1 nM STING ADC-induced Type III IFN in co-cultures using the IFNλ1 mAb

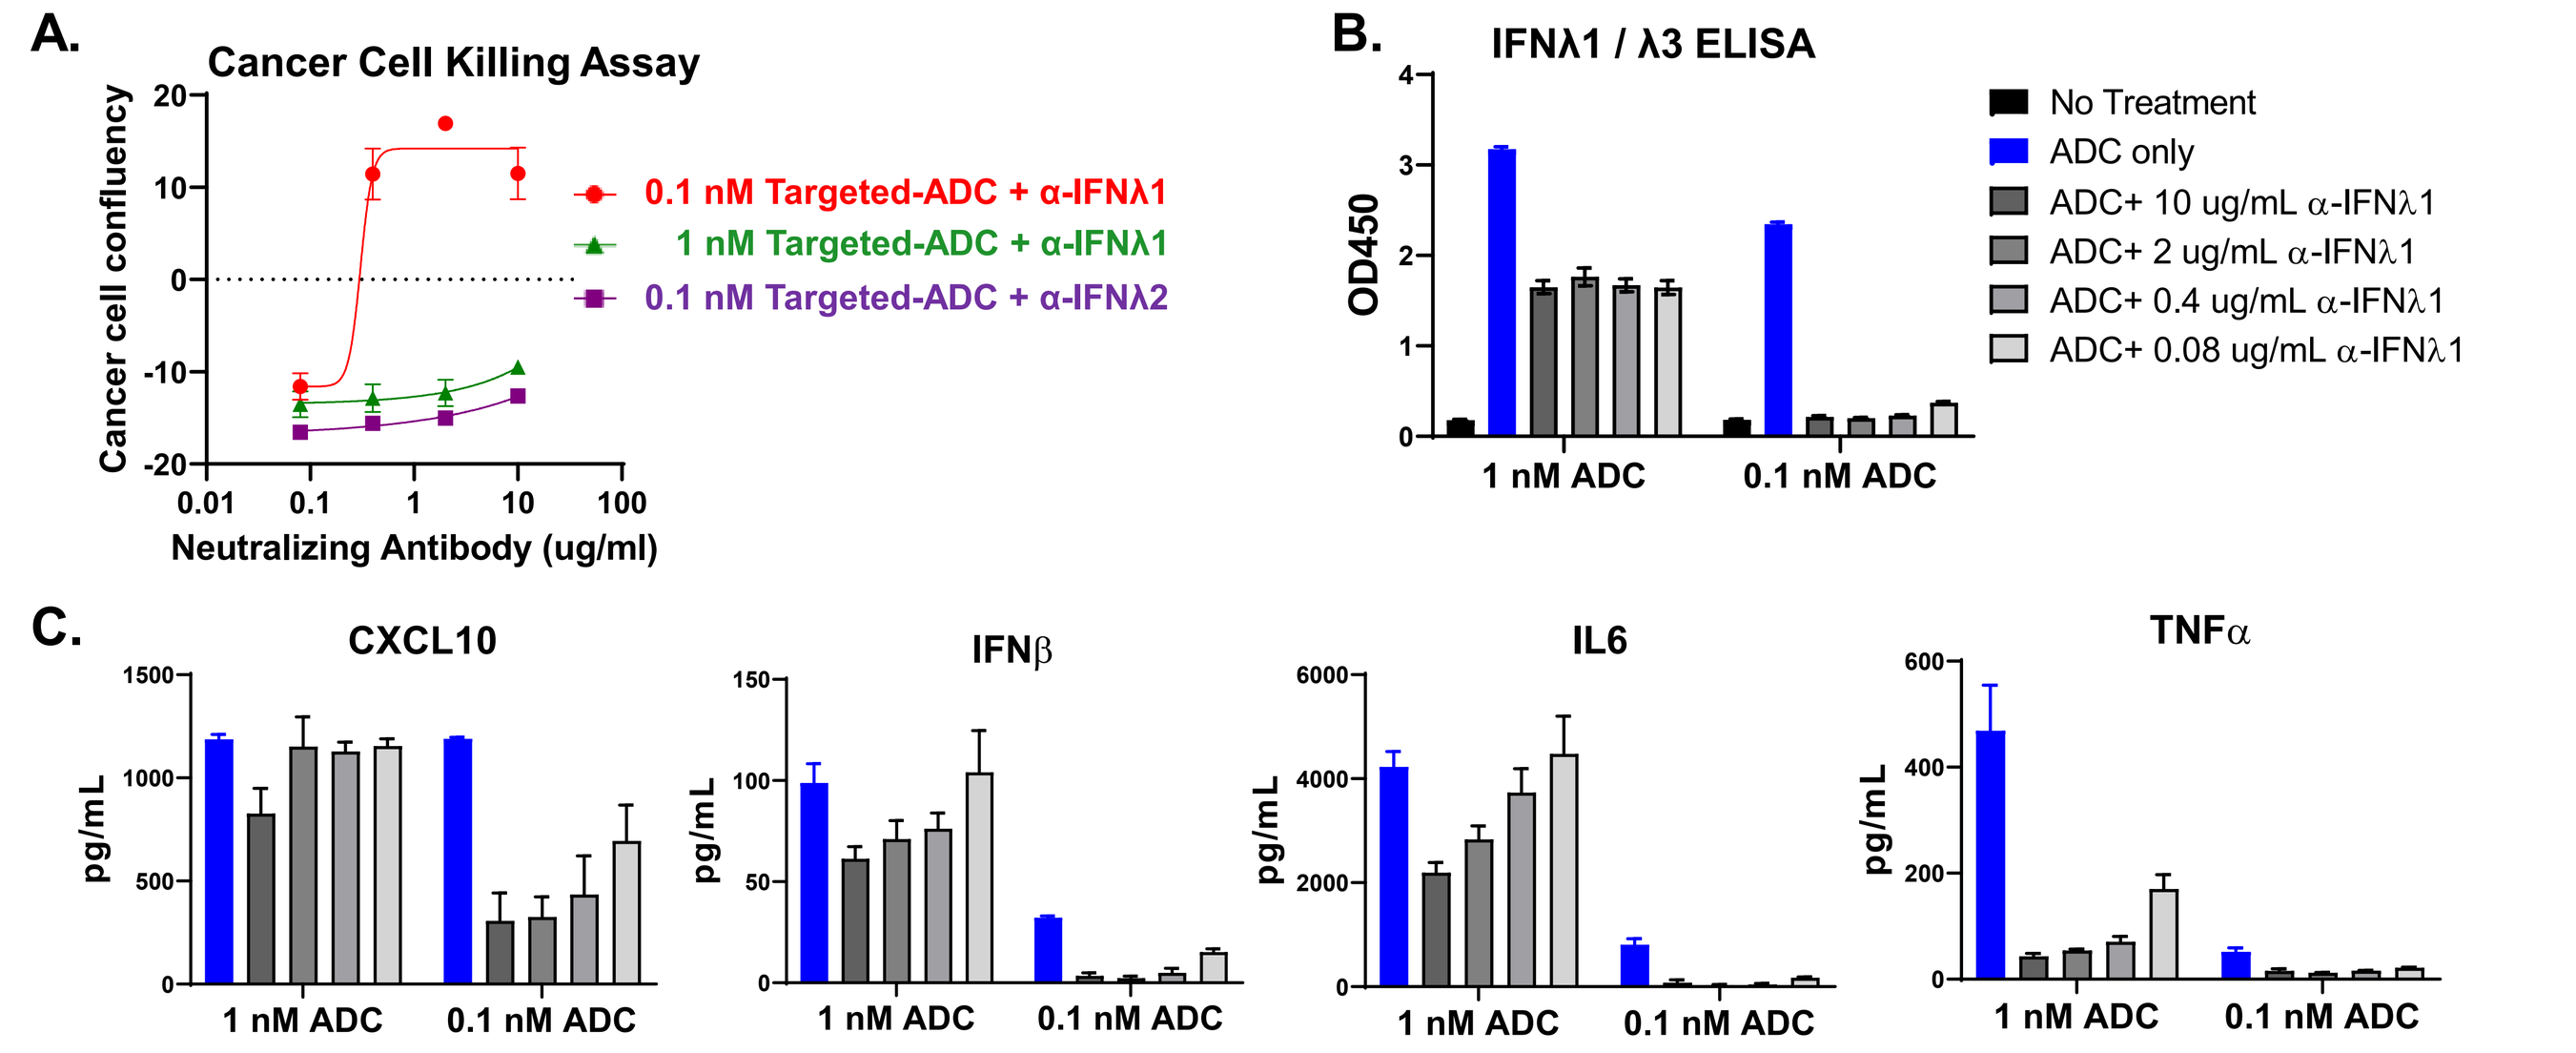
A.
Cancer Cell Killing Assay
Cancer cell confluency
Neutralizing Antibody (ug/ml)
0.1 nM Targeted-ADC + α-IFNλ1
1 nM Targeted-ADC + α-IFNλ1
0.1 nM Targeted-ADC + α-IFNλ2
B.
IFNλ1 / λ3 ELISA
OD450
No Treatment
ADC only
ADC+ 10 ug/mL α-IFNλ1
ADC+ 2 ug/mL α-IFNλ1
ADC+ 0.4 ug/mL α-IFNλ1
ADC+ 0.08 ug/mL α-IFNλ1
1 nM ADC
0.1 nM ADC
C.
CXCL10
IFNβ
IL6
TNFα
pg/mL

Figure 8. IFNλ1-neutralizing antibodies inhibit cancer cell killing and production of key cytokines by STING-ADC treatment in cancer cell : PBMC co-cultures. (A) Cancer cell : PBMC co-cultures were treated with either 1 nM or 0.1 nM (based on payload) of cancer cell-targeted STING-ADC and 0, 0.08, 0.4, 2, and 10 μg/mL IFNλ1 or IFNλ2-neutralizing antibodies. Cancer cell confluency was tracked over time in an IncuCyte instrument. Supernatants from the sister plates after 2 hours of incubation were analyzed by **(B)** IFNλ1 / λ3 ELISA assay and **(C)** Luminex assay. IFNλ1-neutralizing antibodies inhibited cancer cell killing by 0.1 nM ADC treatment but not 1 nM ADC treatment. IFNλ2-neutralizing antibodies had no impact even on the 0.1 nM ADC treatment (note that we did not detect significant levels of IFNλ2 cytokine in the supernatants in response to STING activation in co-cultures). IFNλ1-neutralizing antibodies efficiently reduced IFNλ1 and other key cytokines in co-cultures treated with only 0.1 nM STING-ADC.

CONCLUSIONS

- ❖ The Immunosynthen ADC platform enables tumor-targeted delivery of a STING agonist with improved efficacy and tolerability over a free IV STING agonist.
- ❖ Anti-tumor activity of STING agonist ADCs involves activation of STING pathway in both immune cells and tumor cells.
- ❖ In this study we have demonstrated:
 - Tumor cell-intrinsic STING pathway can be activated in the presence of cues from immune cells.
 - Tumor cell-targeted STING agonist ADCs induce Type III Interferons
 - Tumor cell-intrinsic STING pathway activation is required for a robust Type III Interferon induction
 - Type III Interferons are important for STING-ADC activity especially at low STING agonist exposure conditions

The ability of Immunosynthen-based ADCs to activate STING and Type III Interferons in tumor cells in addition to the immune cells may represent a significant therapeutic advantage of targeting the STING pathway.

References:

1. Malli Cetinbas et al. SITC 2020 Abstract #620
2. Ramanjulu et al. ***Nature*** 2018, 564: 438-443