Results From the Phase 1 Dose Escalation Study of XMT-1592, a NaPi2b-Directed Dolasynthen Antibody-Drug Conjugate (ADC)

Erika P. Hamilton[1]; Kyriakos P. Papadopoulos[2]; Minal A. Barve[3]; Nehal J. Lakhani[4]; Timothy F. Burns[5]; Robert A. Burger[6]; **Adi Reske**[6]; Kelley Wekheye[6]; Anthony Tolcher[7]

[1]Sarah Cannon Research Institute, Nashville, TN, United States; [2]START San Antonio, San Antonio, TX, United States; [3]Mary Crowley Cancer Research Center, Dallas, TX, United States; [4]START Midwest, Grand Rapids, MI, United States; [5]University of Pittsburgh Medical Center (UPMC) Hillman Cancer Center, Pittsburgh, PA, United States; [6]Mersana Therapeutics Inc, Cambridge, MA, United States; [7]Next Oncology, San Antonio, TX, United States

INTRODUCTION

- ADCs have provided substantial benefits to patients; nonetheless, observed platform toxicities and payload resistance limit their use
- NaPi2b is a sodium-dependent phosphate transporter broadly expressed in ovarian cancer (OC), non-small cell lung cancer (NSCLC) and in healthy Type II pneumocytes in the lung, but with limited expression in other healthy tissue (**Table 1**)[1]
- ADCs targeting NaPi2b have demonstrated clinical activity in platinum-resistant OC, but have been associated with both target-related toxicities (ILD/pneumonitis) and platform-related toxicities, including AST increase, thrombocytopenia/platelet count decrease (observed with XMT-1536)[2], and neutropenia and neuropathy (observed with lifatuzumab vedotin)[3]
- Dolasynthen is a next-generation platform designed to produce homogeneous ADCs with precise control of the drug-to-antibody ratio and site-specific bioconjugation with the goal of improving efficacy and reducing platform-related toxicities[4]
 - Improvements in PK, in vivo efficacy and tolerability were observed preclinically with XMT-1592 when compared with an earlier NaPi2b-targeted ADC, XMT-1536 (UpRi; data on file)
- Here we report Phase 1 dose escalation data for XMT-1592, a Dolasynthen NaPi2b-directed ADC in patients with OC and NSCLC

NaPi2b Highly Expressed in Normal Lung and Ovarian Cancer Tissues[1]

Type of tissue	IHC Score[a]			
	0	1	2	3
Normal breast (n=4)	0	0	1	3
Breast cancer (n=10)	8	2	0	0
Normal lung (n=9)	0	0	1	8
Lung cancer (n=11)	5	5	0	1
Normal uterus (n=3)	0	0	1	2
Normal oviduct (n=2)	0	0	0	2
Normal ovary (n=5)	5	0	0	0
Ovarian cancer (n=10)	0	1	1	8

Table 1: Semi-Quantitative Scoring of IHC Staining for NaPi2b (MX35) in a Panel of Normal and Cancer Tissues

- NaPi2b is highly expressed in normal lung tissue at a cumulative IHC score comparable to that for OCs[1]
- Type II pneumocytes in the lungs may be proliferating and thus susceptible to a microtubule inhibitor

Leveraging Learnings to Develop an Improved Cytotoxic ADC Platform

Dolaflexin
(First-generation cytotoxic ADC platform)

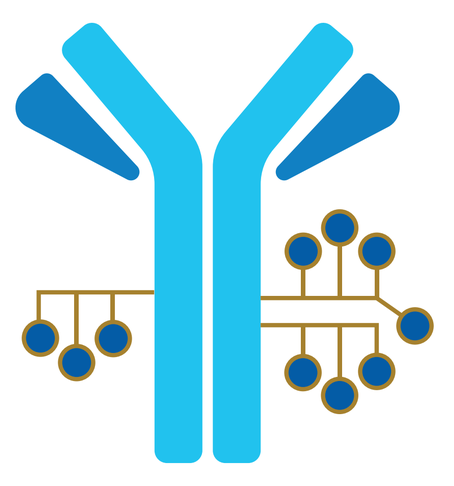

- AF-HPA payload with controlled bystander effect; highly potent anti-tubulin inhibitor selectively toxic to rapidly dividing cells designed to avoid dose-limiting neuropathy or neutropenia
- Stochastic (random) bioconjugation
- Heterogeneous DAR ~10
- Fully preserved Fcγ receptor interaction; could lead to non-target uptake

Goals for Next-Generation Platform

- DAR customization for target
- Antibody-like PK
- Enhance tumor payload delivery
- Increase efficacy
- Reduce platform toxicity
- Expand therapeutic index

Dolasynthen
(Next-generation cytotoxic ADC platform)

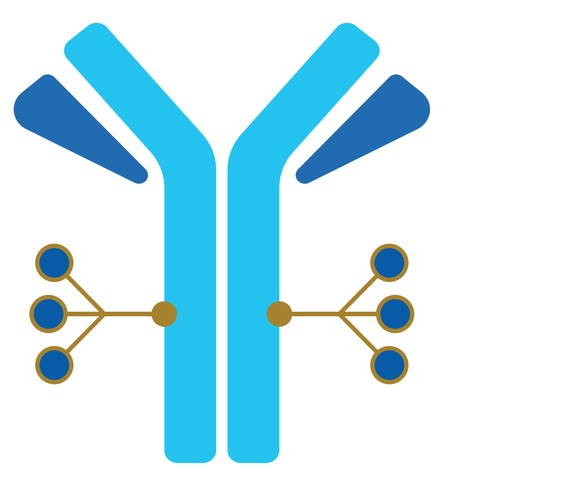

- AF-HPA payload with controlled bystander effect; highly potent anti-tubulin inhibitor selectively toxic to rapidly dividing cells designed to avoid dose-limiting neuropathy or neutropenia
- Site-specific conjugation
- Homogeneous DAR 6
- Significantly reduced Fcγ binding

Preclinical Studies: Dolasynthen Outperforms Dolaflexin at Equal Payload Doses

- Dolasynthen platform enables the creation of single-species ADCs that outperform Dolaflexin's heterogeneous ADC population in preclinical models

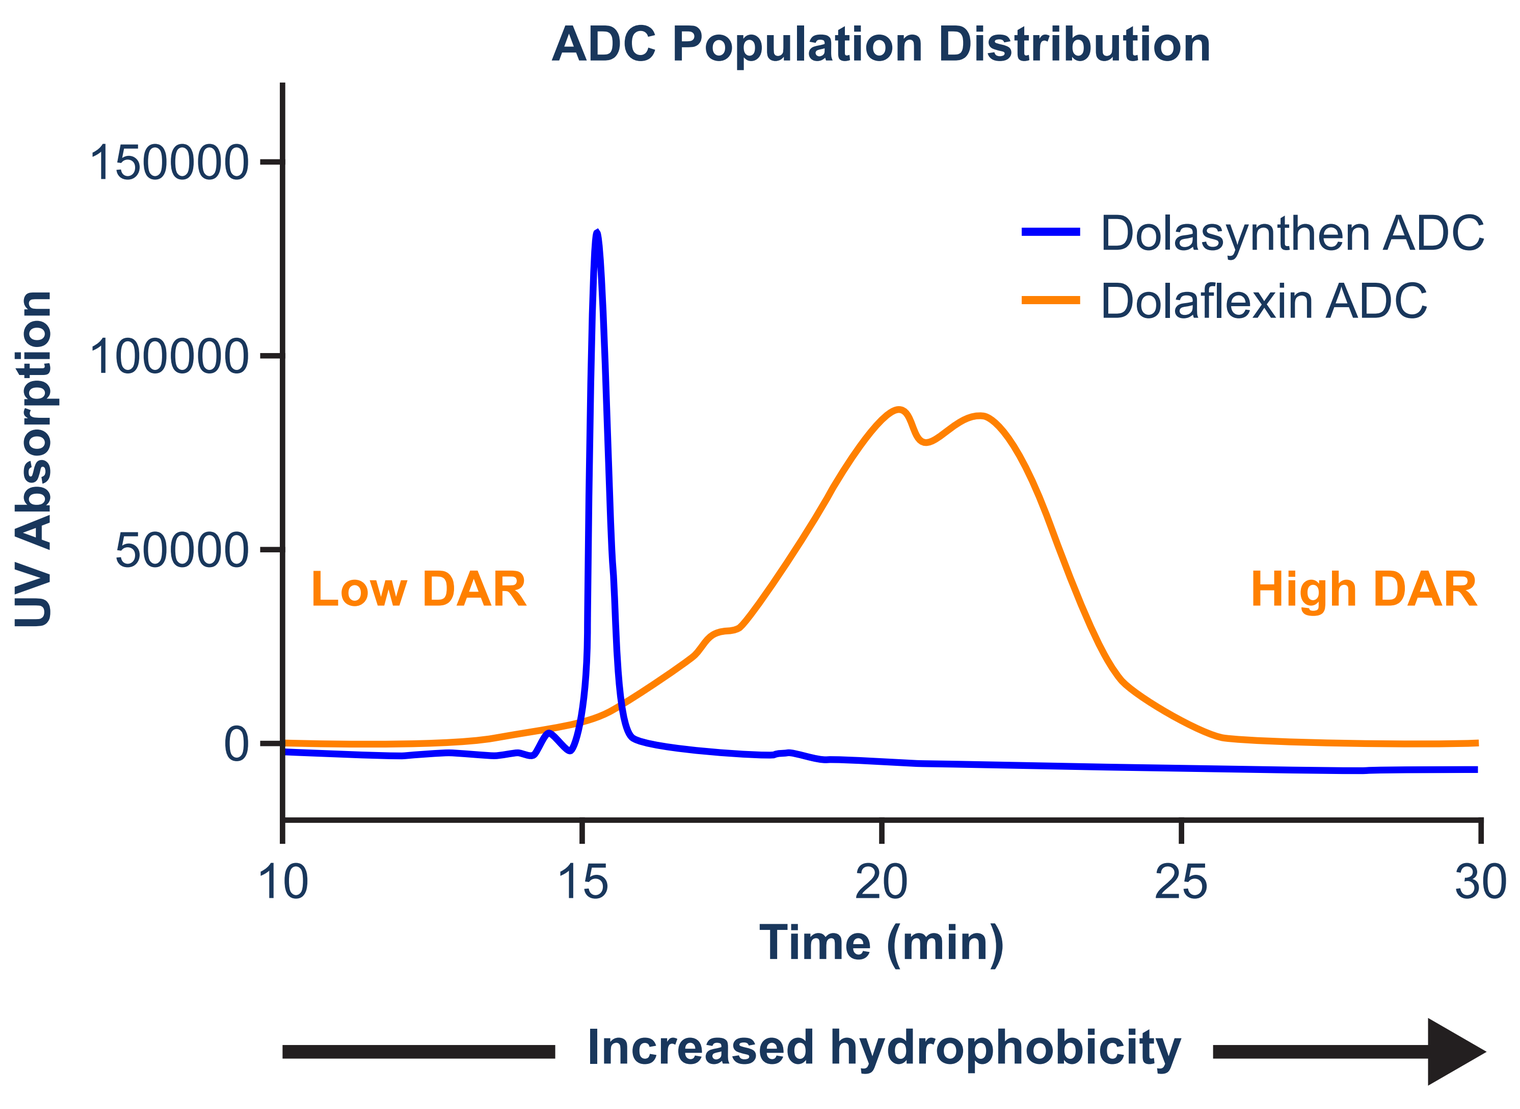
ADC Population Distribution
Dolasynthen ADC
Dolaflexin ADC
UV Absorption
150000
100000
50000
0
Low DAR
High DAR
10
15
20
25
30
Time (min)
Increased hydrophobicity

Figure 1: Heterogeneous Population vs Homogeneous Population.
As measured by hydrophobic interaction chromatography, 280 nanometers

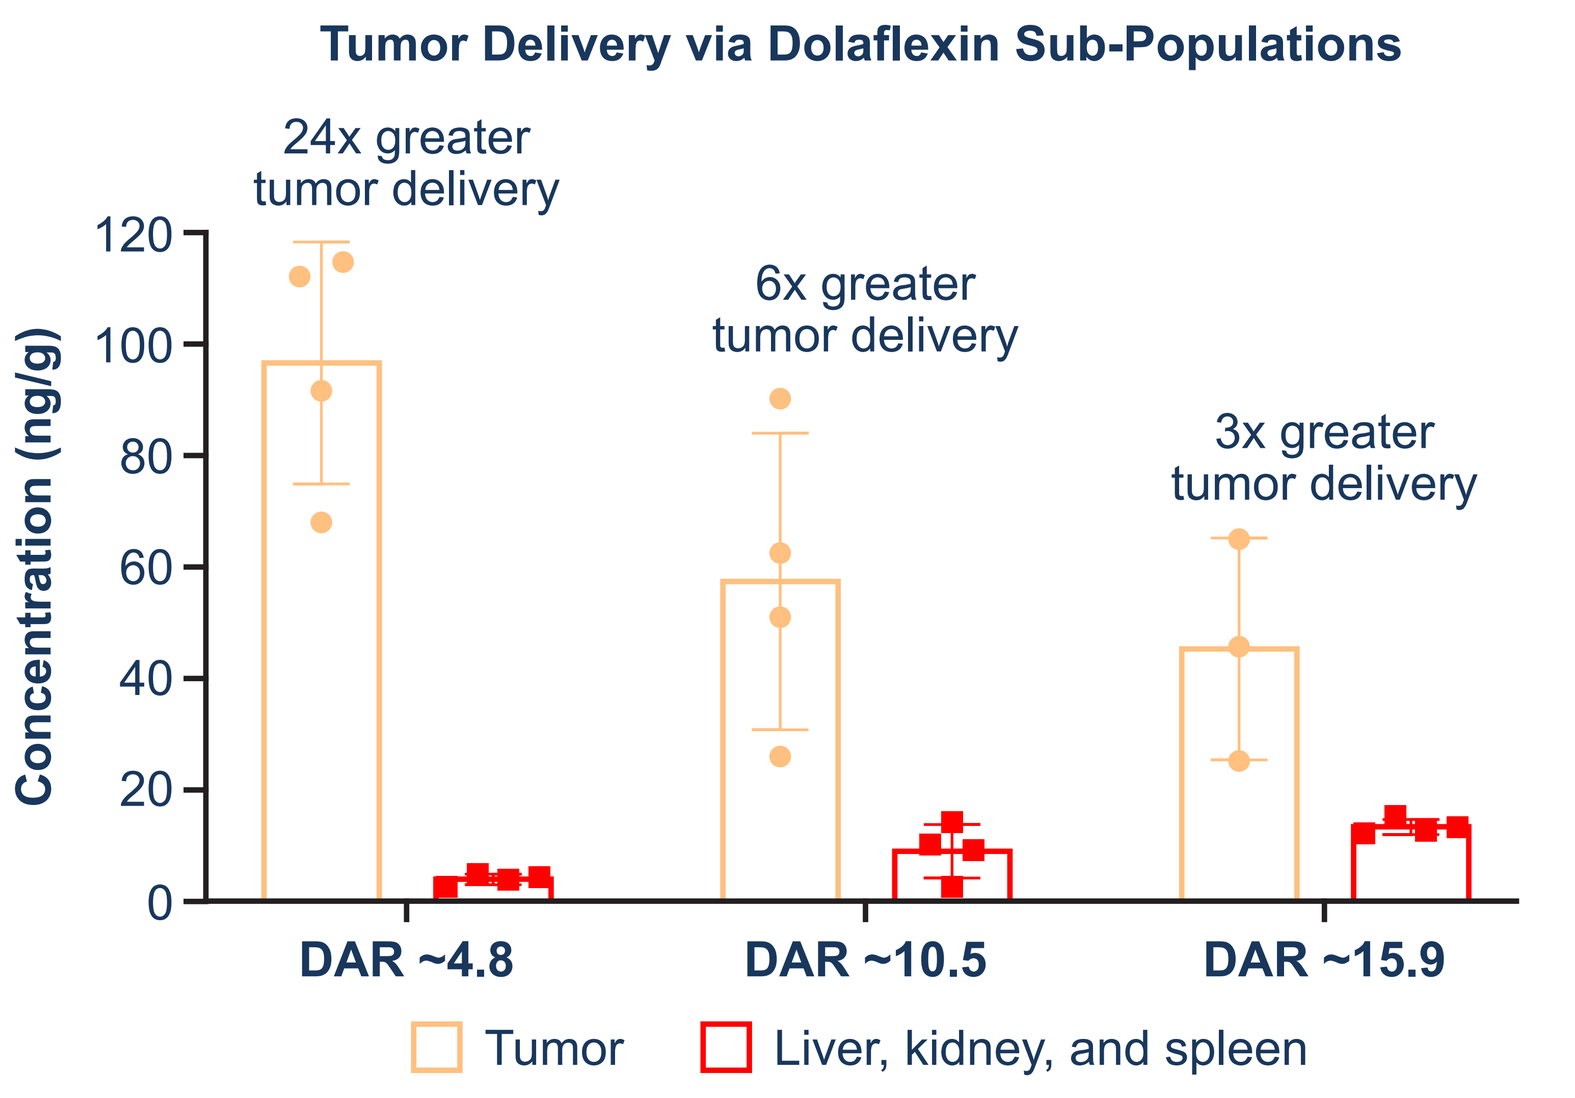
Tumor Delivery via Dolaflexin Sub-Populations
24x greater tumor delivery
6x greater tumor delivery
3x greater tumor delivery
Concentration (ng/g)
120
100
80
60
40
20
0
DAR ~4.8
DAR ~10.5
DAR ~15.9
Tumor
Liver, kidney, and spleen

Figure 2: Dolaflexin High-DAR Subpopulations Show Reduced Tumor-Specific Delivery (Total payload/DAR)

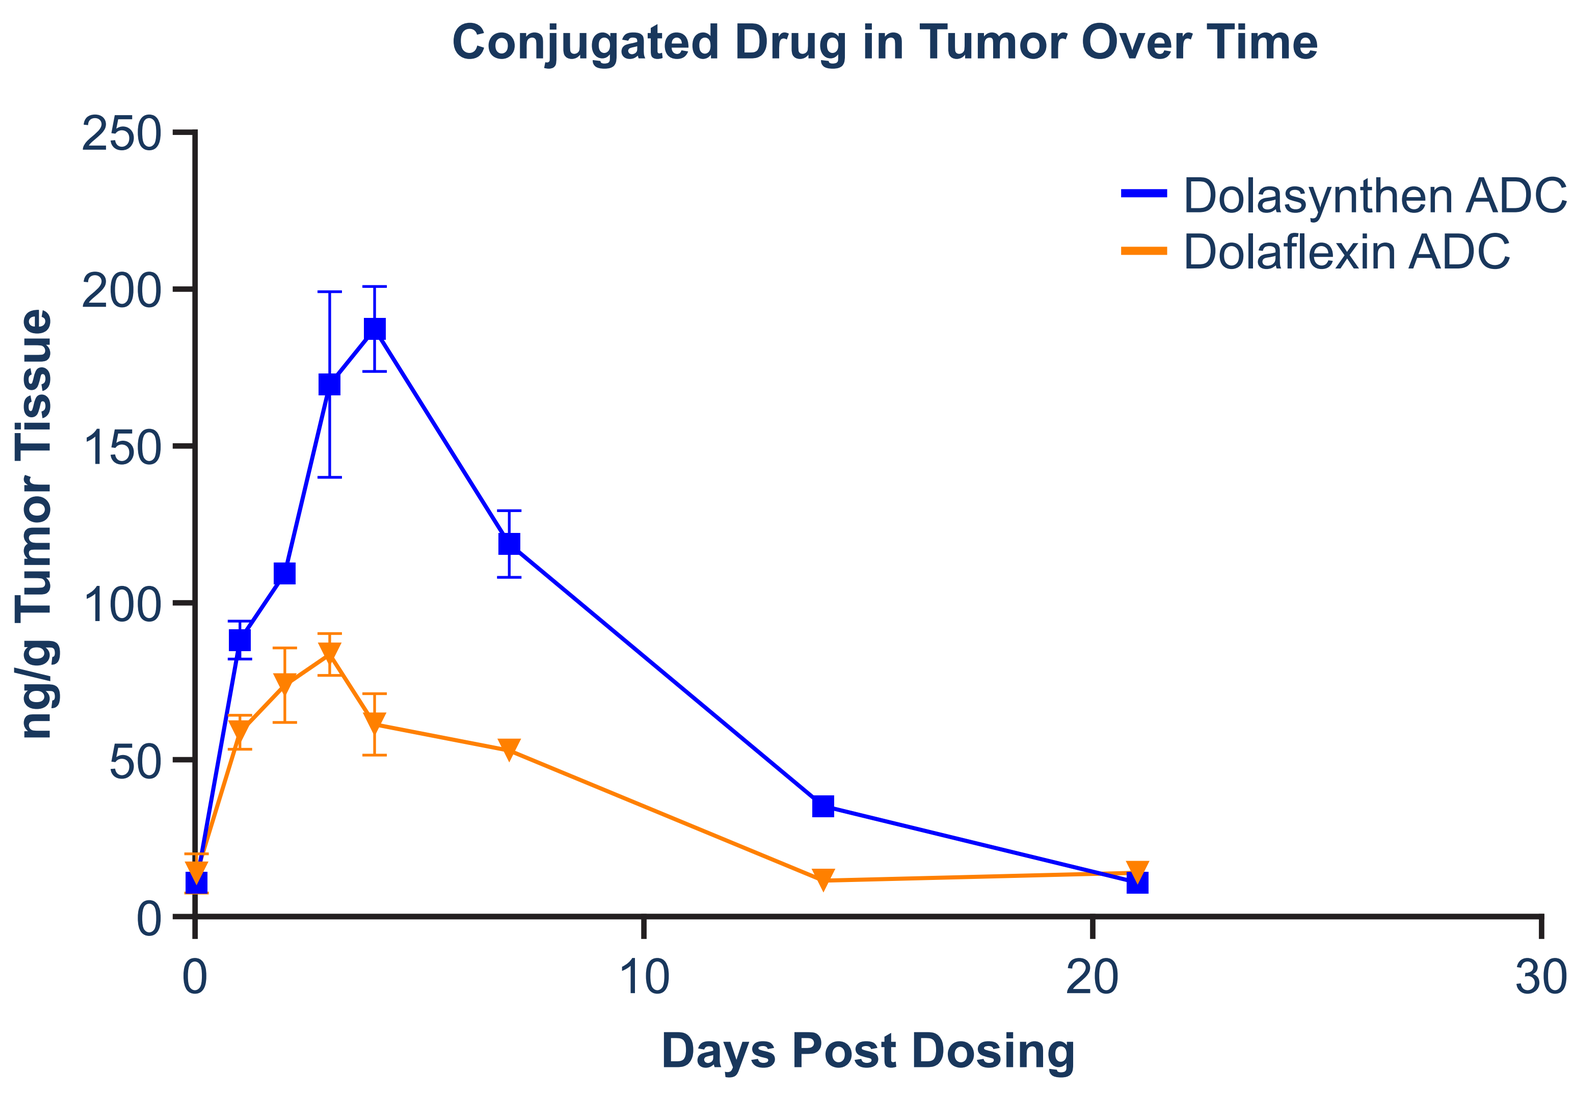
Conjugated Drug in Tumor Over Time
Dolasynthen ADC
Dolaflexin ADC
ng/g Tumor Tissue
250
200
150
100
50
0
0
10
20
30
Days Post Dosing

Figure 3: Higher Payload Exposure in Tumor With Dolasynthen
0.05 mg/kg payload (mouse)

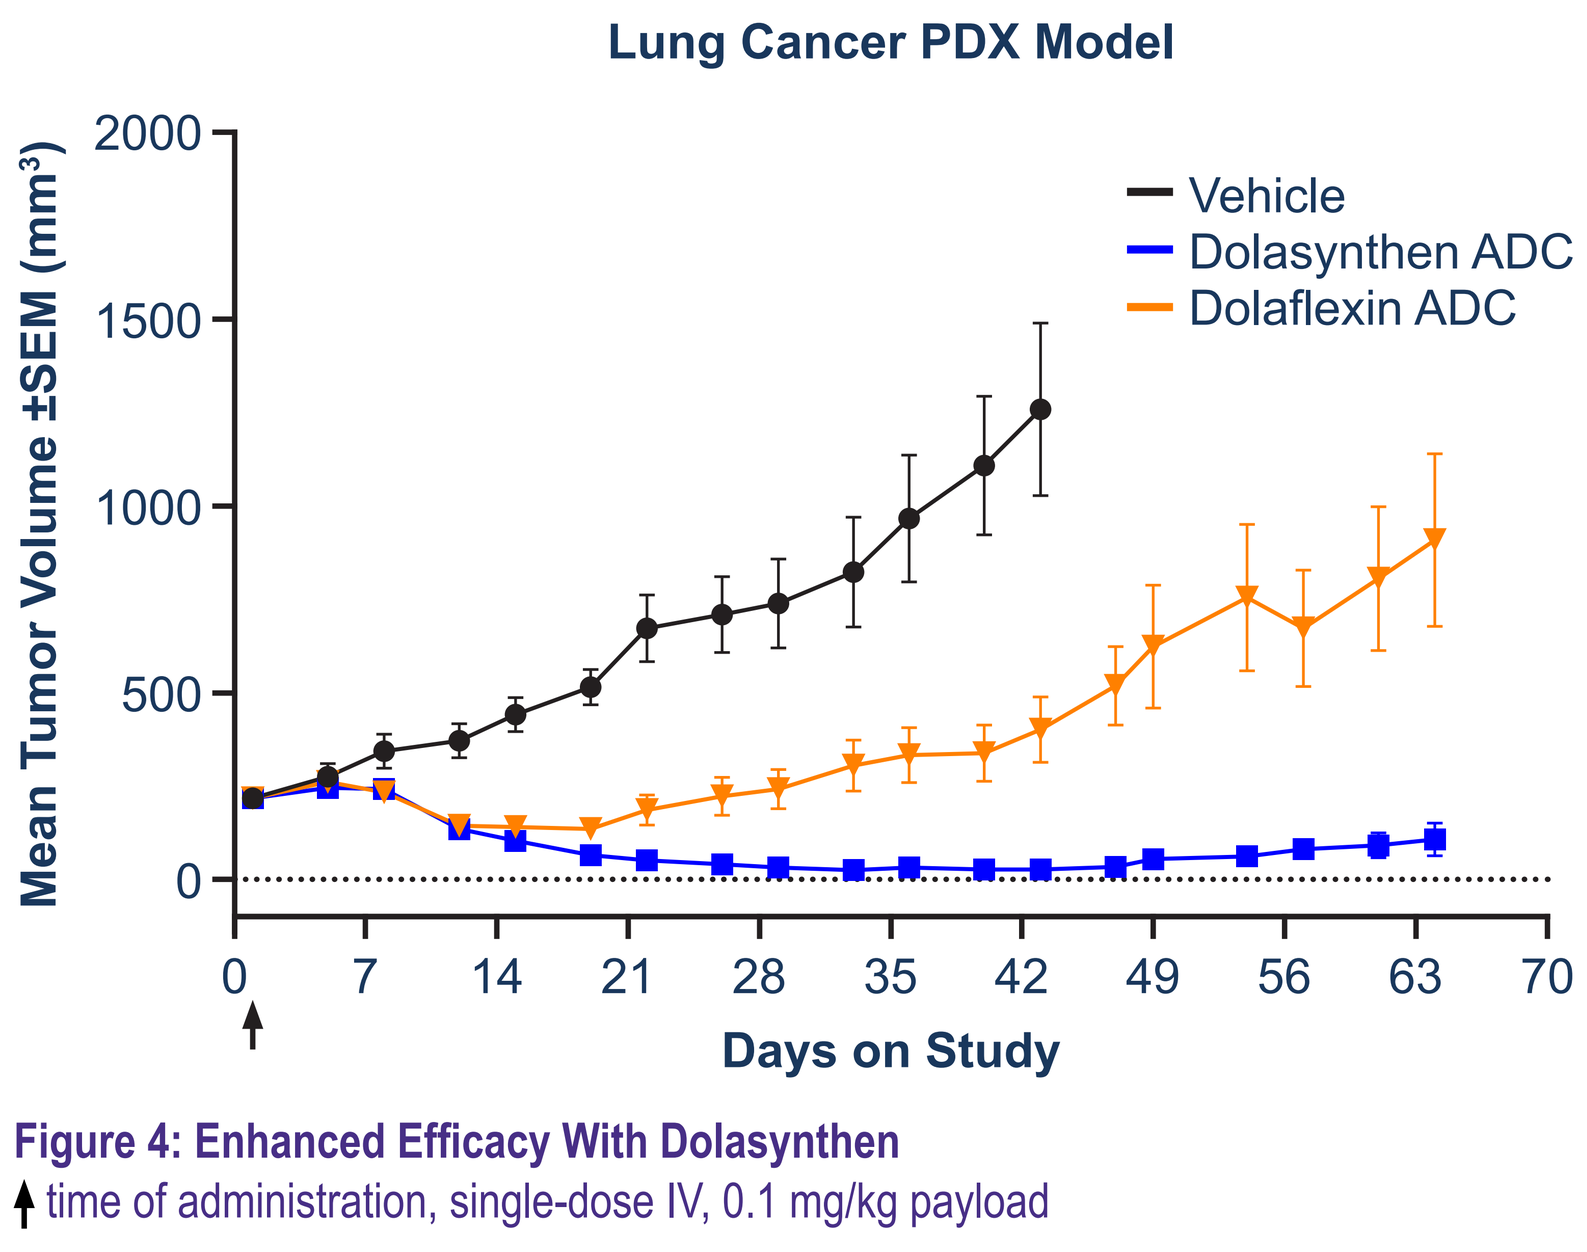
Lung Cancer PDX Model
Vehicle
Dolasynthen ADC
Dolaflexin ADC
Mean Tumor Volume ±SEM (mm³)
2000
1500
1000
500
0
0
7
14
21
28
35
42
49
56
63
70
Days on Study

Figure 4: Enhanced Efficacy With Dolasynthen
↑ time of administration, single-dose IV, 0.1 mg/kg payload

METHODS

XMT-1592 Phase 1 Dose Escalation Trial Design

- Enrolled patients with OC with histological diagnosis of high-grade serous epithelial ovarian, fallopian tube, or primary peritoneal cancers; non-squamous NSCLC
 - Measurable disease by RECIST 1.1
 - ECOG performance status 0 or 1
 - Available archival tumor tissue blocks, or freshly cut tissue slides for retrospective NaPi2b testing
- Dosing: XMT-1592 administered IV every 3 or 4 weeks
- Primary objectives: MTD or RP2D; safety and tolerability
- Escalation design: Modified version of the Simon accelerated titration design

DL1 q3w 3.5 mg/m² n=1	**DL2 q3w** 7 mg/m² n=2	**DL3 q3w** 14 mg/m² n=4	**DL3.5 q3w** 21 mg/m² n=5	**DL4 q3w** 28 mg/m² n=11	**DL5 q3w** 56 mg/m² n=4
		DL3 q4w 14 mg/m² n=4			

Figure 5: Dose Escalation Design

RESULTS *(Data cutoff: November 1, 2022)*

Patient Demographics and Disease Characteristics

Safety analysis set (N=31)	
Median age (range), years	68 (47-82)
ECOG PS, n (%)	
0	4 (12.9)
1	27 (87.1)
Primary tumor type, n (%)	
NSCLC[b]	3 (9.7)
OC	28 (90.3)
Prior lines of therapy, n (%)	
1-3	13 (41.9)
≥4	18 (58.1)
Prior therapy, n (%)	
Bevacizumab	16 (51.6)
PARP inhibitor	18 (58.1)
Platinum-free interval, n (%)	
0-3 Months	11 (35.5)
>3-6 Months	9 (29.0)
>6 Months	7 (22.6)
Unknown	4 (12.9)
NaPi2b H-score, n (%)	
Determined	25 (80.6)
High (H-score ≥110)	12 (48)
Low (H-score <110)	13 (52)
Not determined	6 (19.4)

Safety - TRAEs in ≥15% of Patients

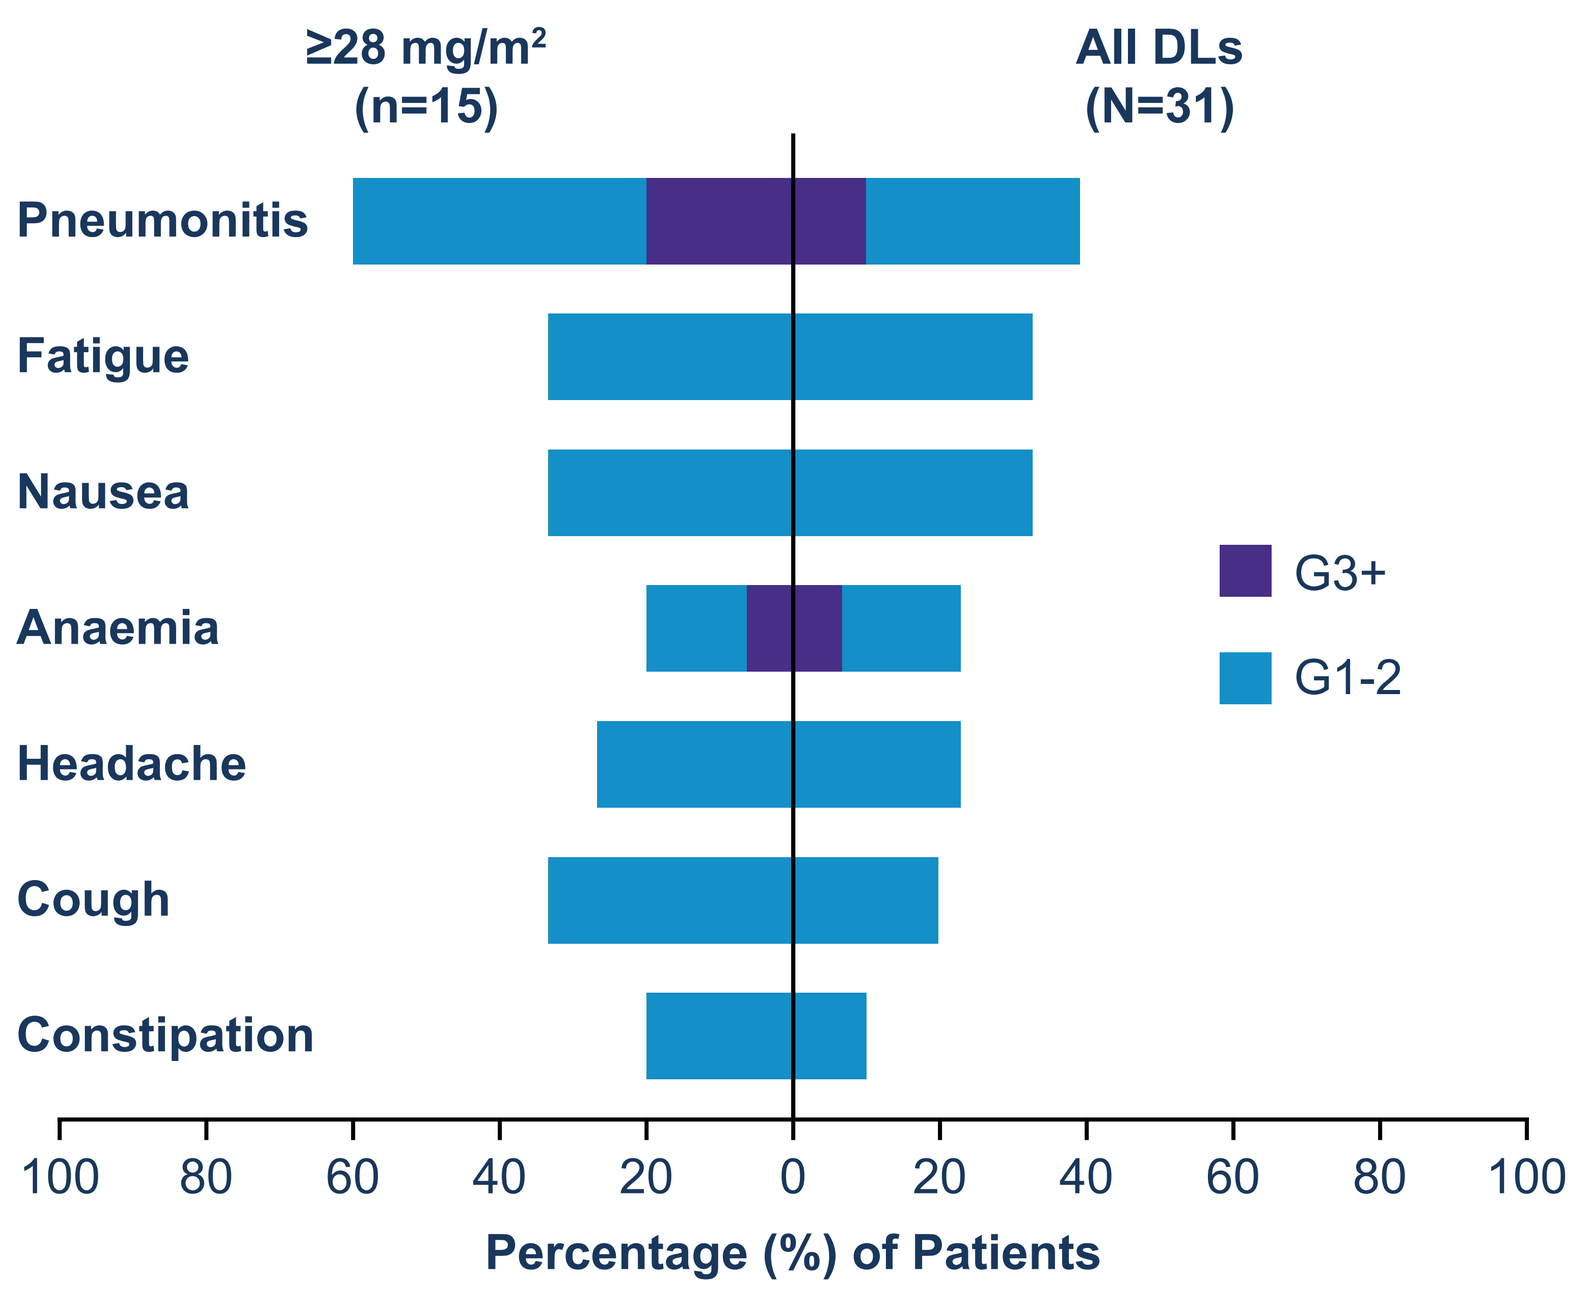
≥28 mg/m² (n=15)
All DLs (N=31)
Pneumonitis
Fatigue
Nausea
Anaemia
Headache
Cough
Constipation
G3+
G1-2
100
80
60
40
20
0
20
40
60
80
100
Percentage (%) of Patients

- TRAEs were mostly low grade
- Pneumonitis observed (including 1 G5) likely an on-target toxicity based on NaPi2b expression in Type II pneumocytes
- No severe peripheral neuropathy, neutropenia, or ocular toxicity
- No thrombocytopenia or treatment-related bleeding events
- XMT-1592 half-life: 7.5-10.3 days

Platform Toxicities Appear to be Greatly Reduced with Next-Generation Dolasynthen Platform[c]

	TRAEs	First-Generation XMT-1536 (N=268)[d] 36 mg/m² n (%)		Next-Generation XMT-1592 (N=31) All DLs n (%)	
		All grades	Grade ≥3	All grades	Grade ≥3
Presumed off-target platform toxicities	**AST elevation, n (%)**	185 (69.0)	124 (46.3)	3 (9.7)	0
	Platelet count decrease/ thrombocytopenia, n (%)	133 (49.6)	32 (11.9)	0	0
	Nausea, n (%)	139 (51.9)	6 (2.2)	10 (32.3)	0
	Fatigue, n (%)	118 (44.0)	26 (9.7)	10 (32.3)	0
Presumed on-target toxicity	**Pneumonitis, n (%)**	26 (9.7)	2 (<1)	12 (38.7)	3 (9.7)

Table 2: Comparison table between XMT-1536 and XMT-1592
Based on two independent studies; select AEs

- Observed pneumonitis believed to be due to target-dependent uptake of the ADC by the Type II pneumocytes in the lungs expressing NaPi2b
- Histologic findings in Type II pneumocytes in animal models have not been seen with multiple other Dolasynthen ADCs directed towards other targets (data on file)

Efficacy

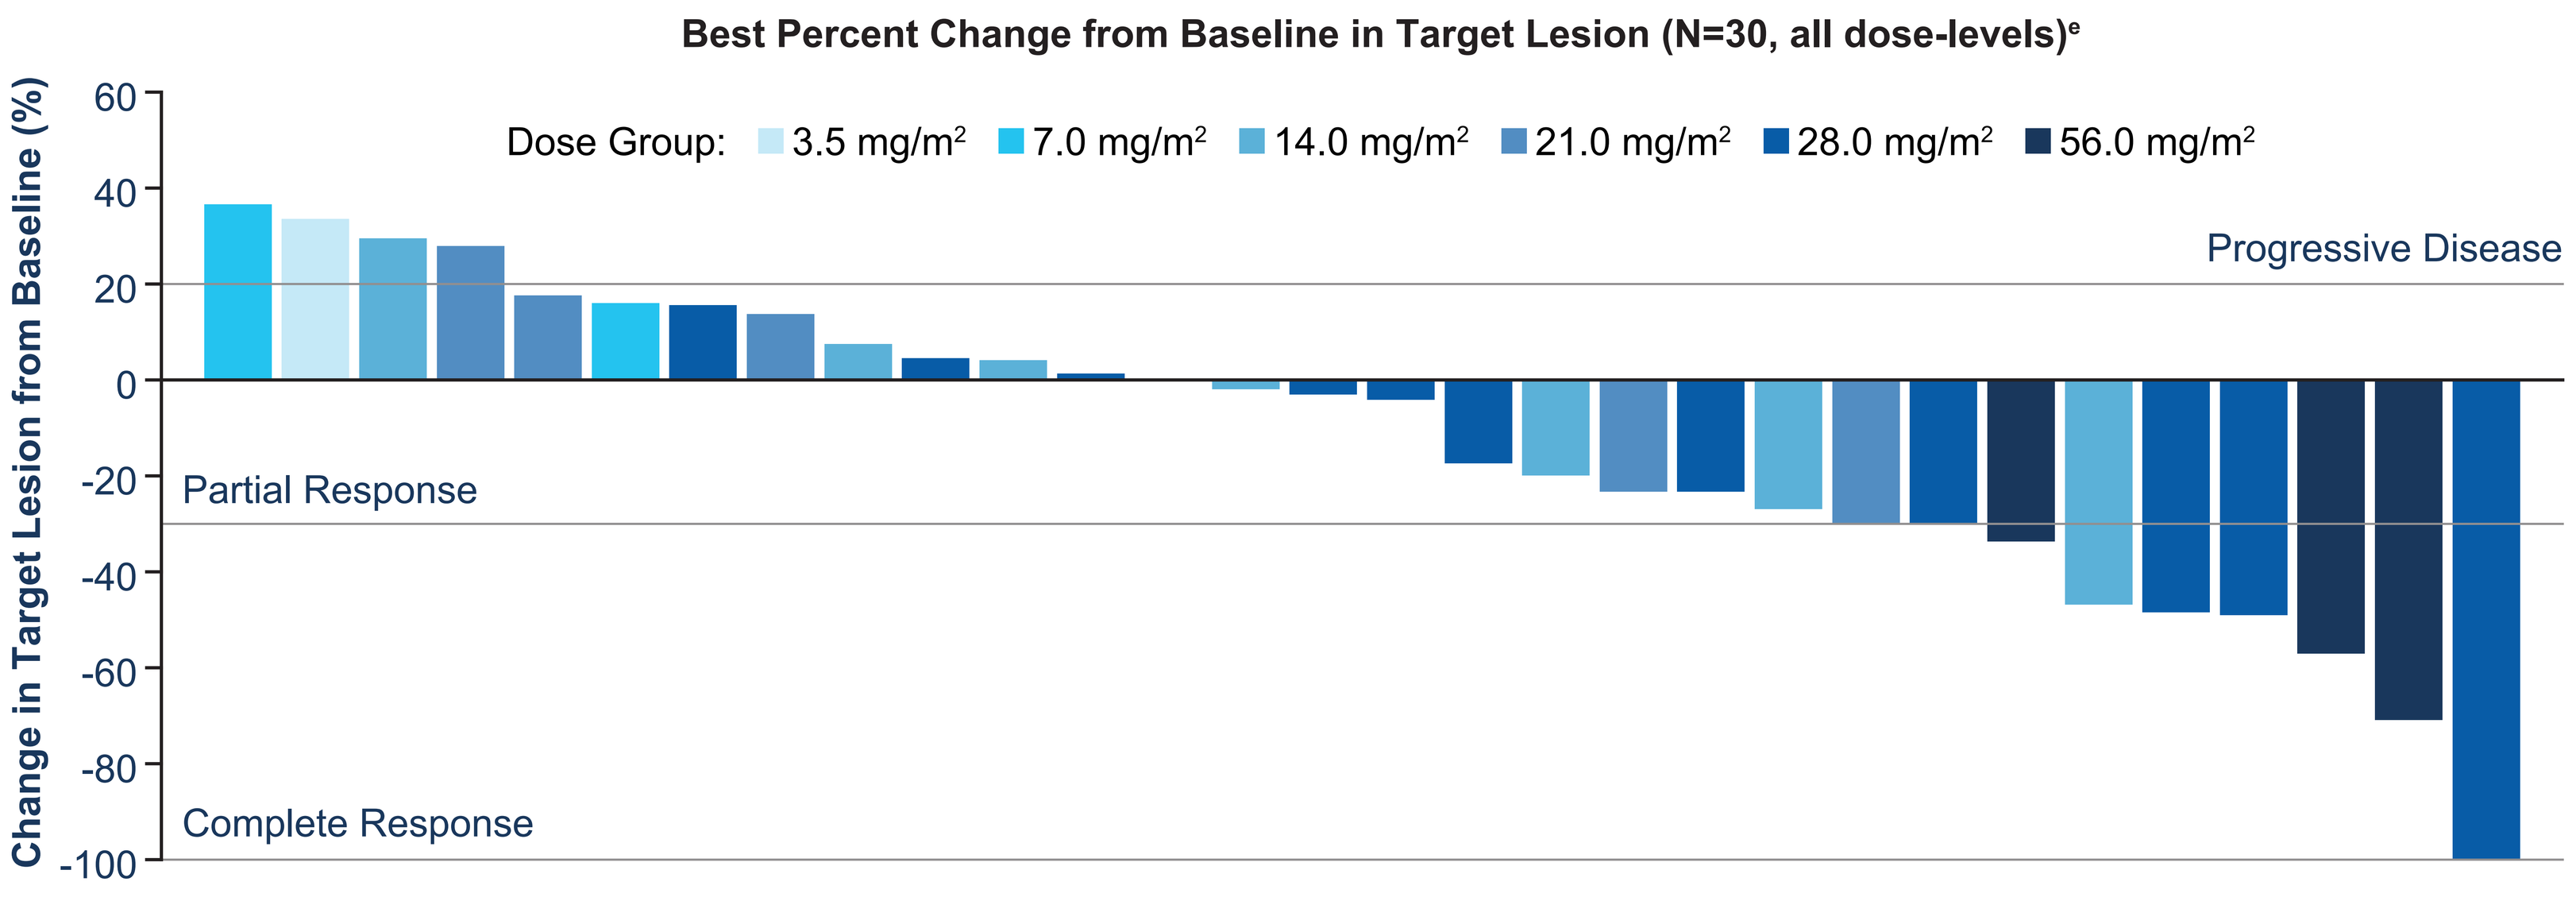
Best Percent Change from Baseline in Target Lesion (N=30, all dose-levels)[e]
Dose Group: 3.5 mg/m² 7.0 mg/m² 14.0 mg/m² 21.0 mg/m² 28.0 mg/m² 56.0 mg/m²
Change in Target Lesion from Baseline (%)
60
40
20
0
-20
-40
-60
-80
-100
Progressive Disease
Partial Response
Complete Response

Figure 6: Waterfall Plot of Evaluable Patients Regardless of NaPi2b Expression

	All Patients All DLs	Ovarian Cancer ≥28 mg/m²
Patients, N	30[e]	13[e]
ORR, n (%)	5 (17%)	4 (31%)
SD, n (%)	19 (63%)	7 (54%)
DCR, n (%)	24 (80%)	11 (85%)
Median DOR, months	7.9	7.9

Table 3: Confirmed Best Overall Response of Evaluable Patients Regardless of NaPi2b Expression

CONCLUSIONS

- XMT-1592 is a NaPi2b-directed Dolasynthen ADC evaluated in a Phase 1 dose escalation trial in patients with platinum-resistant OC and non-squamous NSCLC
- Platform-associated off-target toxicity with XMT-1592 appeared to be reduced relative to first-generation NaPi2b-directed ADCs. Fatigue, nausea, thrombocytopenia, and AST elevation occurred at a lower frequency and severity with XMT-1592 as compared to XMT-1536. No treatment-related bleeding events were reported with XMT-1592
- Pneumonitis was the most common TRAE with XMT-1592 and is believed to be an on-target toxicity due to NaPi2b expression in Type II pneumocytes. Histologic findings in Type II pneumocytes in animal models have not been seen with multiple other Dolasynthen ADCs directed towards other targets
- Responses were observed with XMT-1592 treatment in heavily pretreated patients, including one CR. In evaluable OC patients regardless of NaPi2b expression treated with 28 mg/m² or 56 mg/m², ORR was 31% with median DOR of 7.9 months
- Development of XMT-1592 was discontinued by the sponsor before completing the Phase 1 dose escalation due to portfolio reprioritization considerations

ACKNOWLEDGMENTS

We would like to thank the patients, their families, and the site staff for making this study possible.

This study is sponsored by Mersana Therapeutics, Inc.

Editorial support for this poster was provided by Bio Connections LLC.

REFERENCES

1. Kiyamova R, et al. *Exp Oncol*. 2011;33(3):157_161.
2. Richardson D, et al. *Gynecol Oncol* Rep. 2022;44(2):S16-S17.
3. Banjeree S, et al. *Ann Oncol*. 2018;29(4):917_923.
4. Fessler S, et al. *Can Res*. 2020;80(suppl 16):2894.

ADDITIONAL INFORMATION

Scan the Quick Response (QR) code for downloadable PDF copies of this poster and author disclosures.

PDF copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission.

Presented at the 2024 European Society of Gynecological Oncology Annual Meeting, Barcelona, Spain, Mar 7-10, 2024

[a] IHC labeling intensity: 1+ = weak; 2+ = moderate; 3+ = strong. [b] Adenocarcinoma. [c] Based on two independent studies. [d] Data cut: May 31, 2023 (UPLIFT: Phase 2 trial). [e] Evaluable patients; excludes one patient at 56 mg/m² who was non-evaluable by RECIST due to withdrawal of consent before first scan.

ADC, antibody-drug conjugate; AF-HPA, Auristatin F hydroxypropyl amide; AST, aspartate aminotransferase; CR, complete response; DAR, drug-to-antibody ratio; DCR, disease control rate; DL, dose level; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; Fcγ, fragment crystallizable gamma receptor; G, grade; IHC, immunohistochemistry; ILD, interstitial lung disease; IV, intravenous; MTD, maximum tolerated dose; NaPi2b, sodium-dependent phosphate transporter 2b; NSCLC, non-small cell lung cancer; OC, ovarian cancer; ORR, objective response rate; PARP, poly-ADP ribose polymerase; PDX, patient-derived xenograft; PK, pharmacokinetics; qw, every week; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose; SD, stable disease; SEM, standard error of mean; TRAE, treatment-related adverse event; UV, ultraviolet.

Poster #559