ANNUAL MEETING ON WOMENS' CANCER®

BUILDING BRIDGES // BREAKING BARRIERS

SGO // PHOENIX, ARIZONA // MARCH 18 – 21, 2022

UPGRADE: Phase 1 Combination Trial of the NaPi2b-directed Dolaflexin Antibody Drug Conjugate (ADC) Upifitamab Rilsodotin (UpRi; XMT-1536) in Patients With Ovarian Cancer

Lakhani, Nehal[1]; Barve, Minal[2]; Burns, Timothy[3]; Edenfield, Jeffery[4]; Hays, John[5]; Zarwan, Corrine[6]; Werner, Theresa[7]; Anderson, Charles[8]; Buscema, Joseph[9]; Richards, Paul[10]; Bernardo, Patricia[11]; Keeton, Erika[11]; Carrington, Cassandra[11]; Burger, Robert A[11]

[1]START Midwest, Grand Rapids, MI; [2]Mary Crowley Cancer Research, Dallas, TX; [3]Hillman Cancer Center, University of Pittsburgh, Pittsburgh, PA; [4]Prisma Health Cancer Institute, Greenville, SC; [5]Arthur James Cancer Hospital, Ohio State University, Columbus, OH; [6]Lahey Clinic, Burlington, MA; [7]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT; [8]Willamette Valley Cancer Institute and Research Center, Eugene, OR; [9]Arizona Oncology, Tucson, AZ; [10]US Oncology, Oncology and Hematology Assoc. of Southwest VA, Salem, VA; [11]Mersana Therapeutics, Inc, Cambridge, MA

BACKGROUND

Unmet Need for Platinum-Sensitive Recurrent HGSOC[1–3]

- Standard of care for patients with newly diagnosed and platinum-sensitive recurrent HGSOC consists of platinum-based chemotherapy (often in combination with paclitaxel)
- Although platinum-containing combination therapies offer improvement in outcomes over single-agent platinum therapy, these improvements are associated with additional toxicity, with response rates diminishing through subsequent therapies
 - Paclitaxel is associated with high incidence of TRAEs, including hypersensitivity reactions, hematological toxicity (neutropenia), alopecia, peripheral neuropathy, and myalgia or arthralgia
- Limited response rates beyond first line indicate that there is a continued need for effective therapies in the treatment of platinum-sensitive recurrent ovarian cancer
- Dose-limiting toxicities, such as thrombocytopenia and neutropenia, limit duration of platinum-based therapy (usually 6 cycles) in the recurrent disease setting

Rationale for Combination Therapy With Carboplatin[3–5]

- To address this unmet medical need, novel platinum-based combinations must be developed that:
 - Can be continued as maintenance treatment following completion of platinum-based chemotherapy
 - Specifically contain targeted agents with favorable therapeutic index and lack appreciable overlapping toxicity with carboplatin
 - Have non-overlapping mechanisms of action with other agents typically combined with carboplatin
- Antibody drug conjugates (ADCs) may represent a promising strategy in combination with carboplatin to optimize therapeutic index for patients

Upifitamab Rilsodotin (UpRi): First-in-Class ADC Targeting NaPi2b[6,7]

- **NaPi2b** is a sodium-dependent phosphate transport protein broadly expressed in solid tumors, including high-grade serous epithelial ovarian, fallopian tube, and primary peritoneal cancer, with limited expression in normal tissue
- Estimated **two-thirds** of high-grade serous ovarian cancers express NaPi2b
- A Phase 1b UpRi single-agent study demonstrated a RECIST v1.1 confirmed ORR of 34% including 2 complete responses, and a disease control rate of 84% in 38 patients with tumors expressing high NaPi2b (TPS ≥75) and at least 1 on-study tumor imaging
- Based on the encouraging single-agent safety and efficacy data, we hypothesize that UpRi in combination with other therapies can provide additional clinical benefit and improved tolerability over current standard of care

Hydrophilic Polymer Scaffold

- High DAR of ~10
- Excellent drug-like properties
- Highly stable in circulation
- Dose-proportional exposure
- Very low exposure of free payload

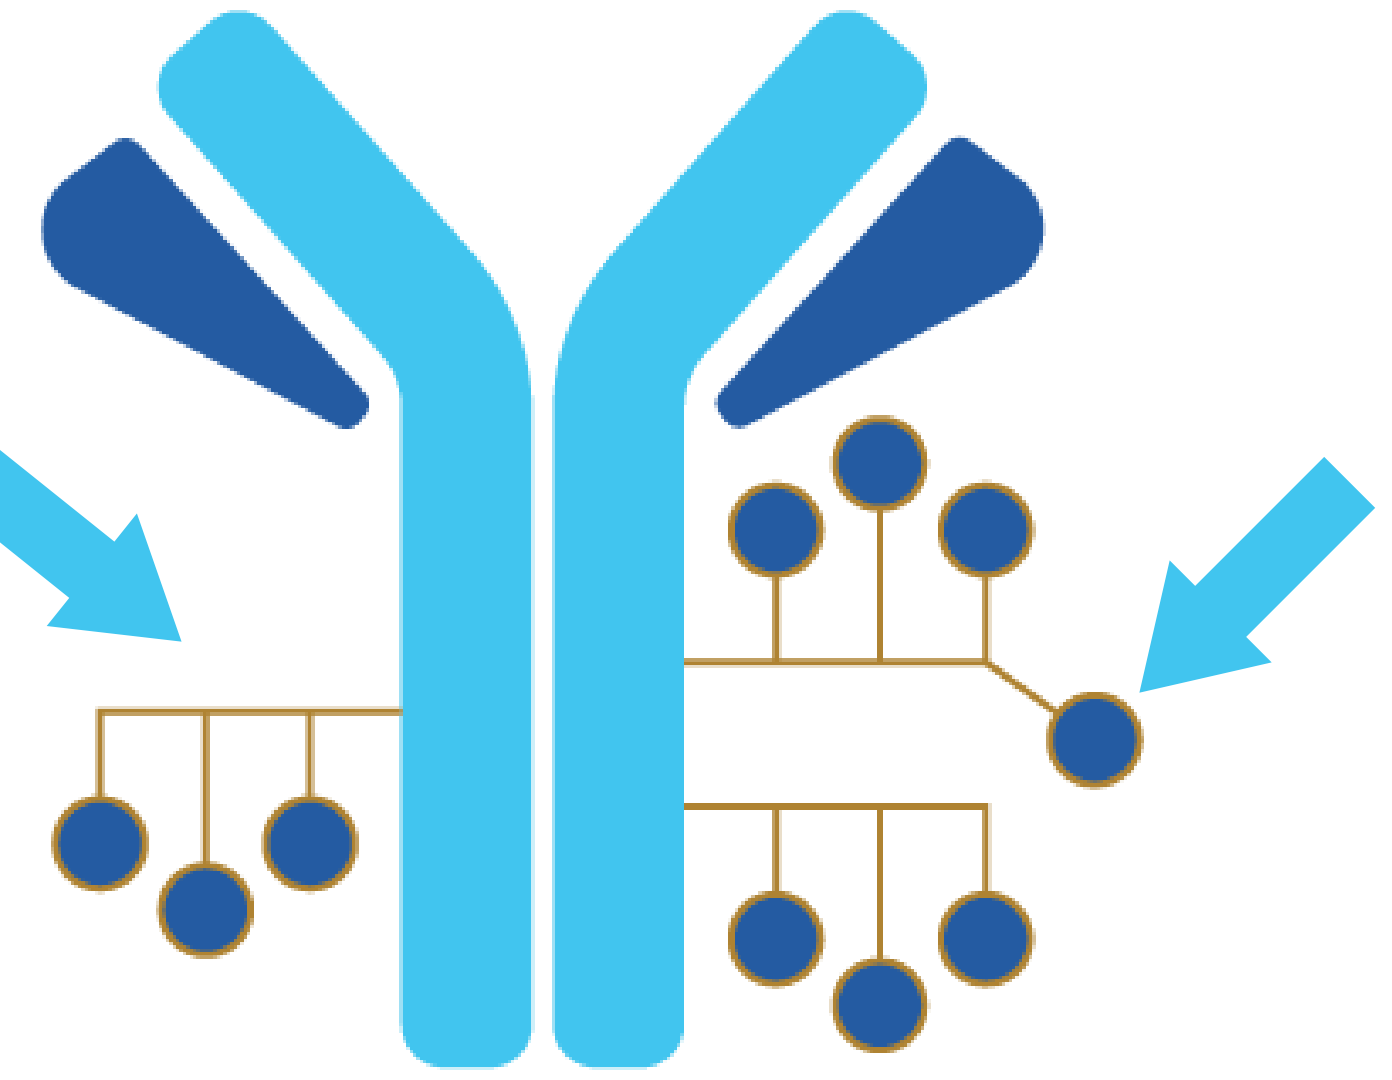

DolaLock Payload With Controlled Bystander Effect

- Selectively toxic to rapidly dividing cells
- Initially released payload (AF-HPA) is freely cell permeable and bystander capable
- Intracellular conversion to AF diminishes permeability and controls bystander effect
- Accumulates in tumor, not a PgP substrate
- Induces immunogenic cell death

METHODS

Study Design and Eligibility

UPGRADE-A is a cohort under the UPGRADE umbrella study evaluating UpRi in combination with other therapies (NCT04907968), specifically a Phase 1 dose escalation and expansion study evaluating UpRi in combination with carboplatin in patients with platinum-sensitive[a] recurrent OC who have received 1–2 prior lines of therapy. Trial currently enrolling patients. Additional combination cohorts will be added.

Key Enrollment Criteria

- Recurrent, platinum-sensitive[a], high-grade serous ovarian cancer, including fallopian tube or primary peritoneal cancer
- 1–2 prior platinum-based regimens
- Patients not selected for NaPi2b expression
- Tissue (fresh or archival) for retrospective assessment of NaPi2b expression
- RECIST v1.1 measurable disease
- ECOG PS = 0–1

Dose Escalation (BOIN design; N=18)

UpRi (3 doses) + carbo AUC 5 Q4W ×6 → **UpRi Q4W** until PD or unacceptable AE

Primary Endpoint

- MTD for UpRi with carboplatin AUC 5

Secondary Objectives

- AEs, PK for UpRi, PK for carboplatin, immunogenicity for UpRi, ORR, PFS, OS

Expansion (N=30)

UpRi MTD + carbo AUC 5 Q4W ×6 → **UpRi Q4W** until PD or unacceptable AE

Primary Endpoint

- Feasibility

Secondary Objectives

- AEs, PK for UpRi, PK for carboplatin, immunogenicity for UpRi, ORR, PFS, OS, efficacy by NaPi2b expression

[a] Platinum-sensitive is defined as having achieved either a partial or complete response to 4 or more cycles in their last platinum-containing regimen and their disease progressing more than 6 months after completion of the last dose of platinum-containing therapy.

Statistical Considerations

Dose Escalation

- Bayesian Optimal Interval (BOIN) design will be used to determine the MTD among the 3 dose levels to be evaluated in this study
- If additional dose levels/schemes are planned to be evaluated, then the maximum sample size will be increased by 6 patients for every dose level/scheme planned to be evaluated

Expansion

- Primary objective of expansion cohort is to determine tolerability at MTD determined by ≥60% of participants completing at least 4 cycles of UpRi/carboplatin combination without discontinuing treatment earlier for reasons other than disease progression
- Secondary objective is to assess correlation of tumor expression of NaPi2b and objective tumor response

CONCLUSIONS

- Upifitamab rilsodotin (UpRi) is an investigational first-in-class ADC targeting the sodium-dependent phosphate transport protein NaPi2b
- Based on available emerging data, we hypothesize that UpRi in combination with other therapies can provide additional clinical benefit and improved tolerability over current standard of care
- UPGRADE-A is a cohort under the umbrella study, UPGRADE, evaluating UpRi in combination with carboplatin in patients with platinum-sensitive (progressing >6 months of last dose of platinum) recurrent OC who have received 1–2 prior lines of therapy
- The primary objectives are to identify the MTD and to assess the feasibility of the combination. Secondary endpoints include safety and tolerability, PK, and preliminary anti-neoplastic activity
- Combining UpRi with carboplatin has the potential of improving tolerability by replacing paclitaxel and other chemotherapies typically combined with carboplatin, which could allow for UpRi maintenance following completion of platinum treatment
- ClinicalTrials.gov registry: NCT04907968

ACKNOWLEDGEMENTS

We would like to thank the patients, their families, and the site staff for making this study possible.

This study is sponsored by Mersana Therapeutics, Inc.

Editorial support for this poster was provided by BluPrint Oncology.

REFERENCES

1. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Ovarian Cancer V.2.2020. Published 18 January 2022. Accessed 27 January 2022. https://www.nccn.org/professionals/physician_gls/pdf/ovarian.pdf
2. Herzog TJ. *Clin Cancer Res*. 2004;10(22):7439–7449.
3. Slaughter KLL et al. *Gynecol Oncol*. 2016;142(2):225–230.
4. Ray-Coquard I et al. *Cancer Treatment Reviews*. 2020; 90:102107.
5. Vasan N et al. *Nature*. 2019;575:299–309.
6. Lin K et al. Clin Cancer Res. 2015;21(22):5139–5150.
7. Mersana. Data on File. 2022.

ADDITIONAL INFORMATION

Downloadable PDF copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from SGO and the author of this poster.

For more information on UPGRADE, visit ClinicalTrials.gov page NCT04907968 via QR code provided.

Abbreviations: ADC, antibody drug conjugate; AE, adverse event; AUC, area under the curve; AF, auristatin F; AF-HPA, auristatin F-hydroxypropylamide; BOIN, Bayesian Optimal Interval; carbo, carboplatin; DAR, drug-to-antibody ratio; ECOG, Eastern Cooperative Oncology Group; FPD, first patient dosed; HGSOC, high-grade serous ovarian cancer; MTD, maximum tolerated dose; NaPi2b, sodium-dependent phosphate transport protein 2B; OC, ovarian cancer; ORR, overall response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PgP, P-glycoprotein; PK, pharmacokinetics; PS, performance score; Q4W, every 4 weeks; RECIST, Response Evaluation Criteria in Solid Tumors; TPS, tumor proportion score; TRAE, treatment-related adverse event; UpRi, upifitamab rilsodotin.

TiP Abstract 588