STING-Agonist ADCs Targeting Tumor-Associated Antigens Coordinate Immune-Mediated Killing of Antigen-Negative Cancer Cells

Phonphimon Wongthida, Kalli C. Catcott, Kelly Lancaster, Keith W. Bentley, Anouk Dirksen, Bingfan Du, Timothy Eitas, Eugene W. Kelleher, Naniye Malli, Rebecca Mosher, Marina Protopopova, Pamela Shaw, Cheri Stevenson, Joshua D. Thomas, Alex Uttard, Jeremy R. Duvall, Dorin Toader, Marc Damelin, Raghida A. Bukhalid, Timothy B. Lowinger

Mersana Therapeutics, Inc. Cambridge, MA

Abstract ID: 785

ABSTRACT

The tumor microenvironment is a complex, multicellular system, composed not only of malignant cancer cells but also of a diversity of stromal cells including vascular cells, immune cells, and fibroblasts that support tumorigenesis. Antigens expressed on these cells tend to be widely expressed across a range of malignancies, presenting unique opportunities for development of anti-cancer therapies.

We have previously demonstrated that STING-agonist antibody-drug conjugates (Immunosynthen ADCs) targeting tumor cell antigens induce target-dependent anti-tumor immune responses *in vitro* and *in vivo,* with excellent tolerability in NHP. To that effect, we hypothesized that Immunosynthen ADCs targeting tumor-associated antigens (TAA) would coordinate immune-mediated killing of cancer cells not expressing the tumor-associated antigens (antigen-negative cancer cells) in the tumor microenvironment and induce anti-tumor activity.

Herein, we demonstrate that targeting tumor-associated antigens with STING-agonist ADCs activate the STING pathway in immune cells via Fcγ receptor-mediated uptake. In addition, due to the intrinsic ability of certain tumor-associated cells to activate the STING pathway, STING-agonist ADCs targeting those cells can induce STING signaling in both the targeted cells and the immune cells, which constitutes a therapeutic advantage of ADCs that activate the STING pathway over other approaches of targeted activation of the innate immune system. In triple co-cultures of antigen-positive tumor-associated cells, antigen-negative cancer cells, and immune cells, the STING-agonist ADC specifically induced potent cell killing of the antigen-negative cancer cells with minimal impact on the immune and tumor-associated cells, thus representing a non-traditional, yet highly effective mechanism of ADC targeting.

In vivo efficacy studies showed that STING-agonist ADCs developed for two tumor-associated antigens induced complete, sustained tumor regressions in syngeneic tumor models and exhibited immunological memory after rechallenge. CD8+ T cells contributed to the anti-tumor activity of the STING-agonist ADCs.

In summary, Immunosynthen STING-agonist ADCs targeting tumor-associated antigens represent a novel approach for ADC-mediated cancer immunotherapy for targeted activation of the innate immune system and enable the multifaceted activation of the STING pathway in a tumor-targeted manner beyond tumor antigens.

BACKGROUND

Immunosynthen STING-Agonist ADCs That Target Tumor-Associated Antigens

- ❖ Antigens broadly expressed across many solid tumor types
- ❖ Benefit from proximity to immune cells in the stroma
- ❖ Potential for dual mechanism of STING activation delivering a 1-2 Punch simultaneously to TAA-expressing cells and to tumor-resident immune cells [1,2]
- ❖ Activation of STING in tumor-associated cells could happen regardless of immune cell infiltration status

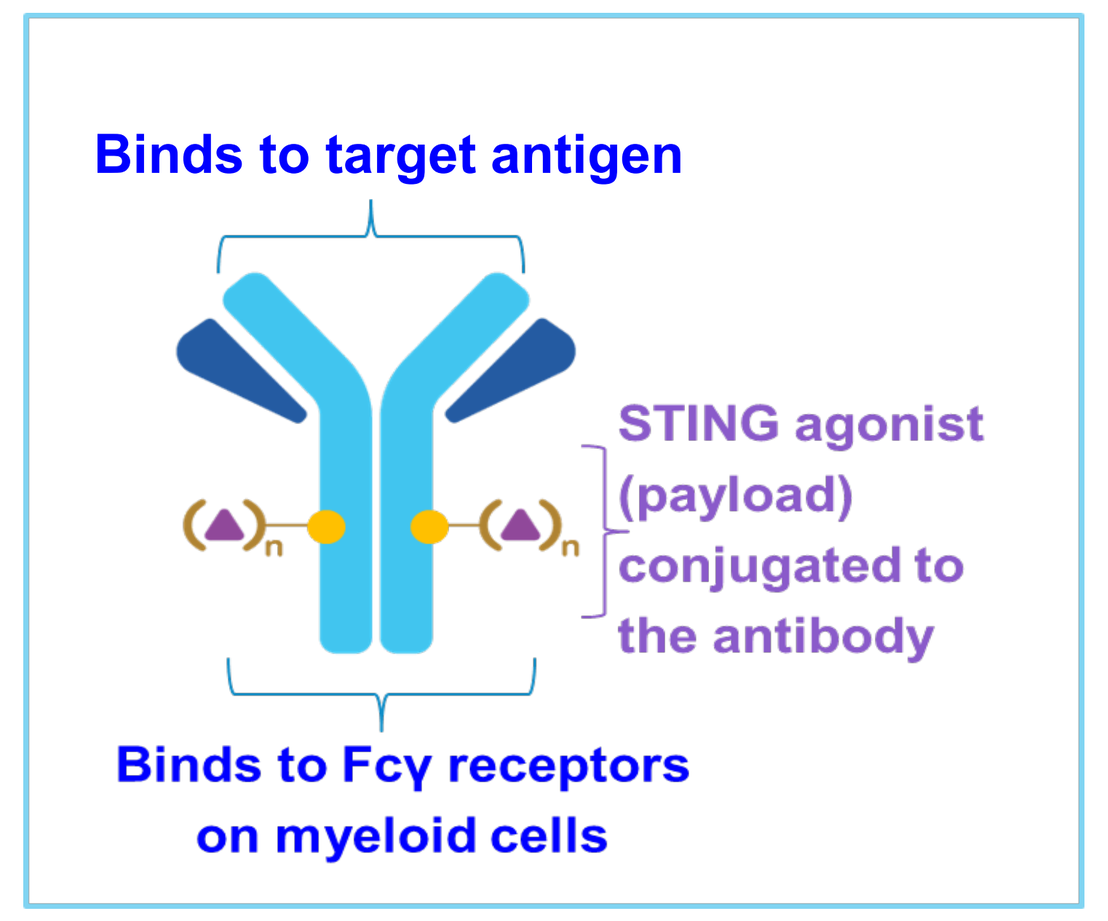
Binds to target antigen
STING agonist (payload) conjugated to the antibody
Binds to Fcγ receptors on myeloid cells

An ADC Is an Ideal Approach for Targeted Innate Immune Activation with STING

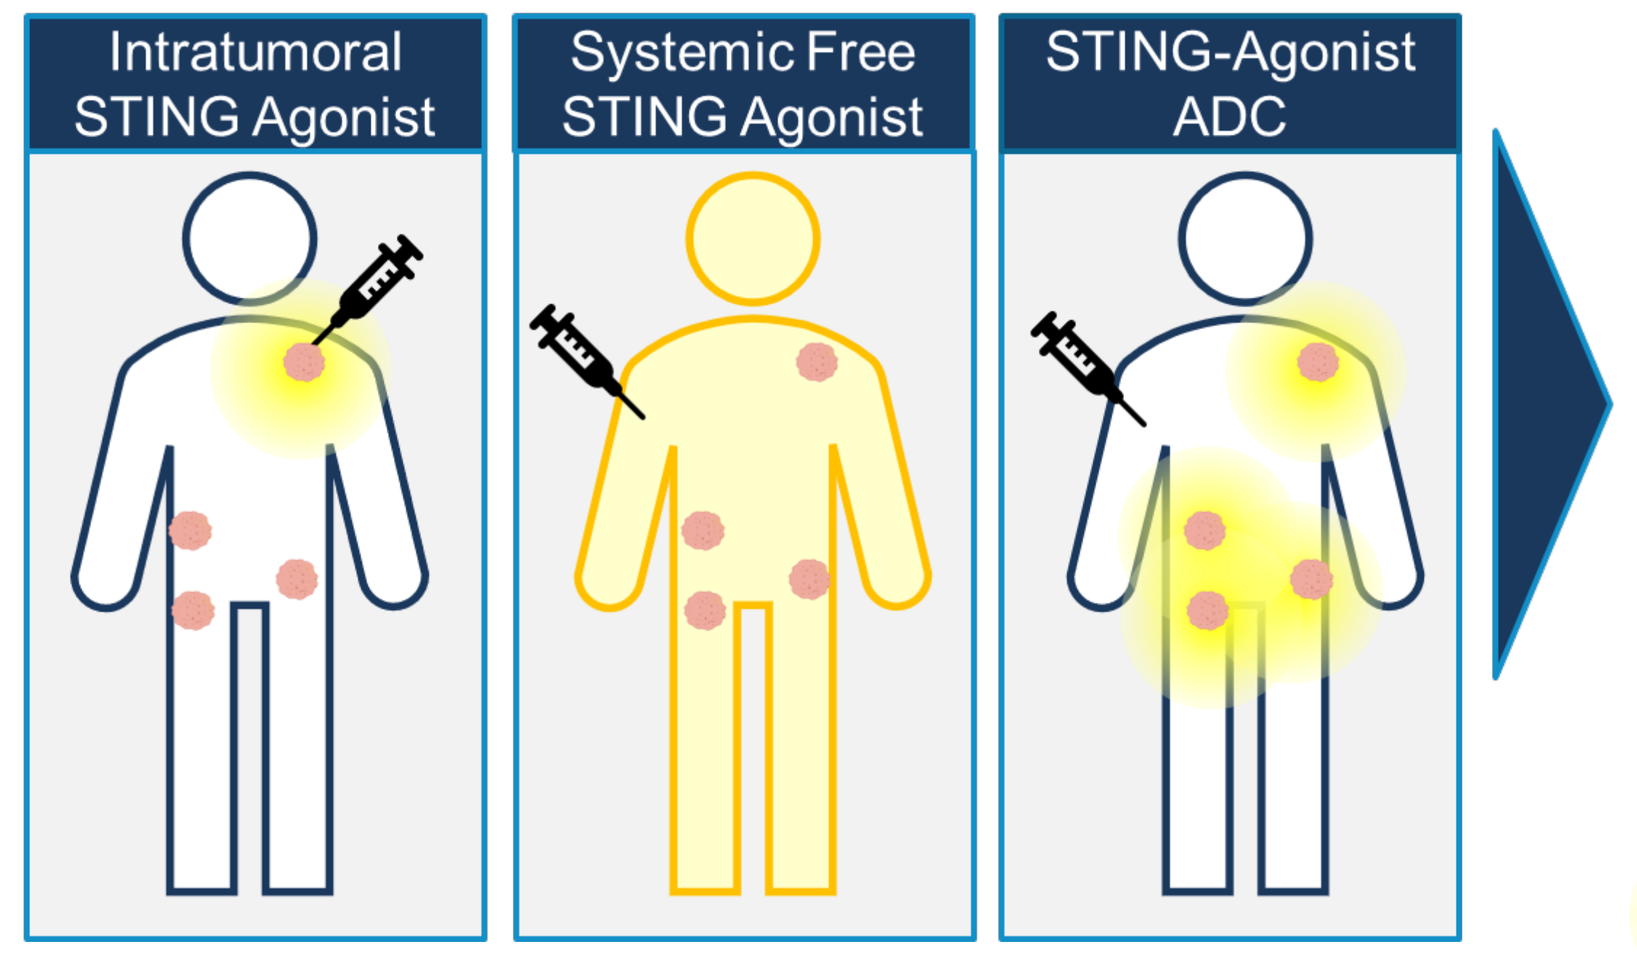
Intratumoral STING Agonist
Systemic Free STING Agonist
STING-Agonist ADC

- ❖ Systemic administration with <u>targeted delivery</u> to all tumor lesions while avoiding healthy tissues
- ❖ <u>Improved anti-tumor activity</u> compared to free agonist
- ❖ <u>Improved tolerability</u> compared to free agonist

Systemic immune activation

Tumor, no immune activation

Tumor with STING-Mediated Innate Immune Activation

Proposed Mechanisms of Action of STING-Agonist ADCs Targeting Tumor-Associated Antigens

Coordinated immune-mediated killing of antigen-negative cancer cells by STING-agonist ADCs

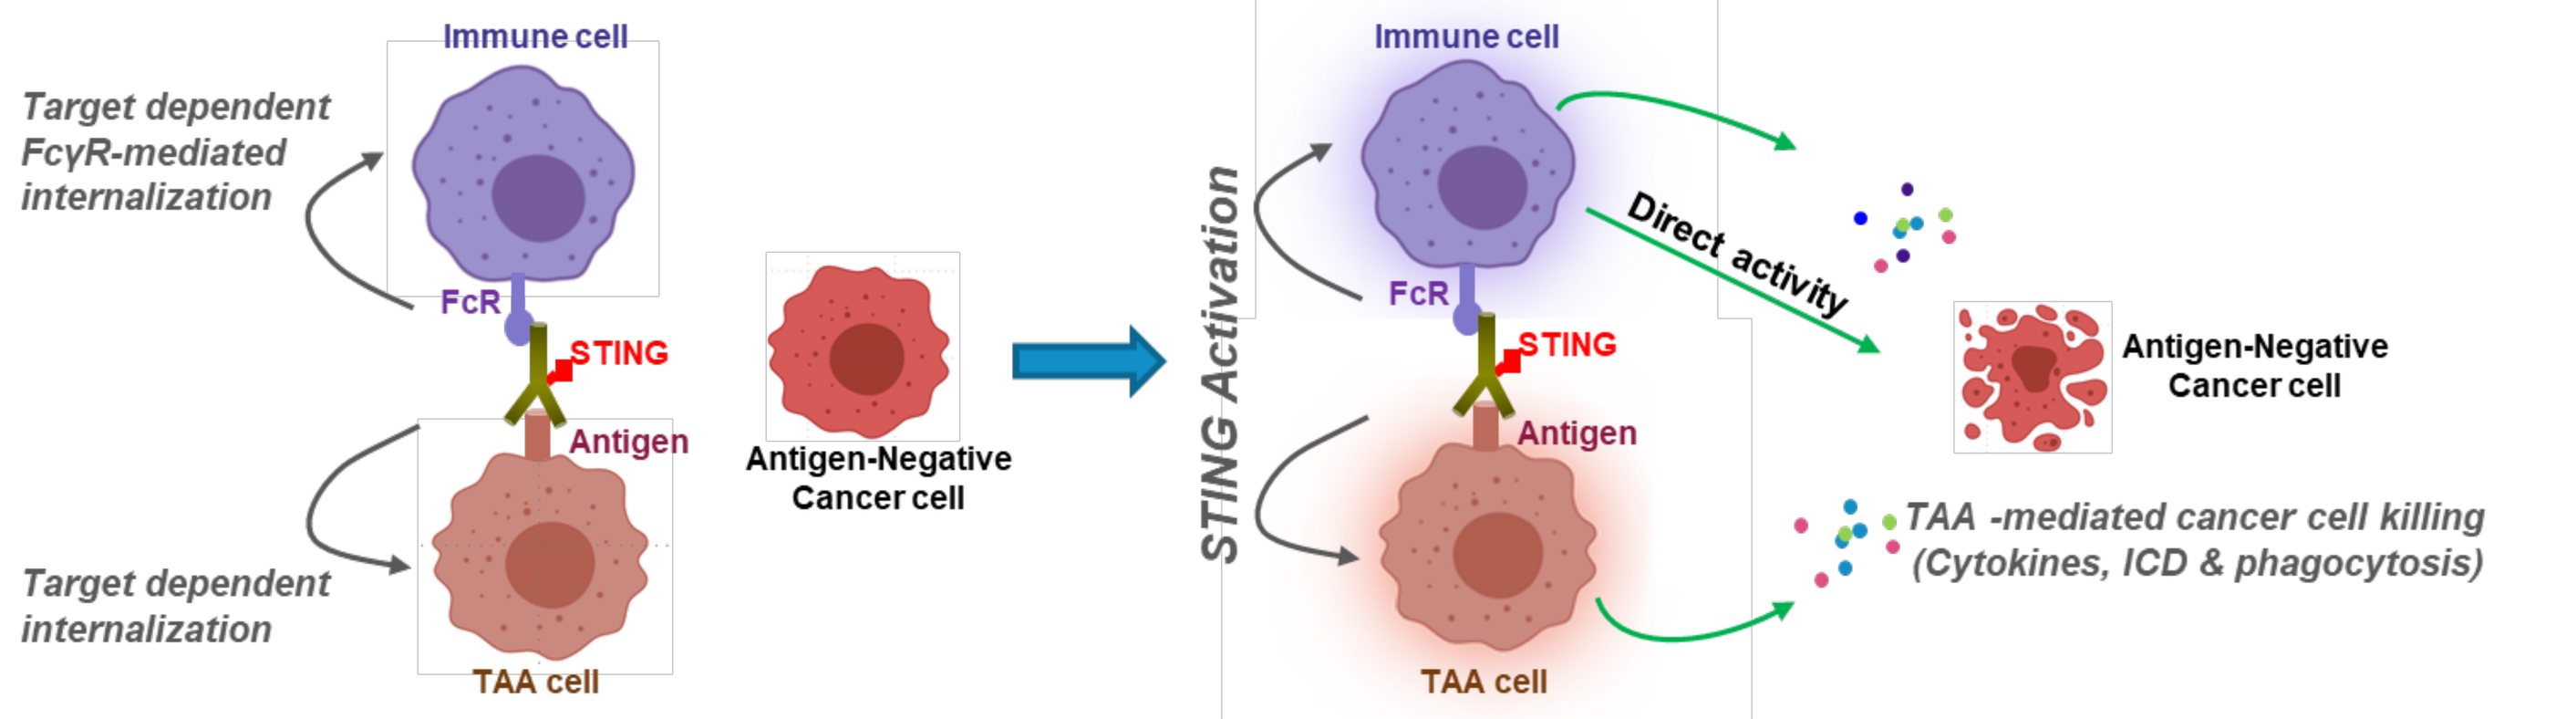
Target dependent FcγR-mediated internalization
Immune cell
FcR
STING
Antigen
Target dependent internalization
TAA cell
Antigen-Negative Cancer cell
STING Activation
Direct activity
TAA -mediated cancer cell killing (Cytokines, ICD & phagocytosis)

Figure 1. Depiction of proposed MOA of TAA targeted ADCs. Dual STING activation in target cells through direct binding of ADC and activation of both TAA cells and immune cells through antigen mediated internalization on the former and FcγR engagement on the latter. Activation of STING pathway results in cytokines/chemokines production and immune cell activation which mediate killing of antigen-negative cancer cells.

RESULTS

STING Activation Demonstrated in Human Tumor-Associated Cells and Mouse Bone Marrow Derived Immune Cells (BMDCs)

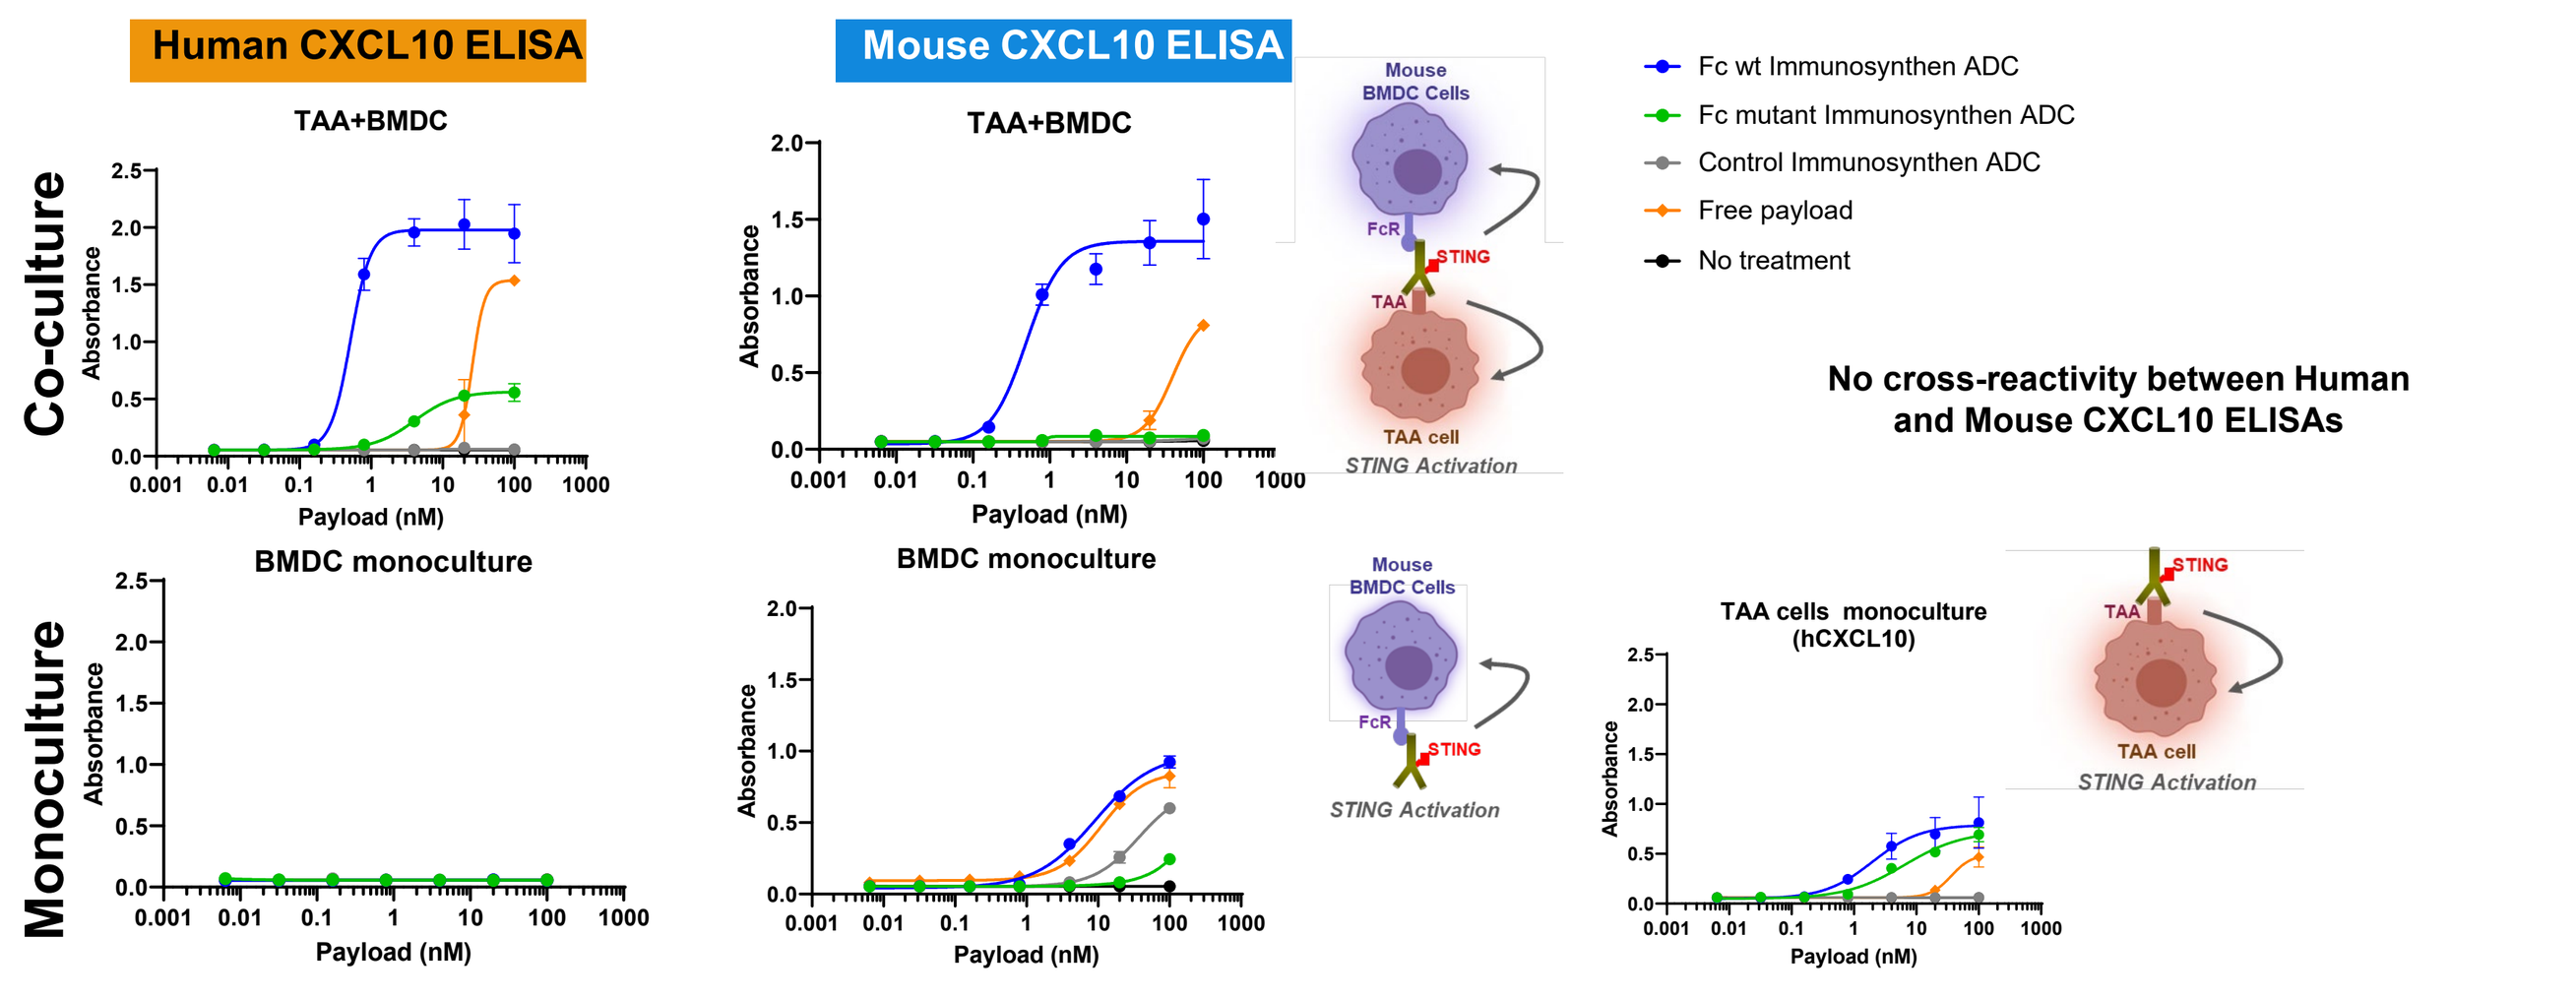
Human CXCL10 ELISA
Mouse CXCL10 ELISA
Co-culture
Monoculture
TAA+BMDC
BMDC monoculture
TAA cells monoculture (hCXCL10)
Payload (nM)
Absorbance
Fc wt Immunosynthen ADC
Fc mutant Immunosynthen ADC
Control Immunosynthen ADC
Free payload
No treatment
No cross-reactivity between Human and Mouse CXCL10 ELISAs

Figure 2. Activation of STING pathway in human target expressing cells and mouse BMDC. TAA cells and BMDCs were co-cultured in the presence of test articles for 24 hrs, and supernatants were harvested for human or mouse CXCL10 ELISA. Wild-type Fc Immunosynthen ADC induced both human and mouse CXCL10 demonstrating STING activation in TAA cells and immune cells, with a clear enhancement in the activation of TAA cells mediated by the immune cell co-culture conditions. As expected, activation by Fc mutant ADC is only observed in the co-culture and monoculture of TAA cells.

TAA Targeted Immunosynthen ADC Induces Coordinated Immune-Mediated Killing of Antigen-Negative Cancer Cells, with No Impact on Immune or TAA cells

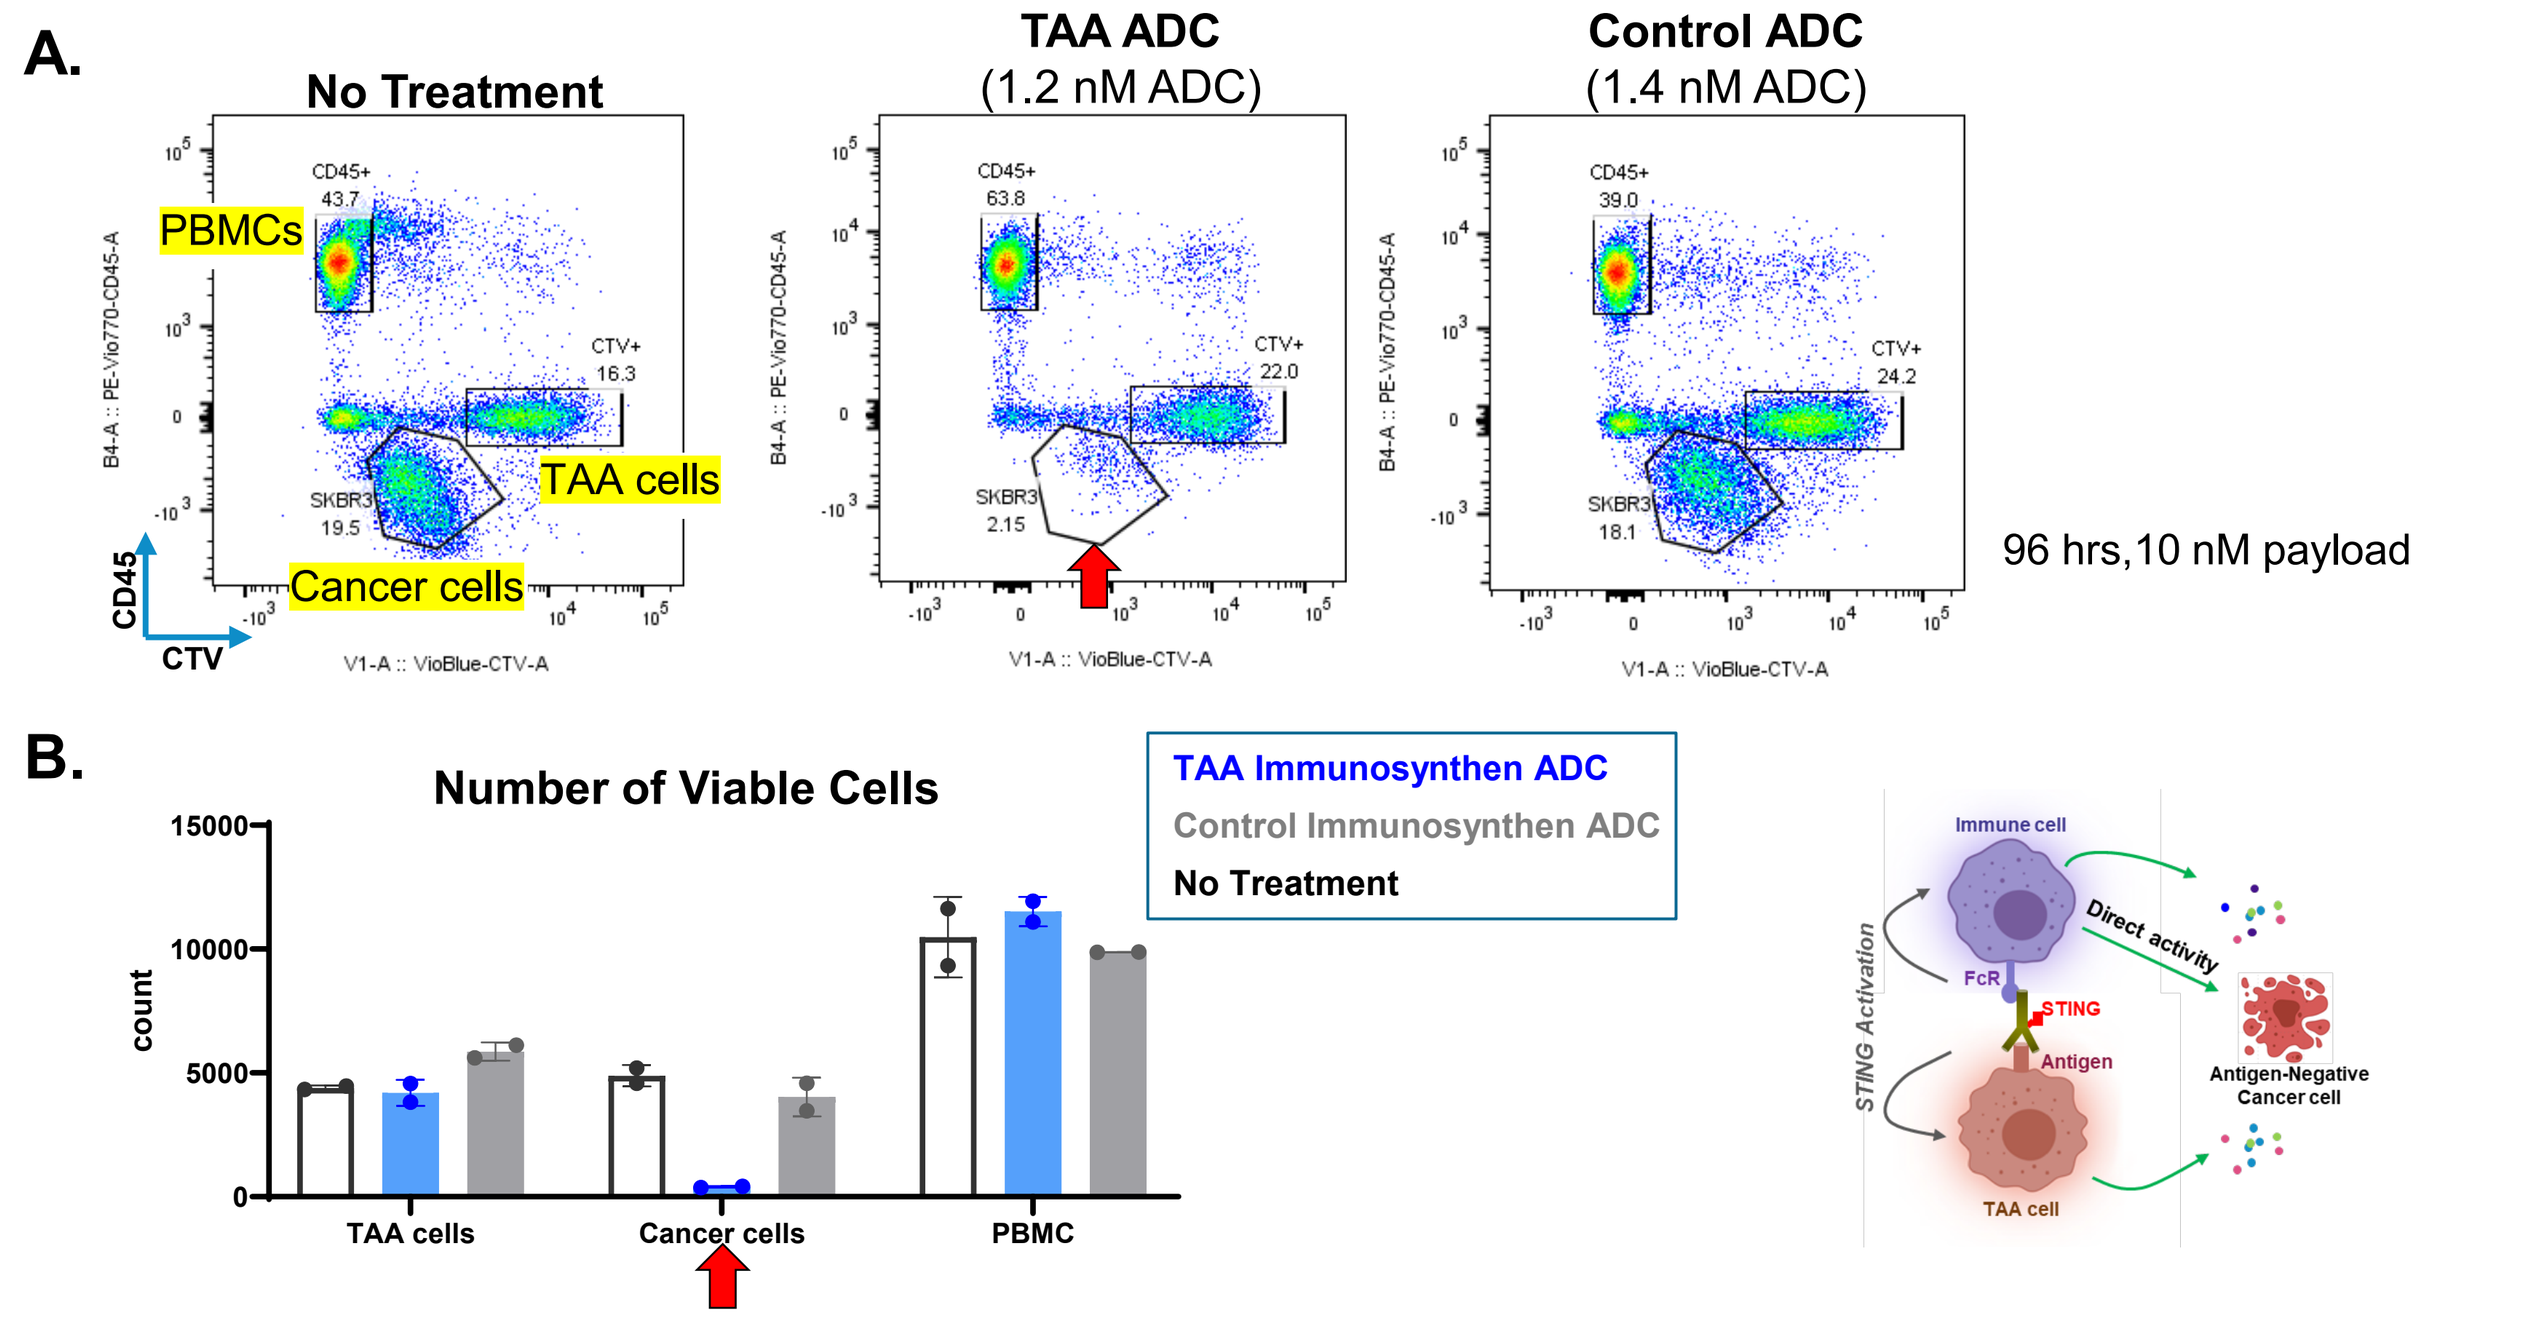
A.
No Treatment
TAA ADC (1.2 nM ADC)
Control ADC (1.4 nM ADC)
PBMCs
TAA cells
Cancer cells
CD45
CTV
96 hrs,10 nM payload
B.
Number of Viable Cells
TAA Immunosynthen ADC
Control Immunosynthen ADC
No Treatment
count
TAA cells
Cancer cells
PBMC

Figure 3. TAA Immunosynthen ADC coordinated immune-mediated antigen-negative cancer cell killing. Triple co-cultures of TAA cells, antigen-negative cancer cells and PBMCs in the presence of TAA ADC or control (non-binding) ADC. **(A)** Representative flow plots showing CD45 and CTV staining. **(B)** Viable cell count of each cell population after treatment.

In Vitro Cytokine Profiles of TAA / PBMC Co-Cultures Demonstrating Activity Relative to Controls

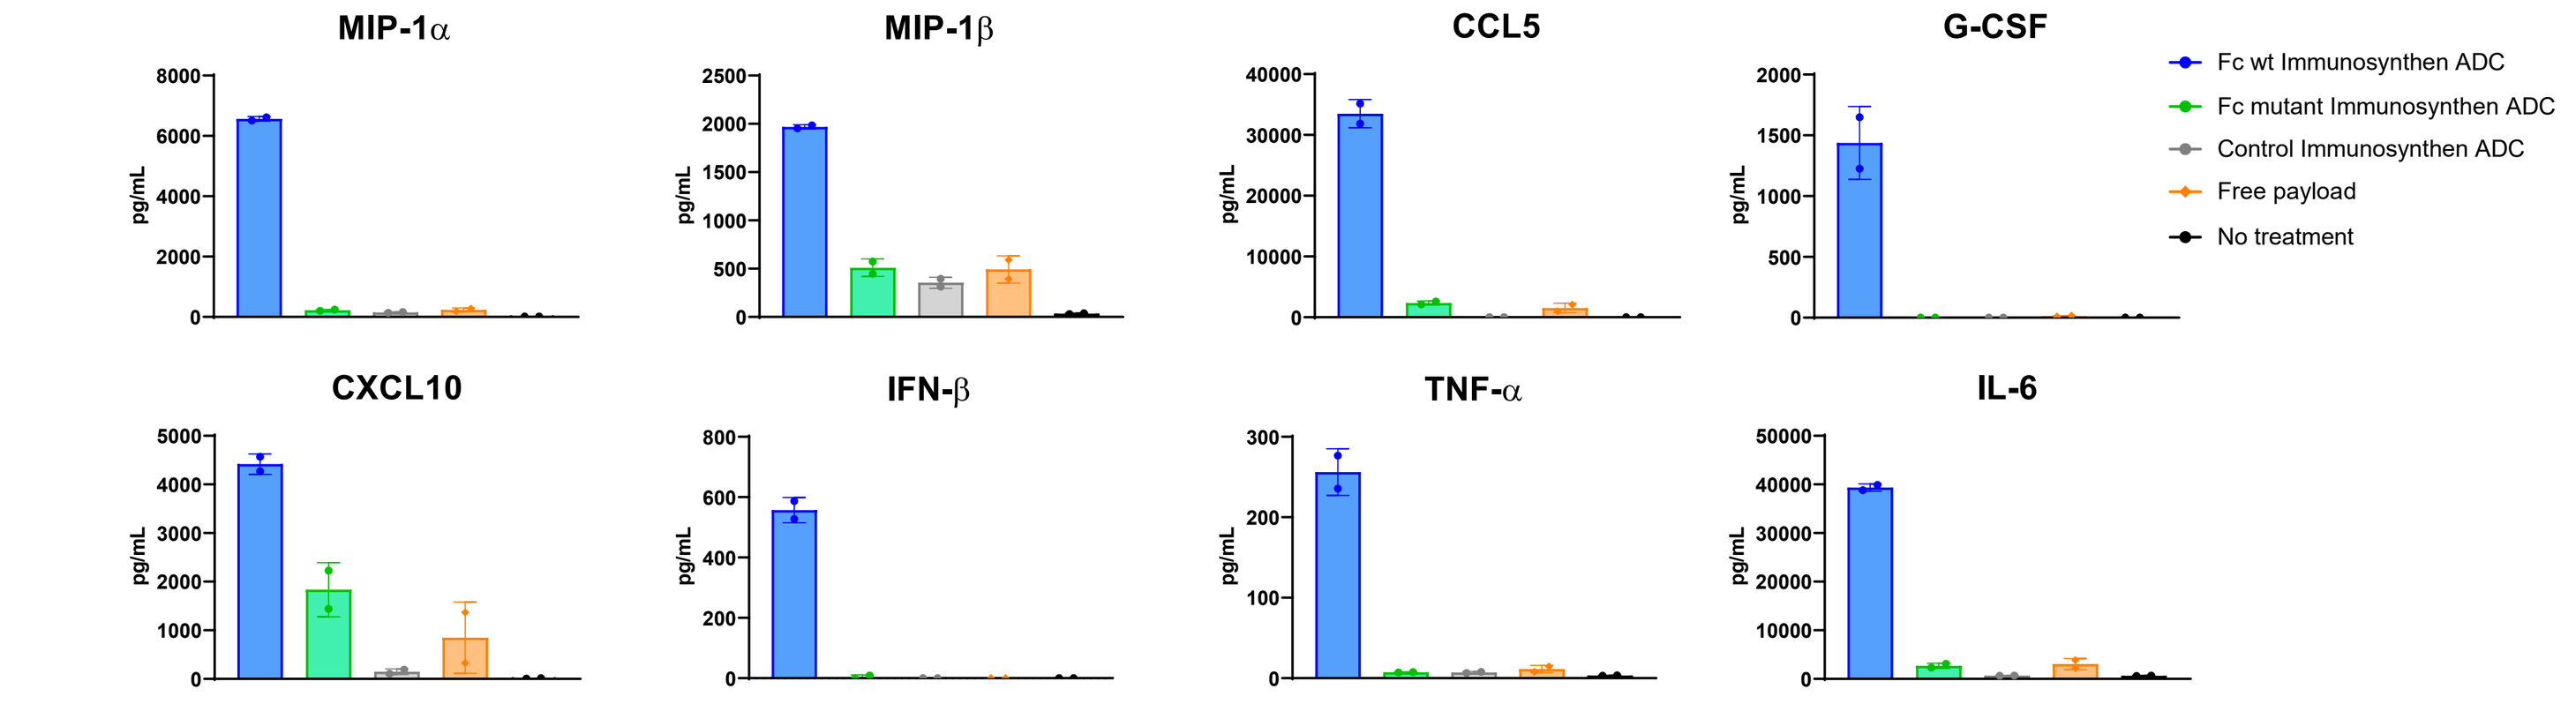
MIP-1α
MIP-1β
CCL5
G-CSF
CXCL10
IFN-β
TNF-α
IL-6
pg/mL
Fc wt Immunosynthen ADC
Fc mutant Immunosynthen ADC
Control Immunosynthen ADC
Free payload
No treatment

Figure 4. Cytokines/chemokines production after TAA Immunosynthen ADC treatment. TAA cells and PBMCs were co-cultured in the presence of test articles for 24 hrs and supernatants were harvested for profiling by Luminex multiplex assay.

Supernatant and Immune Cells Harvested from Co-Cultures of TAA and PBMCs Induced Potent Cancer Cell Killing

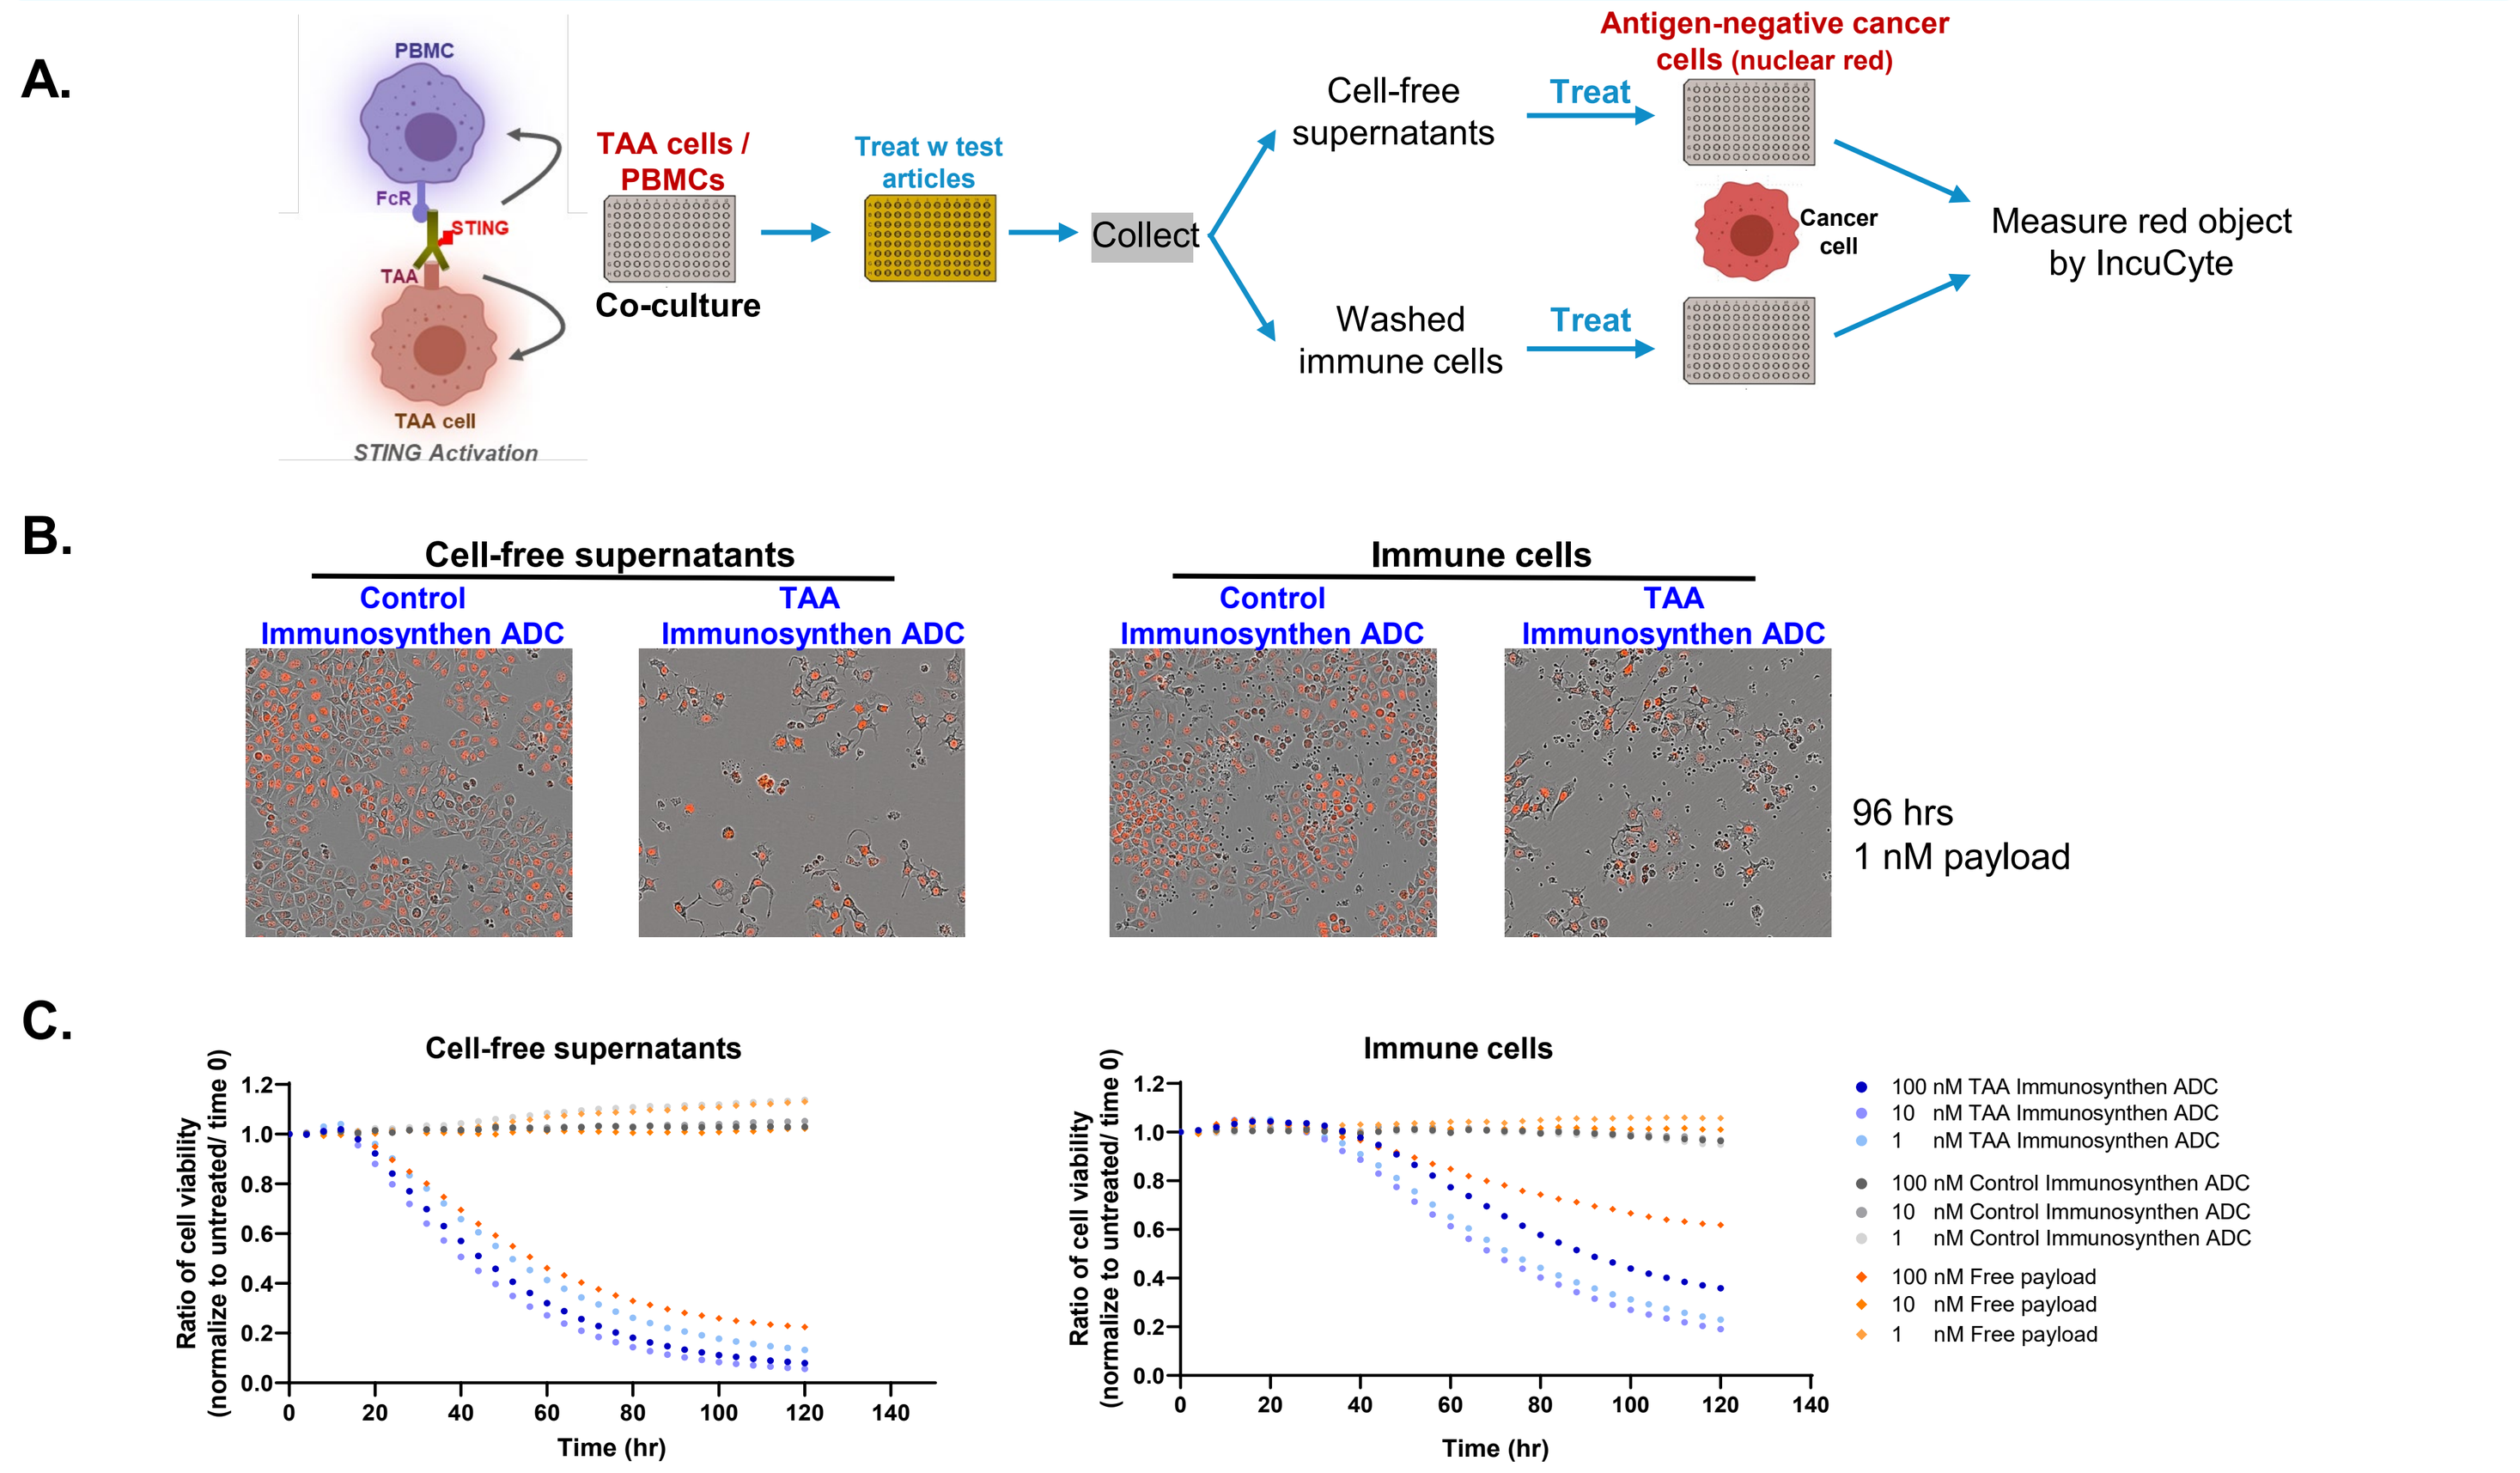
A.
PBMC
TAA cells / PBMCs
Co-culture
Treat w test articles
Collect
Cell-free supernatants
Washed immune cells
Treat
Antigen-negative cancer cells (nuclear red)
Cancer cell
Measure red object by IncuCyte
STING Activation
B.
Cell-free supernatants
Immune cells
Control Immunosynthen ADC
TAA Immunosynthen ADC
96 hrs
1 nM payload
C.
Ratio of cell viability (normalize to untreated/ time 0)
Time (hr)
100 nM TAA Immunosynthen ADC
10 nM TAA Immunosynthen ADC
1 nM TAA Immunosynthen ADC
100 nM Control Immunosynthen ADC
10 nM Control Immunosynthen ADC
1 nM Control Immunosynthen ADC
100 nM Free payload
10 nM Free payload
1 nM Free payload

Figure 5. Activity of cell-free supernatants and activated immune cells from TAA Immunosynthen ADC co-cultures on antigen-negative cancer cell killing. TAA cells and PBMCs were co-cultured in the presence of test articles. Cell-free supernatants and washed immune cells were harvested and added onto antigen-negative cancer cells (labeled with nuclear red) **(A)** Description of experimental design **(B)** Sample IncuCyte images of red cancer cells. **(C)** Ratio of cell viability over time.

CD8+ T Cells Contribute to Anti-Tumor Activity of TAA Targeted ADC in Syngeneic Model

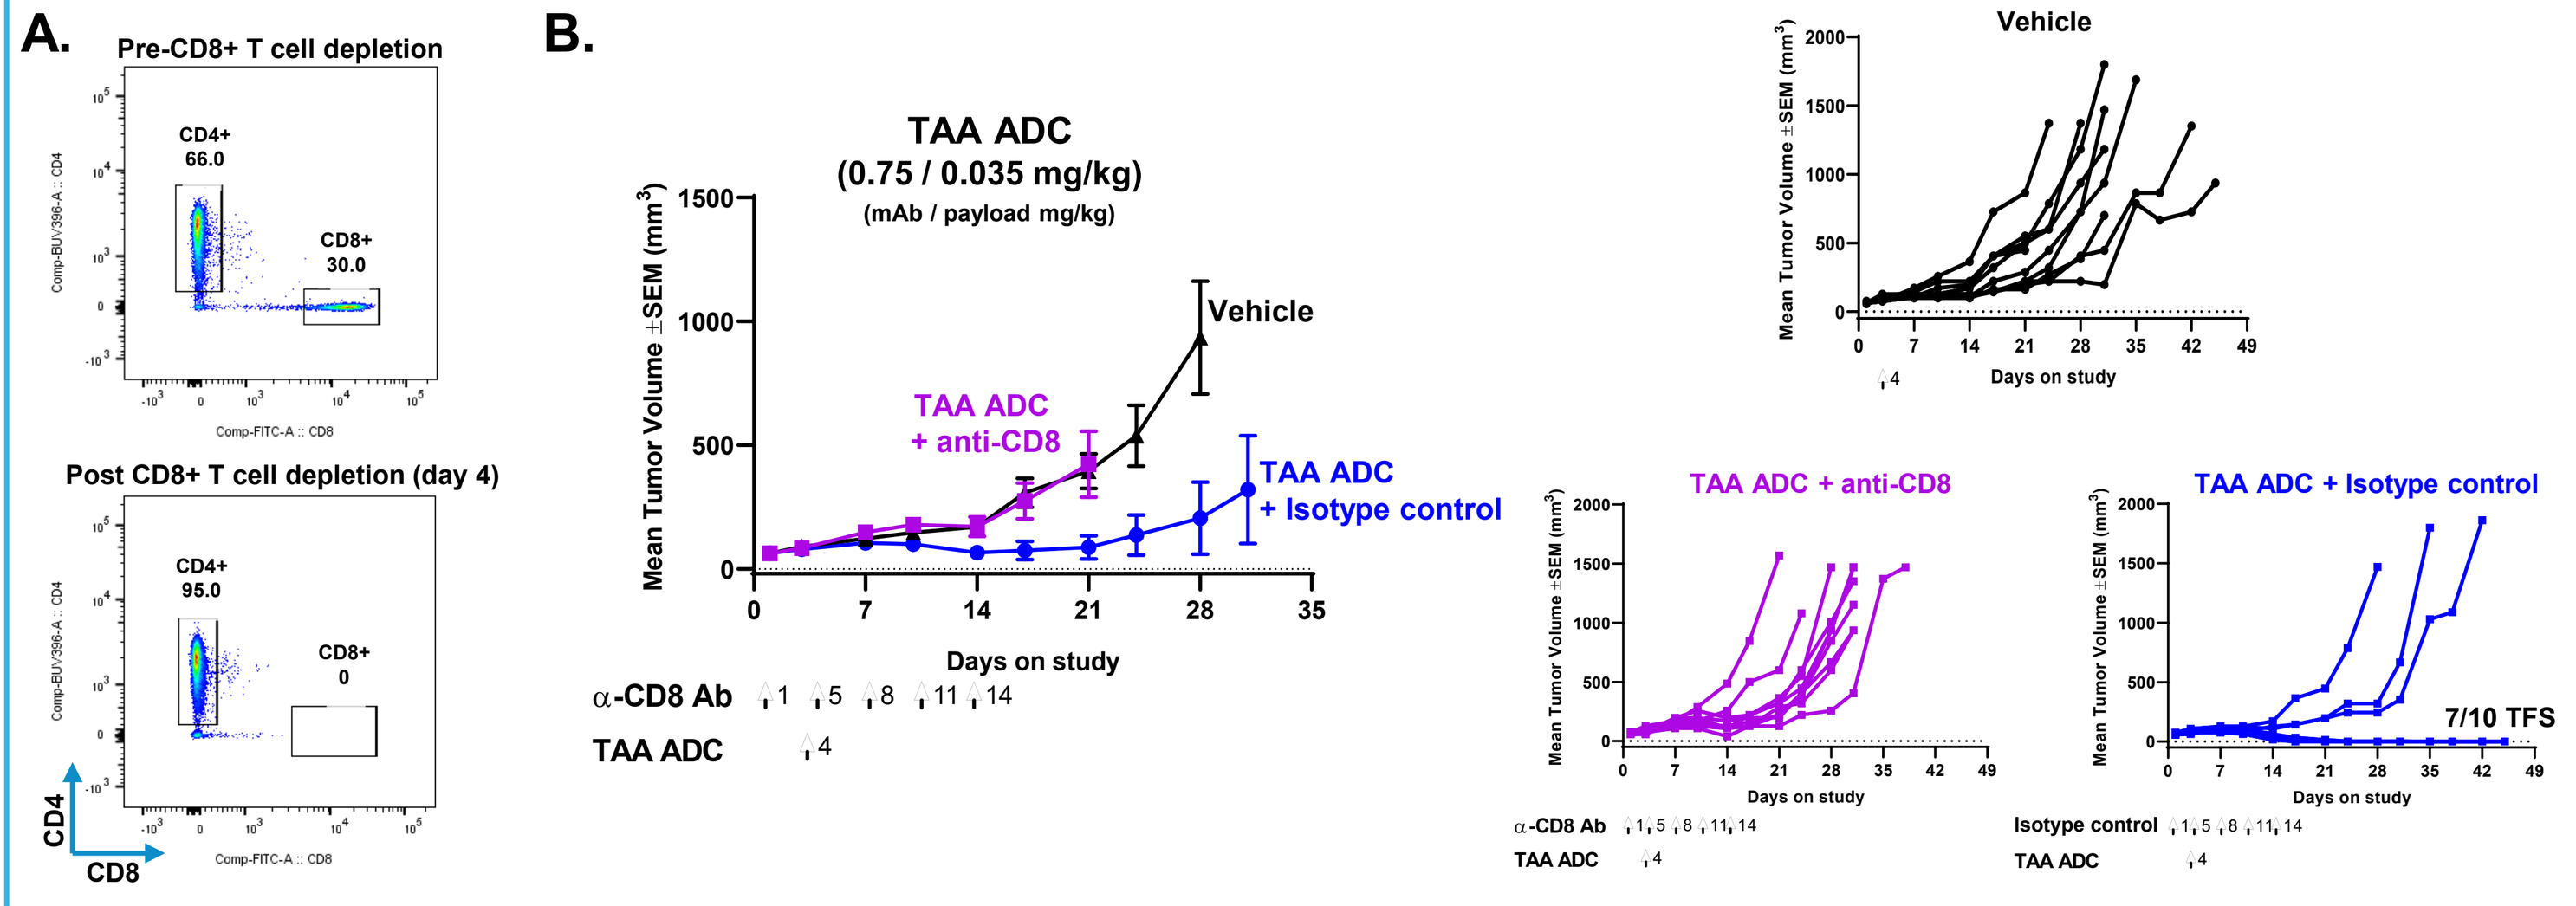
A.
Pre-CD8+ T cell depletion
Post CD8+ T cell depletion (day 4)
CD4+ 66.0
CD8+ 30.0
CD4+ 95.0
CD8+ 0
CD4
CD8
B.
TAA ADC (0.75 / 0.035 mg/kg) (mAb / payload mg/kg)
Vehicle
TAA ADC + anti-CD8
TAA ADC + Isotype control
Mean Tumor Volume ±SEM (mm³)
Days on study
α-CD8 Ab 1 5 8 11 14
TAA ADC 4
7/10 TFS

Figure 6. CD8+ T cells contribute to anti-tumor response of TAA STING-agonist ADC. (A) Flow cytometry of pre and post CD8 T cell depletion. **(B)** CD8+ T cells were depleted in tumor-bearing mice (100 µg/mouse) before single intravenous injection of TAA ADC. Mean tumor volumes for individual treatment groups are shown. TFS, tumor-free survival; Isotype control, control for anti-CD8.

Rapid Pharmacodynamic Effect in the Tumor following TAA Immunosynthen ADC

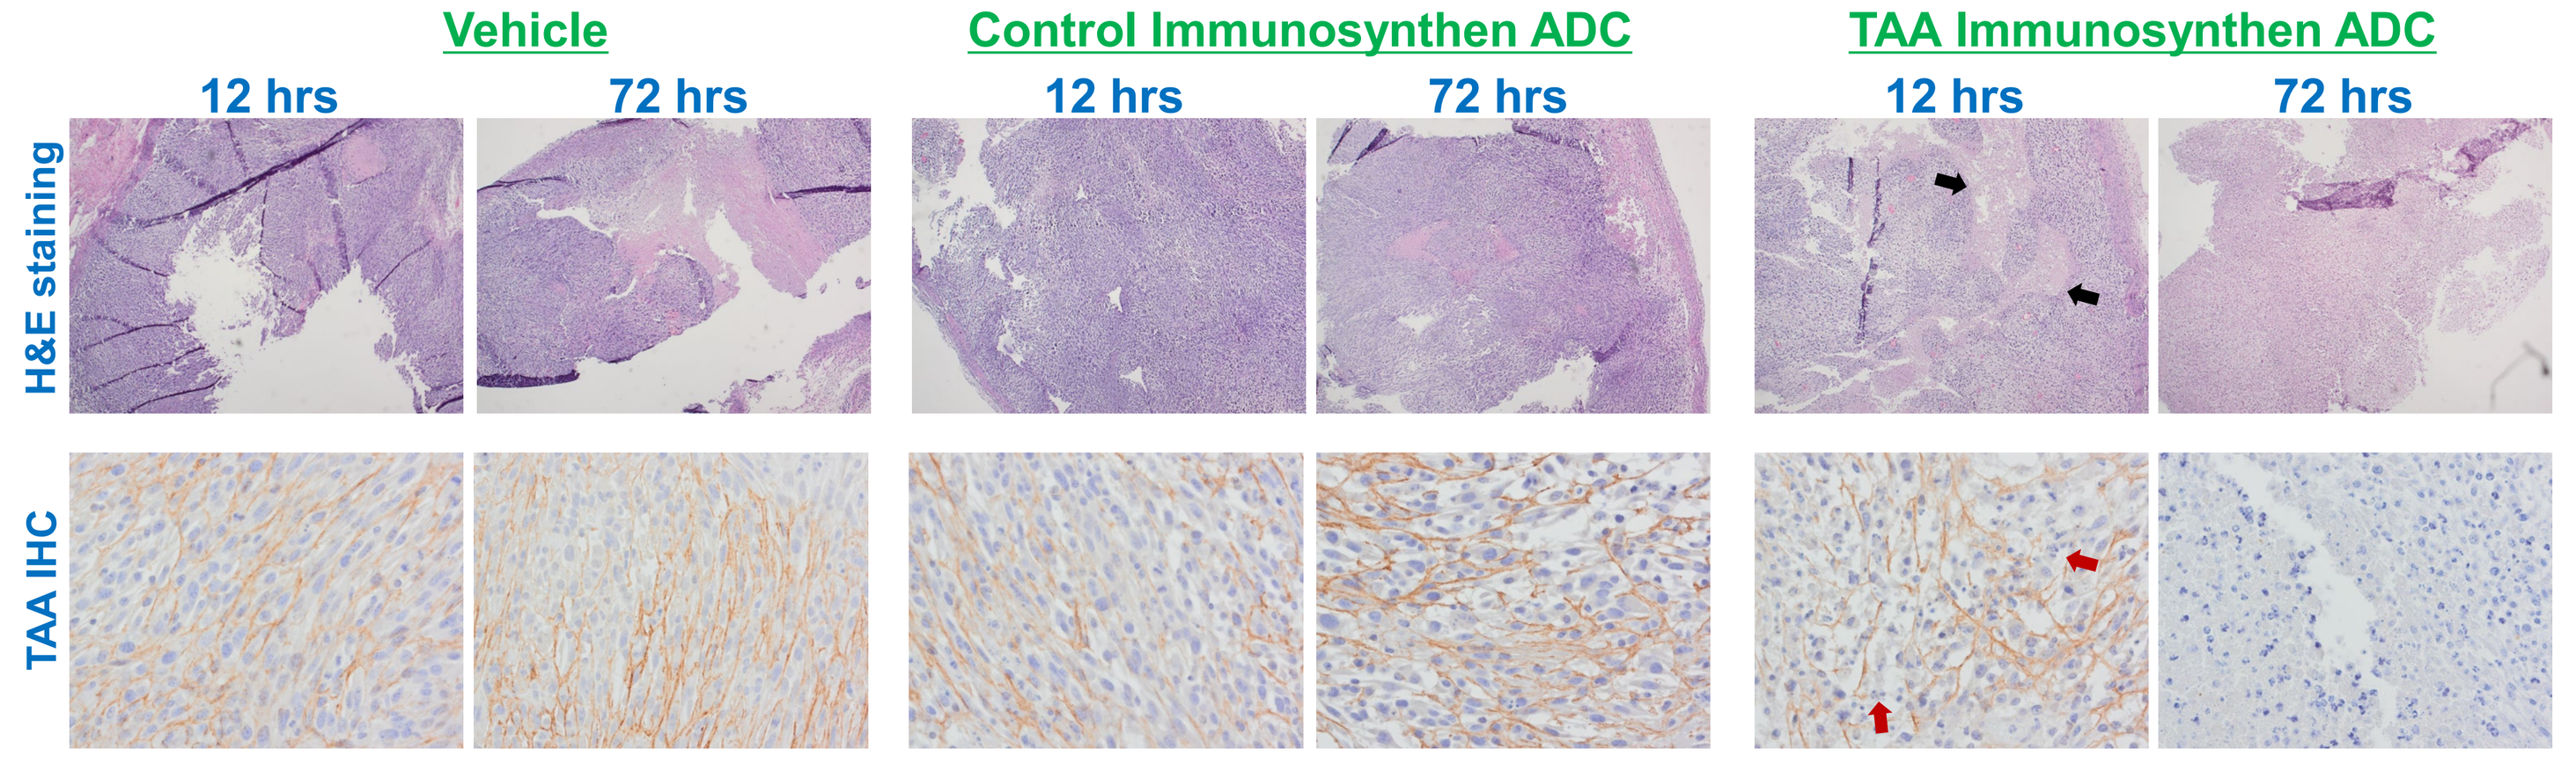
Vehicle
Control Immunosynthen ADC
TAA Immunosynthen ADC
12 hrs
72 hrs
H&E staining
TAA IHC

Figure 7. H&E and IHC staining of TAA cells. Tumors from mice treated with 1 / 0.035 mg/kg (mAb / payload) Immunosynthen ADCs were stained for TAA expression. Antigen expression was sustained outside the necrotic areas (black arrow) following STING activation within the TME 12 hrs post ADC administration indicating limited antigen modulation and intact target expression. Red arrows indicate apoptotic cells. At 72 hrs post TAA ADC administration, antigen expression is very limited due to extensive necrosis; target is no longer detected.

Sustained Tumor Regressions and Induction of Immunological Memory by STING-Agonist ADCs Targeting 2 TAAs

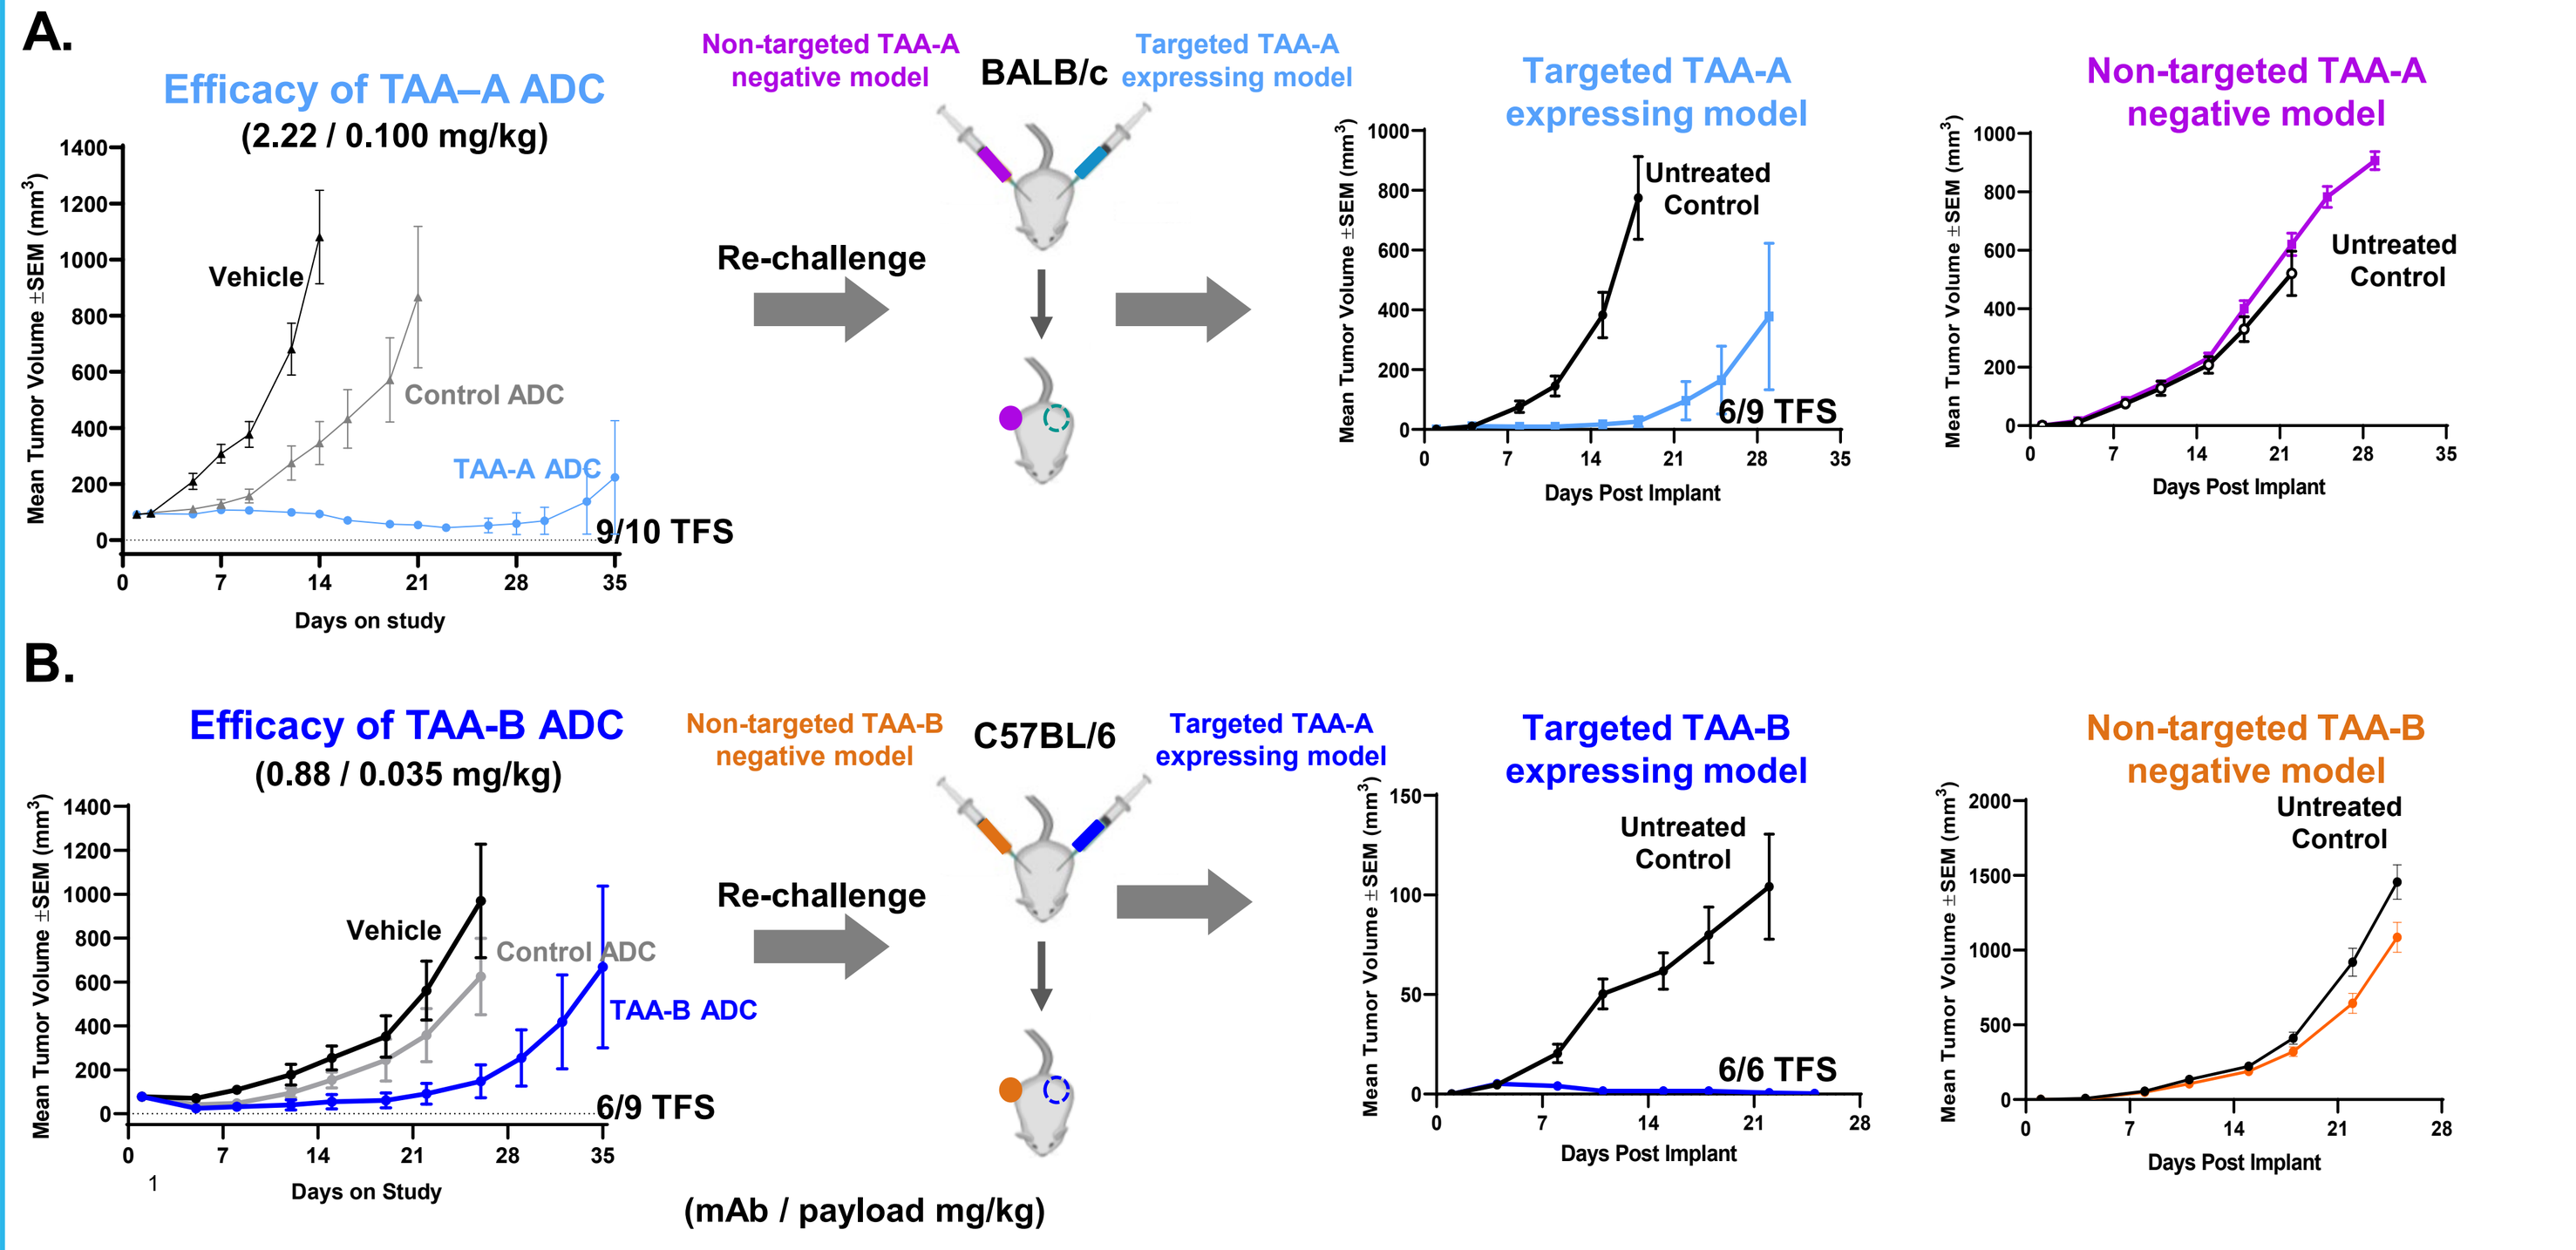
A.
Efficacy of TAA–A ADC (2.22 / 0.100 mg/kg)
Vehicle
Control ADC
TAA-A ADC
9/10 TFS
Non-targeted TAA-A negative model
BALB/c
Targeted TAA-A expressing model
Re-challenge
Targeted TAA-A expressing model
Untreated Control
6/9 TFS
Non-targeted TAA-A negative model
B.
Efficacy of TAA-B ADC (0.88 / 0.035 mg/kg)
TAA-B ADC
6/9 TFS
Non-targeted TAA-B negative model
C57BL/6
Targeted TAA-A expressing model
Targeted TAA-B expressing model
6/6 TFS
Non-targeted TAA-B negative model
(mAb / payload mg/kg)
Mean Tumor Volume ±SEM (mm³)
Days on study
Days Post Implant

Figure 8. Immunological memory of mice treated with TAA STING-agonist ADCs. (A) Tumor-free mice from targeted model expressing TAA-A treated with TAA-A STING-agonist ADC were re-challenged with original model expressing TAA-A on the right flank and non-targeted TAA-A negative model distinct from original model on the left flank. **(B)** Similar approach as shown in **A** but ADC targeting a second antigen, TAA-B, in TAA-B expressing model and re-challenged with original TAA-B model and another TAA-B negative model as indicated.

CONCLUSIONS

- ❖ Targeting tumor-associated antigens with STING-agonist ADCs activate STING pathway in immune cells via Fcγ receptor-mediated uptake
- ❖ Intrinsic activation of STING pathway in tumor-associated cells enables a dual mechanism of STING activation by Immunosynthen in target expressing cells and immune cells, providing a therapeutic advantage of targeting innate immune pathways over other approaches of targeted activation of the innate immune system
- ❖ Targeting of tumor-associated cells with Immunosynthen ADC induces cell killing of antigen-negative cancer cells with no impact on immune cells and minimal impact on target expressing cells
- ❖ STING-agonist ADCs targeting 2 distinct tumor-associated cell antigens induce sustained tumor regressions and induction of immunological memory

REFFERENCES

1. Cetinbas *et al*., SITC 2020 poster (Abstract # 620); Available at www.Mersana.com
2. Cetinbas *et al*., AACR 2021 poster (Abstract # 1773); Available at www.Mersana.com