Tumor cell-targeted STING-agonist antibody-drug conjugates achieve potent anti-tumor activity by delivering STING agonist specifically to tumor cells and Fcγ-RI-expressing subset of myeloid cells

Abstract ID: 4873

Naniye Malli Cetinbas, Travis Monnell, Kalli C. Catcott, Jahna Soomer-James, Pamela Shaw, Rebecca Mosher, Keith Bentley, Susan Clardy, Bingfan Du, Eoin Kelleher, Marina Protopopova, Cheri Stevenson, Joshua D. Thomas, Alex Uttard, Dorin Toader, Jeremy Duvall, Raghida A. Bukhalid, Marc Damelin, Timothy B. Lowinger

Mersana Therapeutics, Inc., Cambridge, MA

Abstract

STING pathway agonism has emerged as a potential therapeutic strategy to stimulate anti-tumor immune responses. We have previously shown that tumor cell-targeted antibody-drug conjugates (ADCs) carrying a novel STING agonist induce anti-tumor activity without causing substantial elevations in systemic cytokine levels, thus suggesting a therapeutic advantage of STING agonist ADCs relative to unconjugated agonists. ADCs constitute a proven therapeutic modality that is ideally suited to enable systemic administration and delivery of the conjugated drug to desired cell types within the tumor microenvironment. In addition to delivering STING agonist into the antigen-expressing tumor cells, antigen-bound ADCs deliver STING agonist to tumor-resident myeloid cells through Fcγ receptor (FcγR)-mediated internalization. In this study we investigated the mechanism of FcγR-mediated internalization of the tumor cell-targeted STING-agonist ADCs into myeloid cells and the nature of the subsequent STING pathway activation. We developed flow cytometry-based assays to determine the changes in FcγRI, FcγRII, and FcγRIII cell surface detection levels in the presence of ADCs specifically designed to be either proficient or deficient in FcγR-binding. Combined with functional assays based on co-culture of cancer cells and FcγRI knock out myeloid cells, we identified FcγRI as the major Fcγ receptor that mediates target-bound ADC internalization into myeloid cells *in vitro*. Even though FcγRI is expressed only by a subset of CD11b+ myeloid cells, tumor cell-targeted ADCs induce greater production of interferons and other cytokines and more potent cancer cell killing than CD11b-targeted-ADCs, which deliver STING agonist into FcγRI- (non-productive) as well as FcγRI+ (productive) myeloid cells. Finally, we demonstrate that myeloid cells within dissociated primary human tumors from multiple donors express FcγRI and are capable of tumor cell killing in response to tumor cell-targeted STING agonist ADCs *in vitro*. In summary, our data indicate that the ADC-mediated delivery of a STING agonist specifically into FcγRI-expressing myeloid cells efficiently activates innate immune responses in the most relevant immune cell types within the tumor microenvironment.

BACKGROUND

Tumor cell-targeted STING-agonist ADCs

- Systemically administered
- Tumor targeted delivery of STING agonist
- Potent target-dependent anti-tumor activity
- Improved efficacy and reduced systemic cytokines compared to a systemically administered free STING agonist
- Well-tolerated in repeat IV dose toxicology studies
- XMT-2056, a HER2-targeted Immunosynthen STING-agonist ADC is scheduled to initiate clinical trials this year. See our AACR poster (#3503) for more details.

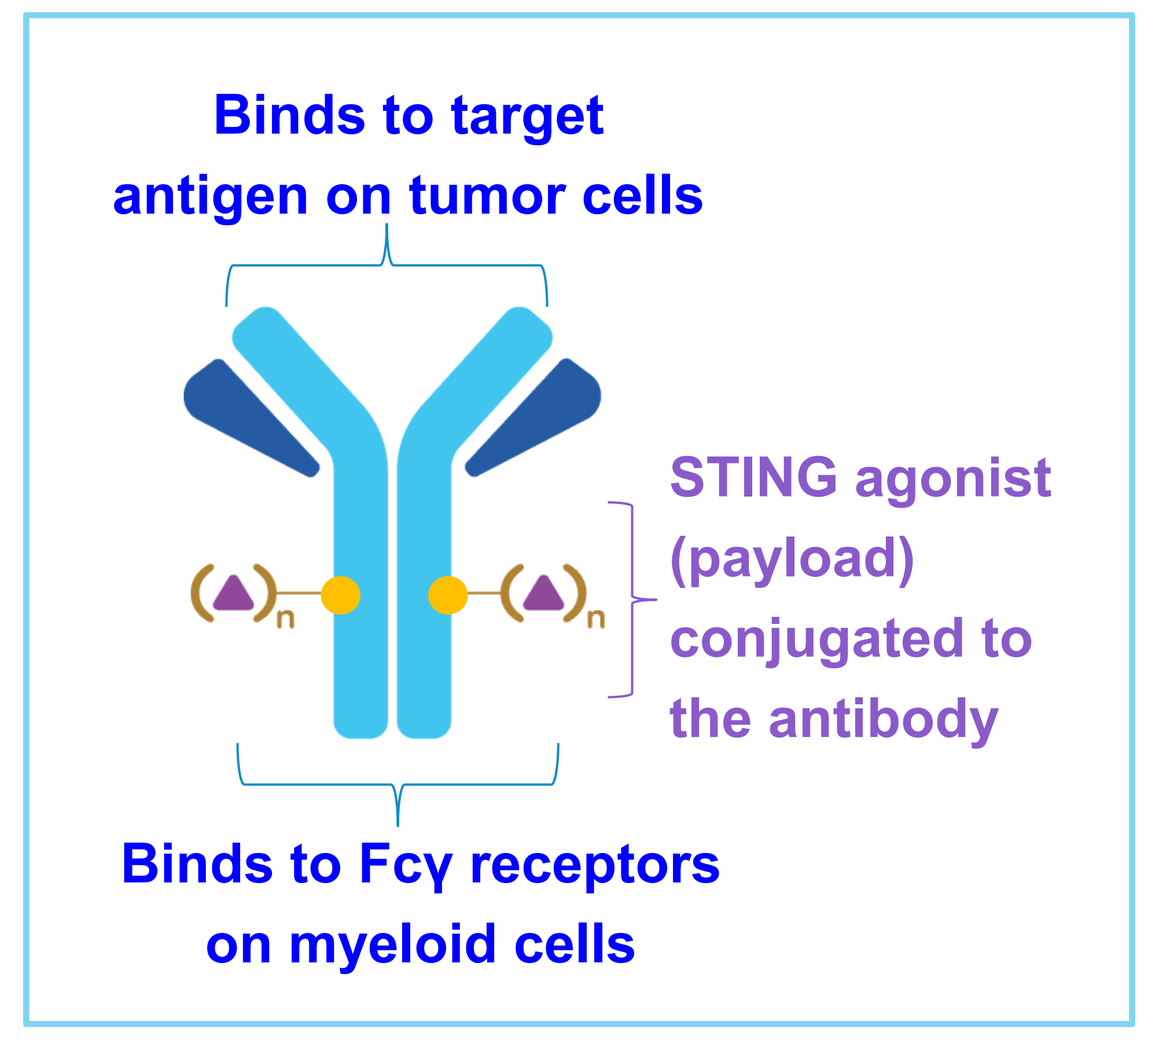
Binds to target antigen on tumor cells
STING agonist (payload) conjugated to the antibody
Binds to Fcγ receptors on myeloid cells

Proposed mechanism of action of tumor cell-targeted STING-agonist ADCs

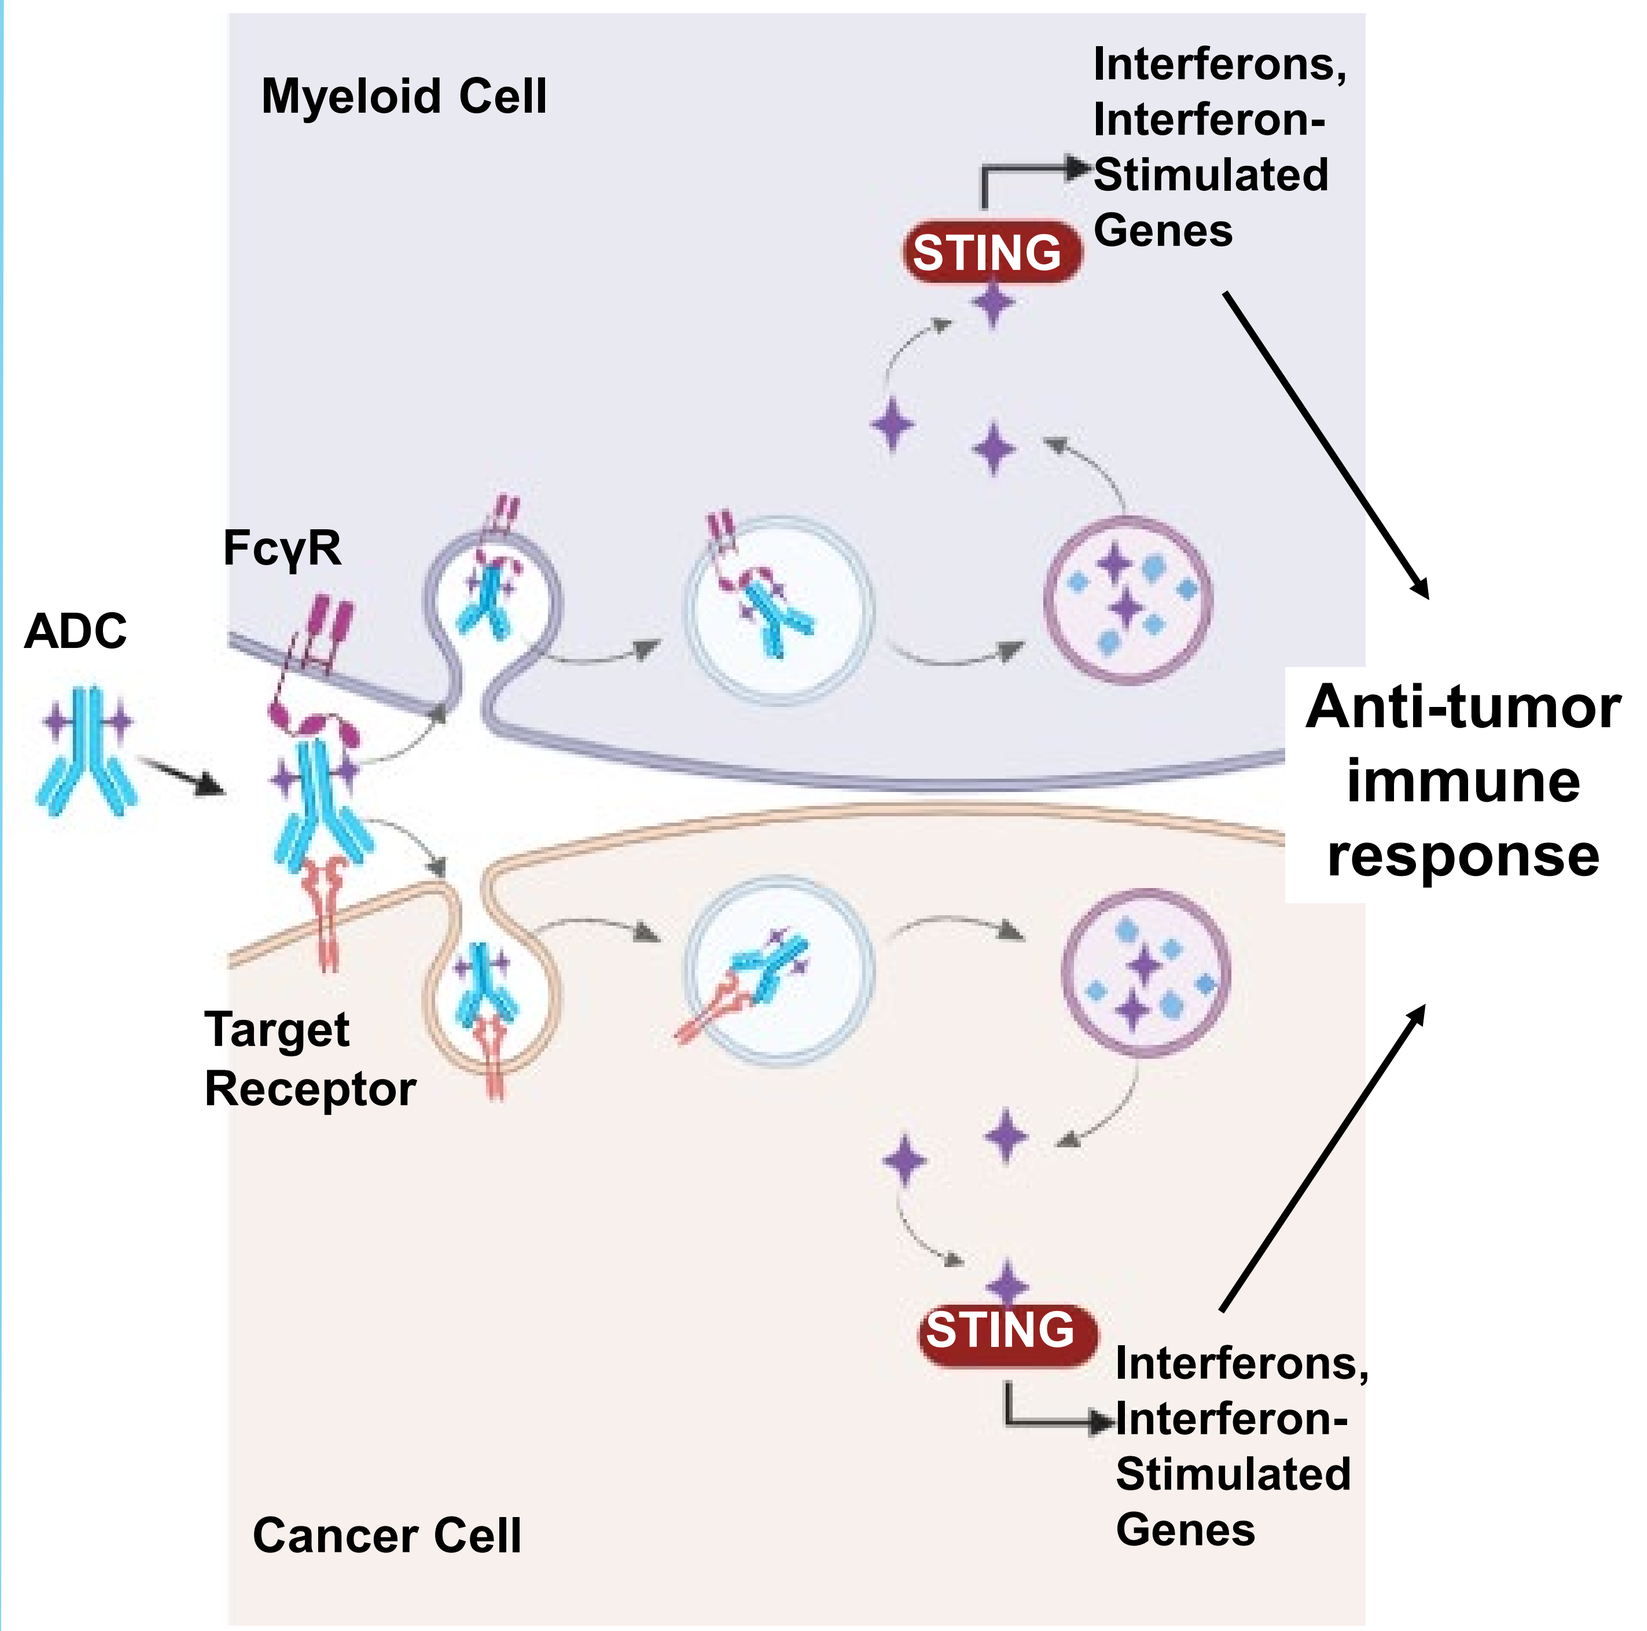
Myeloid Cell
Interferons, Interferon-Stimulated Genes
STING
FcγR
ADC
Anti-tumor immune response
Target Receptor
STING
Interferons, Interferon-Stimulated Genes
Cancer Cell

Figure 1. Delivery of a STING agonist into tumor cells and myeloid cells via a tumor cell-targeted ADC.

- When bound to their target antigens on tumor cells ADCs strongly interact and crosslink Fcγ receptors and internalize into myeloid cells resulting in robust activation of the STING pathway.
- Tumor cell-targeted Immunosynthen ADCs are also internalized into tumor cells and can activate tumor intrinsic STING pathway in the presence of cues from TME or primary immune cells[1,2].
- **In this study we investigated the mechanism of FcγR-mediated internalization of the tumor cell-targeted STING-agonist ADCs into myeloid cells and the nature of the subsequent STING pathway activation.**

RESULTS

Tumor cell-targeted STING-agonist ADCs induce STING pathway in myeloid cells in an Fc- and antigen-dependent manner

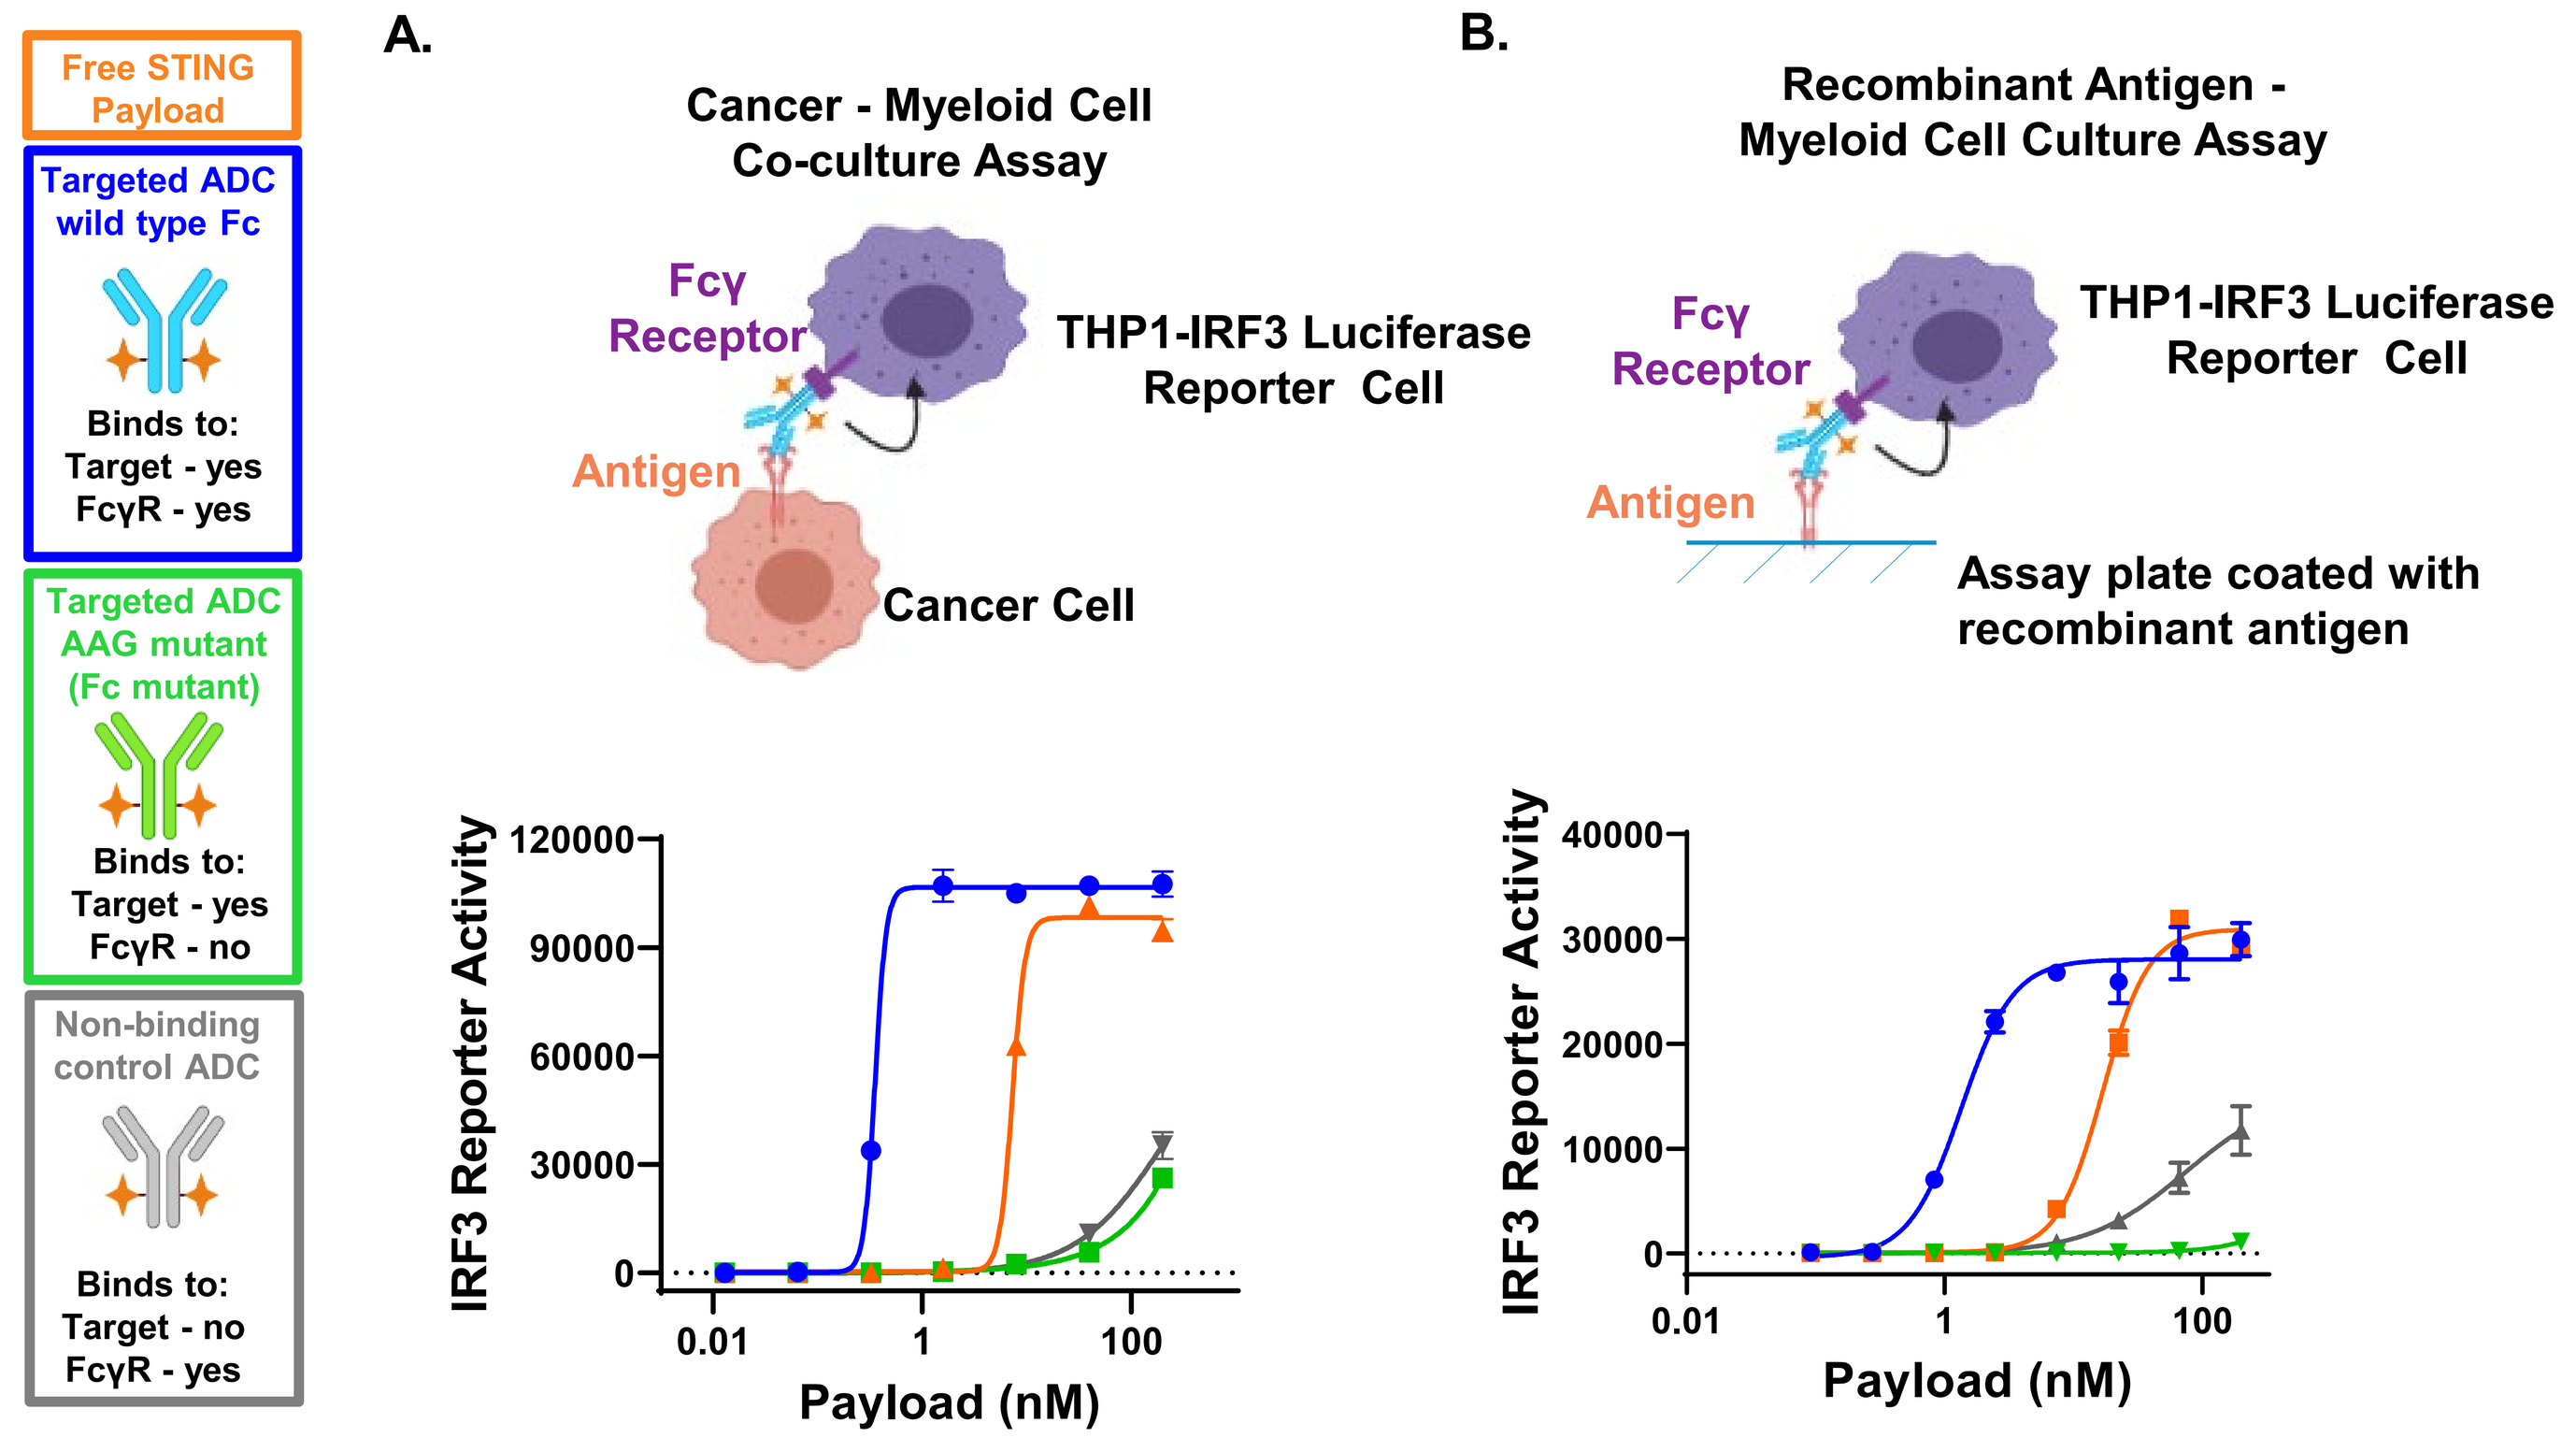
Free STING Payload
Targeted ADC wild type Fc
Binds to: Target - yes FcγR - yes
Targeted ADC AAG mutant (Fc mutant)
Binds to: Target - yes FcγR - no
Non-binding control ADC
Binds to: Target - no FcγR - yes
A.
Cancer - Myeloid Cell Co-culture Assay
Fcγ Receptor
THP1-IRF3 Luciferase Reporter Cell
Antigen
Cancer Cell
B.
Recombinant Antigen - Myeloid Cell Culture Assay
Fcγ Receptor
THP1-IRF3 Luciferase Reporter Cell
Antigen
Assay plate coated with recombinant antigen
IRF3 Reporter Activity
Payload (nM)

Figure 2. Tumor cell-targeted STING-agonist ADCs activate STING pathway in THP1 IRF3-Luciferase reporter cells in an Fc- and antigen-dependent manner. THP1 IRF3-Luciferase cells were **(A)** co-cultured with cancer cells expressing the antigen or **(B)** cultured on recombinant antigen-coated plates in the absence of cancer cells and treated with tumor antigen-targeted ADC with *wt* or mutant Fc, a non-binding control ADC, and free payload for 24 hours and Luciferase activity (as proxy for STING pathway activation downstream of IRF3 activation) was measured. Targeted ADC with *wt* Fc induced potent IRF3 reporter activity in THP1 cells. Neither the targeted ADC with mutant Fc nor the control ADC had significant activity, demonstrating that STING pathway activation in THP1 cells by the tumor cell-targeted STING-agonist ADC requires both FcγR-binding and antigen-binding.

Fcγ-RI is the major receptor mediating internalization of the antigen-bound tumor cell-targeted STING agonist ADC into myeloid cells

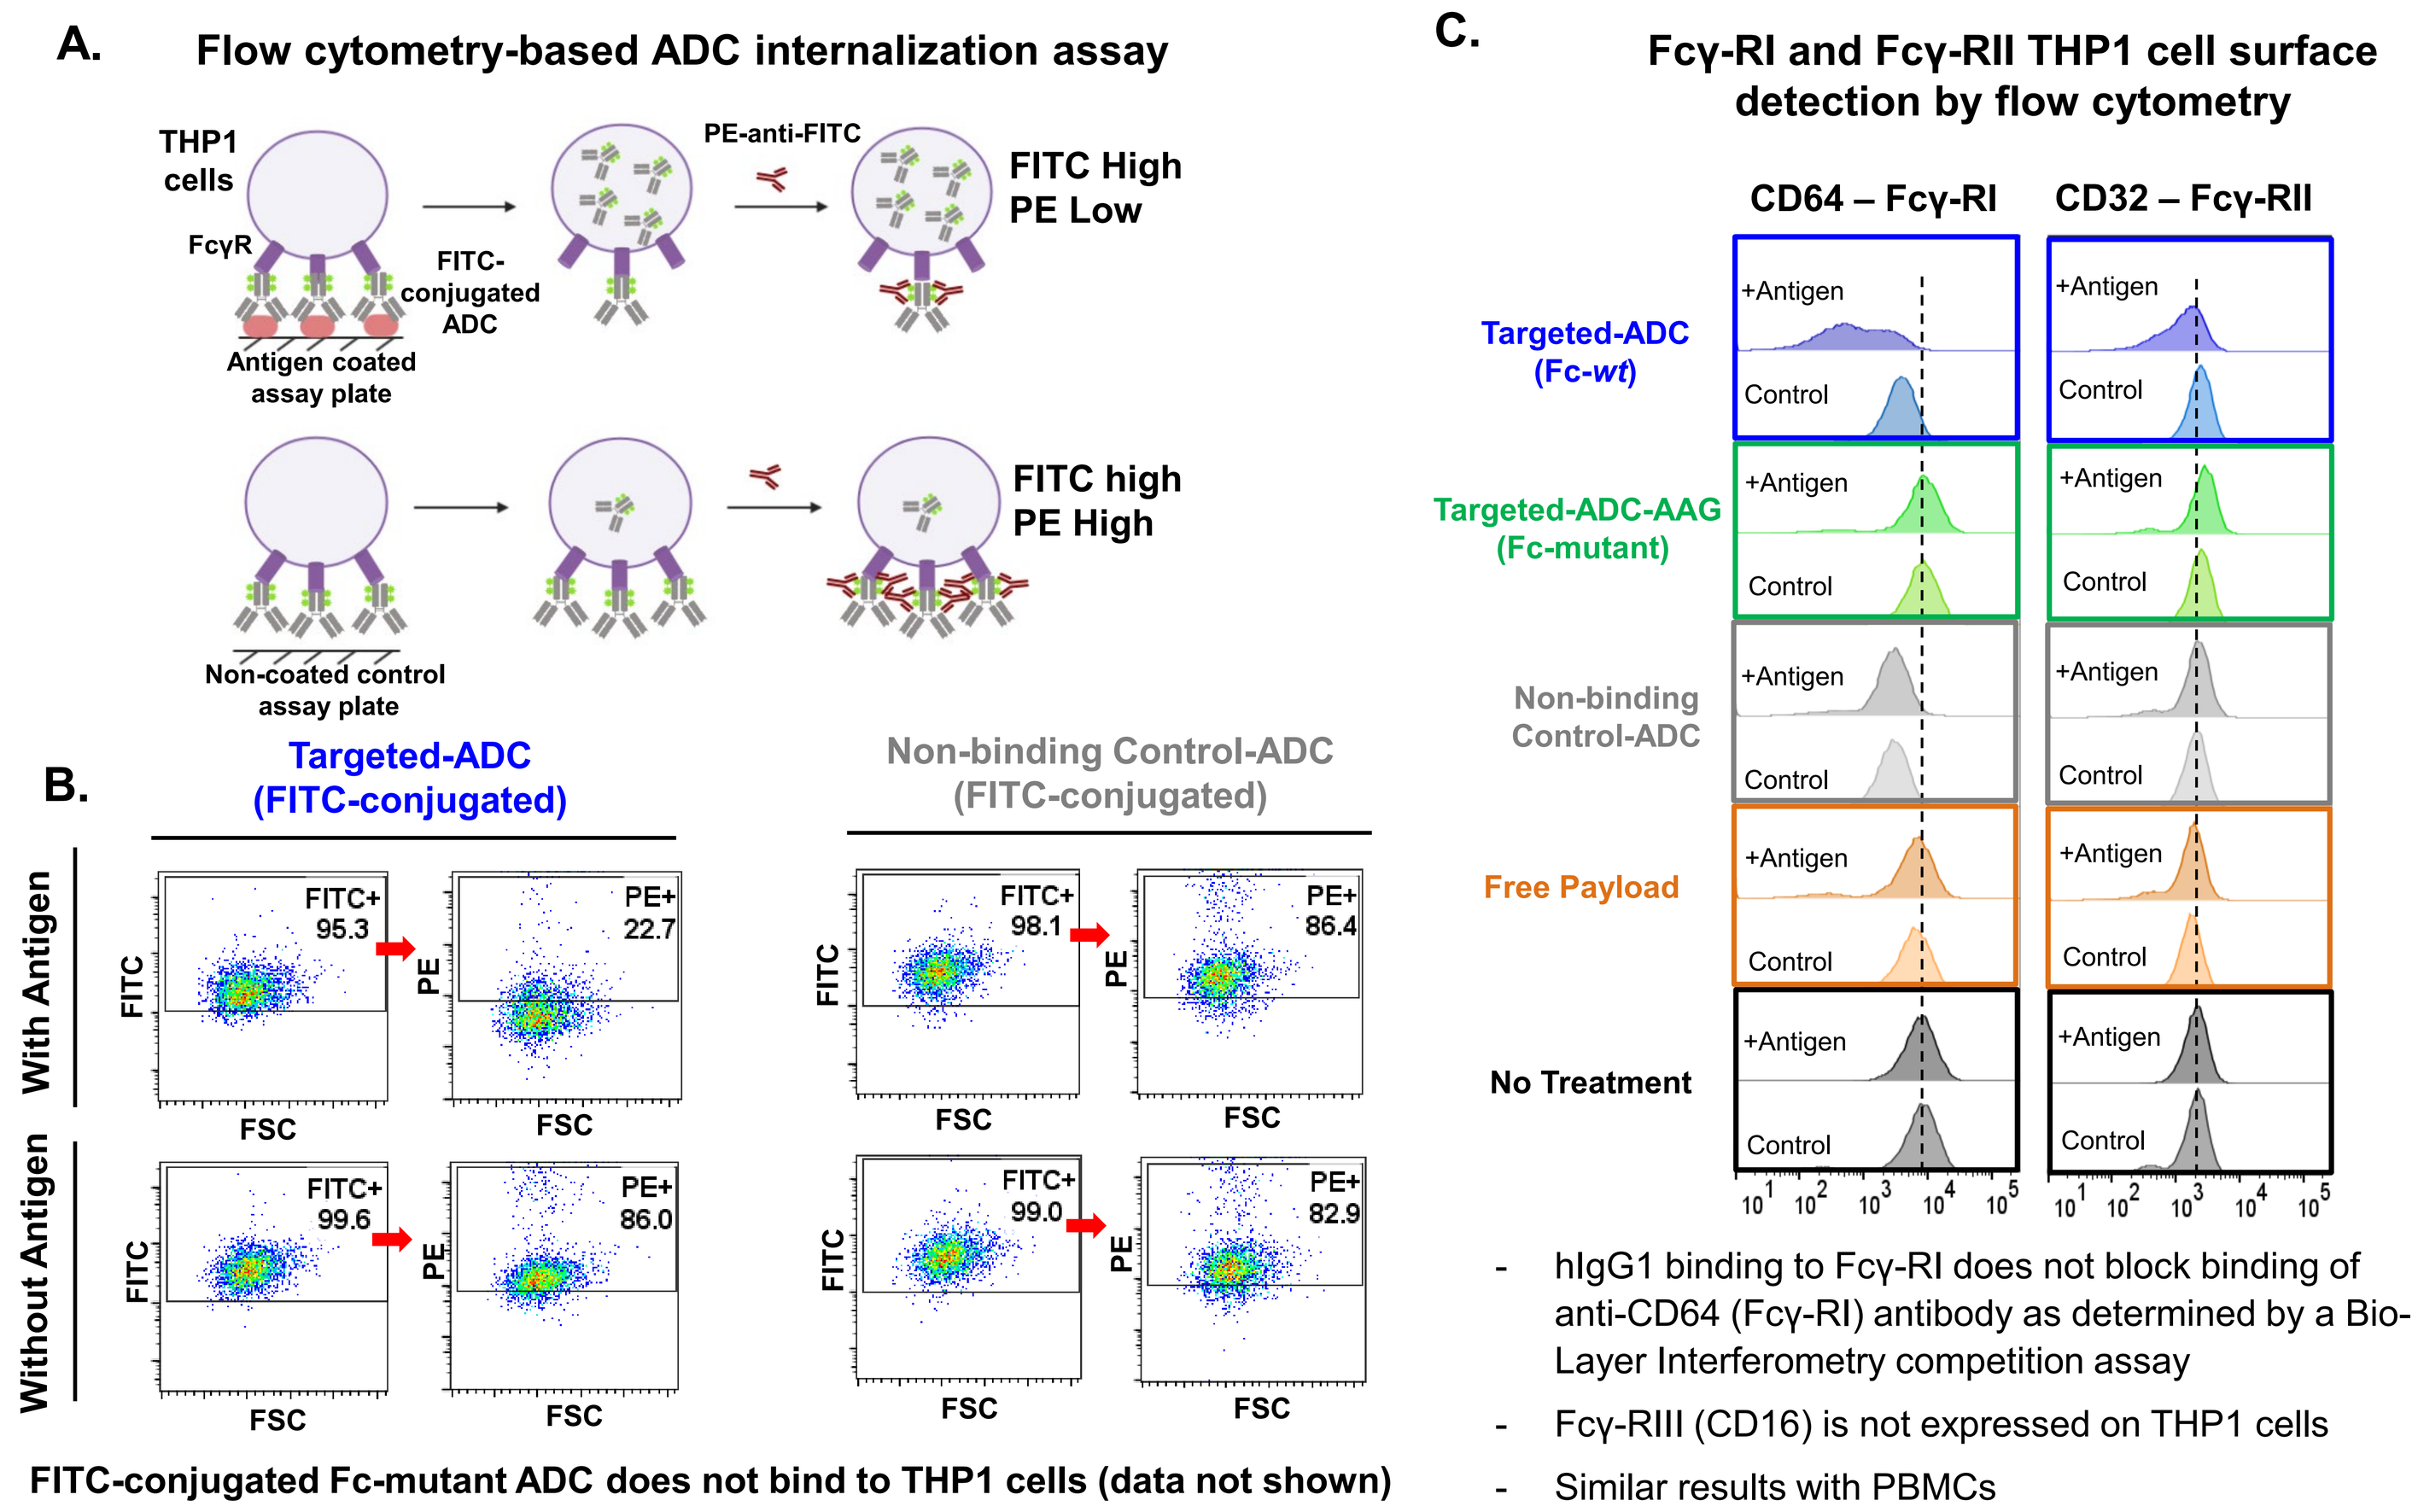
A.
Flow cytometry-based ADC internalization assay
THP1 cells
FcγR
Antigen coated assay plate
FITC-conjugated ADC
PE-anti-FITC
FITC High PE Low
Non-coated control assay plate
FITC high PE High
B.
Targeted-ADC (FITC-conjugated)
Non-binding Control-ADC (FITC-conjugated)
With Antigen
Without Antigen
C.
Fcγ-RI and Fcγ-RII THP1 cell surface detection by flow cytometry
CD64 – Fcγ-RI
CD32 – Fcγ-RII
Targeted-ADC (Fc-wt)
Targeted-ADC-AAG (Fc-mutant)
Non-binding Control-ADC
Free Payload
No Treatment
FITC-conjugated Fc-mutant ADC does not bind to THP1 cells (data not shown)

- hIgG1 binding to Fcγ-RI does not block binding of anti-CD64 (Fcγ-RI) antibody as determined by a Bio-Layer Interferometry competition assay
- Fcγ-RIII (CD16) is not expressed on THP1 cells
- Similar results with PBMCs

Figure 3. Antigen-bound tumor cell-targeted ADCs are internalized into myeloid cells and lead to significant reduction in Fcγ-RI cell surface detection levels. (A) Depiction of flow cytometry-based ADC internalization assay. **(B)** THP1 cells were treated with FITC-conjugated targeted-ADC with *wt* or mutant Fc, or non-binding control-ADC for 5 hours, washed and stained with PE-anti-FITC antibody (non-quenching) followed by flow cytometry analysis. Frequency of FITC+ cells were similar with Fc-*wt* targeted-ADC and control-ADC regardless of the antigen, however frequency of PE+ cells was significantly reduced only with the targeted-ADC treatment in the presence of antigen. Fc-mutant targeted-ADC (FITC-conjugated) did not bind to THP1 cells, demonstrating that tumor cell-targeted ADCs are internalized into myeloid cells in an antigen and Fc-dependent manner. **(C)** THP1 cells were treated with the indicated test articles for 7 hours +/- antigen and stained with anti-Fcγ-RI (CD64) and anti-Fcγ-RII (CD32) antibodies followed by flow cytometry analysis. Fcγ-RI but not Fcγ-RII cell surface detection levels were significantly decreased with Fc-wt targeted-ADC treatment in the presence of antigen, suggesting that Fcγ-RI is the major receptor mediating internalization of the targeted-ADCs into myeloid cells. Similar results were obtained with PBMCs.

* Populations shown were gated on single/live cells.

Tumor cell-targeted STING agonist ADC activity is impaired in Fcγ-RI knock out THP1 cell / cancer cell co-cultures

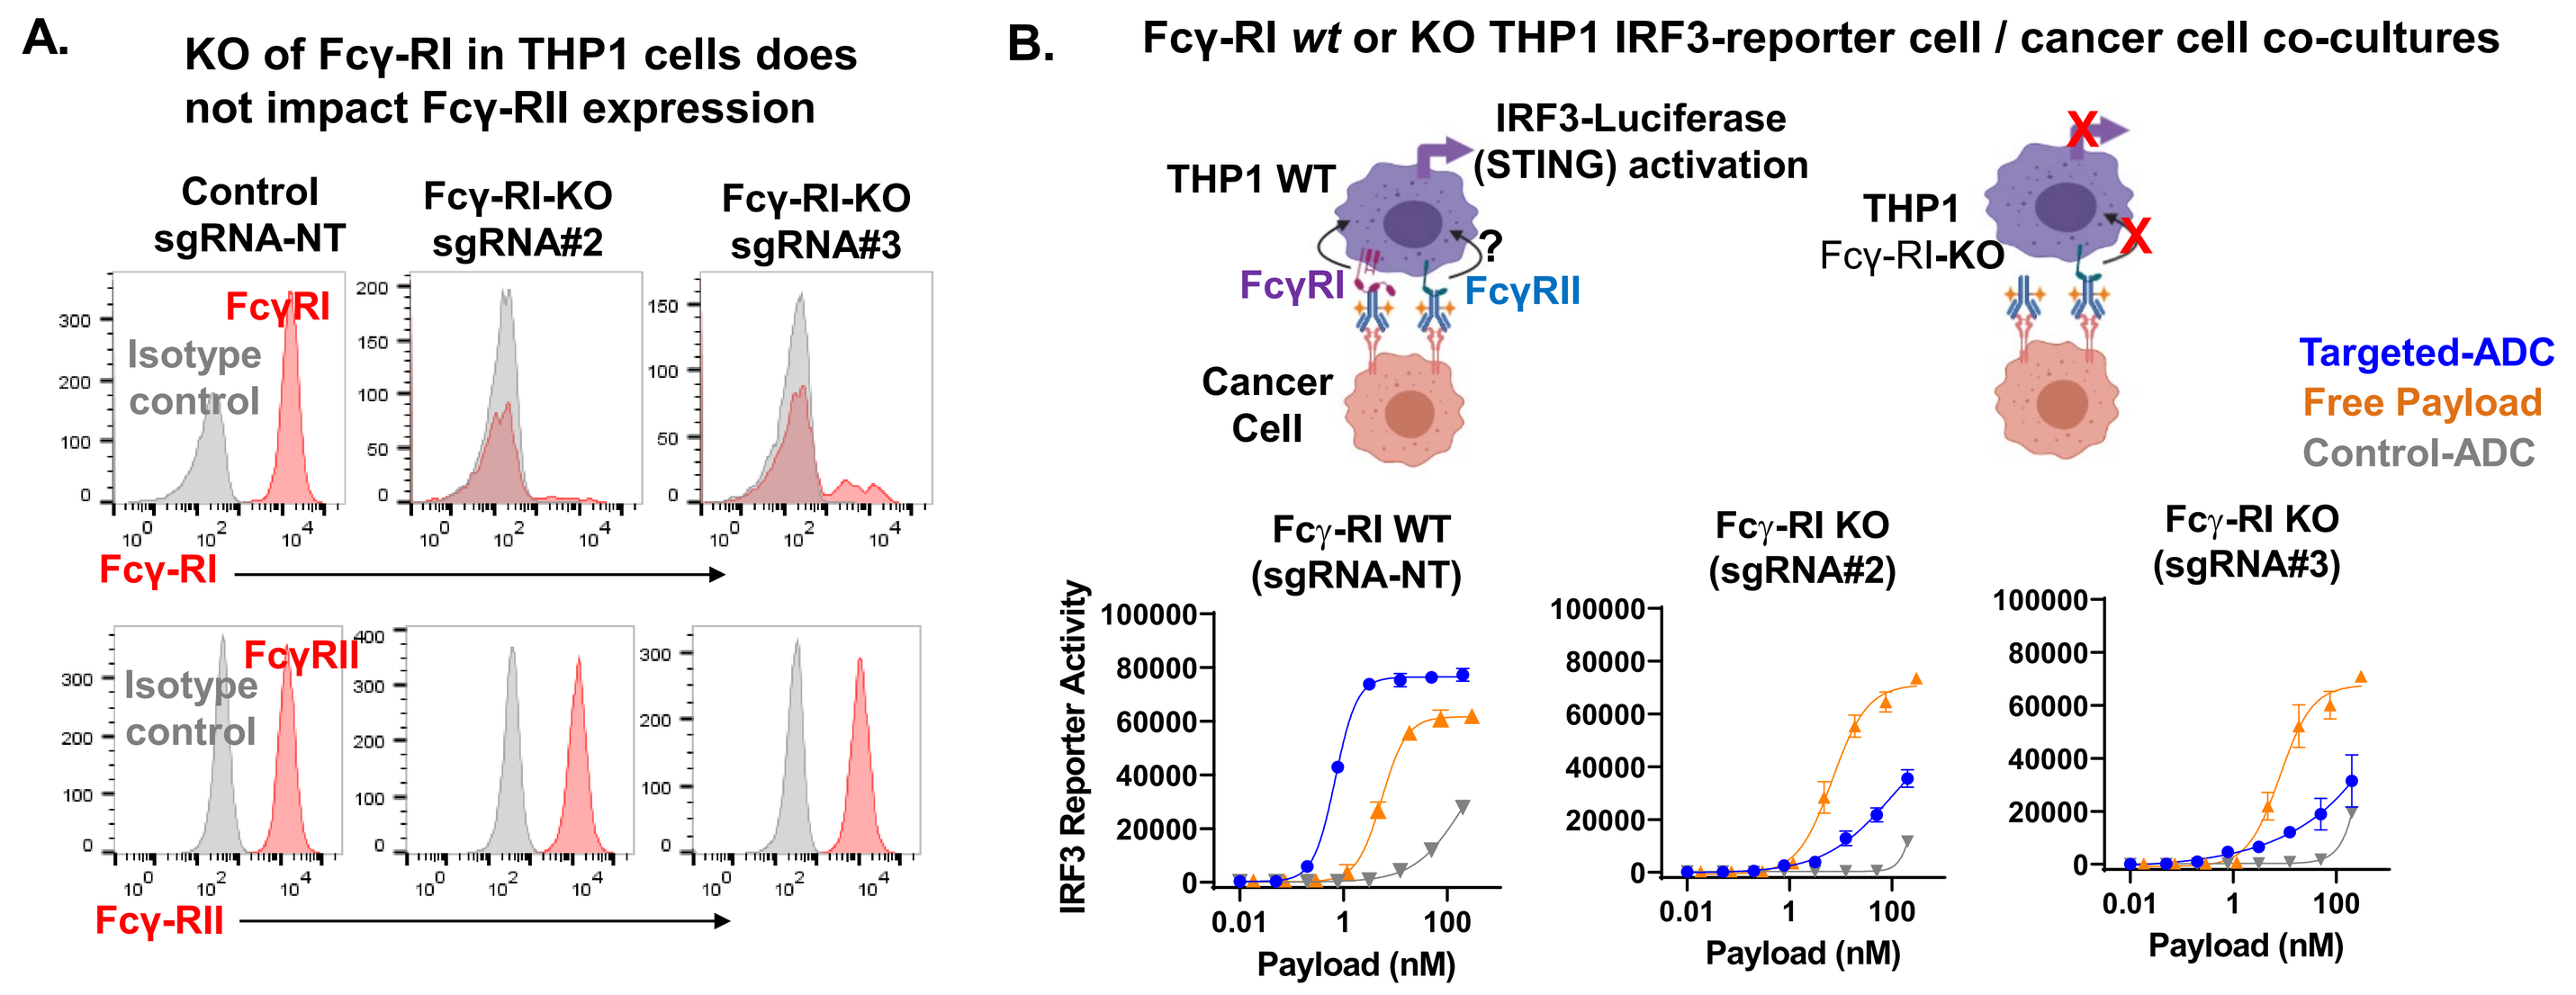
A.
KO of Fcγ-RI in THP1 cells does not impact Fcγ-RII expression
Control sgRNA-NT
Fcγ-RI-KO sgRNA#2
Fcγ-RI-KO sgRNA#3
Isotype control
FcγRI
FcγRII
Fcγ-RI
Fcγ-RII
B.
Fcγ-RI wt or KO THP1 IRF3-reporter cell / cancer cell co-cultures
THP1 WT
IRF3-Luciferase (STING) activation
THP1 Fcγ-RI-KO
Cancer Cell
Targeted-ADC
Free Payload
Control-ADC
Fcγ-RI WT (sgRNA-NT)
Fcγ-RI KO (sgRNA#2)
Fcγ-RI KO (sgRNA#3)
IRF3 Reporter Activity
Payload (nM)

Figure 4. Tumor cell-targeted STING-agonist ADC activity is significantly impaired in Fcγ-RI KO THP1 cell and cancer cell co-cultures despite intact Fcγ-RII expression. (A) Flow cytometry analysis of Fcγ-RI (CD64) and Fcγ-RII (CD32) expression in THP1 *wt* and Fcγ-RI (CD64) KO cells. THP1 Fcγ-RI KO cells were generated using CRISPR/CAS9 and the Fcγ-RI negative population was sorted and further expanded for analysis. **(B)** IRF3-Luciferase reporter assay. Cancer cells were co-cultured with THP1 *wt* or Fcγ-RI KO cells in the presence of the indicated test articles for 24 hours and luciferase activity was measured. Targeted ADC induced robust luciferase activity only in the THP1 wt cells, indicating that Fcγ-RI plays an important role in the internalization of the tumor cell-targeted STING-agonist ADCs into myeloid cells.

Delivery of STING agonist specifically into the Fcγ-RI-expressing subpopulation of myeloid cells via a tumor cell-targeted ADC elicits potent anti-tumor activity

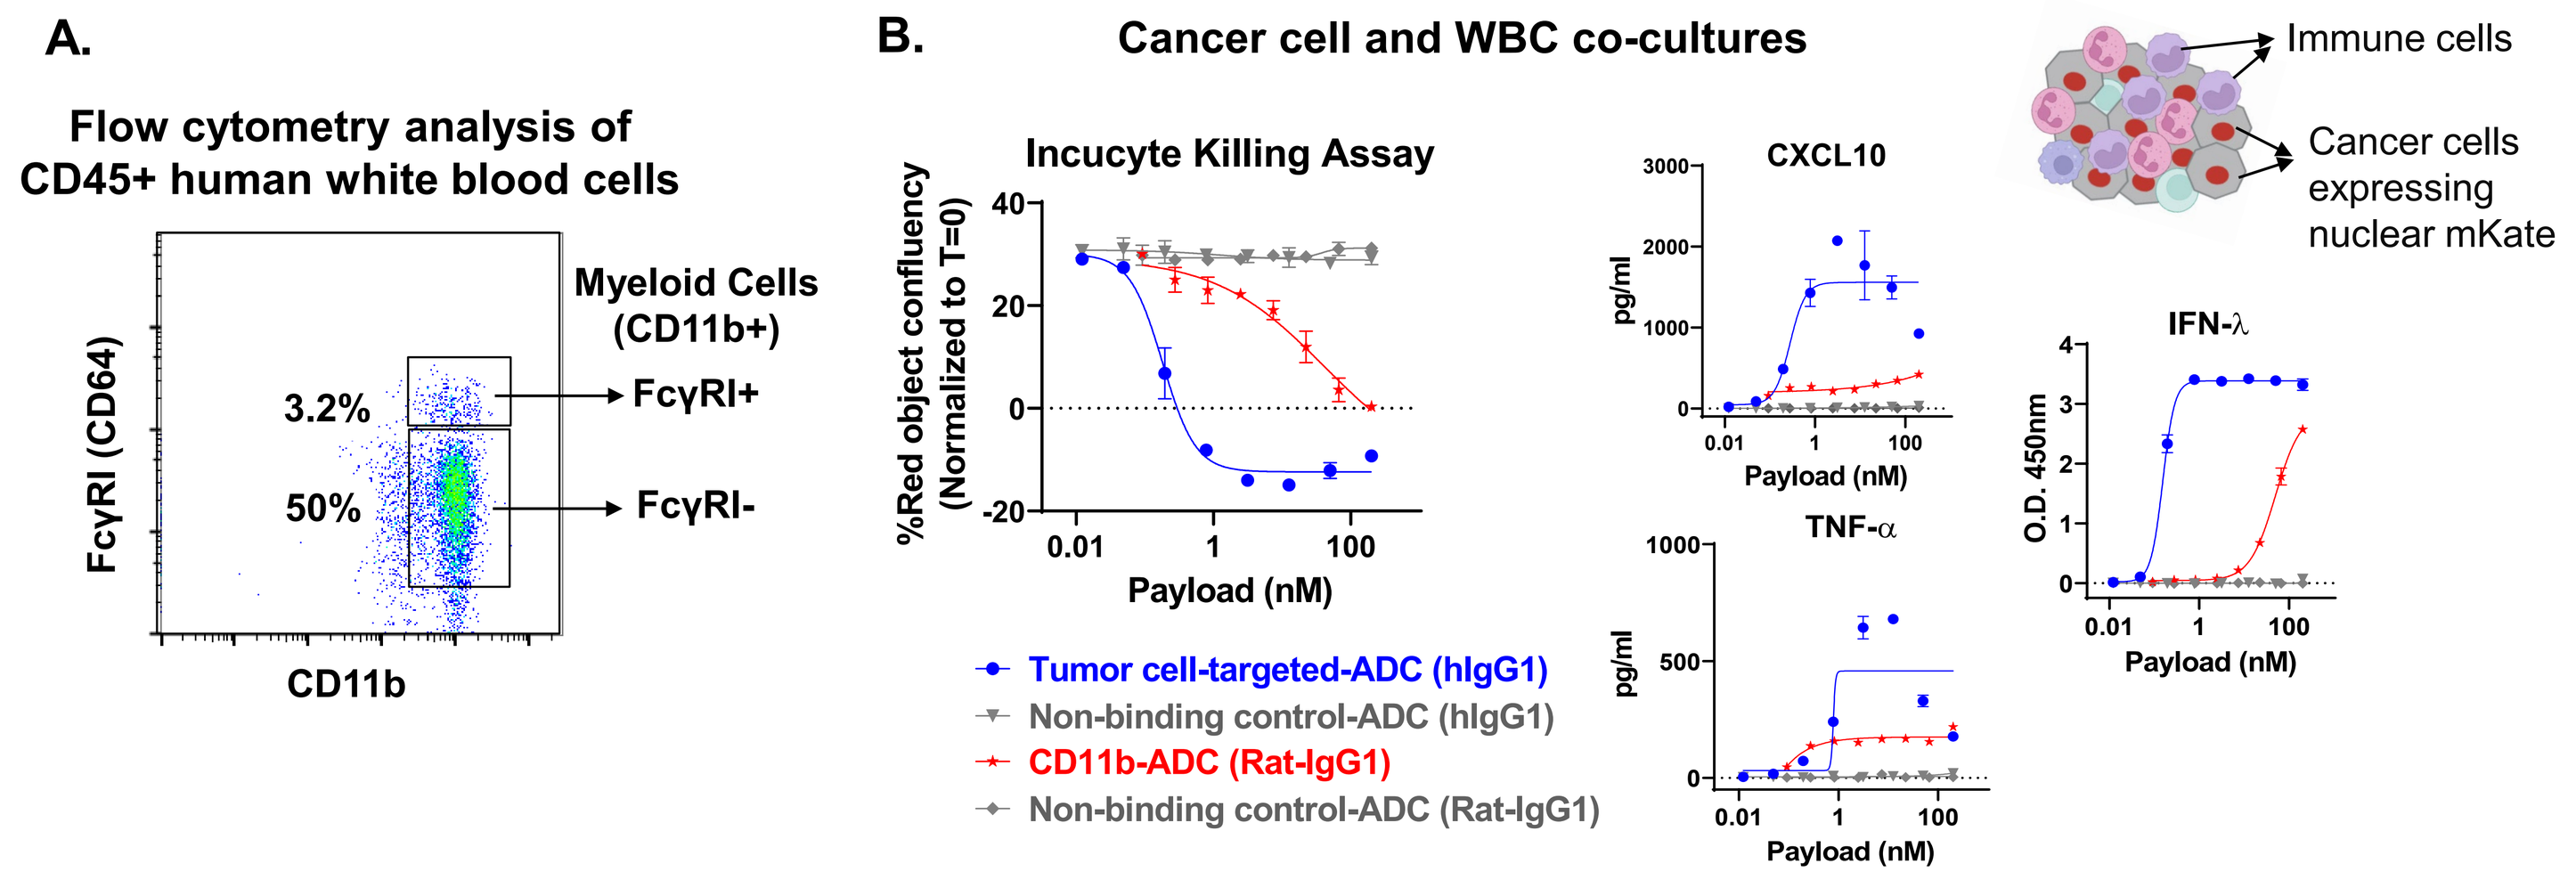
A.
Flow cytometry analysis of CD45+ human white blood cells
Myeloid Cells (CD11b+)
3.2%
FcγRI+
50%
FcγRI-
FcγRI (CD64)
CD11b
B.
Cancer cell and WBC co-cultures
Immune cells
Cancer cells expressing nuclear mKate
Incucyte Killing Assay
%Red object confluency (Normalized to T=0)
Payload (nM)
Tumor cell-targeted-ADC (hIgG1)
Non-binding control-ADC (hIgG1)
CD11b-ADC (Rat-IgG1)
Non-binding control-ADC (Rat-IgG1)
CXCL10
TNF-α
IFN-λ
pg/ml
O.D. 450nm

Figure 5. Tumor cell-targeted STING-agonist ADCs exhibit superior cytokine induction and cancer cell killing activity in cancer cell and white blood cell (WBC) co-cultures compared to CD11b-targeted ADC, which delivers STING agonist into all myeloid cells. (A) Flow cytometry analysis of Fcγ-RI (CD64) and CD11b expression on fresh human WBCs. **(B)** Cancer cells expressing nuclear-restricted mKate protein were co-cultured with WBCs in the presence of tumor cell-targeted or CD11b-targeted ADCs, as well as control-ADCs followed by IncuCyte analysis. %Red object confluency at 84 hours was normalized to T=0. Supernatants (24 hours) were collected from sister plates for CXCL10 and TNF-α analysis by Luminex assay, and IFN-λ1/ λ3 analysis by an ELISA assay. Similar results obtained with WBCs from multiple donors.

Myeloid cells within dissociated tumor cells express Fcγ-RI and can induce killing of cancer cells with tumor cell-targeted STING agonist ADC treatment

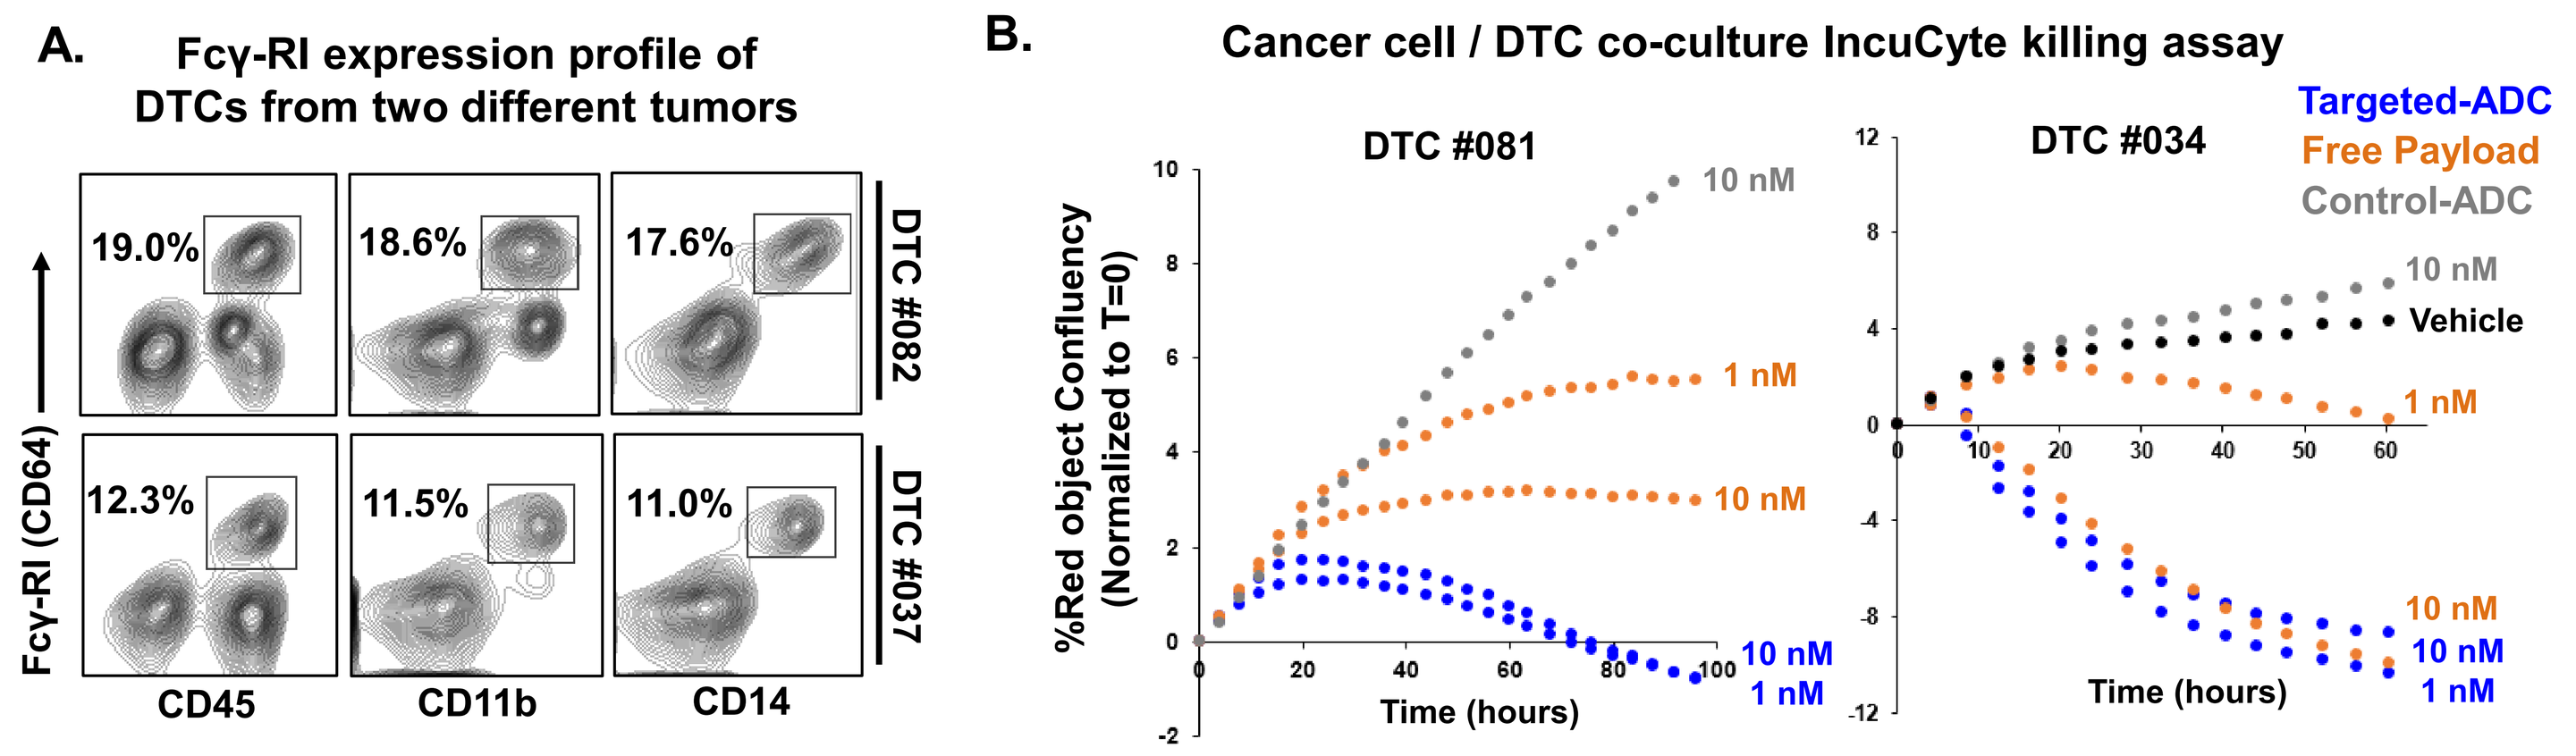
A.
Fcγ-RI expression profile of DTCs from two different tumors
19.0%
18.6%
17.6%
DTC #082
12.3%
11.5%
11.0%
DTC #037
Fcγ-RI (CD64)
CD45
CD11b
CD14
B.
Cancer cell / DTC co-culture IncuCyte killing assay
DTC #081
DTC #034
Targeted-ADC
Free Payload
Control-ADC
Vehicle
%Red object Confluency (Normalized to T=0)
Time (hours)

Figure 6. Dissociated tumor cells (DTCs) express Fcγ-RI and induce killing of cancer cells in co-cultures. (A) Flow cytometry analysis of Fcγ-RI expression within DTCs. **(B)** Frozen DTCs from two different tumors were thawed and co-cultured with cancer cells (expressing nuclear-restricted mKate protein) and treated with the indicated test articles (ADC concentrations shown are based on payload). Plates were scanned in an IncuCyte instrument and the % red object (cancer cell) confluency was plotted against time. Tumor cell-targeted ADCs induced robust killing of cancer cells in co-cultures of DTCs from both donors tested.

Tumor cell-targeted STING agonist ADCs induce STING pathway in fresh human tumor fragments ex-vivo

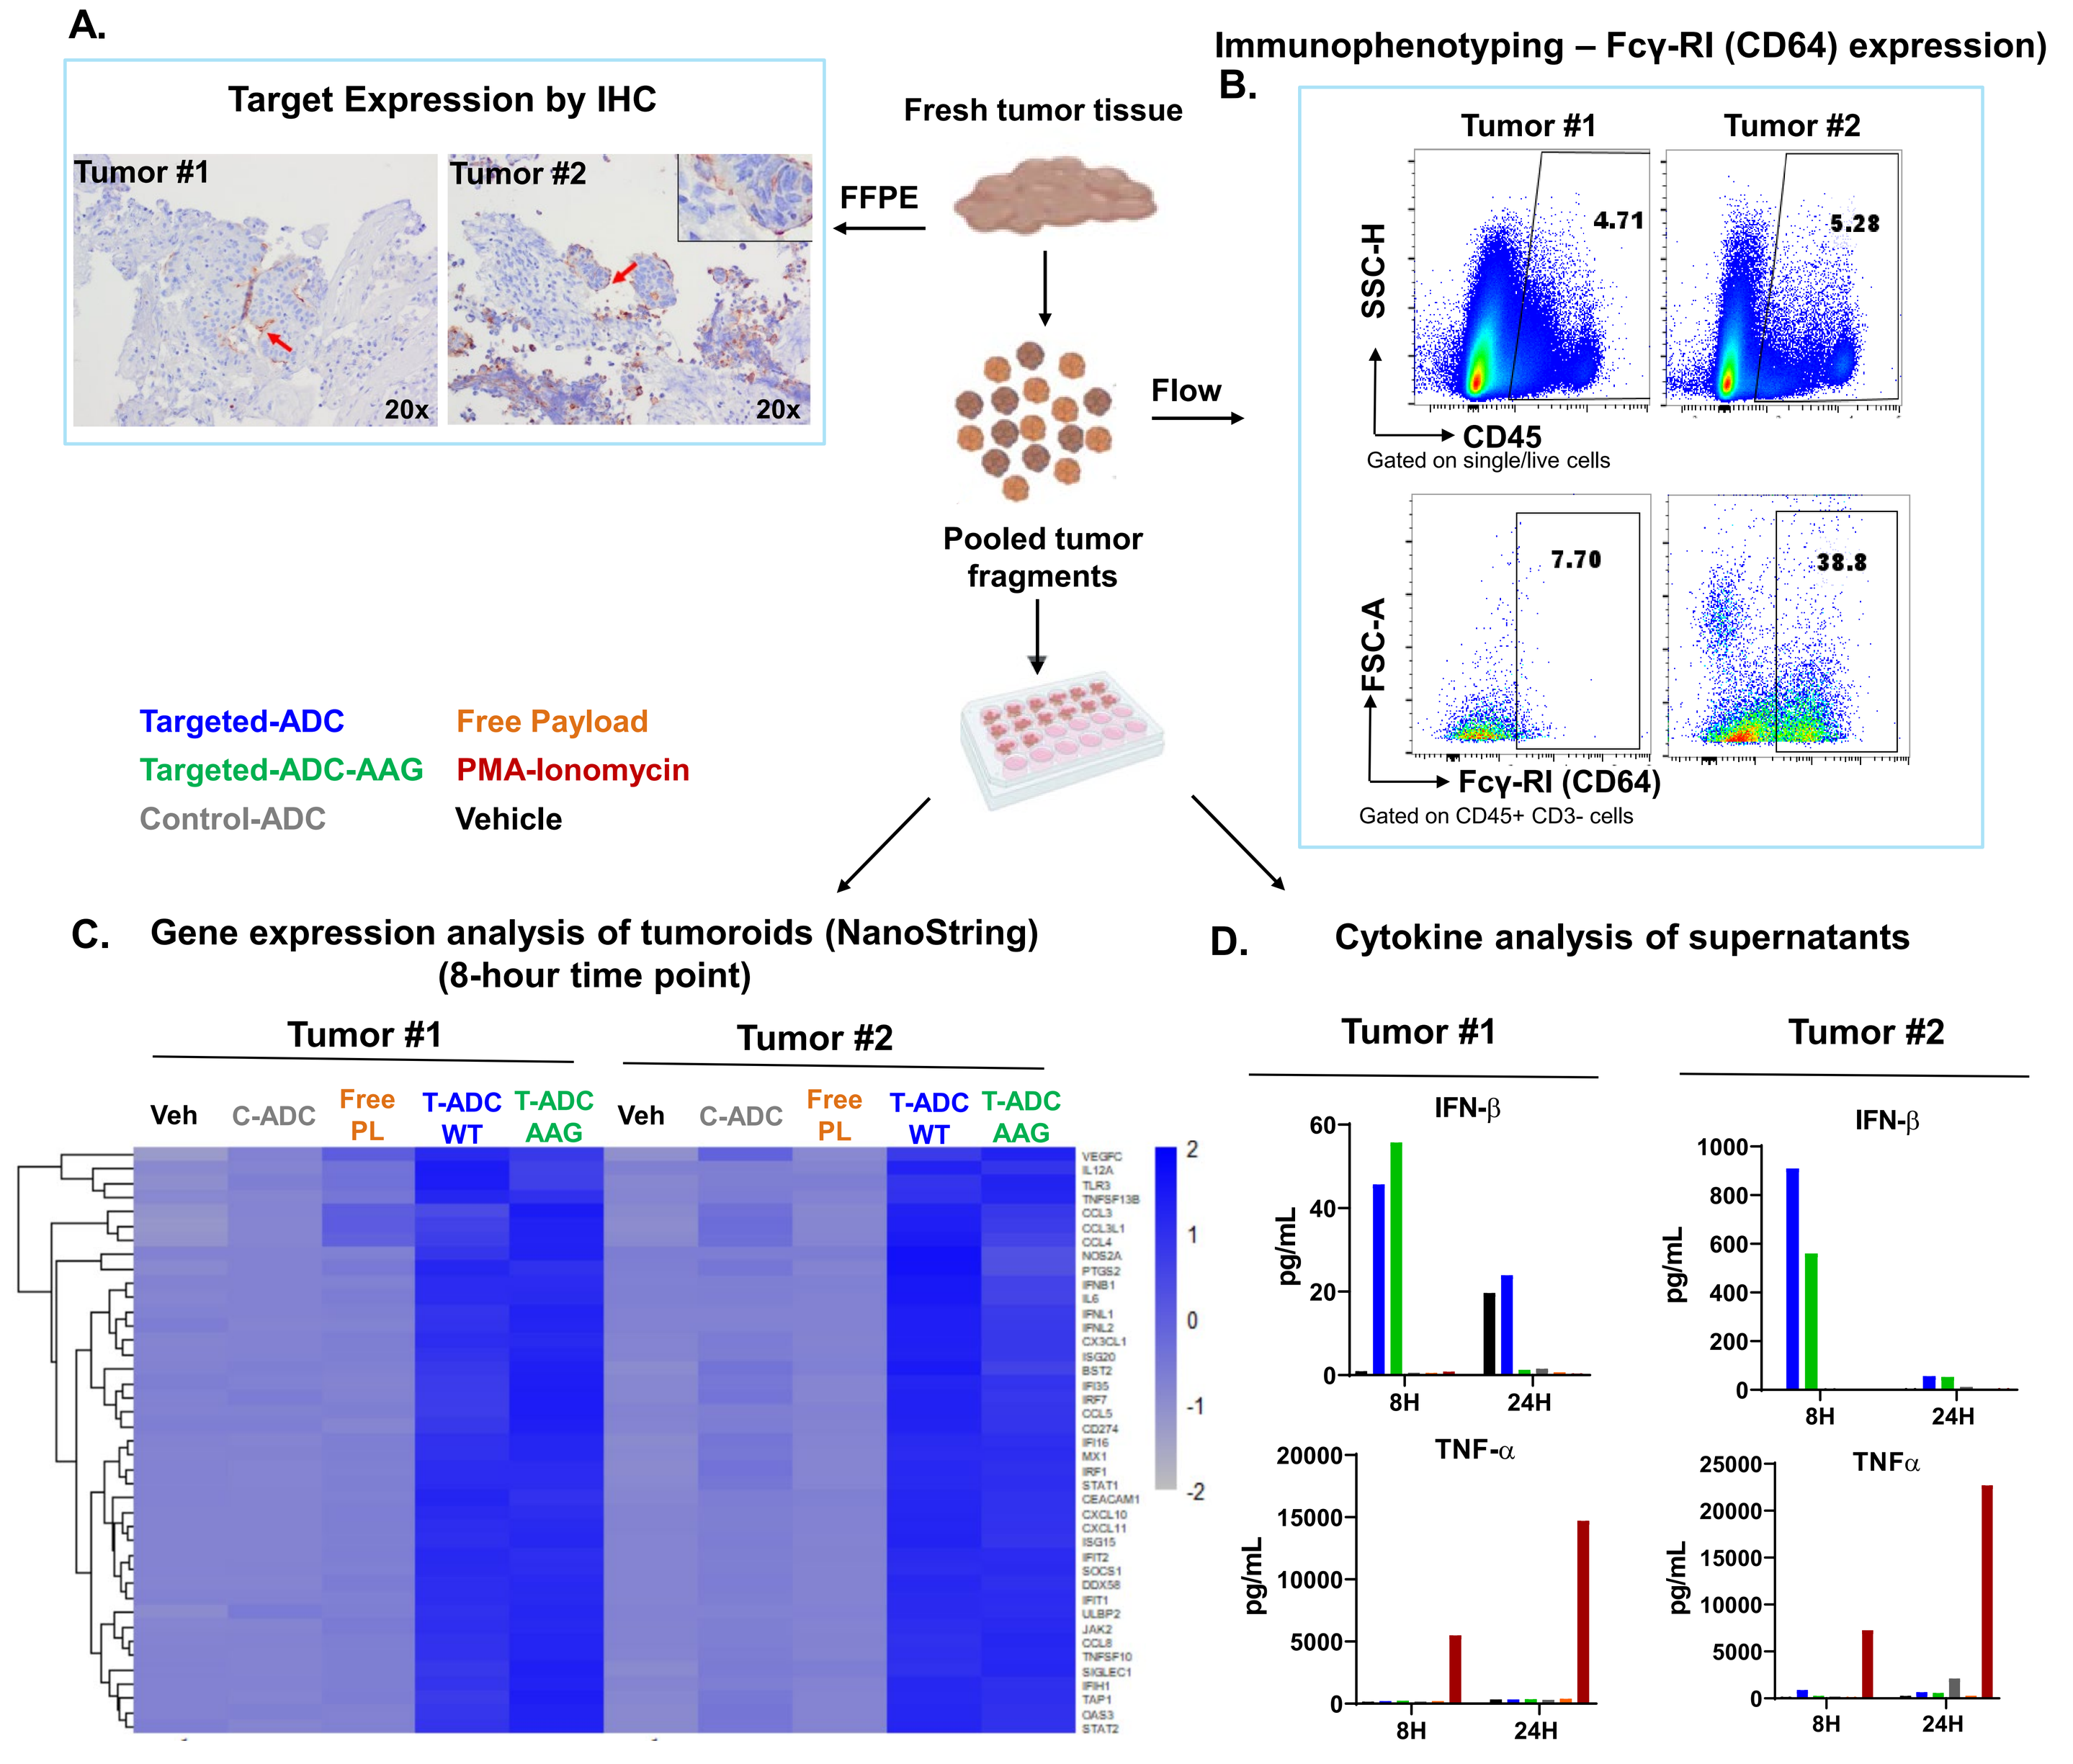
A.
Target Expression by IHC
Tumor #1
Tumor #2
Fresh tumor tissue
FFPE
Flow
Pooled tumor fragments
Targeted-ADC
Targeted-ADC-AAG
Control-ADC
Free Payload
PMA-Ionomycin
Vehicle
B.
Immunophenotyping – Fcγ-RI (CD64) expression)
SSC-H
CD45
Gated on single/live cells
FSC-A
Fcγ-RI (CD64)
Gated on CD45+ CD3- cells
C.
Gene expression analysis of tumoroids (NanoString) (8-hour time point)
Veh
C-ADC
Free PL
T-ADC WT
T-ADC AAG
D.
Cytokine analysis of supernatants
IFN-β
TNF-α
pg/mL

Figure 7. Tumor cell-targeted STING agonist ADCs with *wt* or mutant Fc induce STING pathway in fresh human tumor fragment cultures ex-vivo. Fresh human tumor tissue were collected and portions of them were processed into FFPE to determine target expression by immunohistochemistry staining **(A)**. The tissue was mechanically processed without dissociation and the fragments were pooled to preserve heterogeneity. An aliquot of the pooled fragments was dissociated and stained for immunophenotyping and Fcγ-RI expression by flow cytometry **(B)**. Rest of the fragments were evenly distributed to 96 well plates and treated with the indicated test articles for gene expression analysis by NanoString **(C)** or cytokine analysis by Luminex **(D)** assays. PMA-Ionomycin treatment was included as a control for immune cell activation. Tumor fragmentation, immunophenotyping, and treatment of tumoroid cultures was performed by Nilogen Oncosystems. Both *wt* and Fc-Mutant targeted-ADCs induced similar gene and cytokine induction profiles (tumor intrinsic STING activation). Targeted-ADCs induced type I interferons but not TNF-α in these cultures. Better cytokine induction was observed in Tumor #2 with higher frequency of Fcγ-RI+ cells compared to Tumor#1; however, a correlation cannot be made due to very small sample numbers.

CONCLUSIONS

- Anti-tumor activity of tumor cell-targeted STING agonist ADCs involves activation of STING pathway in both myeloid cells and tumor cells.
- In this study we have demonstrated:
 - Fcγ-RI is the major receptor that mediates internalization of antigen-bound tumor-cell targeted ADCs into myeloid cells in vitro.
 - Delivery of STING agonist specifically into Fcγ-RI-expressing subpopulation of myeloid cells via a tumor cell-targeted ADC induces STING pathway more effectively compared to delivery into all myeloid cells.
 - Fcγ-RI is expressed on myeloid cells in primary human tumor samples.
 - Tumor cell-targeted STING-agonist ADCs activate STING pathway in primary human tumors ex-vivo.

By delivering STING agonist into tumor cells and Fcγ-RI-expressing myeloid cells, tumor cell-targeted STING agonist ADCs induce the STING pathway effectively and therefore may offer a therapeutic advantage

References:

1. Malli Cetinbas et al. SITC 2020 https://www.mersana.com/publications/
2. Malli Cetinbas et al. AACR 2021 https://www.mersana.com/publications/
3. Cartoons were generated using Biorender.